Detection of Genetically Engineered iPSC-derived Natural Killer Cells in Blood and Tissue

Cara Bickers*, Judy Martin*, Steven Castro, Jason Zhang, Thomas Dailey, Eric Sung, Suzanna Gasparian, Jason O'Rourke, Moyar Ge, Tom T Lee, Janel Huffman, Jode Goodridge, Ryan Bjordahl, Bahram Valamehr, Peter M Szabo, Lilly Wong, Sarah Cooley

Fate Therapeutics, Inc., San Diego, CA, USA

Poster 1138

ABSTRACT

Immune cell therapies derived from induced pluripotent stem cells (iPSC) provide a novel opportunity for the treatment of multiple cancer types. Assessment of the persistence and biodistribution of these product candidates requires specific and sensitive methods to detect engineered cells in both liquid and solid biopsies. Here, we present the development and validation of two complimentary nucleic acid-based detection assays for iPSC-derived natural killer (iNK) cell product candidates containing Fate's proprietary high-affinity, non-cleavable CD16 transgene (hnCD16).

The first assay is a droplet digital PCR (ddPCR) method to detect and quantify hnCD16 transgene copies present in a pool of genomic DNA (gDNA). The primers and probe were designed to recognize the optimized codons of hnCD16. Assay linearity and accuracy were assessed through titration studies using 0.024 to 1 ng of hnCD16-containing DNA spiked into different amounts of hnCD16-negative gDNA. Precision was determined through multiple assay runs by different operators on two instruments. The second assay is an *in situ* hybridization based method utilizing RNAscope™ technology to detect cells expressing hnCD16 in fixed tissue. Probes targeting hnCD16 were used to optimize signal specificity. Cells expressing hnCD16 and tissues from in vivo studies treated with iNK products served as positive controls.

For the ddPCR assay, absolute limit of detection (aLoD) was determined to be 4.9 copies of hnCD16 per 20 µL reaction, regardless of total genomic mass input. Absolute limit of quantification (aLoQ) was 12 copies per 20 µL reaction with a %CV ≤30. Relative limit of quantification (rLoQ), assessing transgene to total DNA ratio, is affected by the background gDNA input and is less sensitive with lower input mass. rLoQ for total mass of 70 - 250 ng was 97 - 22 copies/µg gDNA (0.064% - 0.015%) with a %CV ≤30. The sensitivity of this input range allows evaluation of clinical samples with low cellularity. While ddPCR provides robust quantification of the hnCD16 transcript, the RNAscope™ assay informs localization of the iNK product. Specificity of the probe was established by confirming its lack of affinity for endogenous CD16 using a variety of human normal and tumor tissues and by staining hnCD16-positive fixed cell pellets and tissues from in vivo studies. In cell pellets, positive RNAscope™ signal correlated with the known ratio of transgene positive cells. In murine tissues previously confirmed to contain iNK cell product, the RNAscope™ positive staining correlated with NKG2A immunohistochemistry staining, confirming the presence of product NK cells.

The combined use of both the ddPCR and RNAscope™ assays targeted to hnCD16 allows for detection and quantification of transgene-bearing iNK cells in a wide variety of patient samples including tumor biopsies. Both assays are being utilized for cell detection and quantification in our ongoing clinical trials.

GRAPHICAL ABSTRACT

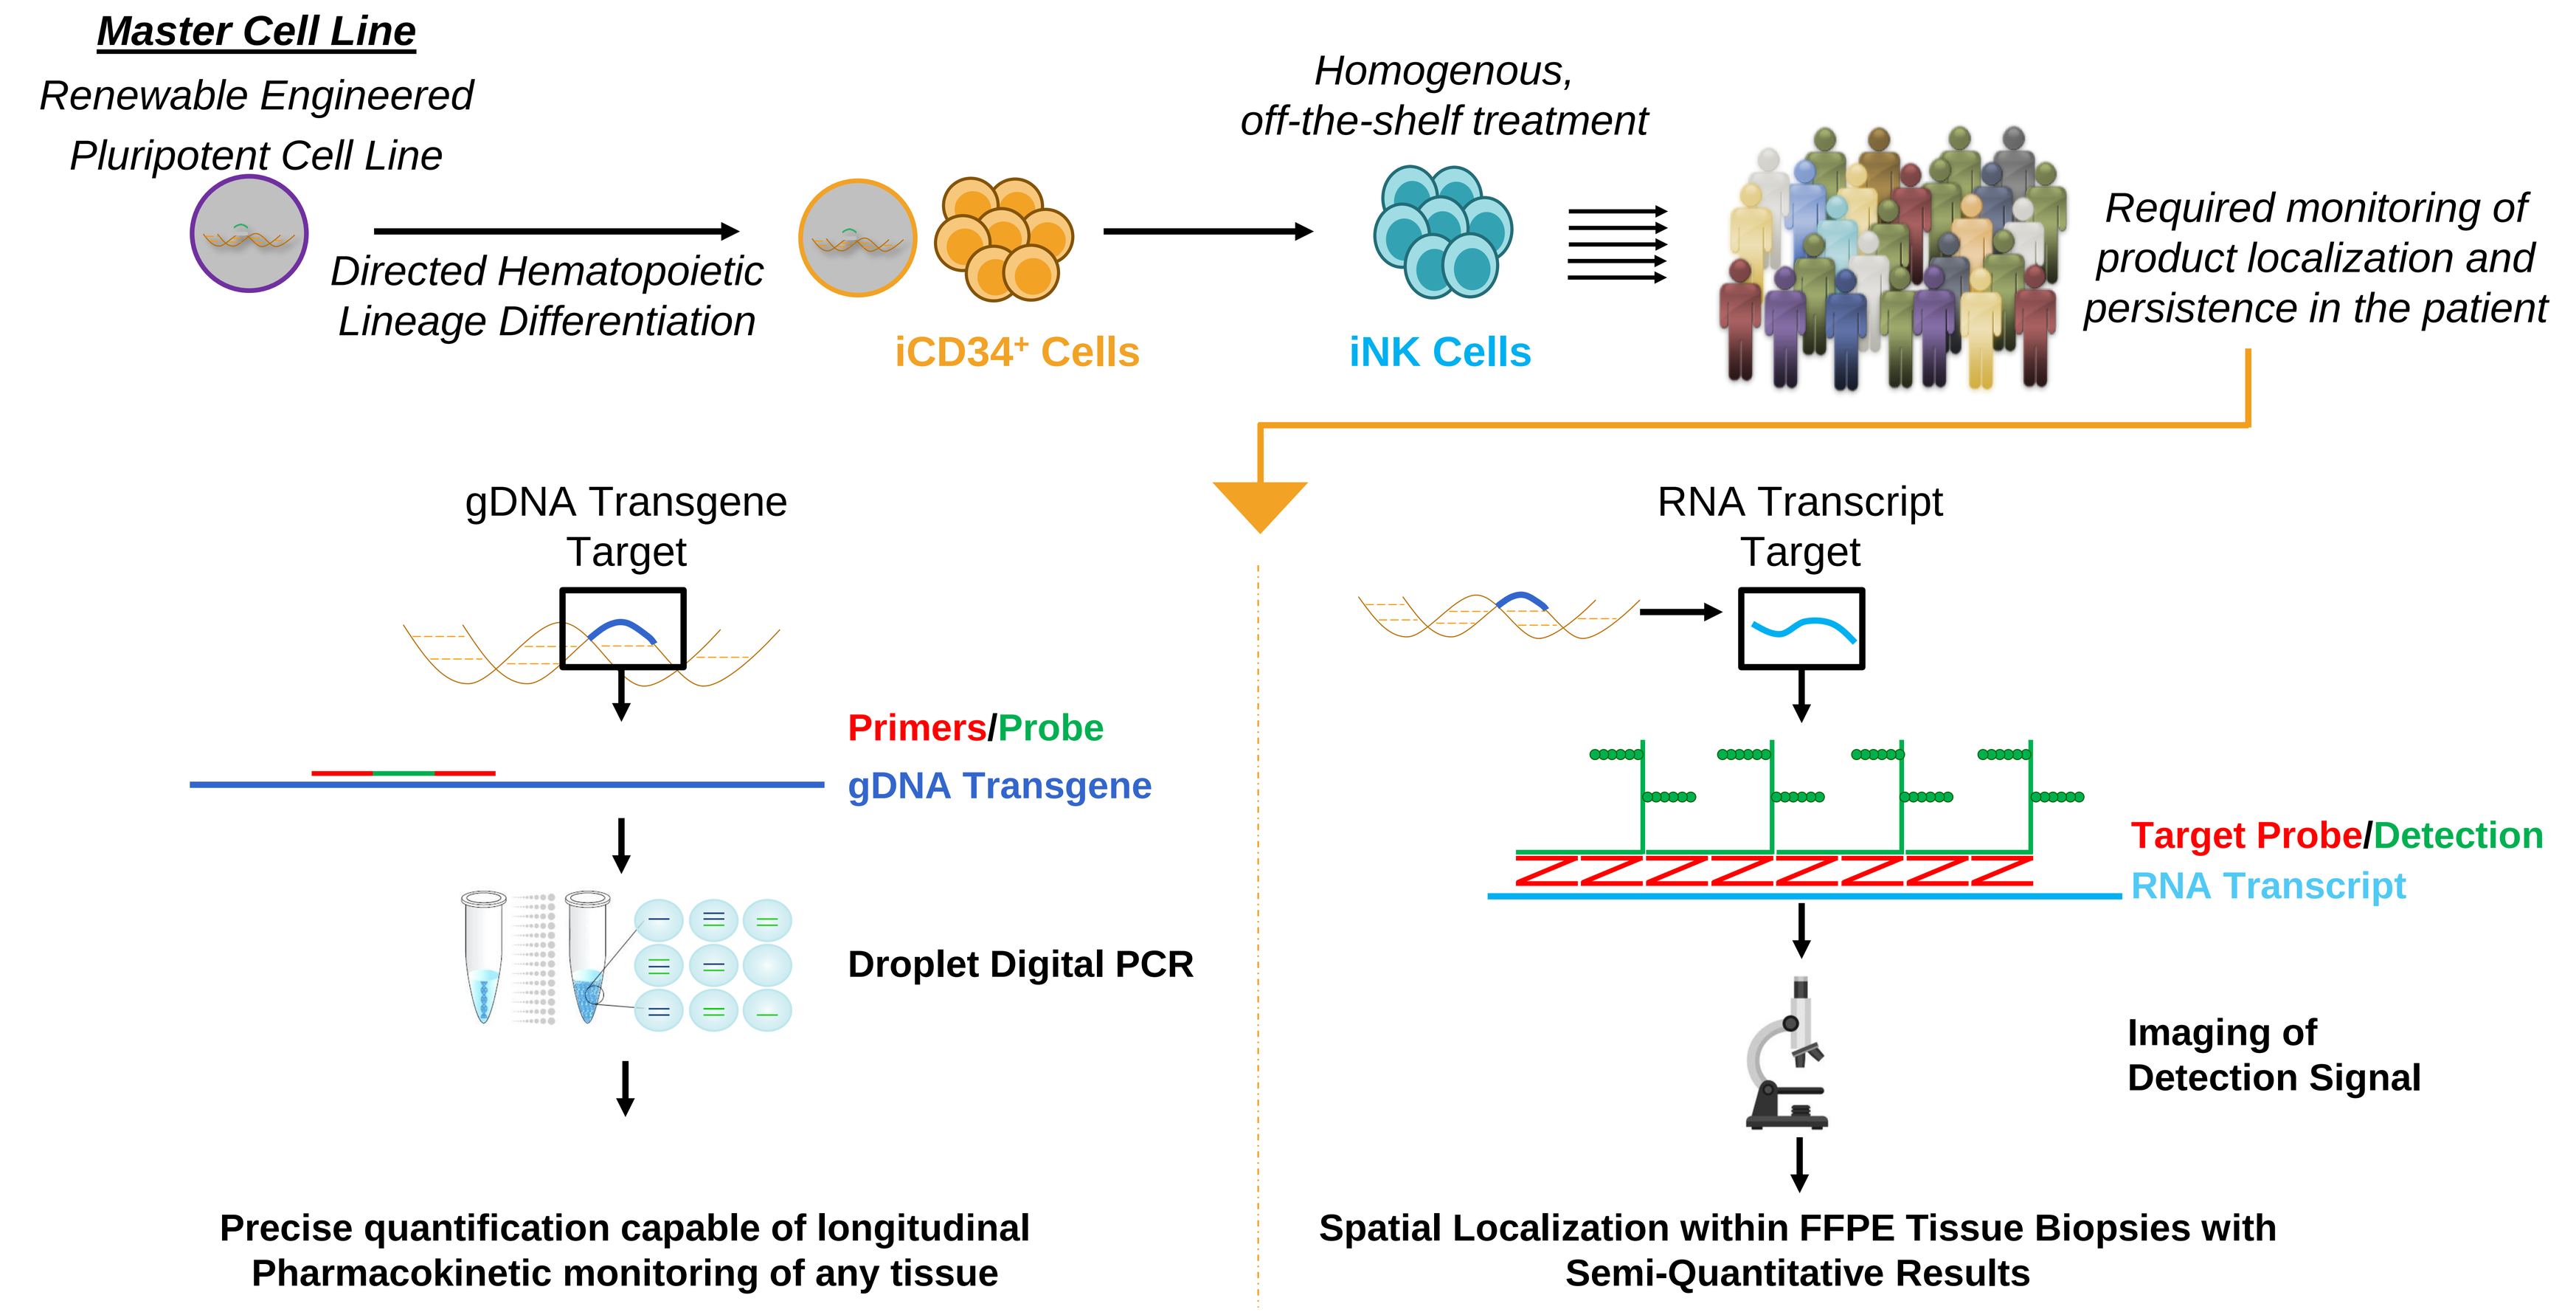
Master Cell Line
Renewable Engineered Pluripotent Cell Line
Directed Hematopoietic Lineage Differentiation
iCD34+ Cells
Homogenous, off-the-shelf treatment
iNK Cells
Required monitoring of product localization and persistence in the patient
gDNA Transgene Target
Primers/Probe
gDNA Transgene
Droplet Digital PCR
Precise quantification capable of longitudinal Pharmacokinetic monitoring of any tissue
RNA Transcript Target
Target Probe/Detection
RNA Transcript
Imaging of Detection Signal
Spatial Localization within FFPE Tissue Biopsies with Semi-Quantitative Results

SUMMARY

- hnCD16 is a novel transgene that induces expression of a high-affinity, non-cleavable CD16a Fc receptor that is incorporated in several of Fate's off-the-shelf, iPSC-derived NK (iNK) cell product candidates.
- The codon optimization of this transgene has allowed us to design and validate parallel probe-based assays for detection of both genomic DNA and RNA transcript that uniquely identify hnCD16 without detection of endogenous CD16. In pre-clinical studies, we have applied our assays to precisely quantify hnCD16-bearing iNK cell products in diverse backgrounds including both human and murine solid and liquid tissue samples.
- With optimal gDNA input, validation established a relative LOQ of 0.028% / 42 copies per microgram.
- We applied our ddPCR assay to quantify hnCD16 transgene copies in peripheral blood samples from a patient administered with FT538 on clinical study NCT04614636. Parallel quantification of FT538 product cells, performed via flow cytometry by using product-specific HLA antibodies mis-matched to the recipient, demonstrated concordance between hnCD16 transgene copies and detectable HLA mis-matched product cells.
- We used RNAscope™ technology to develop a highly specific, semi-quantitative assay to localize hnCD16+ iNK cells in FFPE tissue. This assay will be employed to detect the presence and spatial localization of iNK in tissues derived from Fate clinical trials.

RESULTS

Figure 1. ddPCR Workflow and Representative Data Output

A

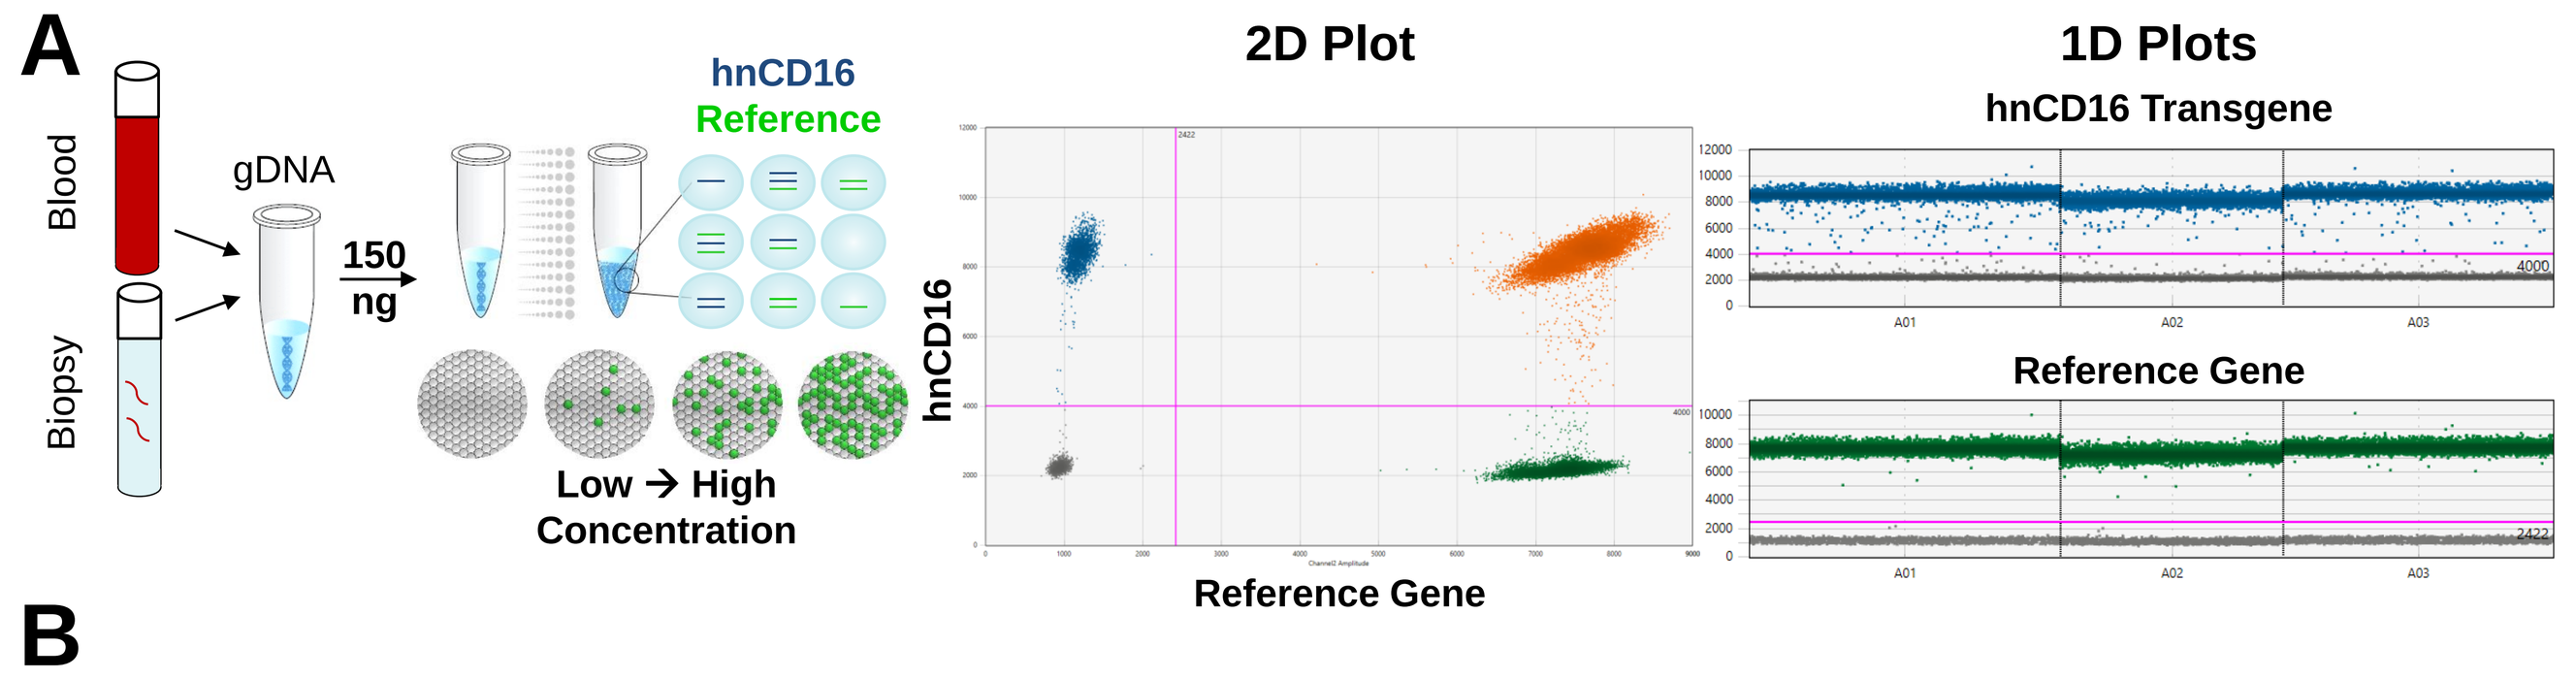
Blood
Biopsy
gDNA
150 ng
hnCD16
Reference
Low → High Concentration
2D Plot
hnCD16
Reference Gene
1D Plots
hnCD16 Transgene
Reference Gene

C

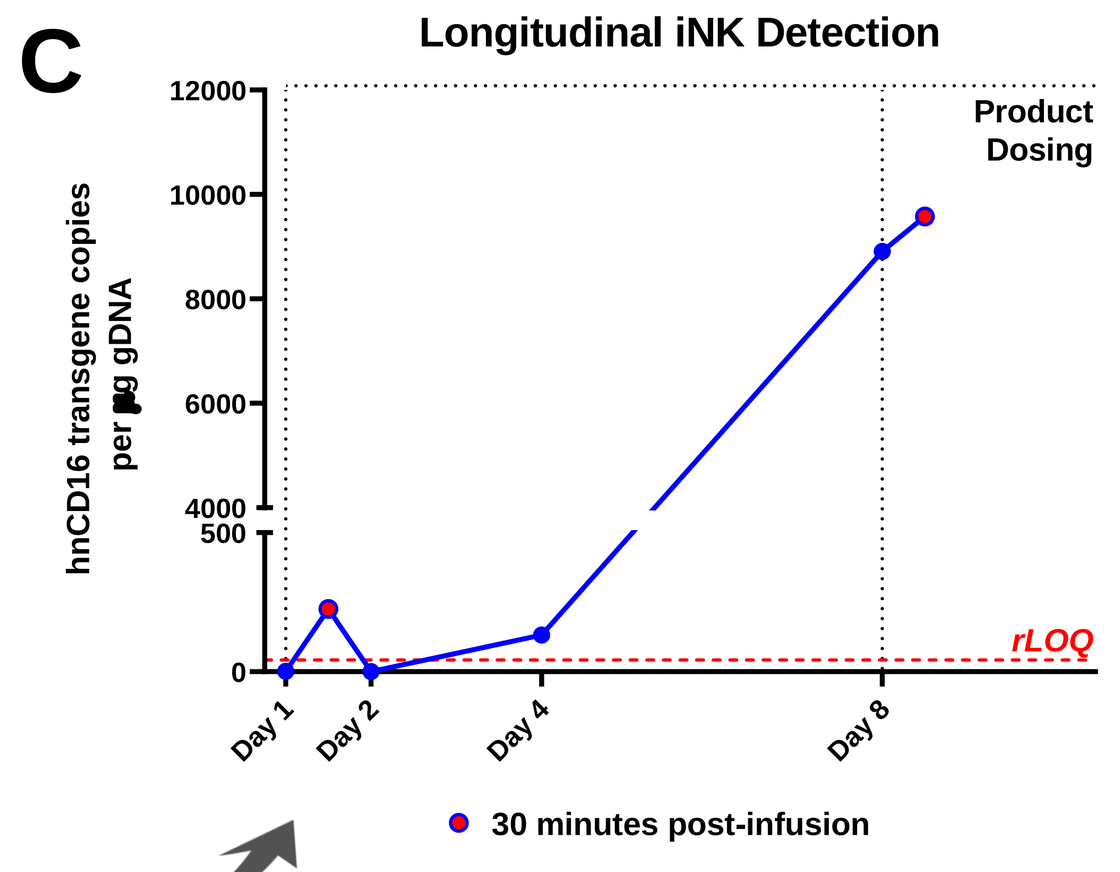
Longitudinal iNK Detection
hnCD16 transgene copies per µg gDNA
Product Dosing
rLOQ
Day 1
Day 2
Day 4
Day 8
30 minutes post-infusion

B

	Reference Gene			iNK Target Gene – hnCD16			Reported Output
Timepoint	Copies / µL	Total Copies per Reaction	Estimated Cell Equivalents	Copies / µL	Total Copies per Reaction	Percentage Positive	Transgene Copies / µg of gDNA
Day 4	1,902.74	38,054.8	19,027	0.83	16.6	0.087	132
Day 8 Pre	1,996.87	39,937.4	19,969	58.70	1,174	5.88	8,908
Day 8 Post	1,182.24	23,644.8	11,822	37.37	747.4	6.32	9,579

Figure 1. (A) gDNA extracted from subject-derived blood or tissue samples is loaded into a ddPCR assay targeting the biallelic reference gene and the iNK transgene, hnCD16. The assay is partitioned into ~20,000 individual droplets and undergoes 45 thermocycles. Droplets are analyzed individually for fluorescence derived from each target hydrolysis-probe and displayed in either a 2D or 1D plot, gated appropriately, and quantified. (B) Utilizing the known copy numbers of both target assays and the fixed mass of 0.0066 ng of gDNA per cell, the target gene concentrations are converted into a ratio of iNK to total genomic mass to report hnCD16 transgene copies/µg of genomic DNA. (C) Pharmacokinetics of FT538 in the peripheral blood (transgene copies/µg genomic DNA) days 1-8 of cycle 2 in a patient with AML treated with FT538 monotherapy (NCT04614636). rLOQ = relative limit of quantification.

Figure 2. hnCD16 ddPCR Assay Validation

A

Validation Design

Specificity
- Negative gDNA – 10 donors
- 5 Total Mass Inputs

Precision/Robustness
- Technical Triplicates
- Four Experimental Runs
- 2 Operators
- 2 machines

Accuracy/Linearity
- 5-point hnCD16+ gDNA Titration
- 5 total mass inputs

B

Precision and Robustness of Absolute Quantification

Titration hnCD16 Input (ng)	Expected Concentration (copies / µL)	Observed Average Concentration (copies / µL)	Relative Bias (%)	Btw-Run SD	Within-Run SD	Total SD	%CV
0.024	0.165	0.092	44.2	0.029	0.078	0.083	89.6
0.268	1.846	1.845	0.03	0.190	0.316	0.369	20.0
0.512	3.526	3.648	3.45	0.279	0.494	0.567	16.1
0.756	5.207	5.275	1.31	0.266	0.554	0.615	11.8
1	6.887	6.922	0.51	0.553	0.683	0.879	12.8
aLOD: 4.9 copies/reaction							

C

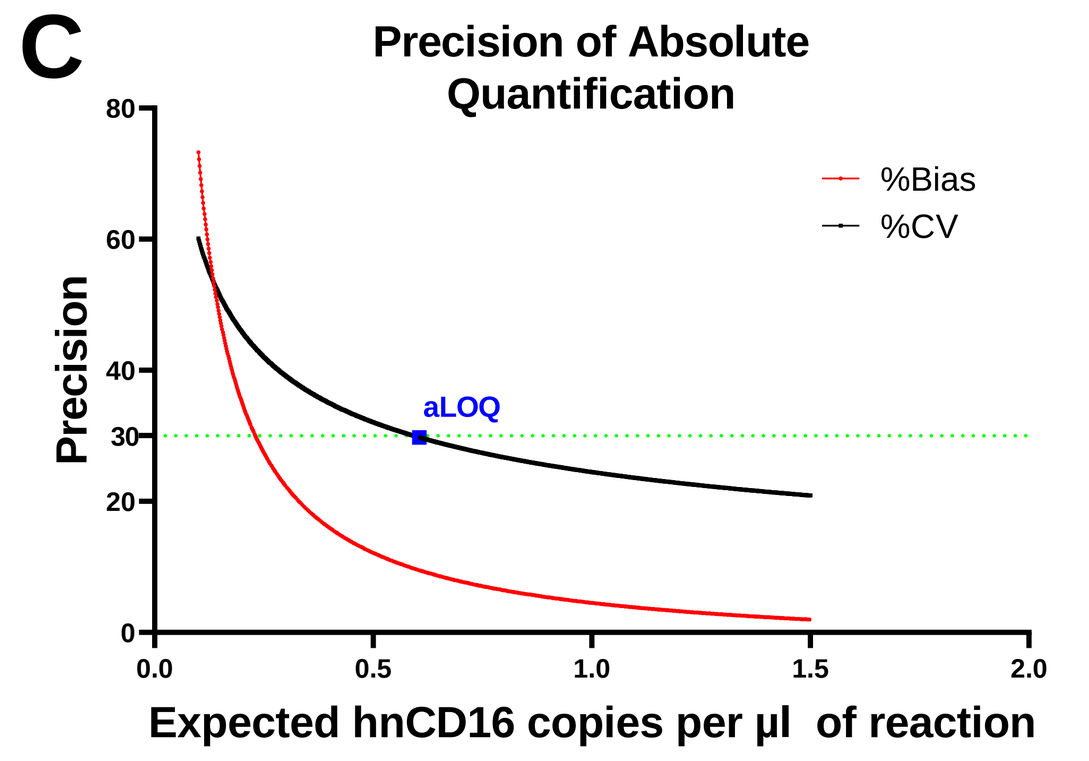
Precision of Absolute Quantification
%Bias
%CV
aLOQ
Precision
Expected hnCD16 copies per µl of reaction

D

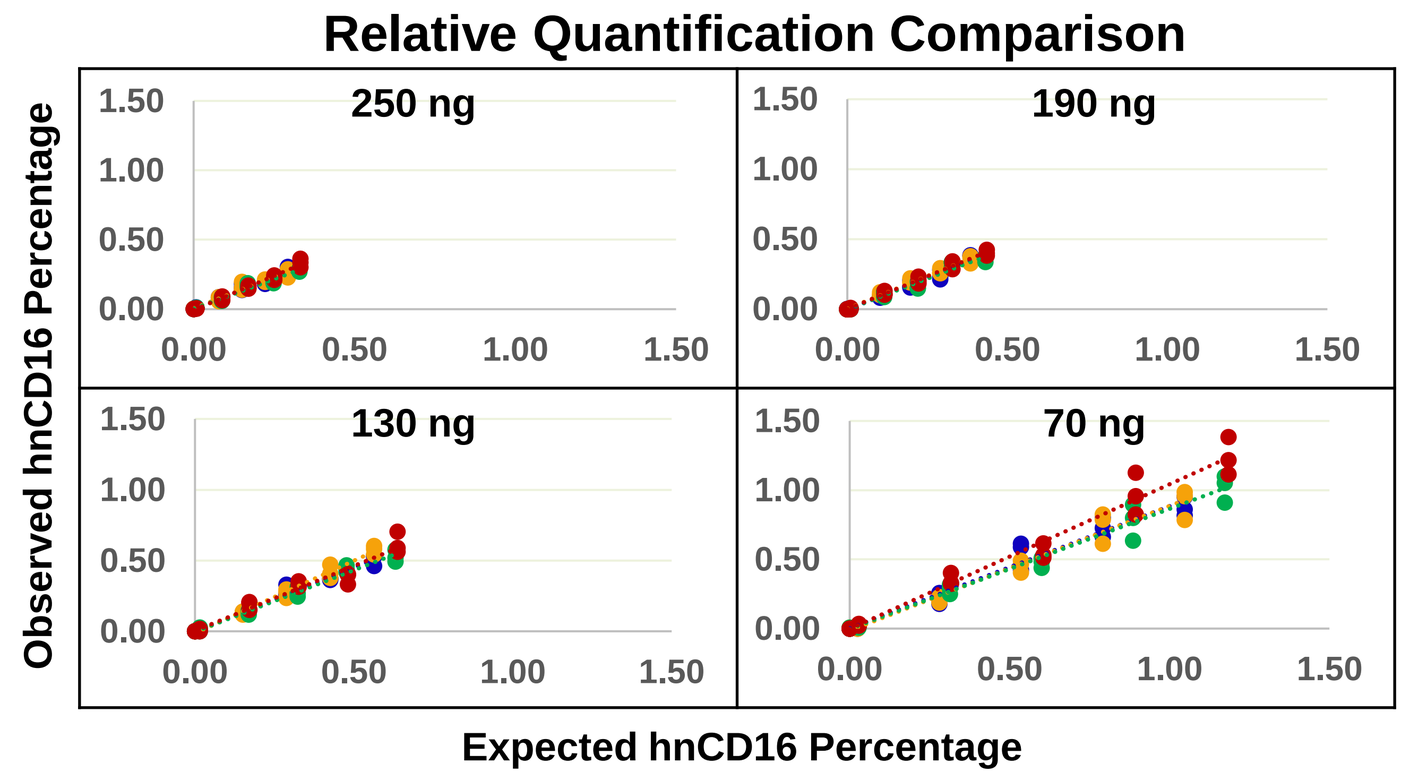
Relative Quantification Comparison
250 ng
190 ng
130 ng
70 ng
Observed hnCD16 Percentage
Expected hnCD16 Percentage

E

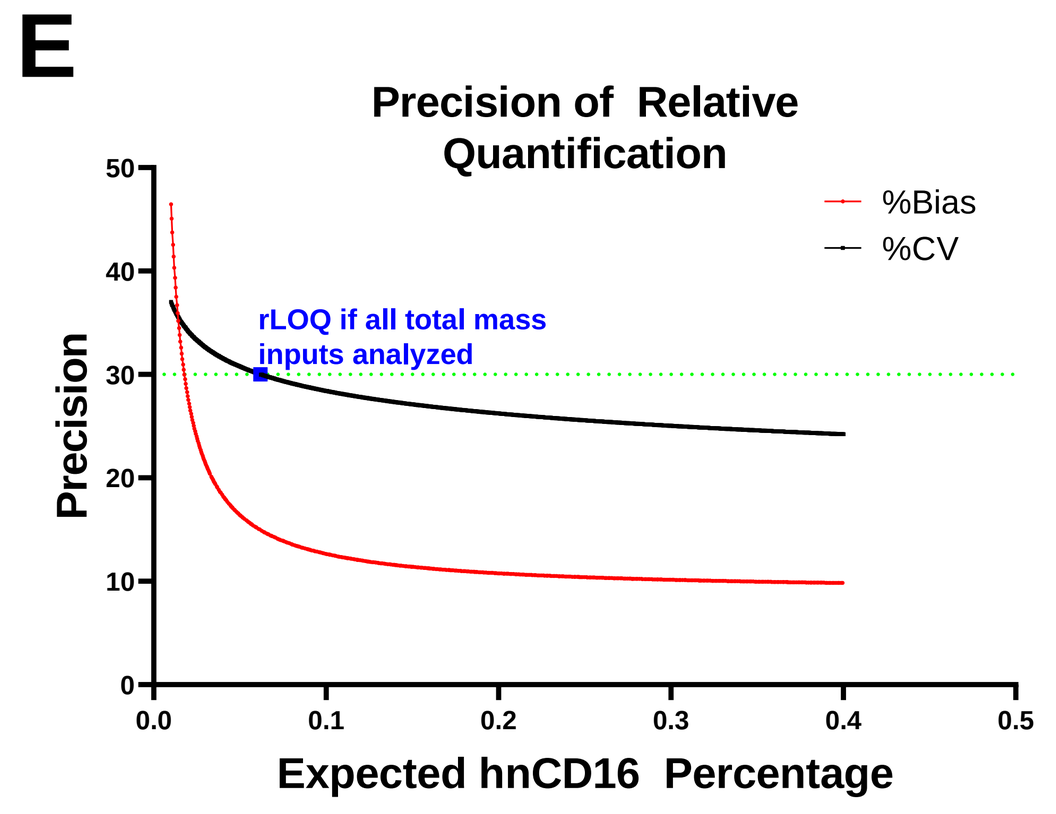
Precision of Relative Quantification
%Bias
%CV
rLOQ if all total mass inputs analyzed
Precision
Expected hnCD16 Percentage

F

Relative Quantification LOQ

Data Used for Analysis	LOQ (%)	LOQ (Copies / µg gDNA)
All Mass Inputs	0.062	93.9
Exclude 10 ng	0.034	51.5
250 ng	0.015	22.3
> 100 ng	0.028	42.0

G

%CV ≤ 30

aLOD: 4.9 copies / 20 µl reaction
aLOQ: 12 copies / 20 µl reaction
rLOQ*: 42 copies per µg gDNA 0.028%

Figure 2. (A) General validation design. (B) Table of statistical analysis of precision between replicates, operators, and machines summarized for each point in the titration curve. aLOD: absolute LOD. (C) Graphical representation of precision calculated by %Bias and %CV for absolute quantification output. aLOQ: absolute LOQ. (D) Comparison of relative quantification across both operators and both machines focusing on the higher range of mass inputs that minimally affect relative quantification. Note the higher variation and output values at the lowest input. (E) Graphical representation of precision calculated by %Bias and %CV for relative quantification output. rLOQ: relative LOQ. (F) LOQ results calculated from different subsets of the data. (G) Validation derived absolute LOD and LOQ and relative LOQ with a %CV ≤ 30. rLOQ*: relative LOQ with >100 ng of input mass.

Figure 3. Utilization of ddPCR in Active Fate Therapeutics Clinical Trials

A

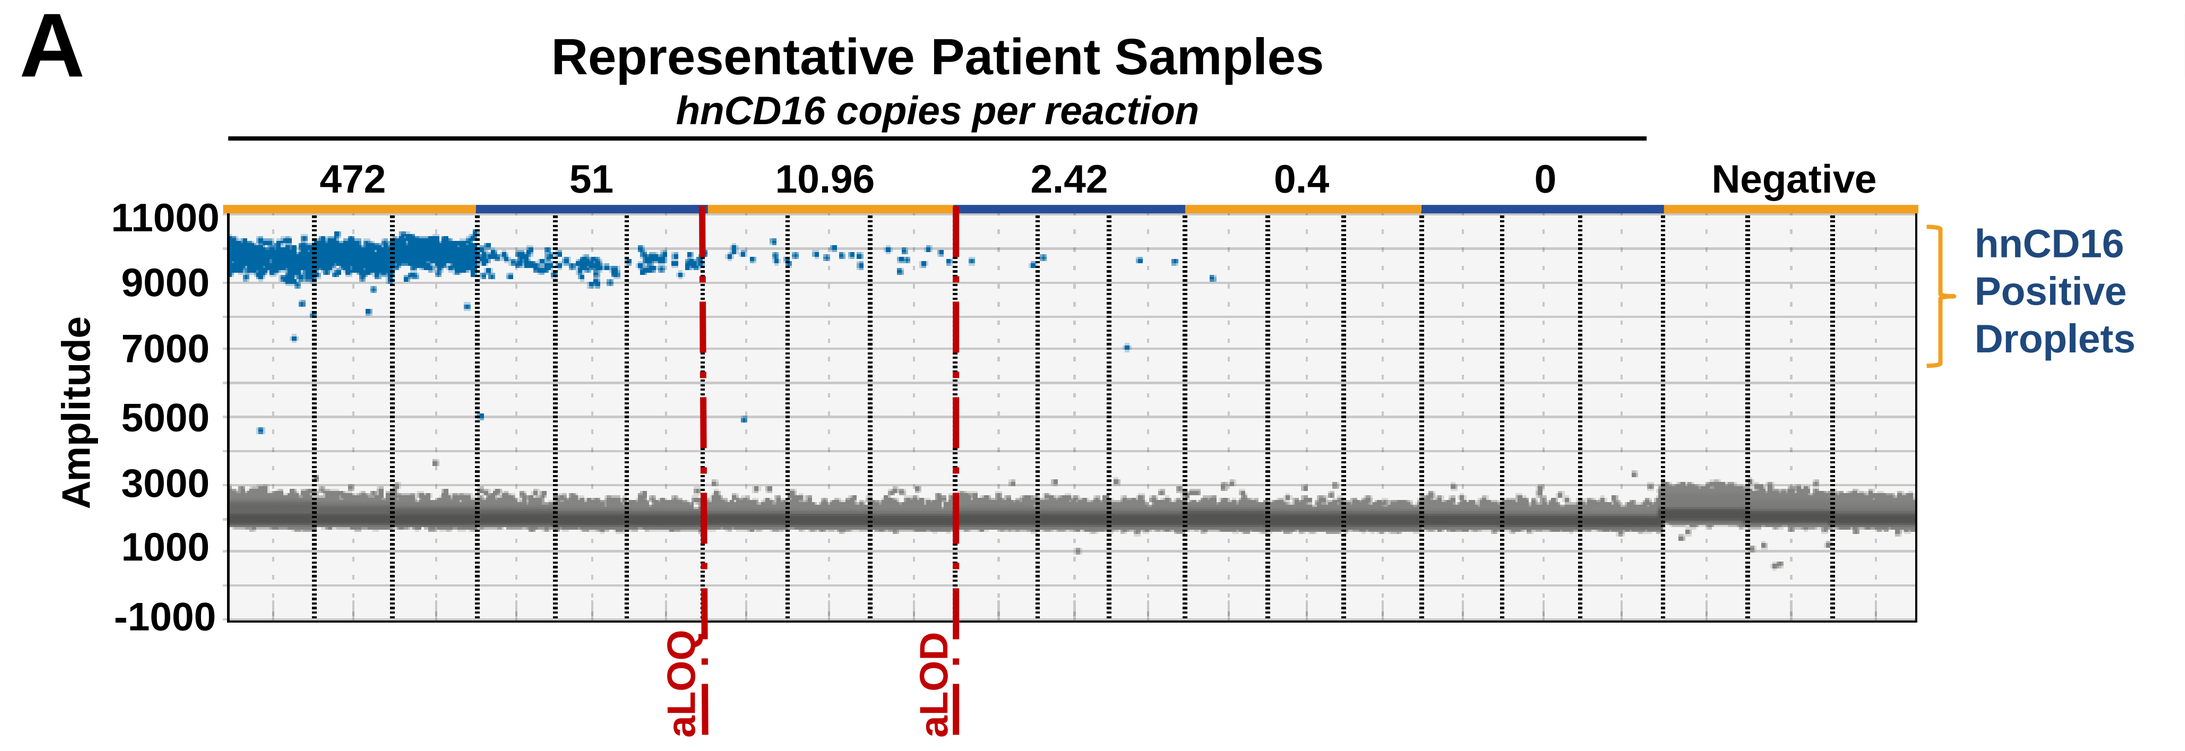
Representative Patient Samples
hnCD16 copies per reaction
472
51
10.96
2.42
0.4
0
Negative
Amplitude
hnCD16 Positive Droplets
aLOQ
aLOD

B

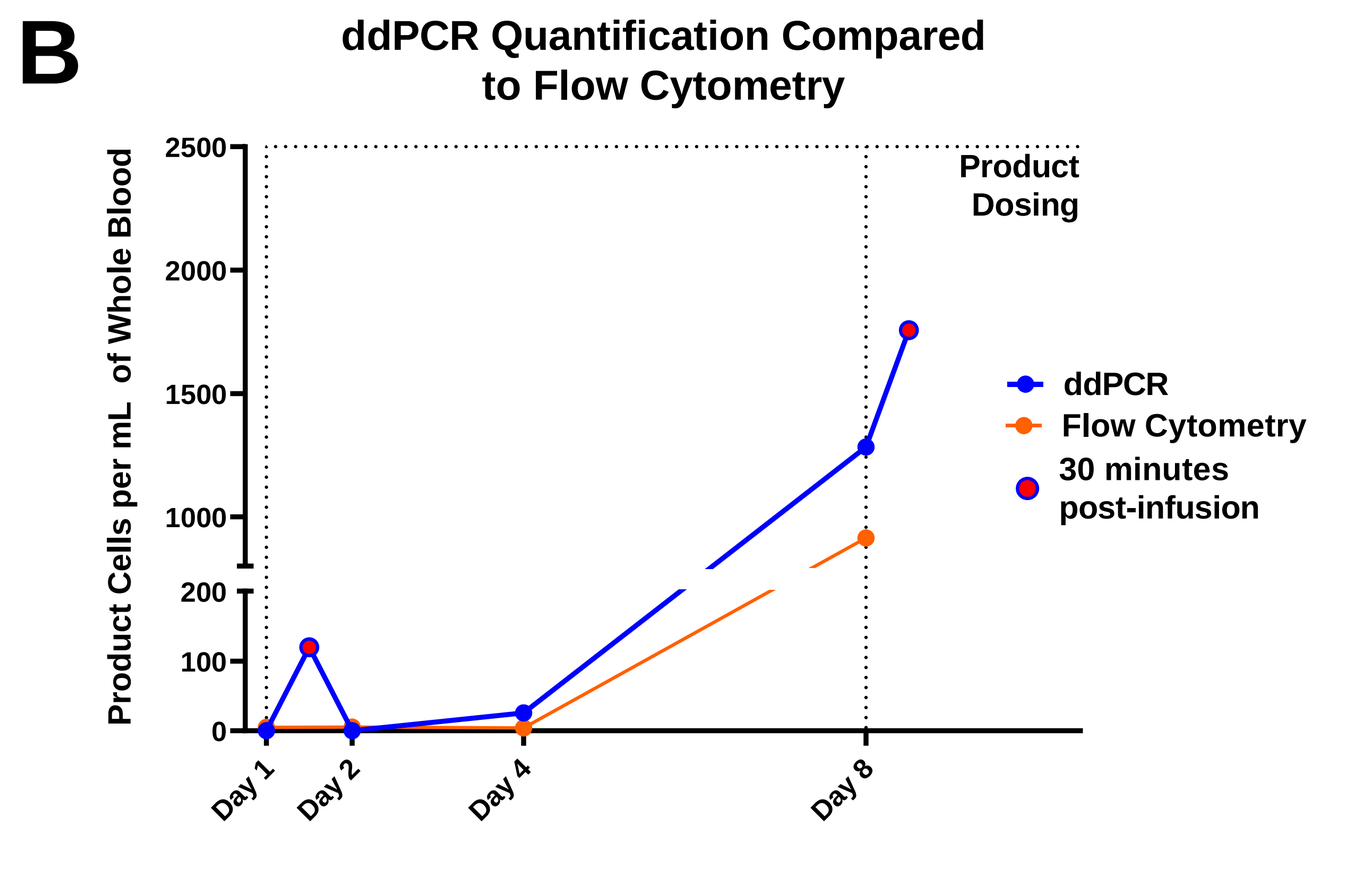
ddPCR Quantification Compared to Flow Cytometry
Product Cells per mL of Whole Blood
Product Dosing
ddPCR
Flow Cytometry
30 minutes post-infusion
Day 1
Day 2
Day 4
Day 8

Figure 3. (A) 1D visual representation of hnCD16 positive droplets derived from patient peripheral blood mononuclear cell (PBMC) samples with a range of product cell detection showing real world sensitivity of the assay. Red dashed lines represent general location of the aLOD and aLOQ as determined in Figure 2. (B) Quantification of FT538 product cells from peripheral blood samples is concordant when analyzed in parallel by ddPCR and flow cytometry. The ddPCR output (hnCD16 transgene copies/mg genomic DNA) was converted to estimate the number of FT538 cells/mL whole blood. In parallel, HLA antibodies that differentiate patient vs. product cells were used to identify and quantify the number of FT538 cells/mL whole blood by flow cytometry. Plotted are the pharmacokinetics of FT538 (cells/mL whole blood) in the peripheral blood by ddPCR or by flow cytometry days 1-8 of cycle 2 in a patient with AML treated with FT538 monotherapy (NCT04614636).

Figure 4. hnCD16 Probe Does Not Detect Endogenous NKs in Pre-Clinical Validation

A

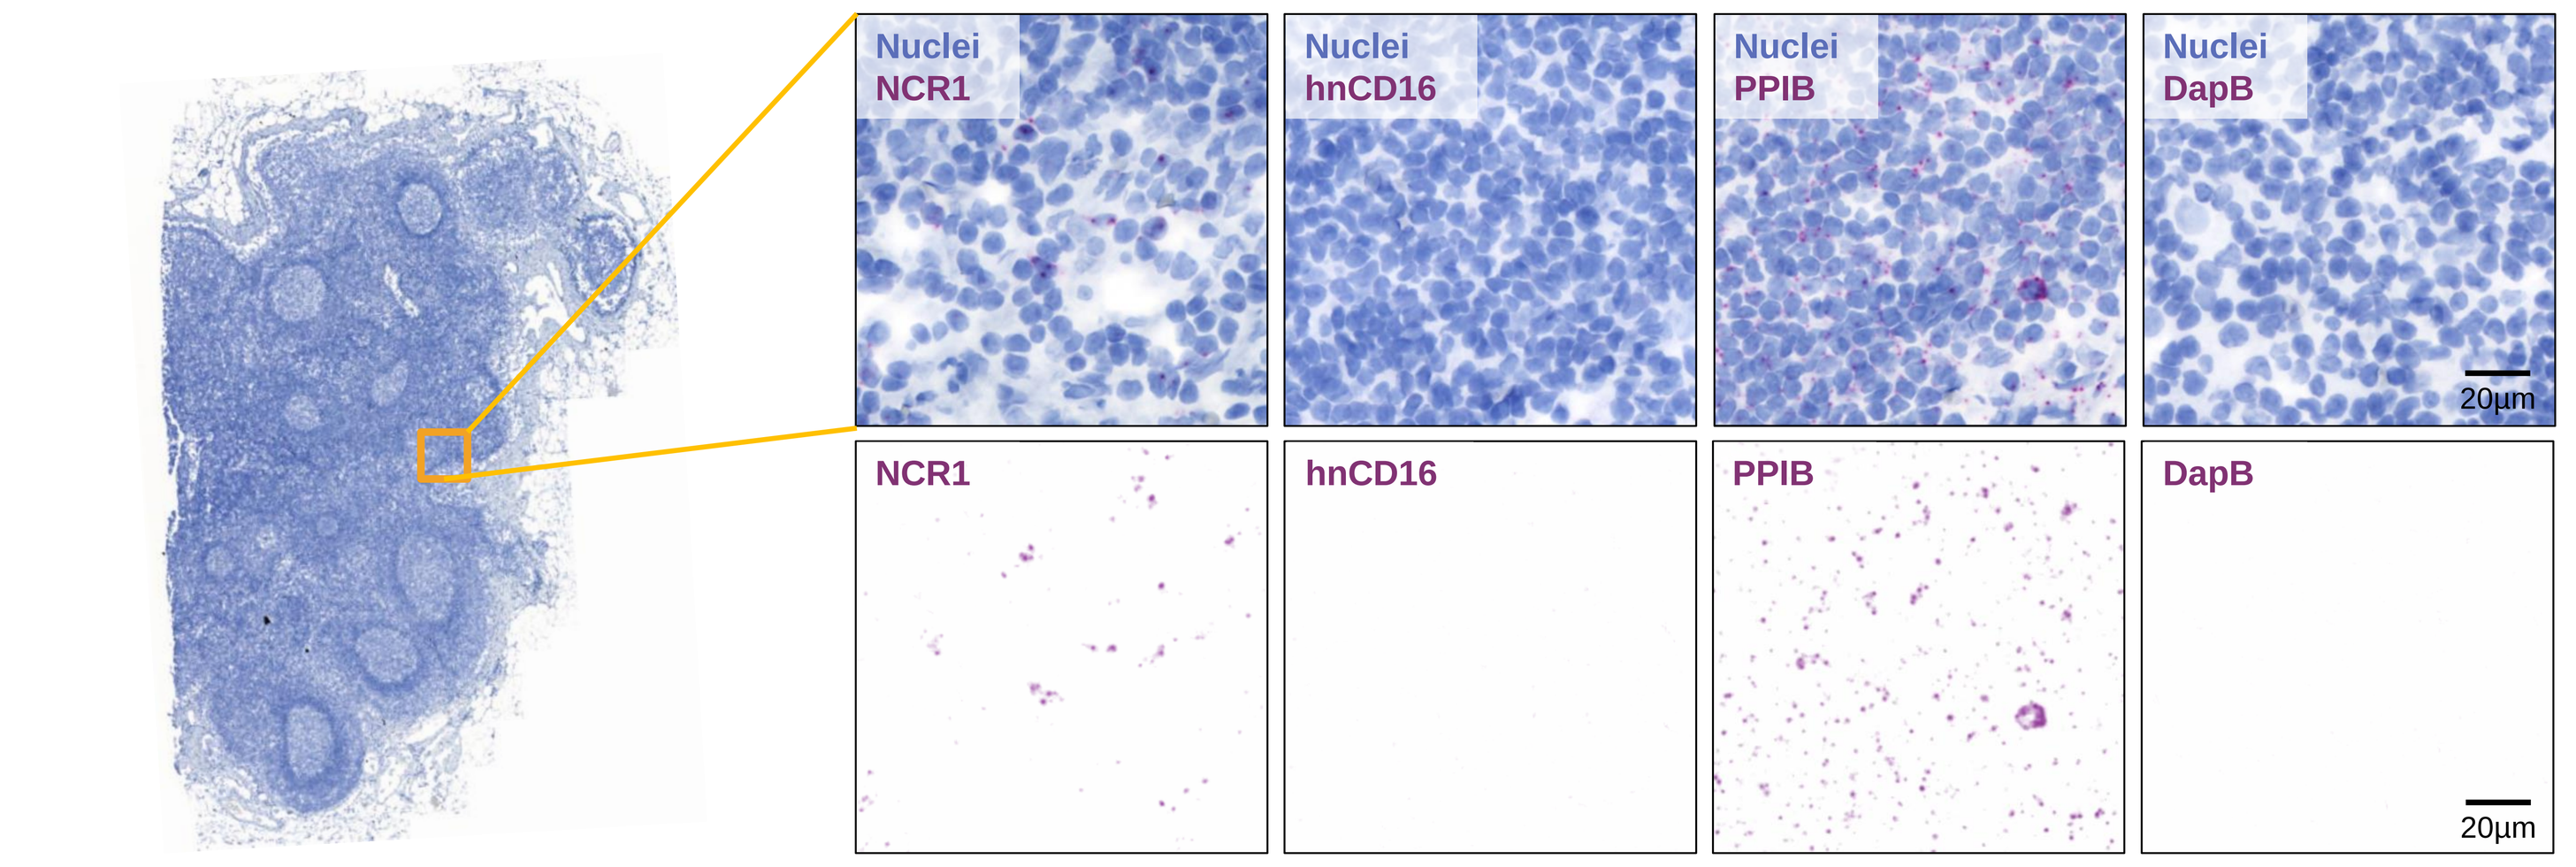
Nuclei NCR1
Nuclei hnCD16
Nuclei PPIB
Nuclei DapB
20µm
NCR1
hnCD16
PPIB
DapB
20µm

Figure 4. (A) FFPE samples of untreated, commercially-available human lymph nodes were used as controls to evaluate RNAscope™ probes targeting the endogenous NK cell marker NCR1, iNK specific hnCD16 transgene, positive control PPIB, and negative control bacterial DapB. Robust staining with PPIB indicates a successful run with a clean DapB negative result. Positive staining for NCR1 shows that the lymph node is infiltrated with endogenous NK cells, but native CD16 is not detected by the hnCD16-specific assay. The bottom row shows red target signal digitally separated from hematoxylin nuclear counterstain. Representative regions of interest were collected from the same tissue area across serial slides as indicated by orange box.

Figure 5. hnCD16 ISH Probe Reproduces huNKG2A IHC Staining of iNKs in NSG Mice

A

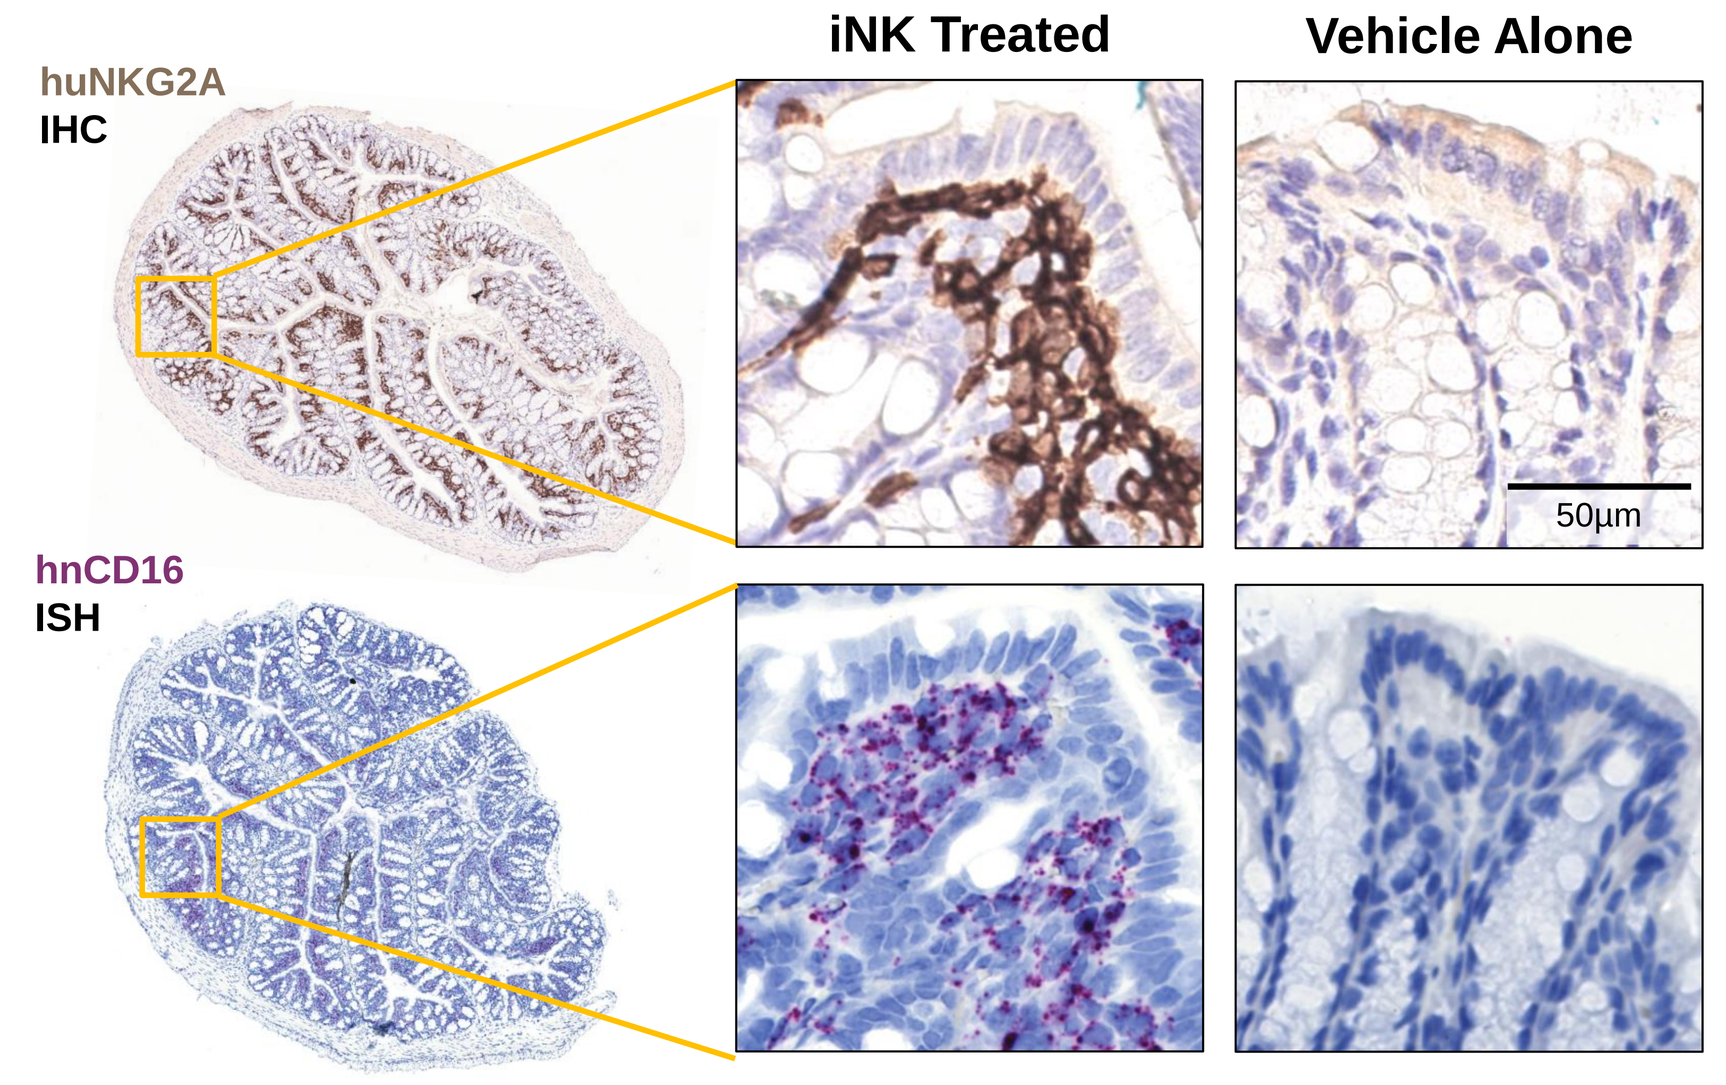
iNK Treated
Vehicle Alone
huNKG2A
IHC
hnCD16
ISH
50µm

Figure 5. FFPE tissue derived from NSG mice treated with hnCD16 positive iNK cells was stained in parallel with human NKG2A antibody (huNKG2A) via chromogenic IHC and hnCD16 RNAscope™ ISH probes. (A) Staining in non-adjacent serial colon sections show strong positive signal in homologous tissue regions with clear morphological similarity indicating qualitative concordance across assays and specificity of the hnCD16 probe in the detection of iNK product cells.

Figure 6. Pre-Clinical Validation Displays hnCD16 ISH Probes Can Be Used for Semi-Quantitative Applications

A

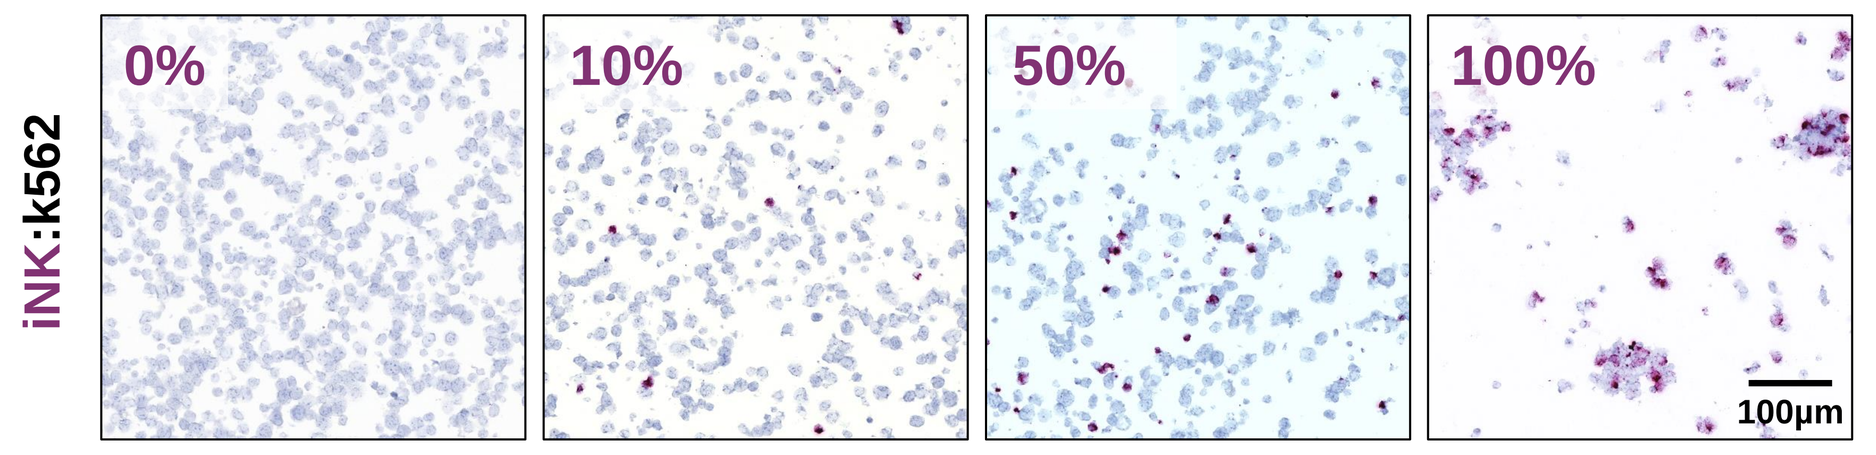
iNK:k562
0%
10%
50%
100%
100µm

B

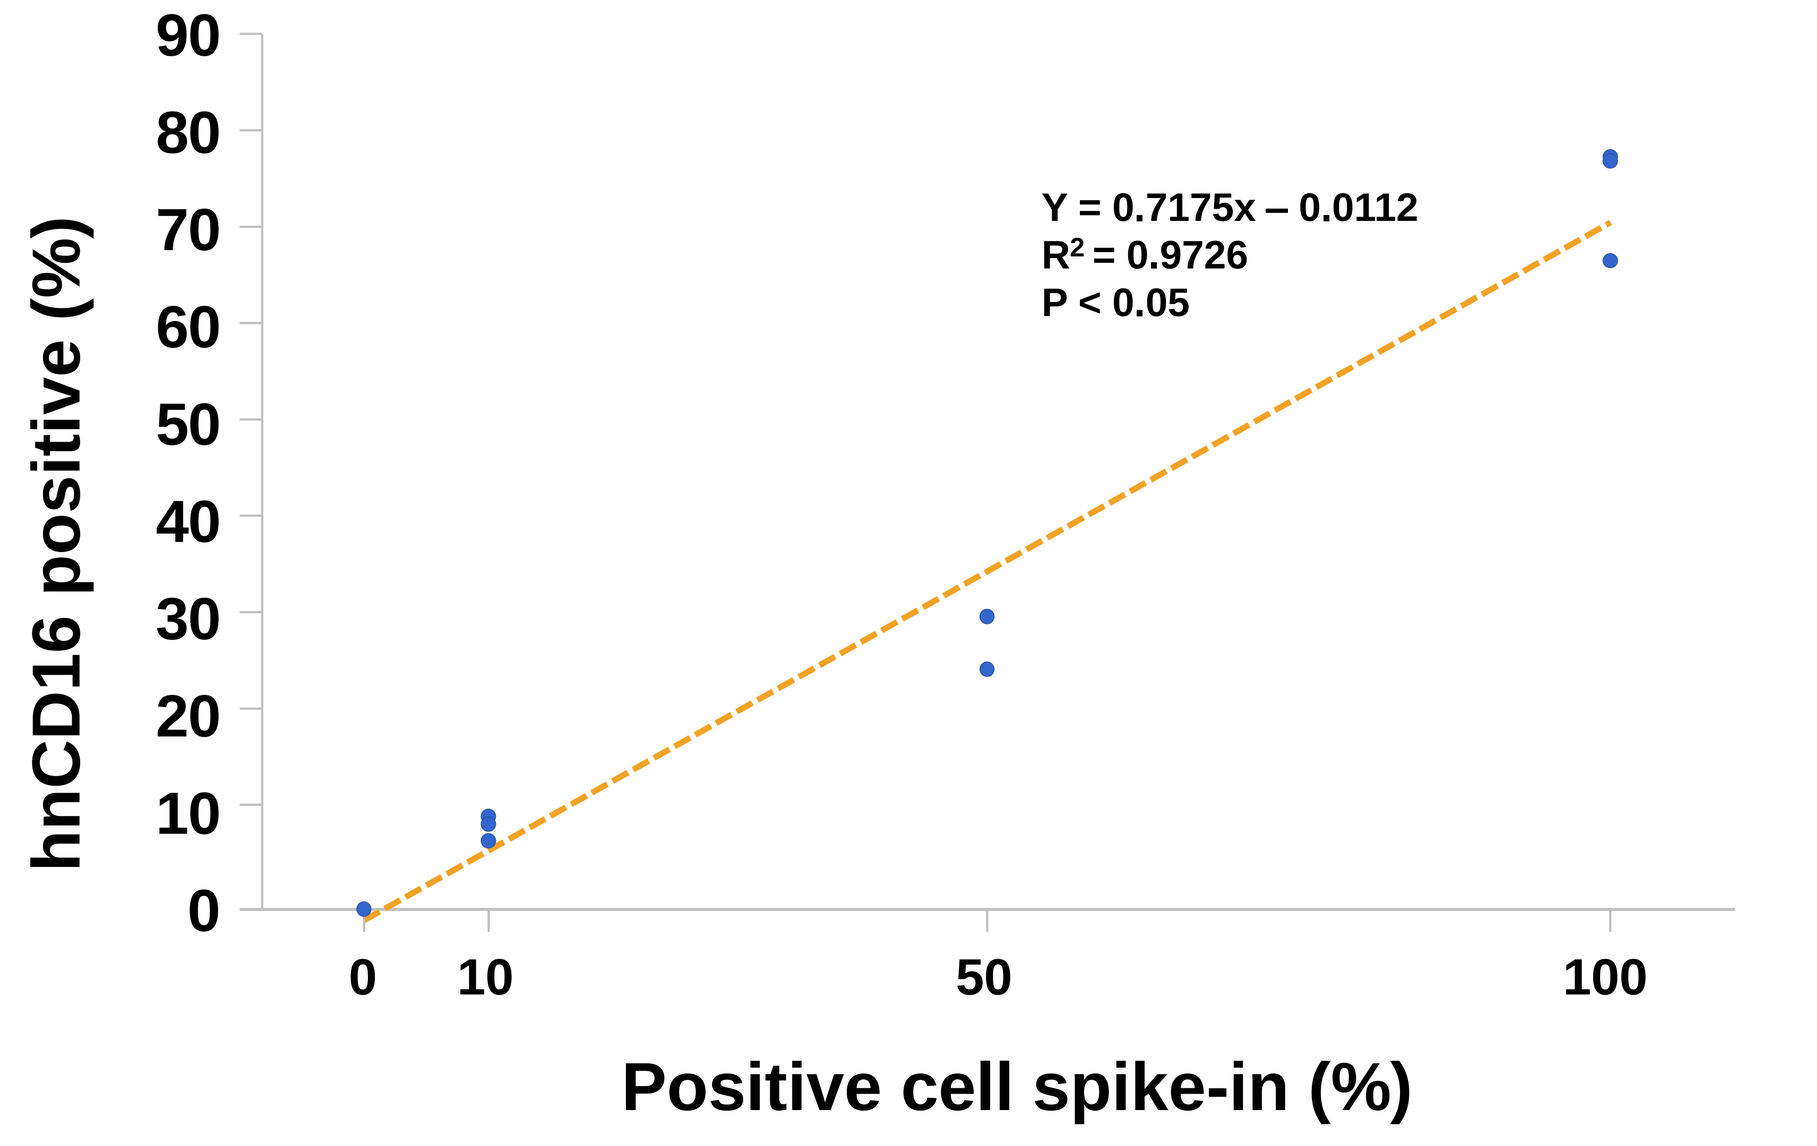
Y = 0.7175x – 0.0112
R2 = 0.9726
P < 0.05
hnCD16 positive (%)
Positive cell spike-in (%)

C

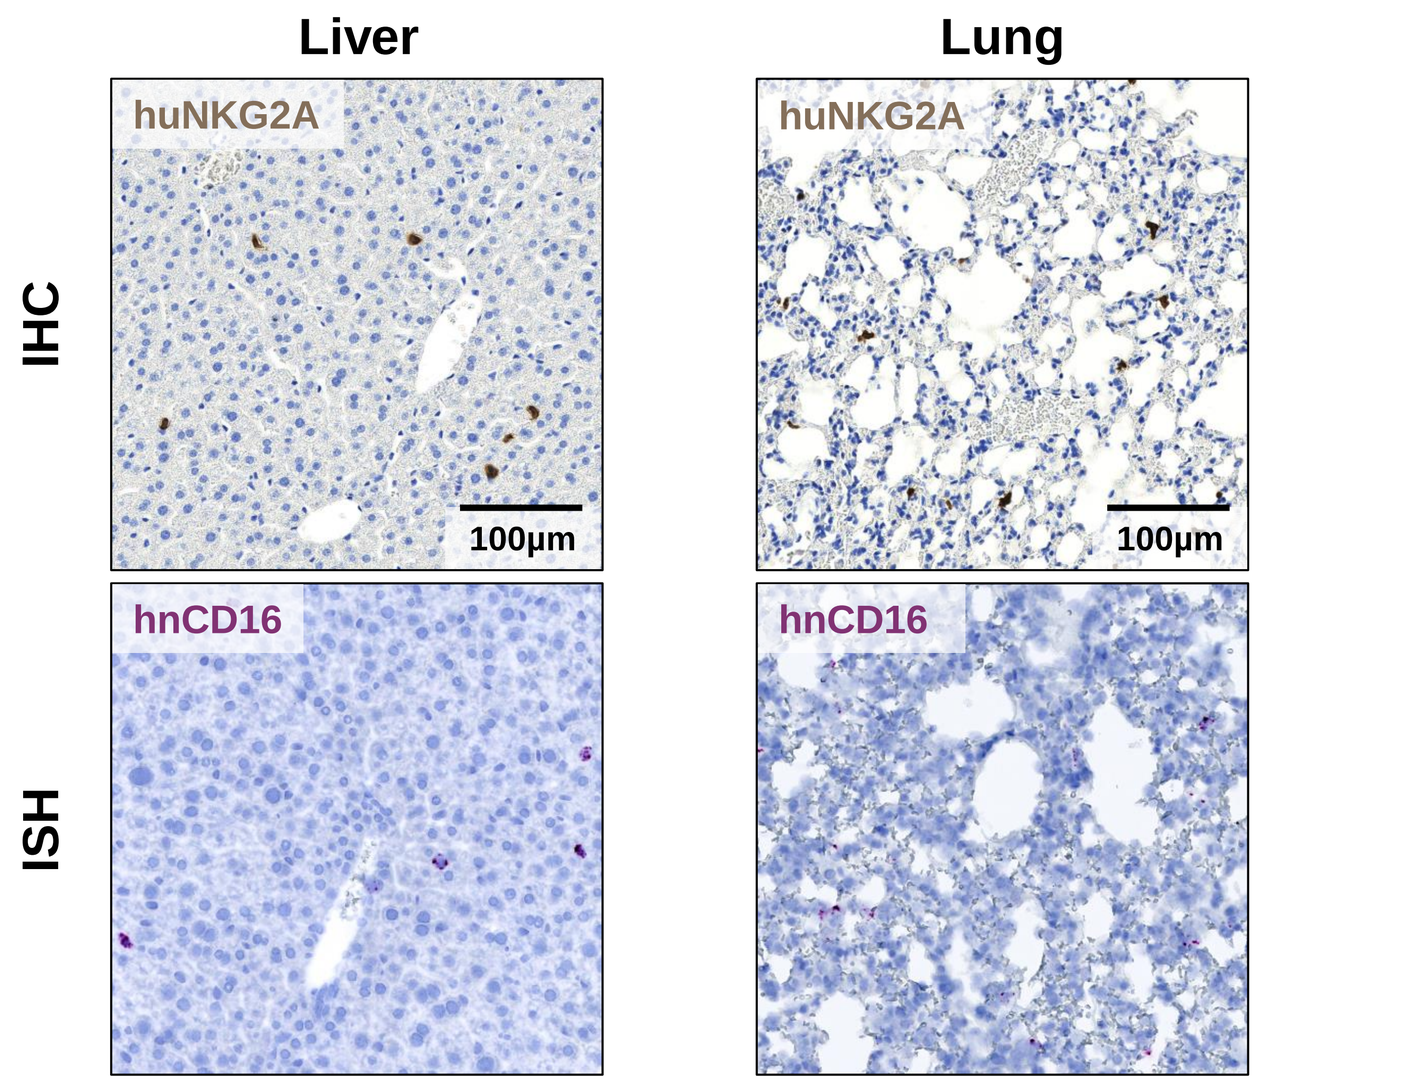
Liver
Lung
IHC
ISH
huNKG2A
hnCD16
100µm

D

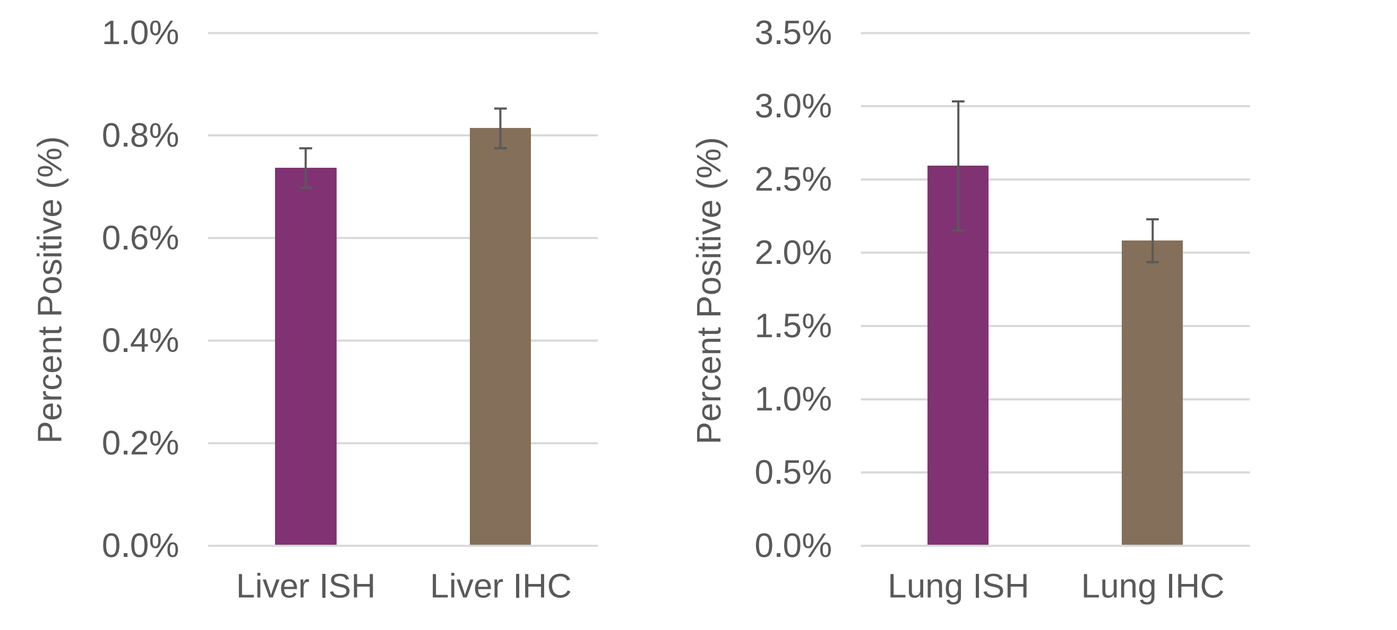
Percent Positive (%)
Liver ISH
Liver IHC
Lung ISH
Lung IHC

Figure 6. (A) Cell pellets were created with known spike-in percentages of hnCD16 positive iNK cells in a K562 tumor cell background. Pellets were embedded and fixed for sectioning similar to FFPE tissue. (B) The percentage of hnCD16 positive cells was determined across three regions of intererest per condition using a counting algorithm generated in ImageJ (algorithm was verified via statistical comparison with manual counts as completed by three operators, data not shown). Quantitative comparison of the percent positive cell spike-in to the percent positive cells observed shows a dose-dependent linear association with a p-value of $p < 0.05$. (C) Quantification of ISH was also completed in murine liver and lung tissue from mice treated with a hnCD16 positive iNK. This quantification was compared to IHC staining of huNKG2A in the same tissue and shows equivalent quantitative readout (D).