Development of Master Multiplexed-Engineered iPSC Bank for Off-the-Shelf Cell-Based Cancer Immunotherapy with Reduced Conditioning Chemotherapy

Yijia Pan, **Mark Jelcic**, Jode Goodridge, Rina Mbofung, Alan Williams, Ken Hayama, Brian Groff, Pei-Fang Tsai, Tom T. Lee, and Bahram Valamehr[1]

Fate Therapeutics Inc, San Diego, CA, USA. [1]Correspondence: bob.valamehr@fatetherapeutics.com

GRAPHICAL ABSTRACT & INTRODUCTION

A Novel Platform for Creating Off-the-shelf Multiplex-Engineered Hematopoietic Cell Products

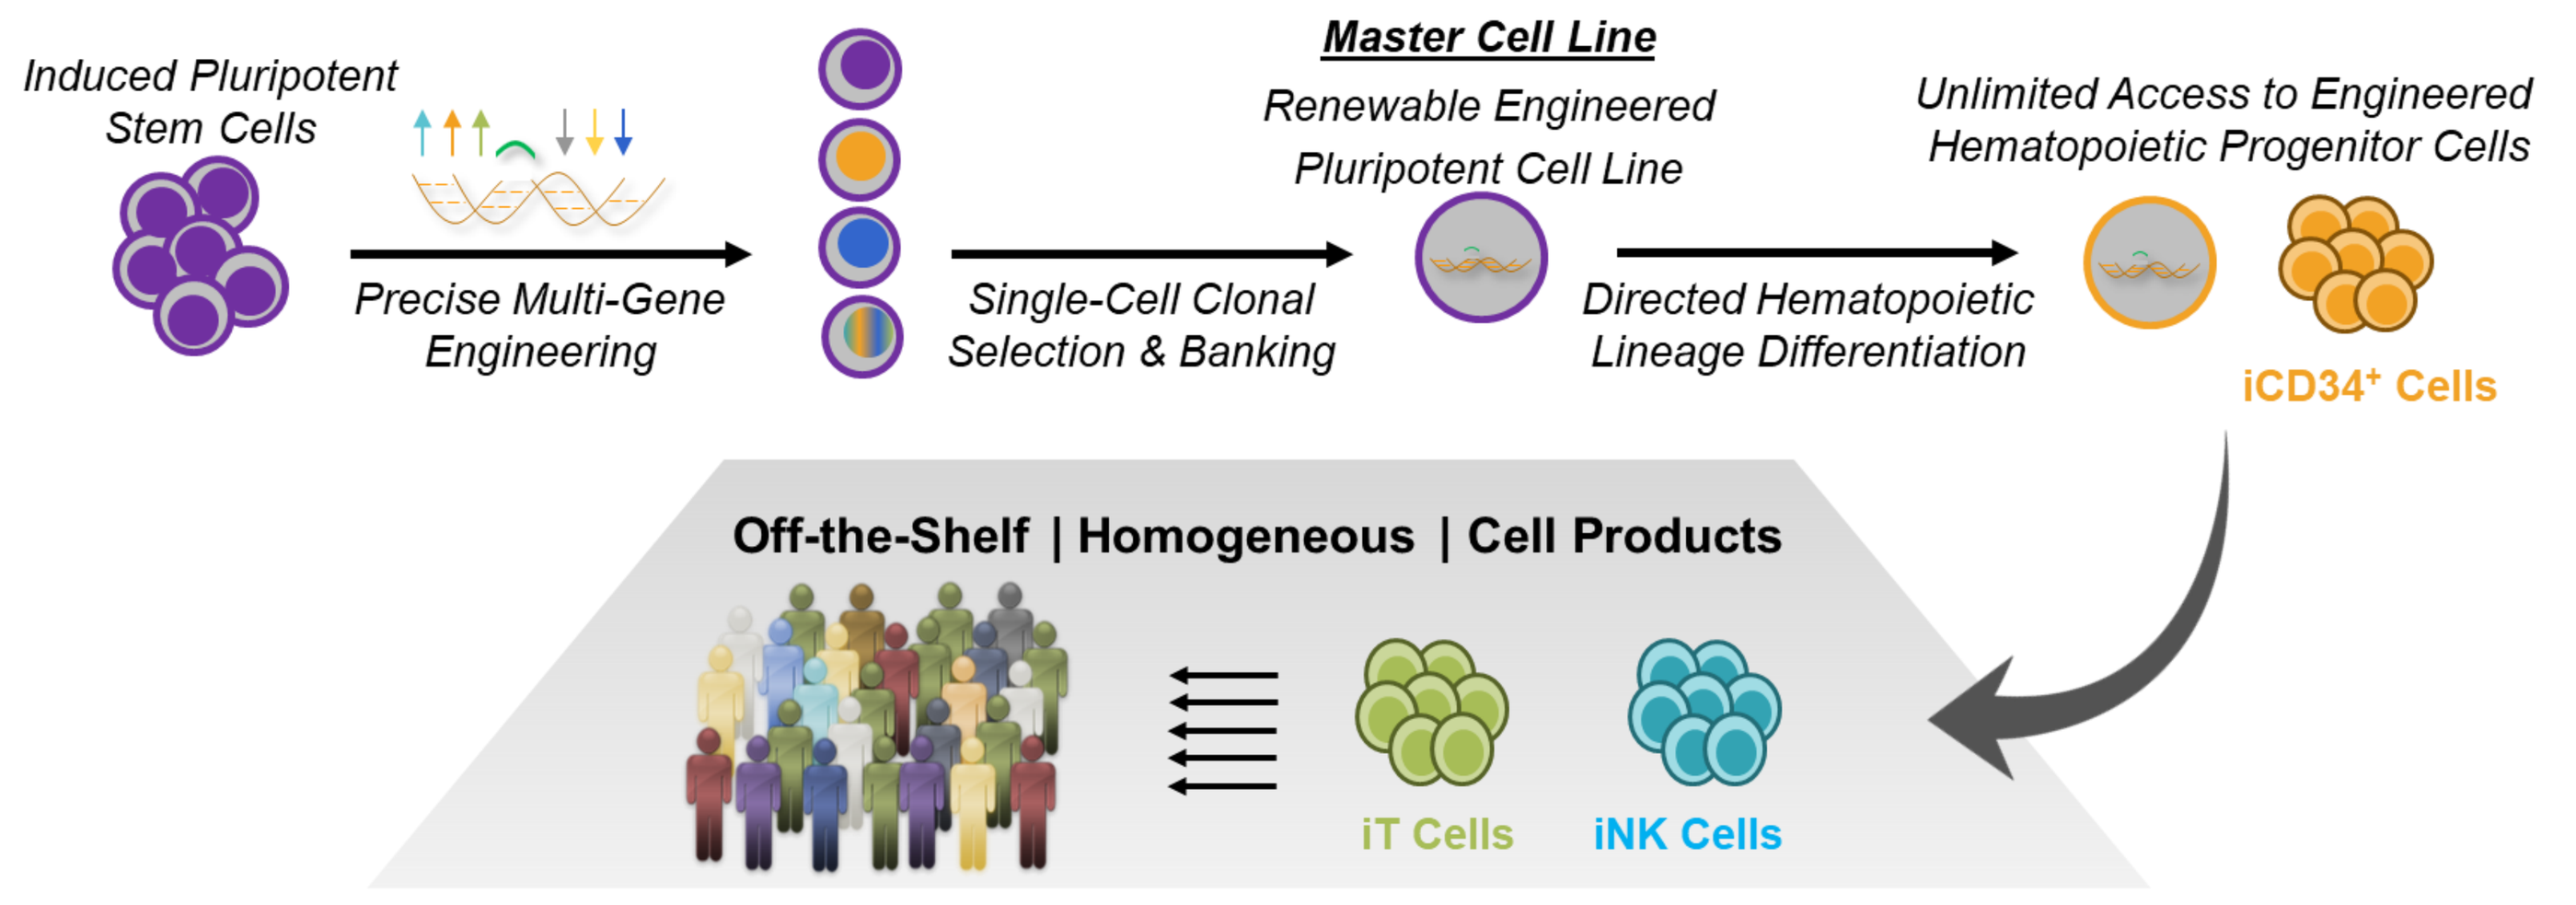
Induced Pluripotent Stem Cells
Precise Multi-Gene Engineering
Master Cell Line
Renewable Engineered Pluripotent Cell Line
Single-Cell Clonal Selection & Banking
Unlimited Access to Engineered Hematopoietic Progenitor Cells
Directed Hematopoietic Lineage Differentiation
iCD34⁺ Cells
Off-the-Shelf | Homogeneous | Cell Products
iT Cells
iNK Cells

- Autologous and allogeneic cell-based cancer immunotherapy requires administration of systemic conditioning chemotherapy to suppress the patient's immune system to potentiate the adoptively-transferred immune cells.
- Protracted immune suppression can lead to increased risk of complications such as severe infections and affects immune reconstitution
- New therapeutic strategies to prevent allorejection with minimal chemotherapy conditioning may significantly improve the application and efficacy of adoptively-transferred immune cells while preserving the patient's immune system

Multiplexed-Engineering of Off-the-Shelf Cell-Based Cancer Immunotherapy to Reduce the Need for Conditioning Chemotherapy

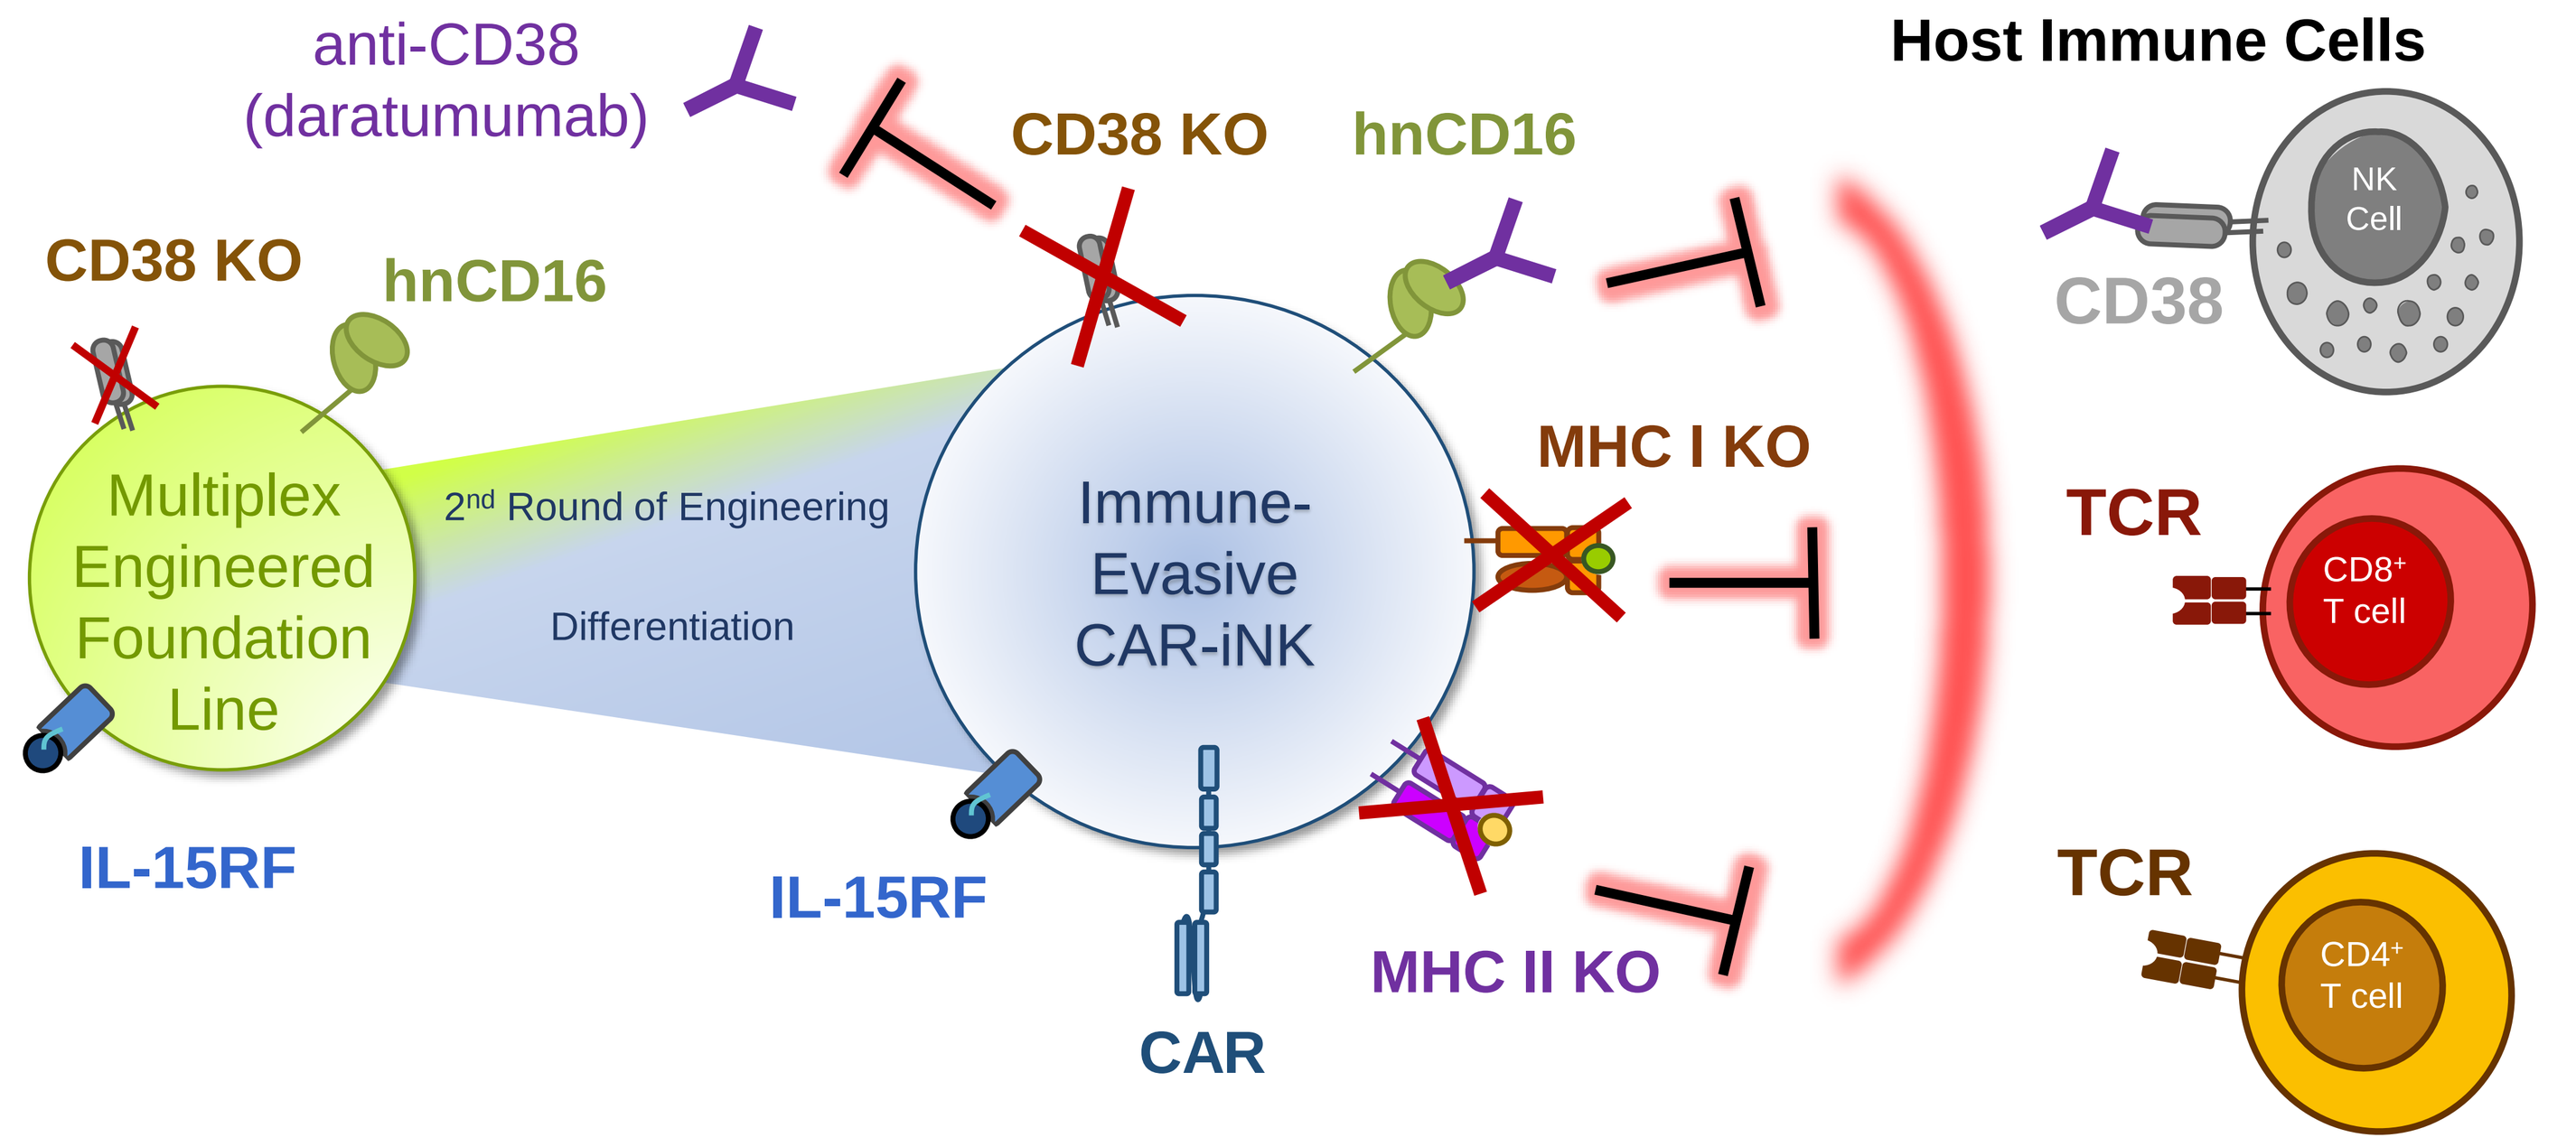
anti-CD38 (daratumumab)
CD38 KO
hnCD16
Host Immune Cells
NK Cell
CD38
Multiplex Engineered Foundation Line
2nd Round of Engineering
Differentiation
Immune-Evasive CAR-iNK
MHC I KO
MHC II KO
TCR
CD8⁺ T cell
CD4⁺ T cell
IL-15RF
CAR

- iPSC derived NK cell (iNK cell) therapy is multiplex-engineered with a novel combination of immune-evasion modalities
 1. CD38 deletion for resistance to fratricide when combined with anti-CD38 monoclonal antibody to selectively eliminate host alloreactive lymphocytes
 2. Ablation of MHC class I molecules via knockout of Beta-2-Microglobulin (B2M) to prevent CD8⁺ T cell mediated rejection
 3. Ablation of MHC class II molecules via knockout of the Class II Transactivator (CIITA) to prevent CD4⁺ T cell mediated rejection
- In the first stage, a clonal foundation iPSC line was established incorporating IL-15/IL-15 receptor α fusion (IL-15RF) and a high-affinity, non-cleavable CD16 (hnCD16) at the CD38 locus for enhanced NK cell activity and antibody-dependent cellular cytotoxicity (ADCC), respectively
- In the second stage, iPSCs from the clonal master cell line were further engineered and subcloned to knockout B2M and CIITA and knock-in a tumor-targeting CAR simultaneously

RESULTS

Multi-Loci Engineering of Clonal iPSC Master Cell Lines with Six Edits to Drive Anti-Tumor Activity and Persistence

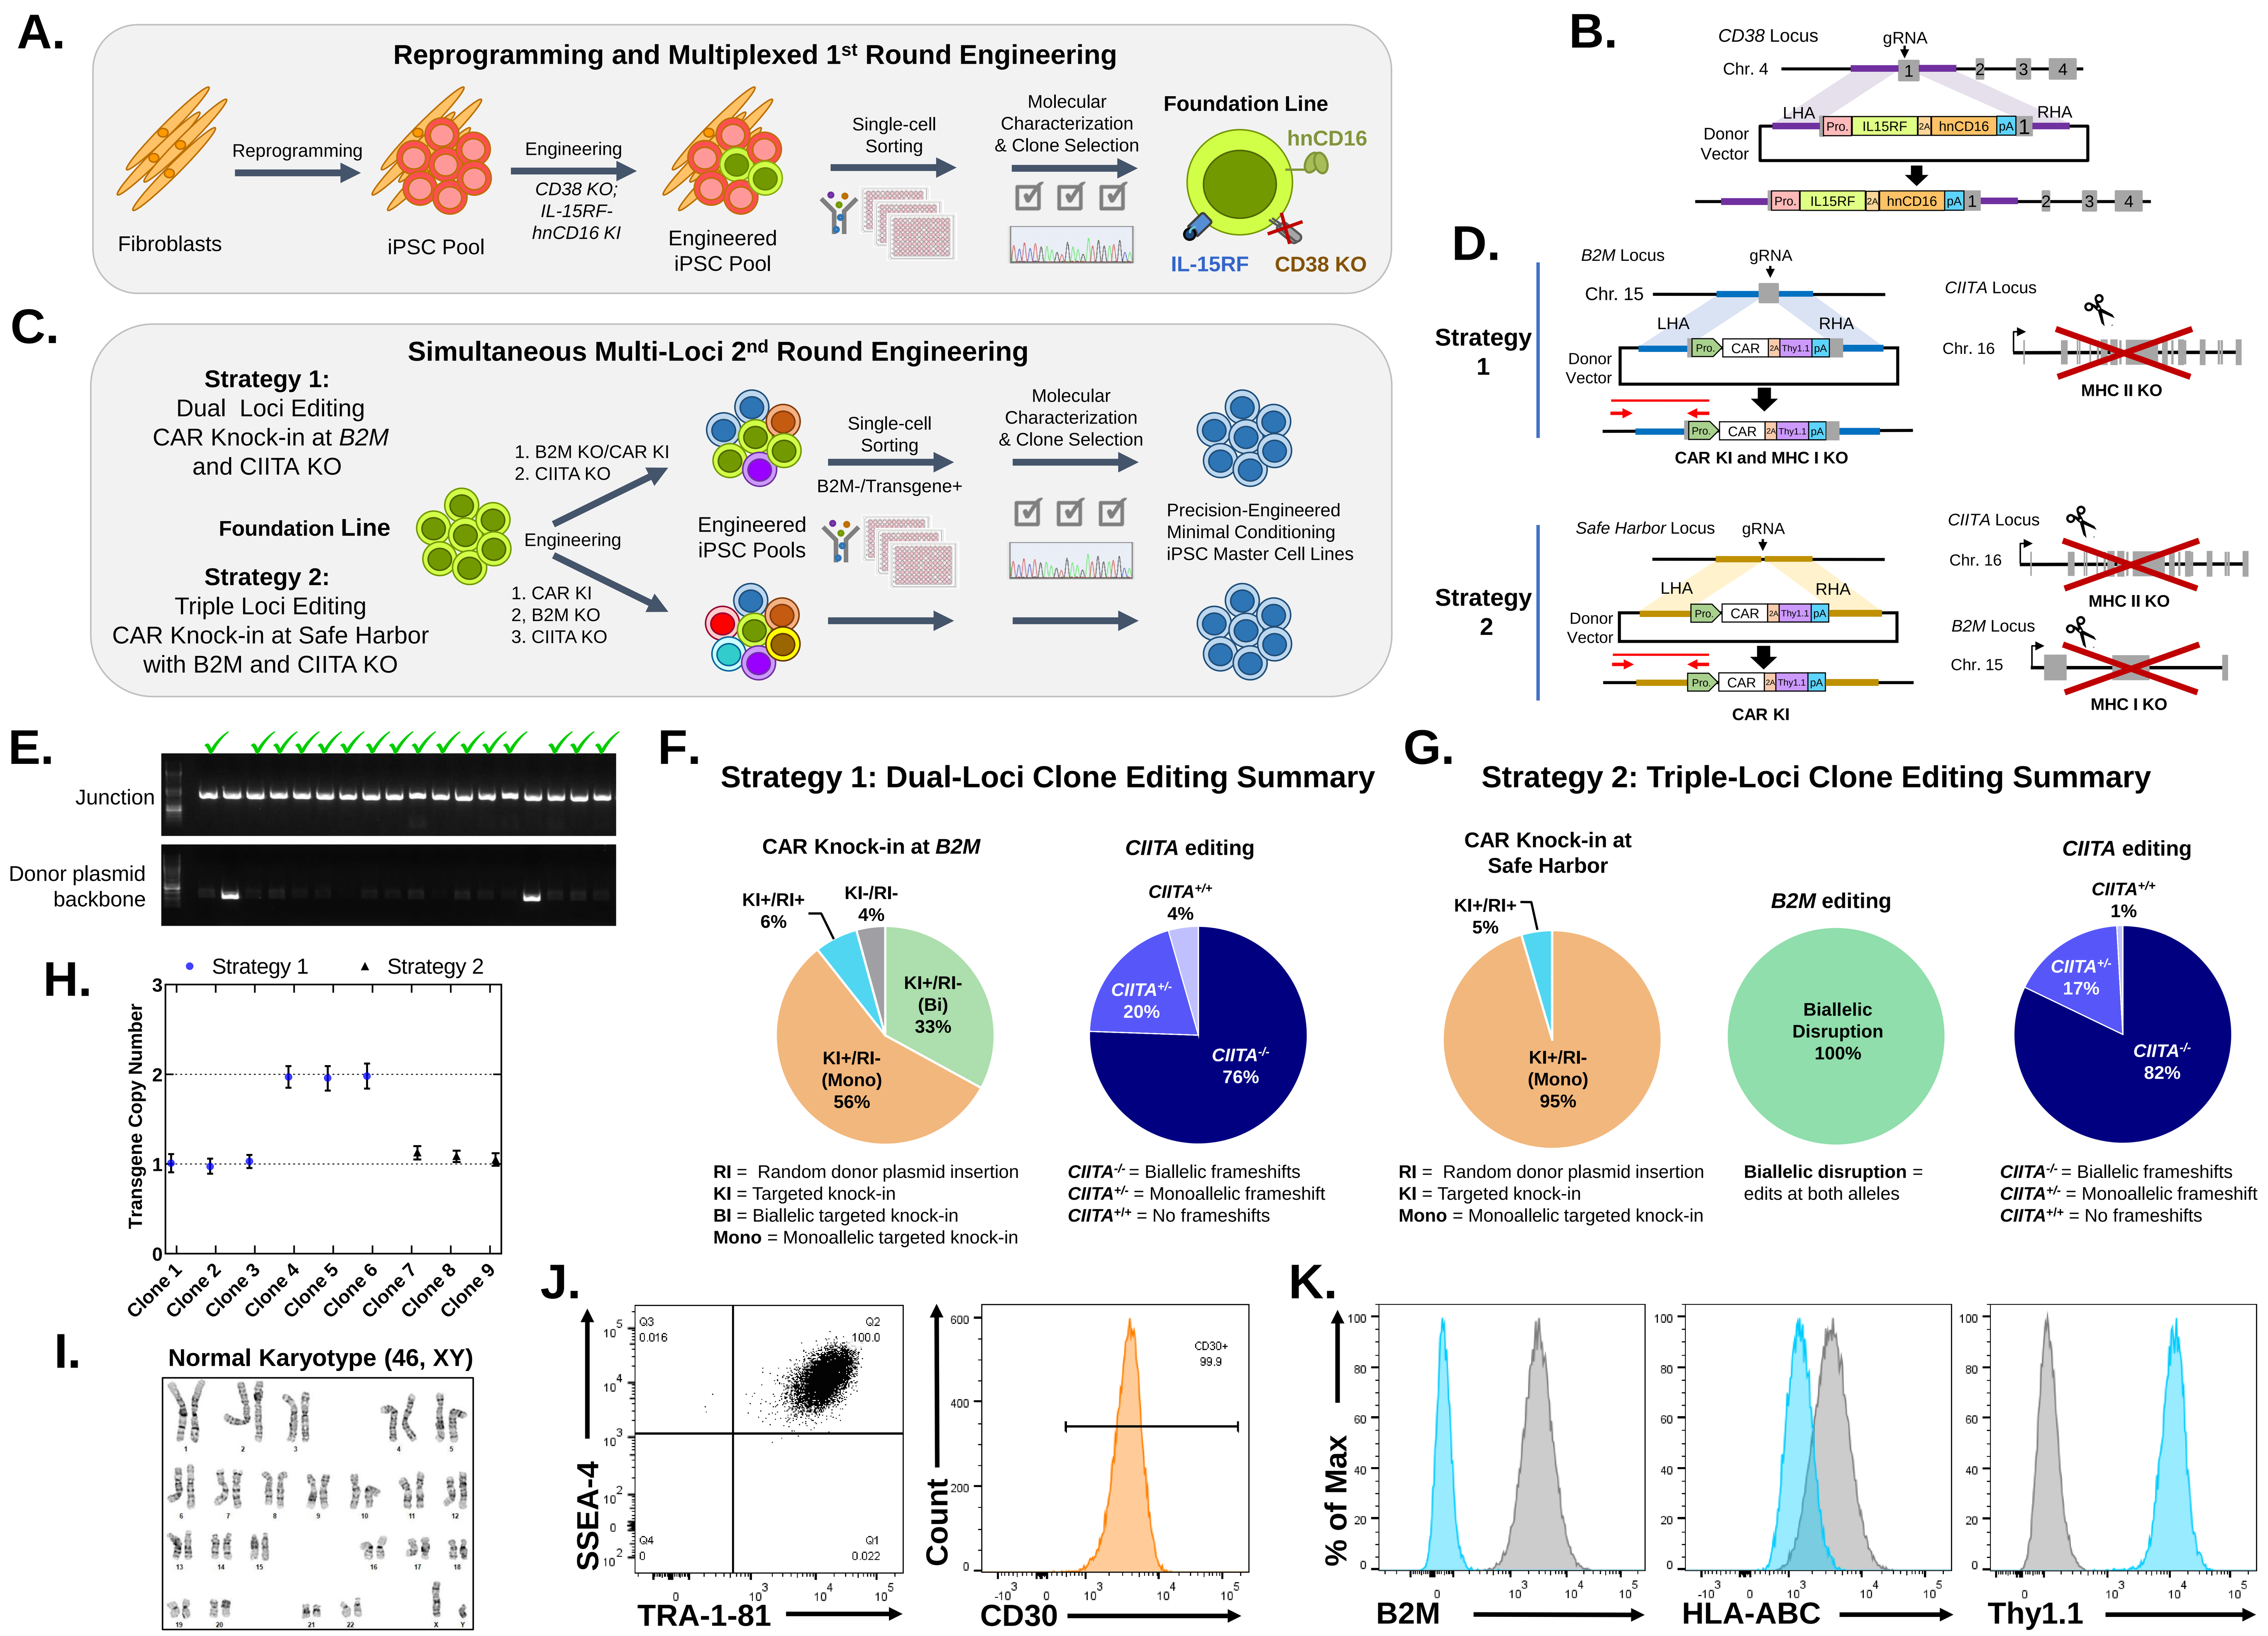
A. Reprogramming and Multiplexed 1st Round Engineering
Fibroblasts
Reprogramming
iPSC Pool
Engineering
CD38 KO; IL-15RF-hnCD16 KI
Engineered iPSC Pool
Single-cell Sorting
Molecular Characterization & Clone Selection
Foundation Line
hnCD16
IL-15RF
CD38 KO
B. CD38 Locus
gRNA
Chr. 4
LHA
RHA
Donor Vector
Pro. IL15RF 2A hnCD16 pA
C. Simultaneous Multi-Loci 2nd Round Engineering
Strategy 1: Dual Loci Editing CAR Knock-in at B2M and CIITA KO
Foundation Line
Strategy 2: Triple Loci Editing CAR Knock-in at Safe Harbor with B2M and CIITA KO
1. B2M KO/CAR KI
2. CIITA KO
1. CAR KI
2, B2M KO
3. CIITA KO
Engineering
Engineered iPSC Pools
Single-cell Sorting
B2M-/Transgene+
Molecular Characterization & Clone Selection
Precision-Engineered Minimal Conditioning iPSC Master Cell Lines
D. Strategy 1
B2M Locus
Chr. 15
CIITA Locus
Chr. 16
MHC II KO
CAR KI and MHC I KO
Strategy 2
Safe Harbor Locus
CAR KI
MHC I KO
E. Junction
Donor plasmid backbone
F. Strategy 1: Dual-Loci Clone Editing Summary
CAR Knock-in at B2M
KI+/RI+ 6%
KI-/RI- 4%
KI+/RI- (Bi) 33%
KI+/RI- (Mono) 56%
CIITA editing
CIITA+/+ 4%
CIITA+/- 20%
CIITA-/- 76%
RI = Random donor plasmid insertion
KI = Targeted knock-in
BI = Biallelic targeted knock-in
Mono = Monoallelic targeted knock-in
CIITA-/- = Biallelic frameshifts
CIITA+/- = Monoallelic frameshift
CIITA+/+ = No frameshifts
G. Strategy 2: Triple-Loci Clone Editing Summary
CAR Knock-in at Safe Harbor
KI+/RI+ 5%
KI+/RI- (Mono) 95%
B2M editing
Biallelic Disruption 100%
CIITA editing
CIITA+/+ 1%
CIITA+/- 17%
CIITA-/- 82%
Biallelic disruption = edits at both alleles
H. Strategy 1
Strategy 2
Transgene Copy Number
Clone 1
Clone 2
Clone 3
Clone 4
Clone 5
Clone 6
Clone 7
Clone 8
Clone 9
I. Normal Karyotype (46, XY)
J. SSEA-4
TRA-1-81
Q3 0.016
Q2 100.0
Q4 0
Q1 0.022
Count
CD30
CD30+ 99.9
K. % of Max
B2M
HLA-ABC
Thy1.1

Figure 1. High precision, simultaneous multi-loci engineering of minimal conditioning iPSC clones from a multiplexed-engineered foundation line. (A) A multiplexed-engineered iPSC foundation line was generated from reprogrammed healthy donor fibroblasts that were engineered for simultaneous knock-in of the IL-15RF and hnCD16 at the CD38 locus. **(B)** Design of donor construct for simultaneous transgene knock-in into the CD38 locus to facilitate CD38 gene disruption. **(C)** Two multi-loci 2nd round engineering strategies for simultaneous knockout of B2M and CIITA and CAR knock-in were conducted in parallel to generate iPSC master cell lines with additional immune-evasive edits from the multiplexed-engineered foundation line. Strategy 1 consists of dual-loci engineering in which CAR knock-in was directed to the *B2M* locus for synchronized knockout of B2M with concurrent CIITA knockout via a *CIITA* targeted gRNA. Strategy 2 consists of triple-loci engineering in which CAR knock-in was directed to a safe harbor locus while gRNAs targeting *B2M* and *CIITA* were simultaneously administered for knockout. Engineered iPSCs were single cell sorted for transgene marker expression and loss of B2M surface expression in both strategies. **(D)** Multi-loci targeting layouts for strategies 1 and 2 with designs of donor constructs for simultaneous CAR knock-in and B2M knockout (strategy 1) and CAR knock-in at a safe harbor locus (strategy 2). **(E)** PCR assay to screen for clones with targeted integration of the transgene cassette into the *B2M* locus (Junction) and without the random integration of the donor plasmid (Donor plasmid backbone). A similar assay was used to screen for integration at the safe harbor locus. **(F, G)** Molecular characterization of sorted clones show highly efficient and precise transgene targeting (>90% of clones with on-target integration without random donor plasmid integration in both strategies) and high efficiency, *B2M* and *CIITA* editing. **(H)** Transgene copy numbers in isolated clones were determined using ddPCR. Both mono- and biallelic clones were identified. **(I)** Representative cytogenic analyses of engineered iPSC clones demonstrate normal, diploid karyotypes post multi-loci engineering for both strategies. **(J)** Representative flow cytometry profile of clonal iPSCs demonstrates uniform expression of pluripotency markers SSEA-4, TRA-1-81, and CD30 for all isolated clones. **(K)** Representative flow cytometry profile of engineered iPSCs demonstrates loss of B2M and MHC class I expression and uniform expression of the transgene marker (blue) compared to non-engineered iPSCs (grey).

iPSCs with Engineered Immune-Evasion Modalities Robustly Differentiate into NK cells

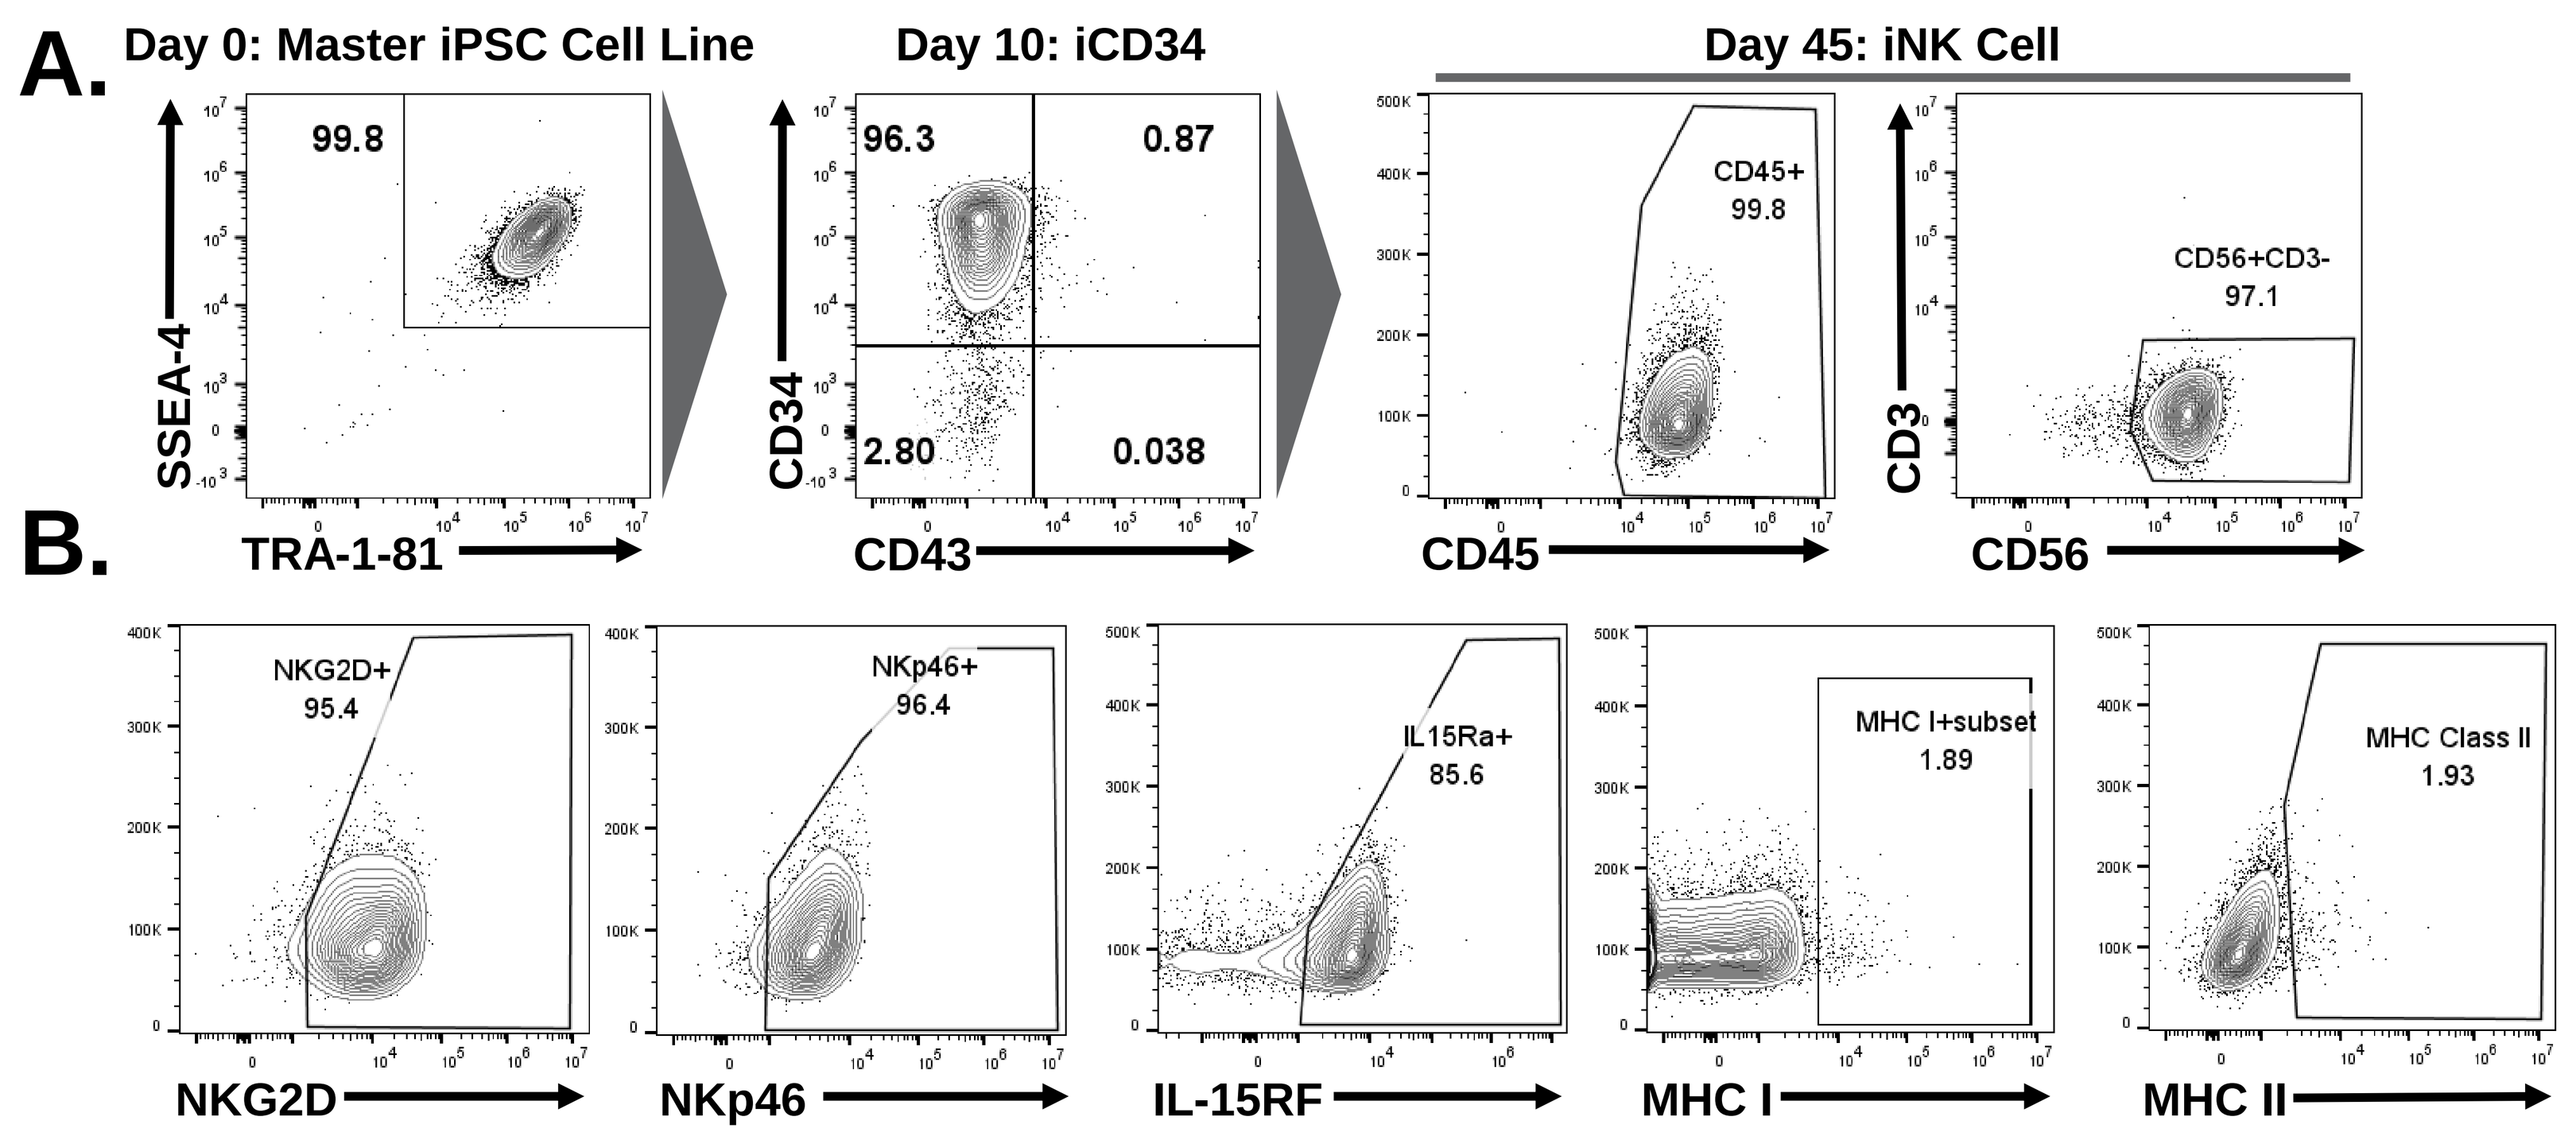
A. Day 0: Master iPSC Cell Line
Day 10: iCD34
Day 45: iNK Cell
SSEA-4
TRA-1-81
99.8
CD34
CD43
96.3
0.87
2.80
0.038
CD45
CD45+ 99.8
CD3
CD56
CD56+CD3- 97.1
B. NKG2D
NKG2D+ 95.4
NKp46
NKp46+ 96.4
IL-15RF
IL15Ra+ 85.6
MHC I
MHC I+subset 1.89
MHC II
MHC Class II 1.93

Figure 2. Phenotype of off-the-shelf NK product engineered with anti-tumor and immune-evasive elements. (A) Immune-evasive NK cells are produced by directed differentiation of SSEA4⁺TRA-1-81⁺ iPSCs to CD34⁺ hematopoietic progenitors and then to CD45⁺CD56⁺ NK cells. **(B)** CD45⁺/CD56⁺ immune-evasive iNK cells express the activating receptors NKG2D and NKp46 with expression of IL-15RF and knockout of all MHC class I (HLA-A, B, and C) and MHC class II (HLA-DR, DP, and DQ) receptor subtypes.

Engineered Immune-Evasion Modalities Prevent the Emergence of Alloreactive NK and T Cells

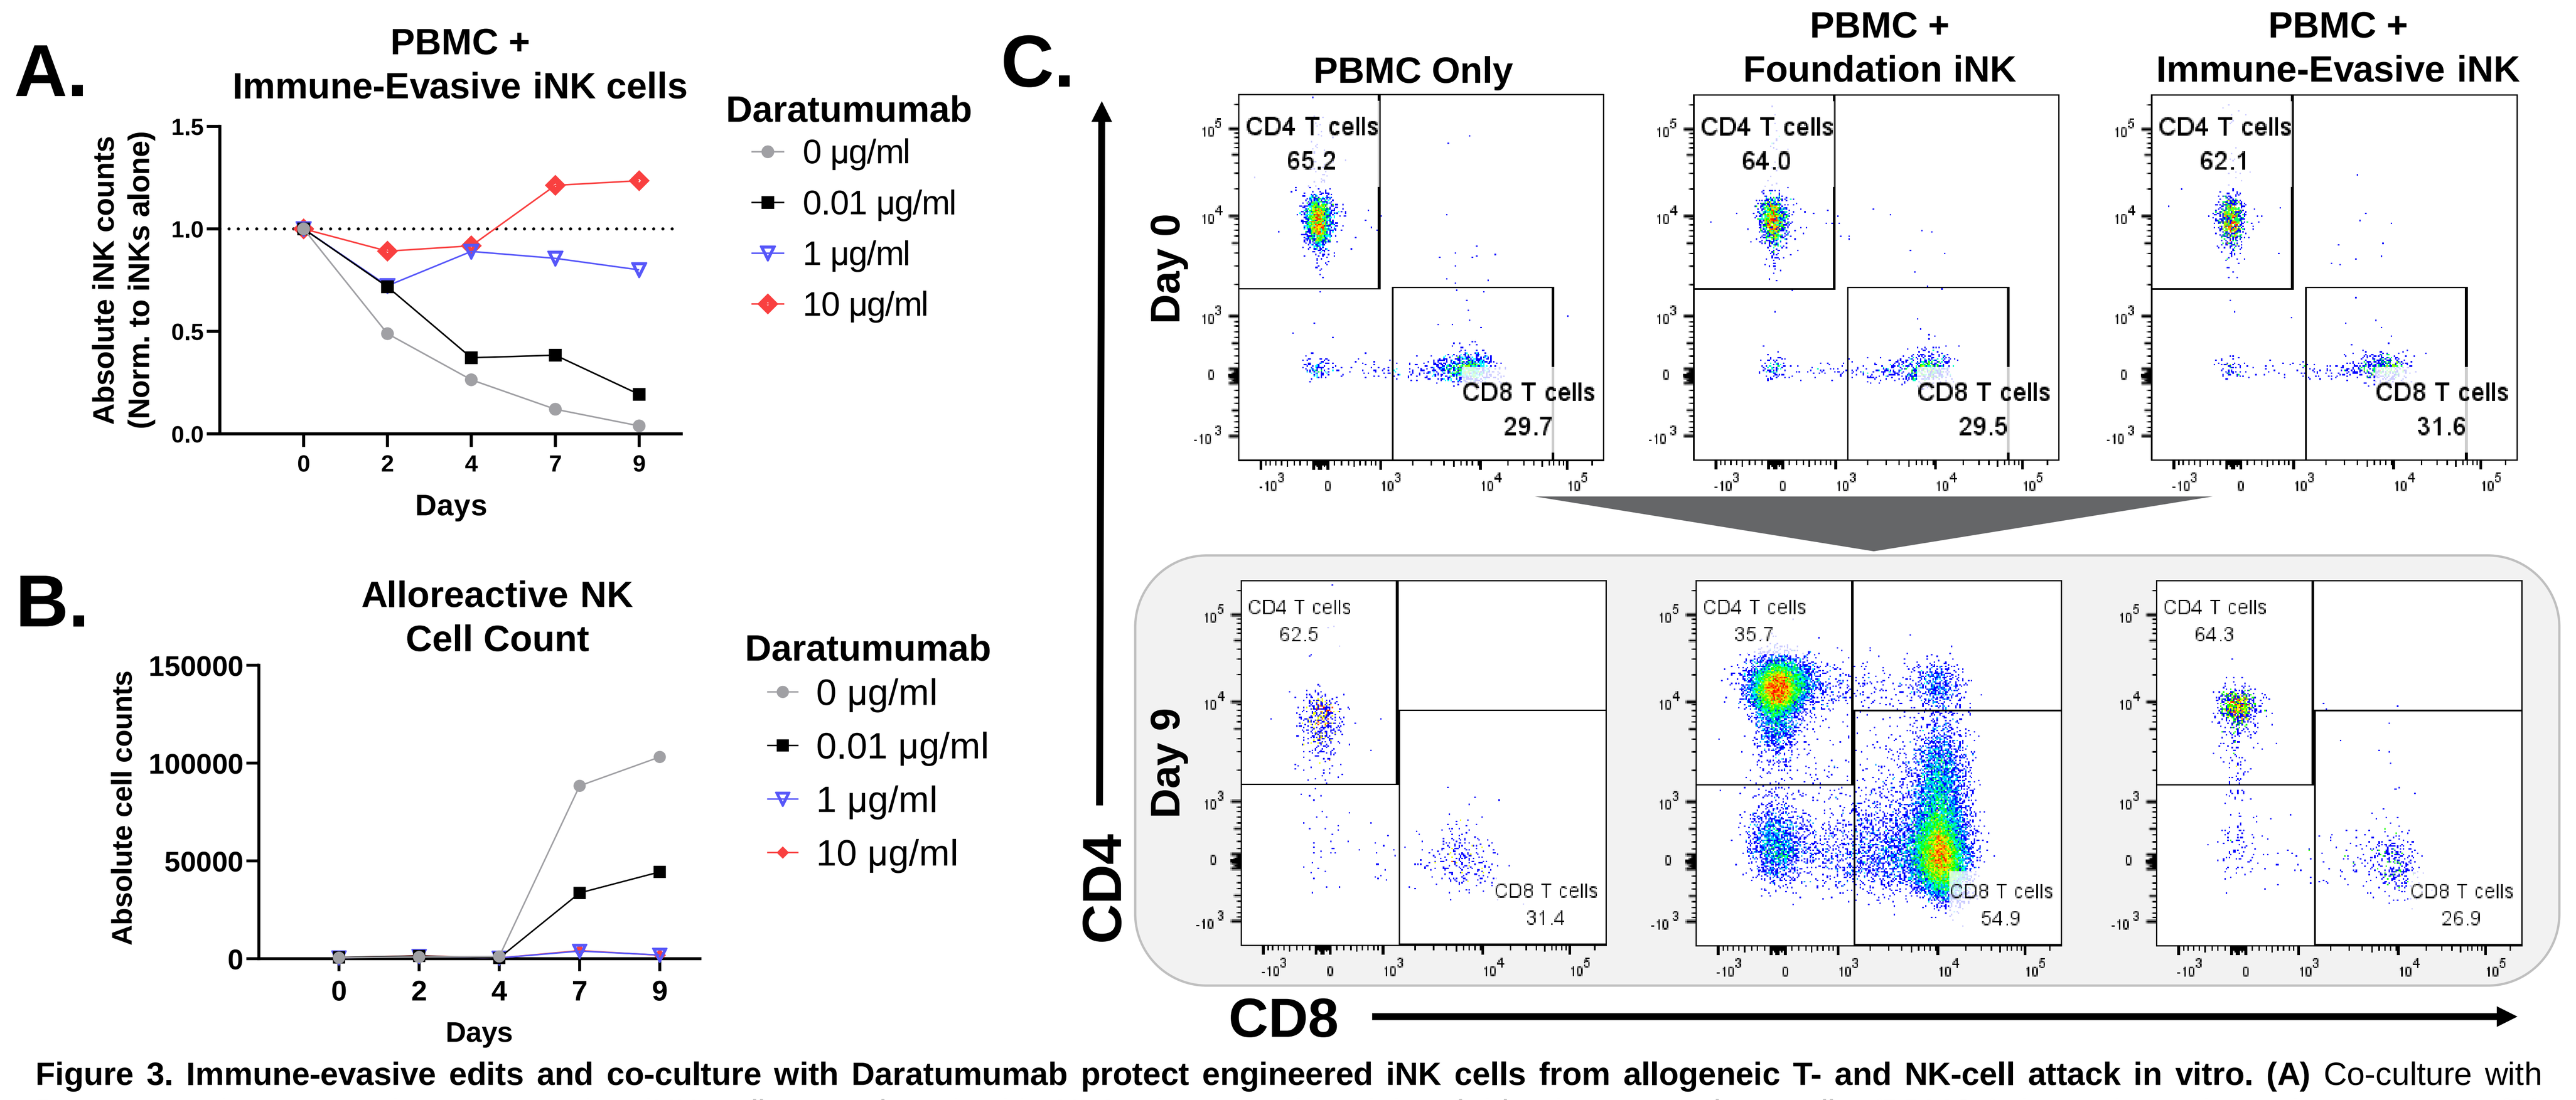
A. PBMC + Immune-Evasive iNK cells
Absolute iNK counts (Norm. to iNKs alone)
Days
Daratumumab
0 μg/ml
0.01 μg/ml
1 μg/ml
10 μg/ml
B. Alloreactive NK Cell Count
Absolute cell counts
Days
C. PBMC Only
PBMC + Foundation iNK
PBMC + Immune-Evasive iNK
Day 0
Day 9
CD4
CD8
CD4 T cells 65.2
CD8 T cells 29.7
CD4 T cells 64.0
CD8 T cells 29.5
CD4 T cells 62.1
CD8 T cells 31.6
CD4 T cells 62.5
CD8 T cells 31.4
CD4 T cells 35.7
CD8 T cells 54.9
CD4 T cells 64.3
CD8 T cells 26.9

Figure 3. Immune-evasive edits and co-culture with Daratumumab protect engineered iNK cells from allogeneic T- and NK-cell attack in vitro. (A) Co-culture with Daratumumab protects immune-evasive iNK cells co-cultured with HLA-mismatched peripheral blood mononuclear cells (PBMCs) in a dose dependent manner. **(B)** Daratumumab decreases the number of alloreactive NK cells in a dose dependent manner when immune-evasive iNK cells are co-cultured with HLA-mismatched PBMCs. **(C)** iNK cells engineered with B2M and CIITA knockout edits do not stimulate the expansion of CD4⁺ and CD8⁺ T cells when co-cultured with HLA-mismatched PBMCs.

Summary

- As a first-of-kind demonstration, we successfully generated precision-engineered iPSC master cell lines incorporating six unique functional elements (including 3 bi-allelic disruptions) at clonal level that drive anti-tumor activity and functional persistence through multiple rounds of highly efficient, on-target engineering with simultaneous editing at multiple loci
- CD38 knockout combined with anti-CD38 Daratumumab treatment protects iNK cells from allorejection in vitro by selectively depleting alloreactive CD38+ lymphocytes
- Ablation of MHC class I and II via simultaneous knockout of B2M and CIITA prevents the activation of alloreactive CD4⁺ and CD8⁺ T cells by multiplex-engineered iNK cells
- These data validate the robustness of our proprietary iPSC product platform to support high-precision, multi-loci engineering of iPSCs to create clonally derived, multiplexed-engineered iPSC master cell lines to produce next-generation, off-the-shelf immune-evasive CAR-iNK cell therapies for use in patients with minimal requirements for immune suppression chemotherapy conditioning