OFF-THE-SHELF IMMUNOREGULATORY CELLS GENERATED FROM HUMAN INDUCED PLURIPOTENT STEM CELLS FOR THE TREATMENT OF AUTOIMMUNE AND INFLAMMATORY DISEASES

Robert Tacke[1], Martin Hosking[1], Christian Maine[1], Lisa Guerrettaz[1], Tom Lee[1], Emily Driver[1], Matthieu Baeur[1], Greg Bonello[1], Heather Foster[1], Thuy Le[1], Stacey Moreno[1], Newsha Shahaf[1], Ramzey Abujarour[1], Raedun Clarke[1], Betsy Rezner[1], Dave Robbins[1], Bahram Valamehr[1], Daniel Shoemaker[1†] and Philippe Parone[1†]

[1]Fate Therapeutics Inc., San Diego, CA, USA; † Co-senior authors

Fate THERAPEUTICS

Better Cells For Better Therapies™

INTRODUCTION

Hematopoietic progenitor cells are potent immune-regulators that suppress T cell activity through multiple mechanisms, including: Programmed death-ligand 1 (PD-L1) and Indoleamine 2,3-Dioxygenase (IDO1). Of this progenitor cell population, myeloid derived suppressor cells (MDSC) have the most potent immunoregulatory activity, which is underscored by the key role these cells play in promoting immunosuppressive tumor microenvironments. Exogenously administered MDSCs have been shown to have profound efficacy across multiple murine models of inflammatory and autoimmune disease. However, the translation of MDSCs to the clinic has been hampered by a lack of scalable manufacturing. To address this issue, we have developed a robust process to generate “off-the-shelf” iMDSCs from human induced pluripotent stem cells (iPSCs). This GMP-compatible, feeder-free differentiation protocol is highly scalable, enabling the potential production of thousands of therapeutic doses from a single manufacturing run. The iMDSCs resulting from this process are over 93% pure and express surface markers characterizing MDSCs ($CD33^+$, $CD14^+$, $CD11b^+$, HLA-DR^{low}). In co-culture assays, iMDSCs potently suppress TCR-driven T cell activity independently of HLA matching, and a single injection of iMDSCs attenuates disease in a xenogeneic mouse model of Graft-versus-Host-Disease (GvHD). Together, our results demonstrate that iMDSCs may serve as a scalable, “off-the-shelf” source of immunoregulatory cells for the treatment of autoimmune and inflammatory diseases.

iMDSC MANUFACTURING

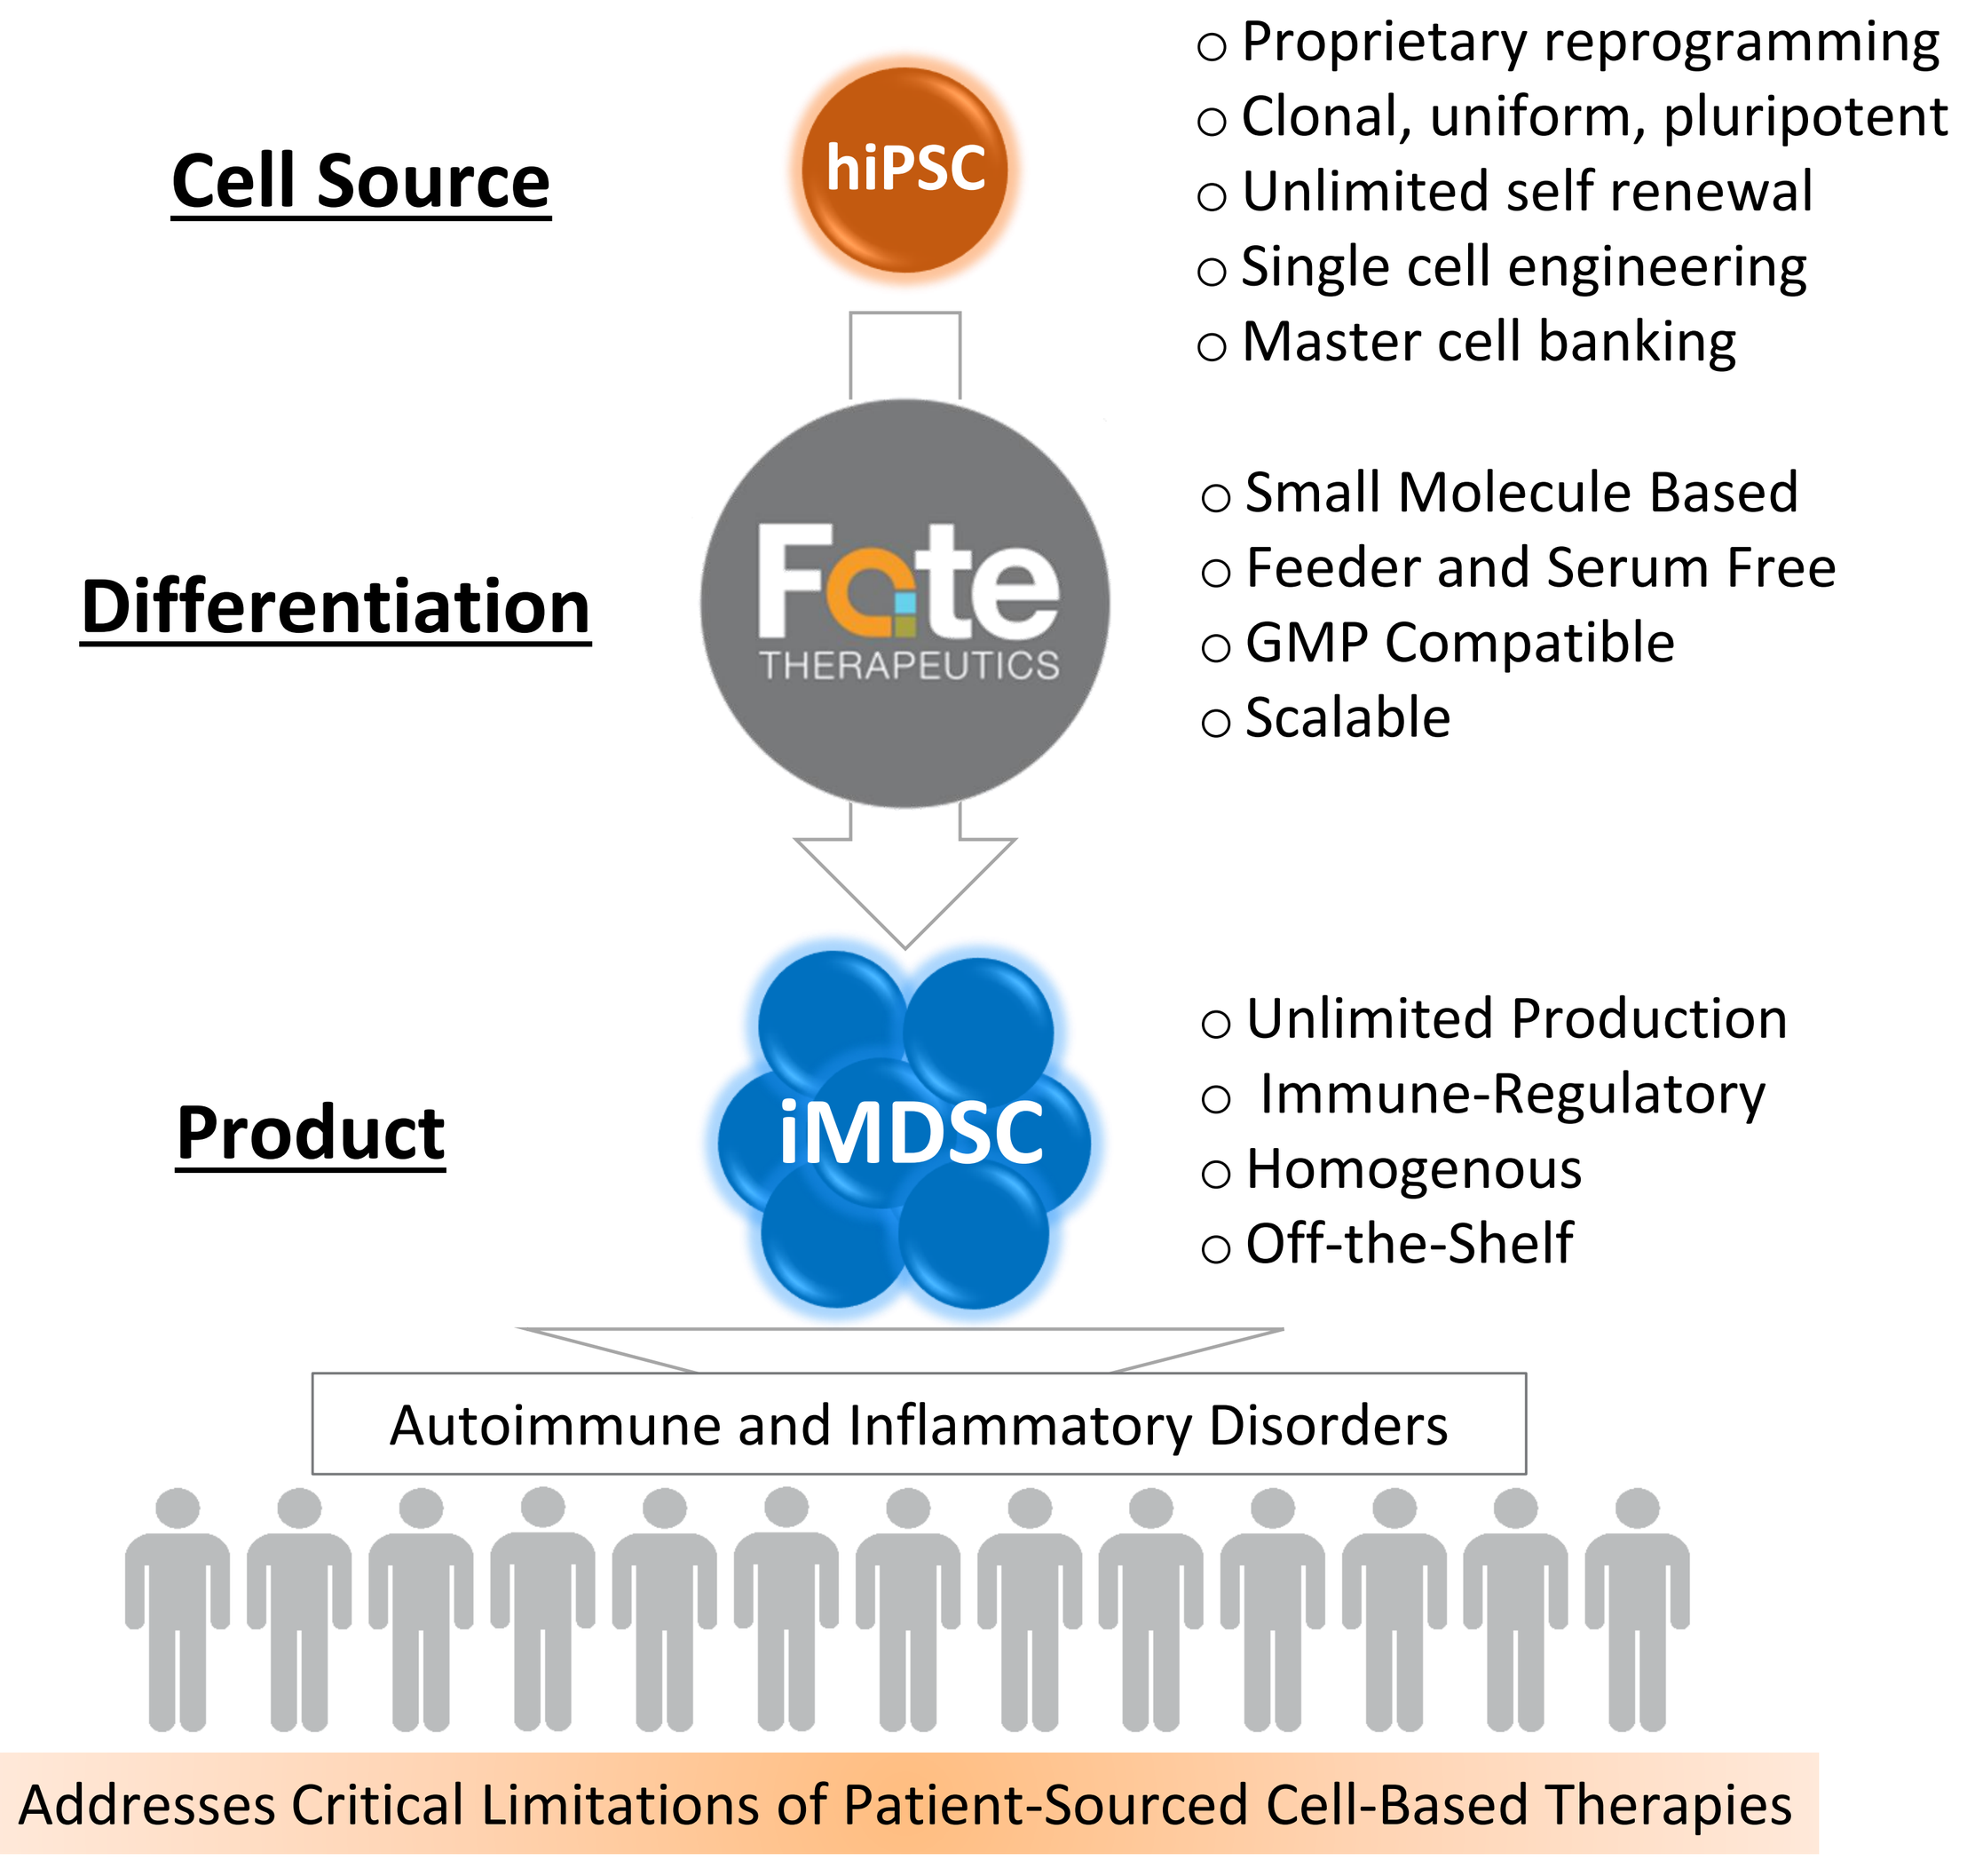
Cell Source
hiPSC
○ Proprietary reprogramming
○ Clonal, uniform, pluripotent
○ Unlimited self renewal
○ Single cell engineering
○ Master cell banking
Differentiation
Fate THERAPEUTICS
○ Small Molecule Based
○ Feeder and Serum Free
○ GMP Compatible
○ Scalable
Product
iMDSC
○ Unlimited Production
○ Immune-Regulatory
○ Homogenous
○ Off-the-Shelf
Autoimmune and Inflammatory Disorders
Addresses Critical Limitations of Patient-Sourced Cell-Based Therapies

CONCLUSIONS

- **Established scalable, feeder free manufacture of myeloid derived suppressor cells (iMDSCs) from human induced pluripotent stem cells**
- **iMDSCs potently suppressed T cell proliferation and effector function independently of HLA matching**
- **A single injection of iMDSCs attenuated disease and prolonged survival in a xenogenic mouse model of GvHD, and is associated with increased levels of Tregs**

RESULTS

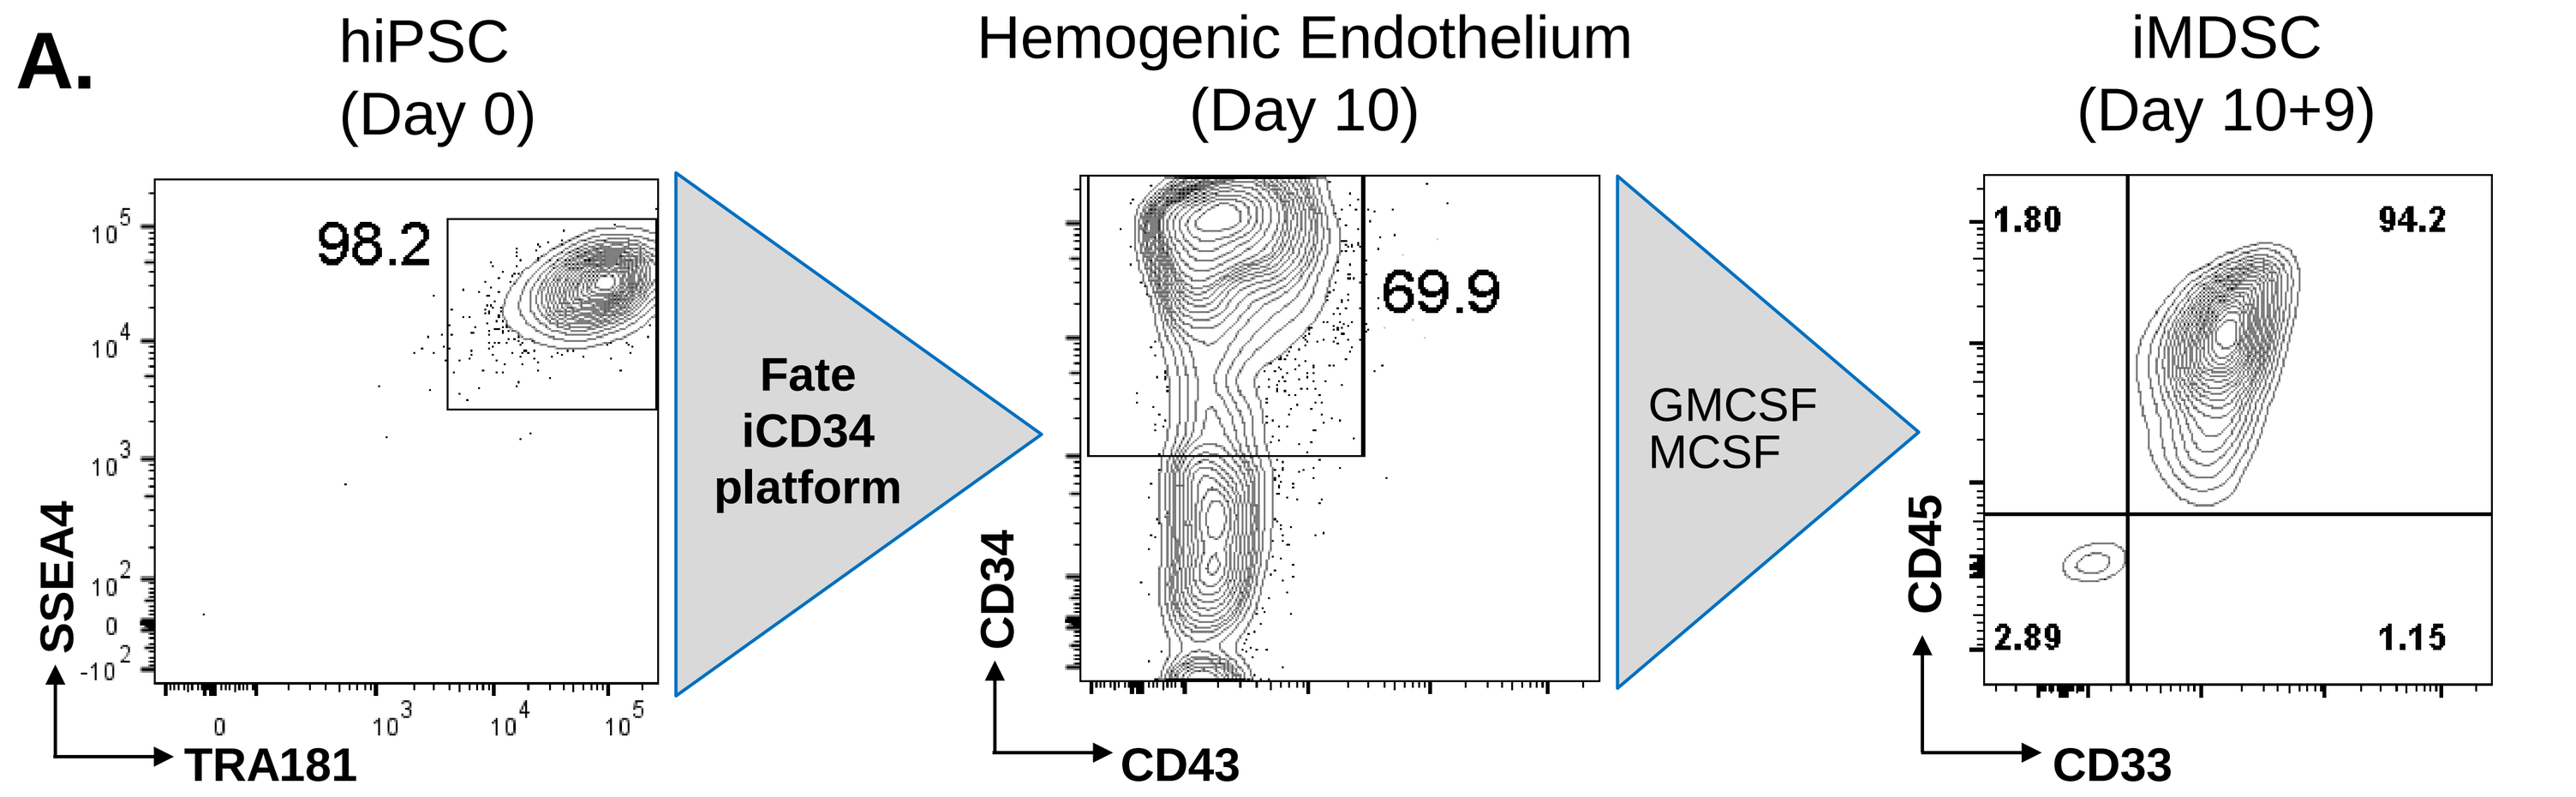
A.
hiPSC (Day 0)
Hemogenic Endothelium (Day 10)
iMDSC (Day 10+9)
98.2
Fate iCD34 platform
69.9
GMCSF MCSF
1.80
94.2
2.89
1.15
SSEA4
TRA181
CD34
CD43
CD45
CD33

B.

	MDSC (% non adherent cells ± SEM)
$CD45^+$	95.15 ± 1.57
$CD45^+CD33^+$	93.75 ± 1.77
Erythrocytes ($CD45^-CD235^+$)	0.084 ± 0.05
Lymphoid ($CD45^+CD7^+$)	0.015 ± 0.01
Granulocytes ($CD45^+CD33^+CD66b^+$)	3.833 ± 2.19
Monocytes ($CD45^+CD33^+CD14^+$)	52.8 ± 8.93
PDL1 expression ($CD45^+PDL1^+$)	56.66 ± 9.297

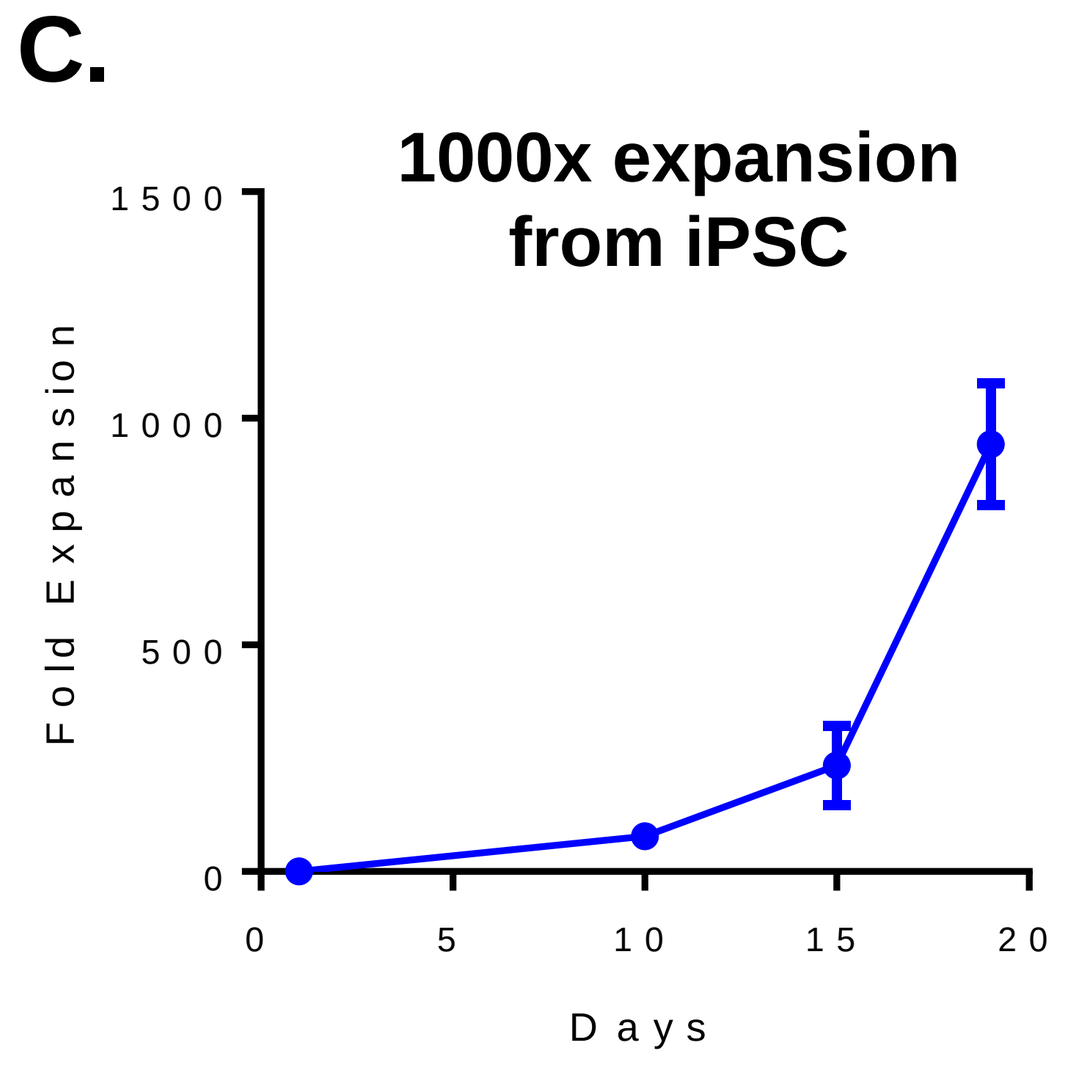
C.
1000x expansion from iPSC
Fold Expansion
Days

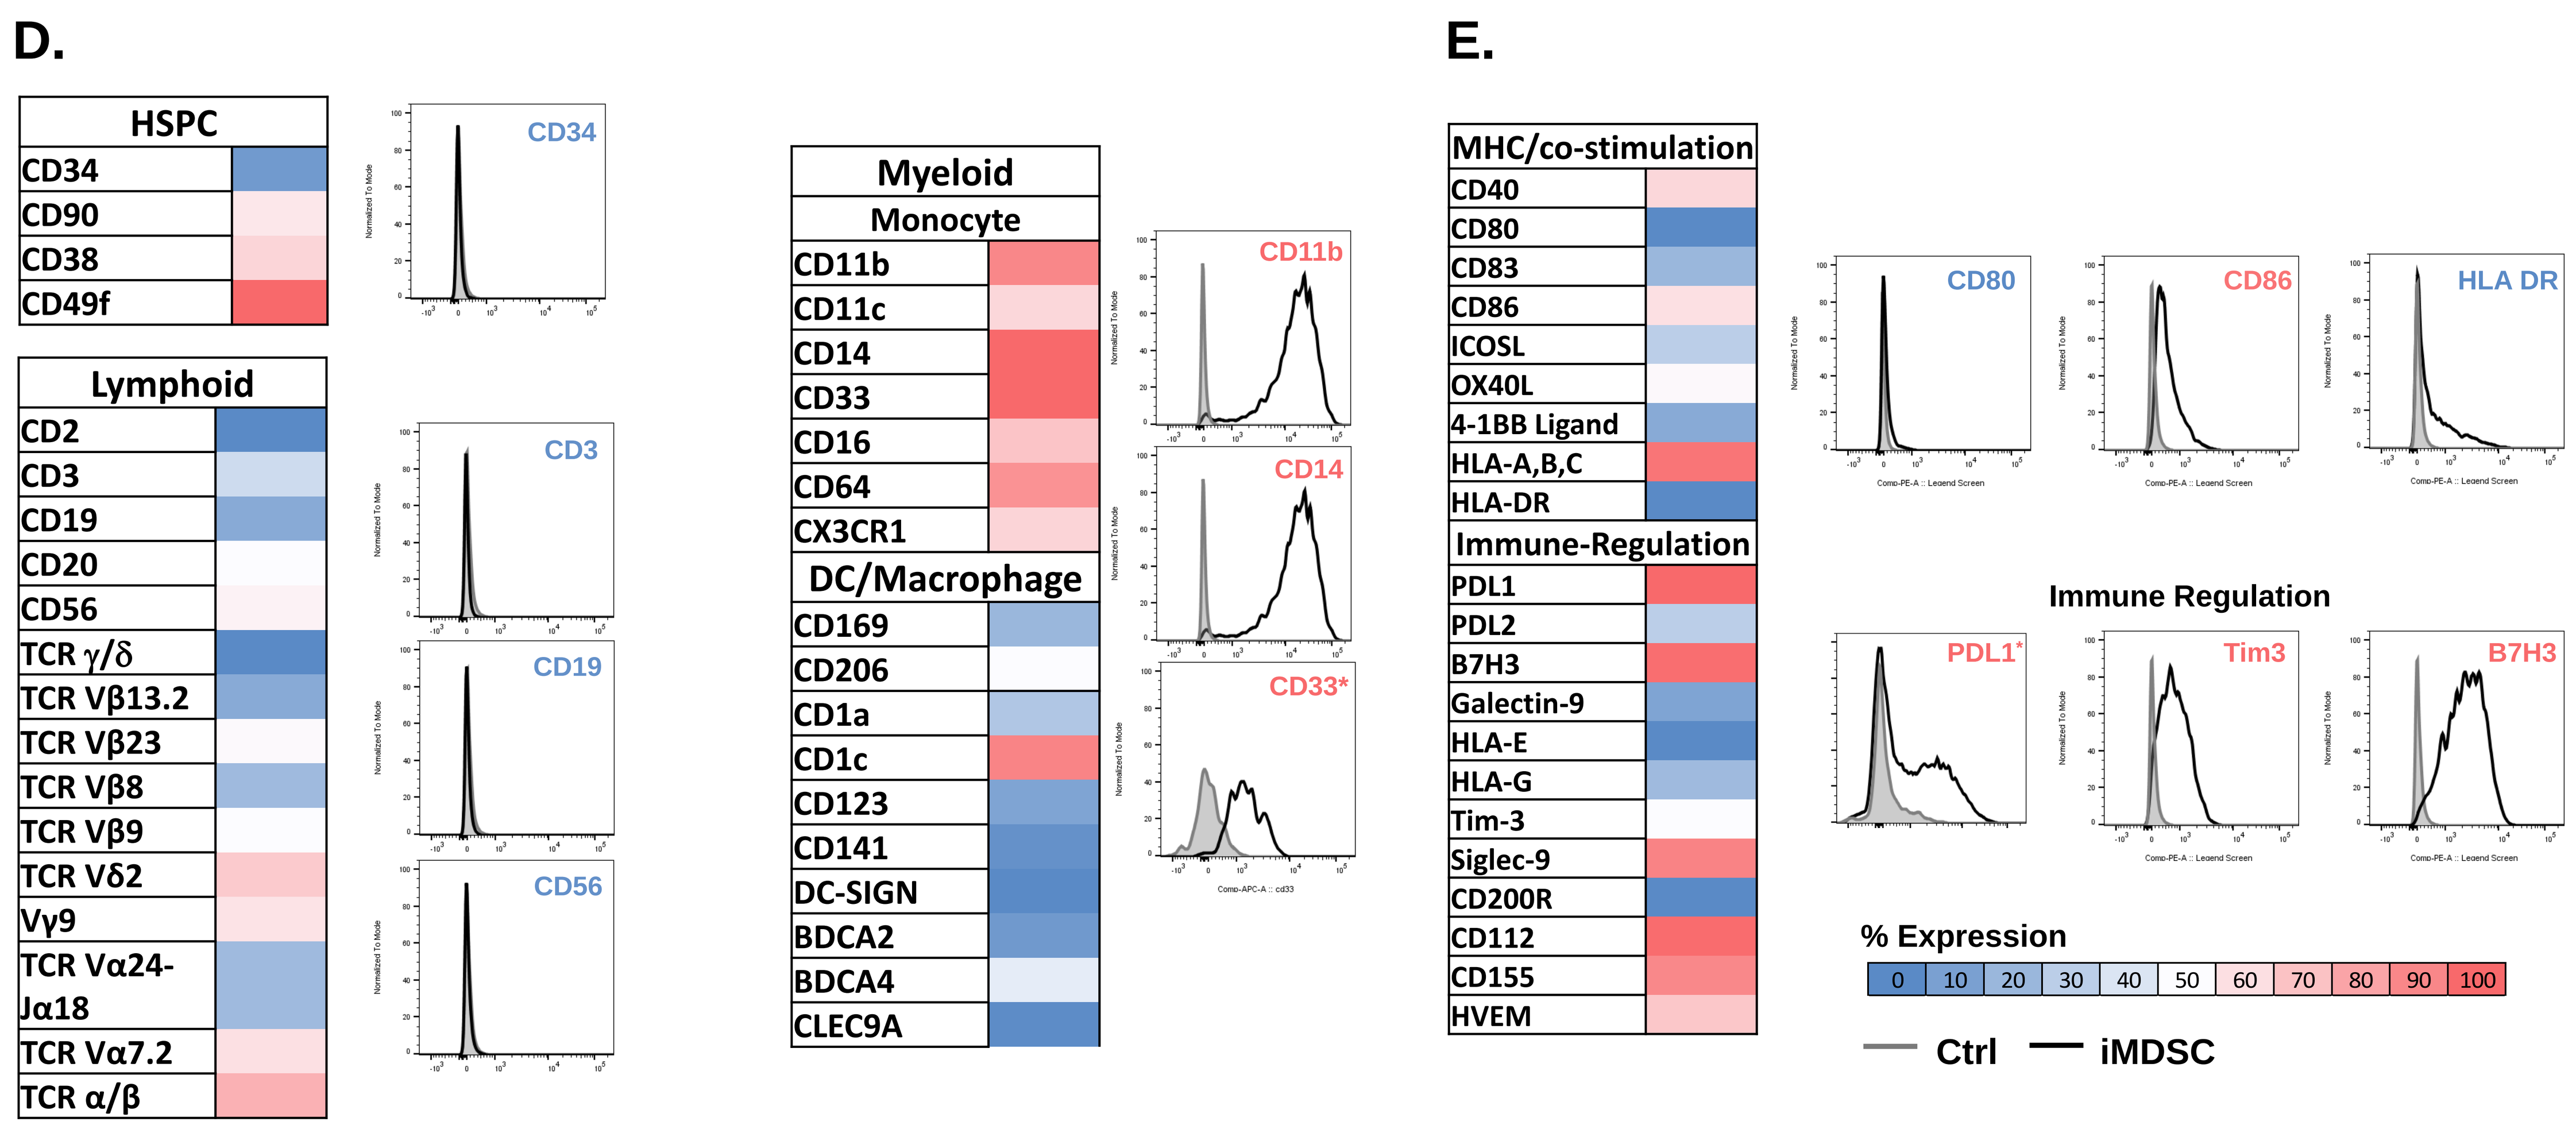
D.
HSPC
CD34
CD90
CD38
CD49f
Lymphoid
CD2
CD3
CD19
CD20
CD56
TCR γ/δ
TCR Vβ13.2
TCR Vβ23
TCR Vβ8
TCR Vβ9
TCR Vδ2
Vγ9
TCR Vα24-Jα18
TCR Vα7.2
TCR α/β
Myeloid
Monocyte
CD11b
CD11c
CD14
CD33
CD16
CD64
CX3CR1
DC/Macrophage
CD169
CD206
CD1a
CD1c
CD123
CD141
DC-SIGN
BDCA2
BDCA4
CLEC9A
CD33*
E.
MHC/co-stimulation
CD40
CD80
CD83
CD86
ICOSL
OX40L
4-1BB Ligand
HLA-A,B,C
HLA-DR
Immune-Regulation
PDL1
PDL2
B7H3
Galectin-9
HLA-E
HLA-G
Tim-3
Siglec-9
CD200R
CD112
CD155
HVEM
HLA DR
Immune Regulation
PDL1*
Tim3
% Expression
0 10 20 30 40 50 60 70 80 90 100
Ctrl
iMDSC

Figure 1. Scalable generation of immune-regulatory myeloid cells from human induced pluripotent stem cells under serum free and feeder-free conditions. A. Parental human induced pluripotent stem cells (hiPSCs) are differentiated through a 10 day multi-staged monolayer culture process to $CD34^+$ hemogenic endothelium (HE, CD34+). Day 10 $iCD34^+$ HE cells are isolated by FACS and cultured in myeloid promoting conditions resulting $CD45^+$ cells emerging from the endothelial layer starting day 3 after plating and acquiring CD33 by day 9. **B.** The $CD45^+$ cell population manufactured using this 19 day process is highly enriched for myeloid cells (>90%) and is free of granulocytes, erythrocytes, and lymphoid cells. **C.** This process enables the production of over 1,000 $CD45^+CD33^+$ cells for 1 iPSC. **D-E.** Immunophenotyping by LEGENDScreen™ demonstrates that day 19 $CD45^+CD33^+$ cells express predominantly early myeloid markers and several immune-regulatory surface proteins. In light of the similarities of this marker profile to that of MDSCs, day 19 $CD45^+CD33^+$ cells are termed iMDSCs.

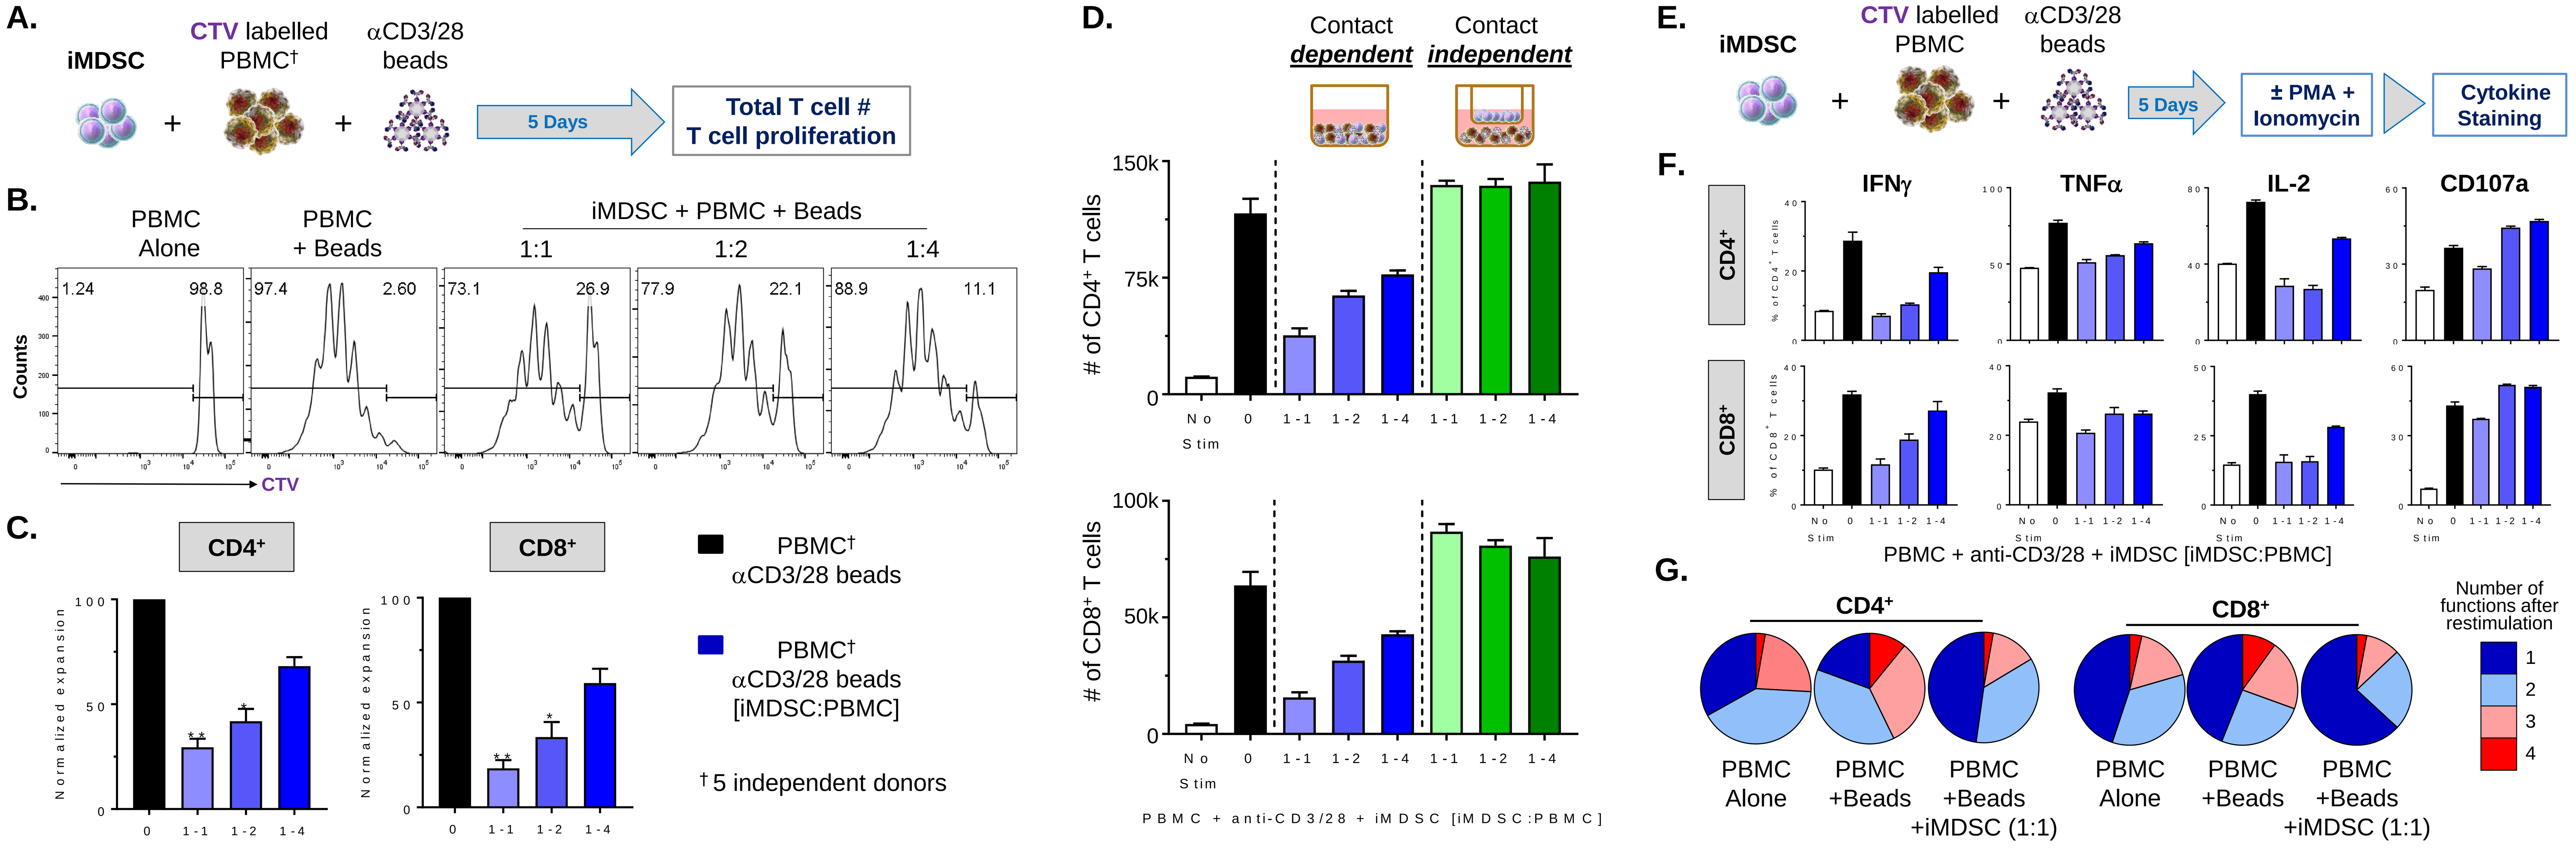
A.
iMDSC
CTV labelled PBMC†
αCD3/28 beads
5 Days
Total T cell #
T cell proliferation
B.
PBMC Alone
PBMC + Beads
iMDSC + PBMC + Beads
1:1
1:2
1:4
1.24
98.8
97.4
2.60
73.1
26.9
77.9
22.1
88.9
11.1
Counts
CTV
C.
CD4+
CD8+
Normalized expansion
PBMC† αCD3/28 beads
PBMC† αCD3/28 beads [iMDSC:PBMC]
† 5 independent donors
D.
Contact dependent
Contact independent
of CD4+ T cells
of CD8+ T cells
No Stim
PBMC + anti-CD3/28 + iMDSC [iMDSC:PBMC]
E.
iMDSC
CTV labelled PBMC
αCD3/28 beads
5 Days
± PMA + Ionomycin
Cytokine Staining
F.
IFNγ
TNFα
IL-2
CD107a
CD4+
CD8+
PBMC + anti-CD3/28 + iMDSC [iMDSC:PBMC]
G.
CD4+
CD8+
Number of functions after restimulation
PBMC Alone
PBMC +Beads
PBMC +Beads +iMDSC (1:1)

Figure 2. iMDSCs potently suppresses TCR-driven T cell proliferation and effector function independently of HLA matching. A. Overview of the *in vitro* T cell suppression assay **B.** Representative CTV histograms of $CD4^+$ T cells cultured ± iMDSCs suggest that iMDSCs limit T cell proliferation. **C.** T cell expansion following iMDSCs coculture was quantified for five independent donors and reveals significant and titratable reductions in T cell expansion independent of HLA matching. **D.** Reduction in T cell expansion was observed when iMDSCs were co-cultured alongside PBMCs (blue bars) and not when separated in a 0.4µm transwell (green bars), thus demonstrating that iMDSCs mediated suppression is contact dependent. **E.** Overview of the *in vitro* cytokine release assay. **F.** iMDSCs attenuate the ability of both $CD4^+$ and $CD8^+$ T cells to produce IFNγ, TNF, IL-2, and express CD107a. **F.** T cells cocultured with iMDSCs are less capable of producing multiple effector functions (red) and are mostly limited to one or two functions (blue). All data are presented as averages +/- SEM. ***p<0.001, **p<0.01, and *p<0.05

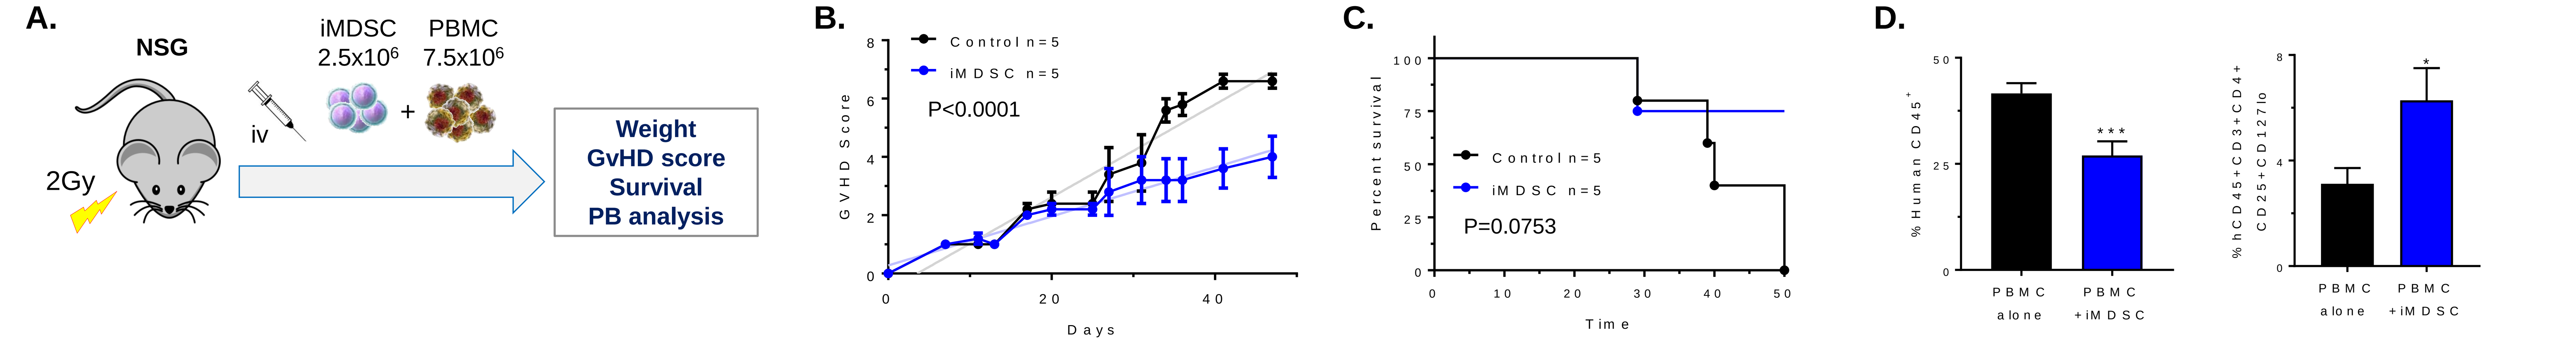
A.
NSG
2Gy
iv
iMDSC 2.5x10^6
PBMC 7.5x10^6
Weight
GvHD score
Survival
PB analysis
B.
Control n=5
iMDSC n=5
P<0.0001
GVHD Score
Days
C.
Percent survival
Control n=5
iMDSC n=5
P=0.0753
Time
D.
% Human CD45+
% hCD45+CD3+CD4+CD25+CD127lo
PBMC alone
PBMC +iMDSC

Figure 3. iMDSCs attenuate disease in a xenogenic model of Graft versus Host Disease (GvHD). A. Overview of xenogeneic acute GvHD model. **B.** iMDSCs attenuate the severity of disease as measured by GVHD score. Mice were observed weekly for symptoms of clinical GvHD. Weight was monitored as an independent parameter. **C.** iMDSCs prolong survival. Kaplan-Meier survival curve were generated using disease endstage (loss of > 25% of body weight) as the termination criteria. **D.** iMDSCs led to a significant reduction in human $CD45^+$ expansion at day 14 with an increase in Tregs ($CD4^+CD25^{hi}CD127^{lo}$) in peripheral blood. Data are presented as averages +/- SEM. ***p<0.001, **p<0.01, and *p<0.05