10-year Stability Assessment of Cryopreserved, Engineered iPSC Banks: Genetic and Phenotypic Characterization

Nicholas Brookhouser, Lauren K. Fong, Aimee R. Pankonin, Brent Wilkinson, Michael V. Pollante, Megan Robinson, Yi-Shin Lai, Janel Huffman, Jerome Bressi, Tom T. Lee, Ramzey Abujarour, Bahram Valamehr*

Fate Therapeutics Inc, San Diego, CA, USA; *bob.valamehr@fatetherapeutics.com

GRAPHICAL ABSTRACT & INTRODUCTION

Changing the Game in Cell Therapy

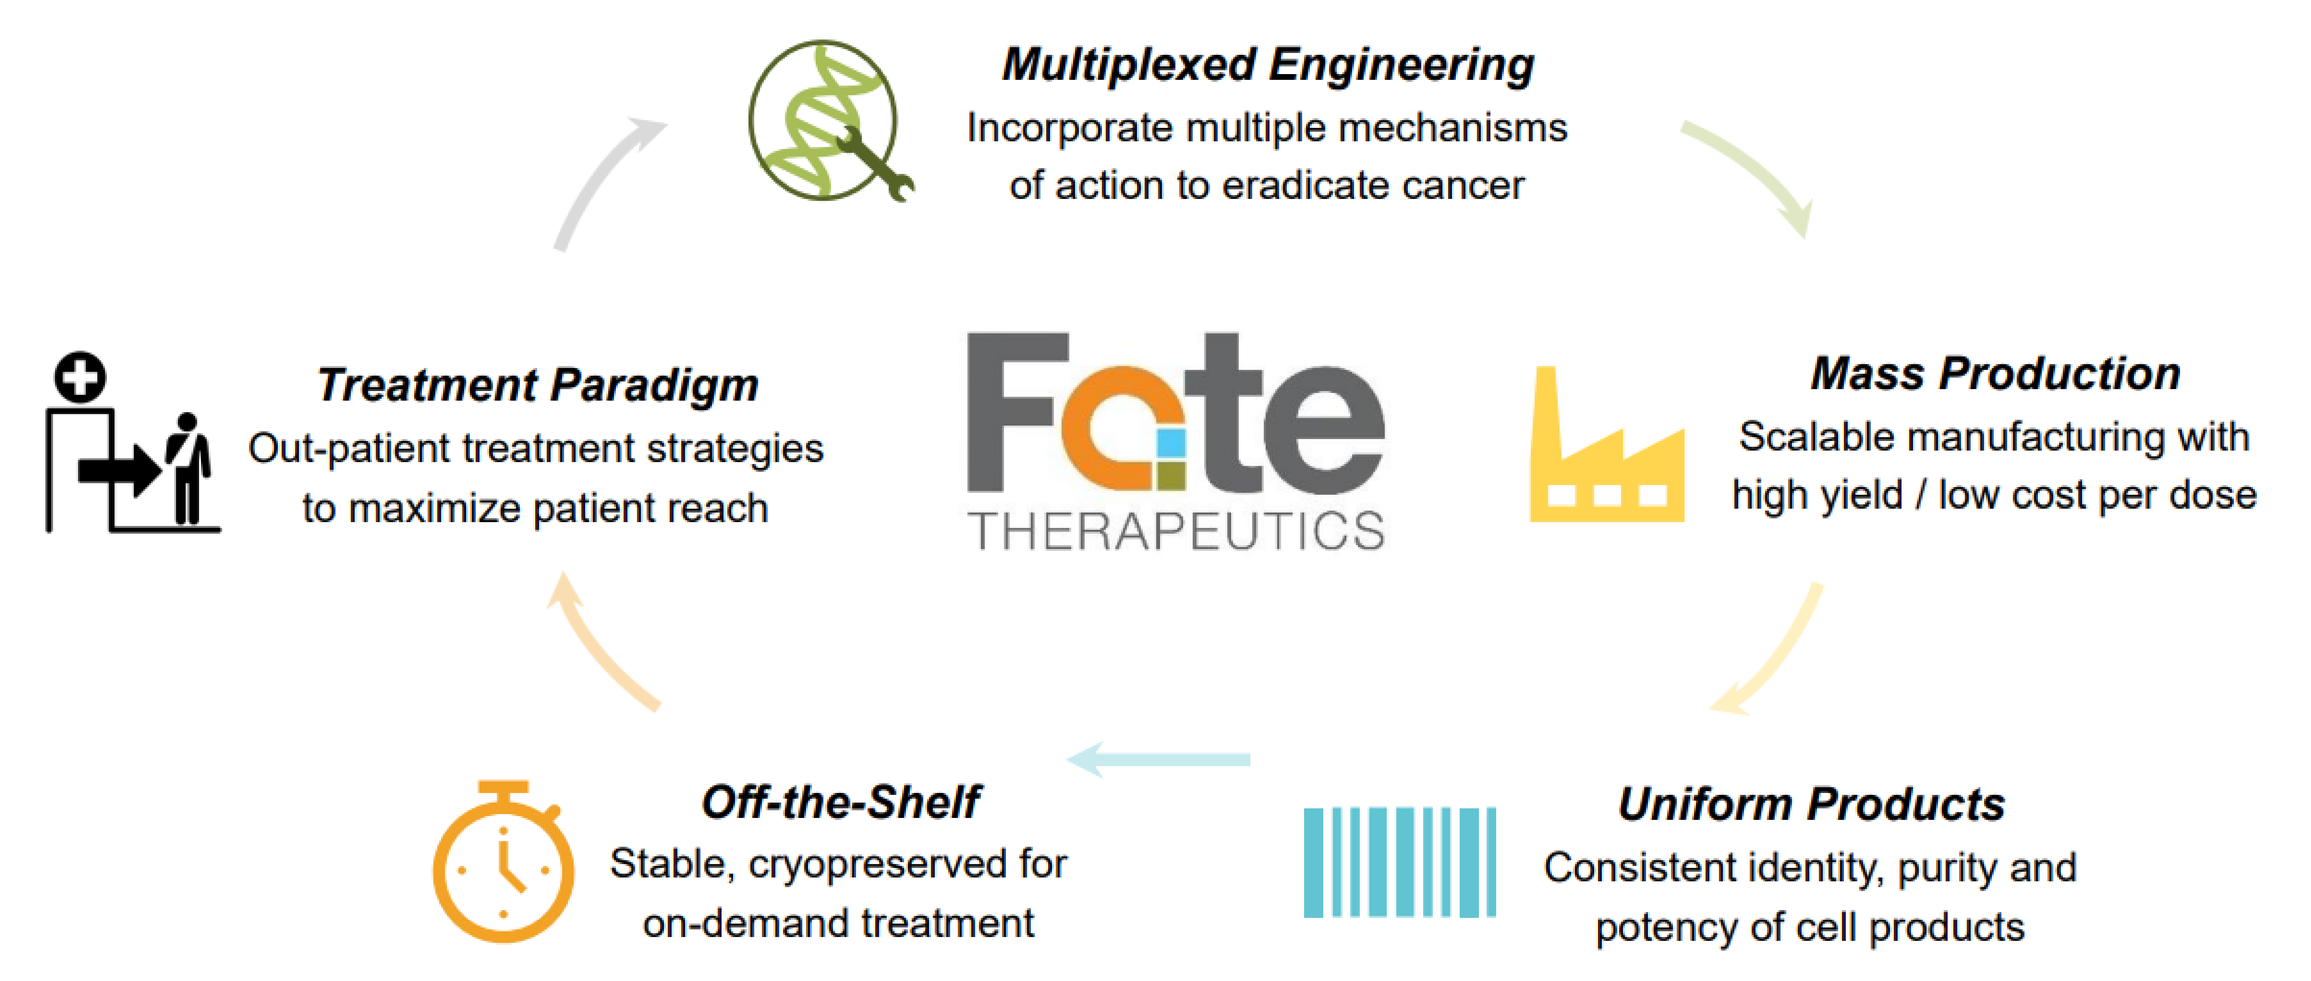
Multiplexed Engineering
Incorporate multiple mechanisms of action to eradicate cancer
Mass Production
Scalable manufacturing with high yield / low cost per dose
Uniform Products
Consistent identity, purity and potency of cell products
Off-the-Shelf
Stable, cryopreserved for on-demand treatment
Treatment Paradigm
Out-patient treatment strategies to maximize patient reach
Fate THERAPEUTICS

Mass Production of Off-the-shelf, Cell-based Cancer Immunotherapies

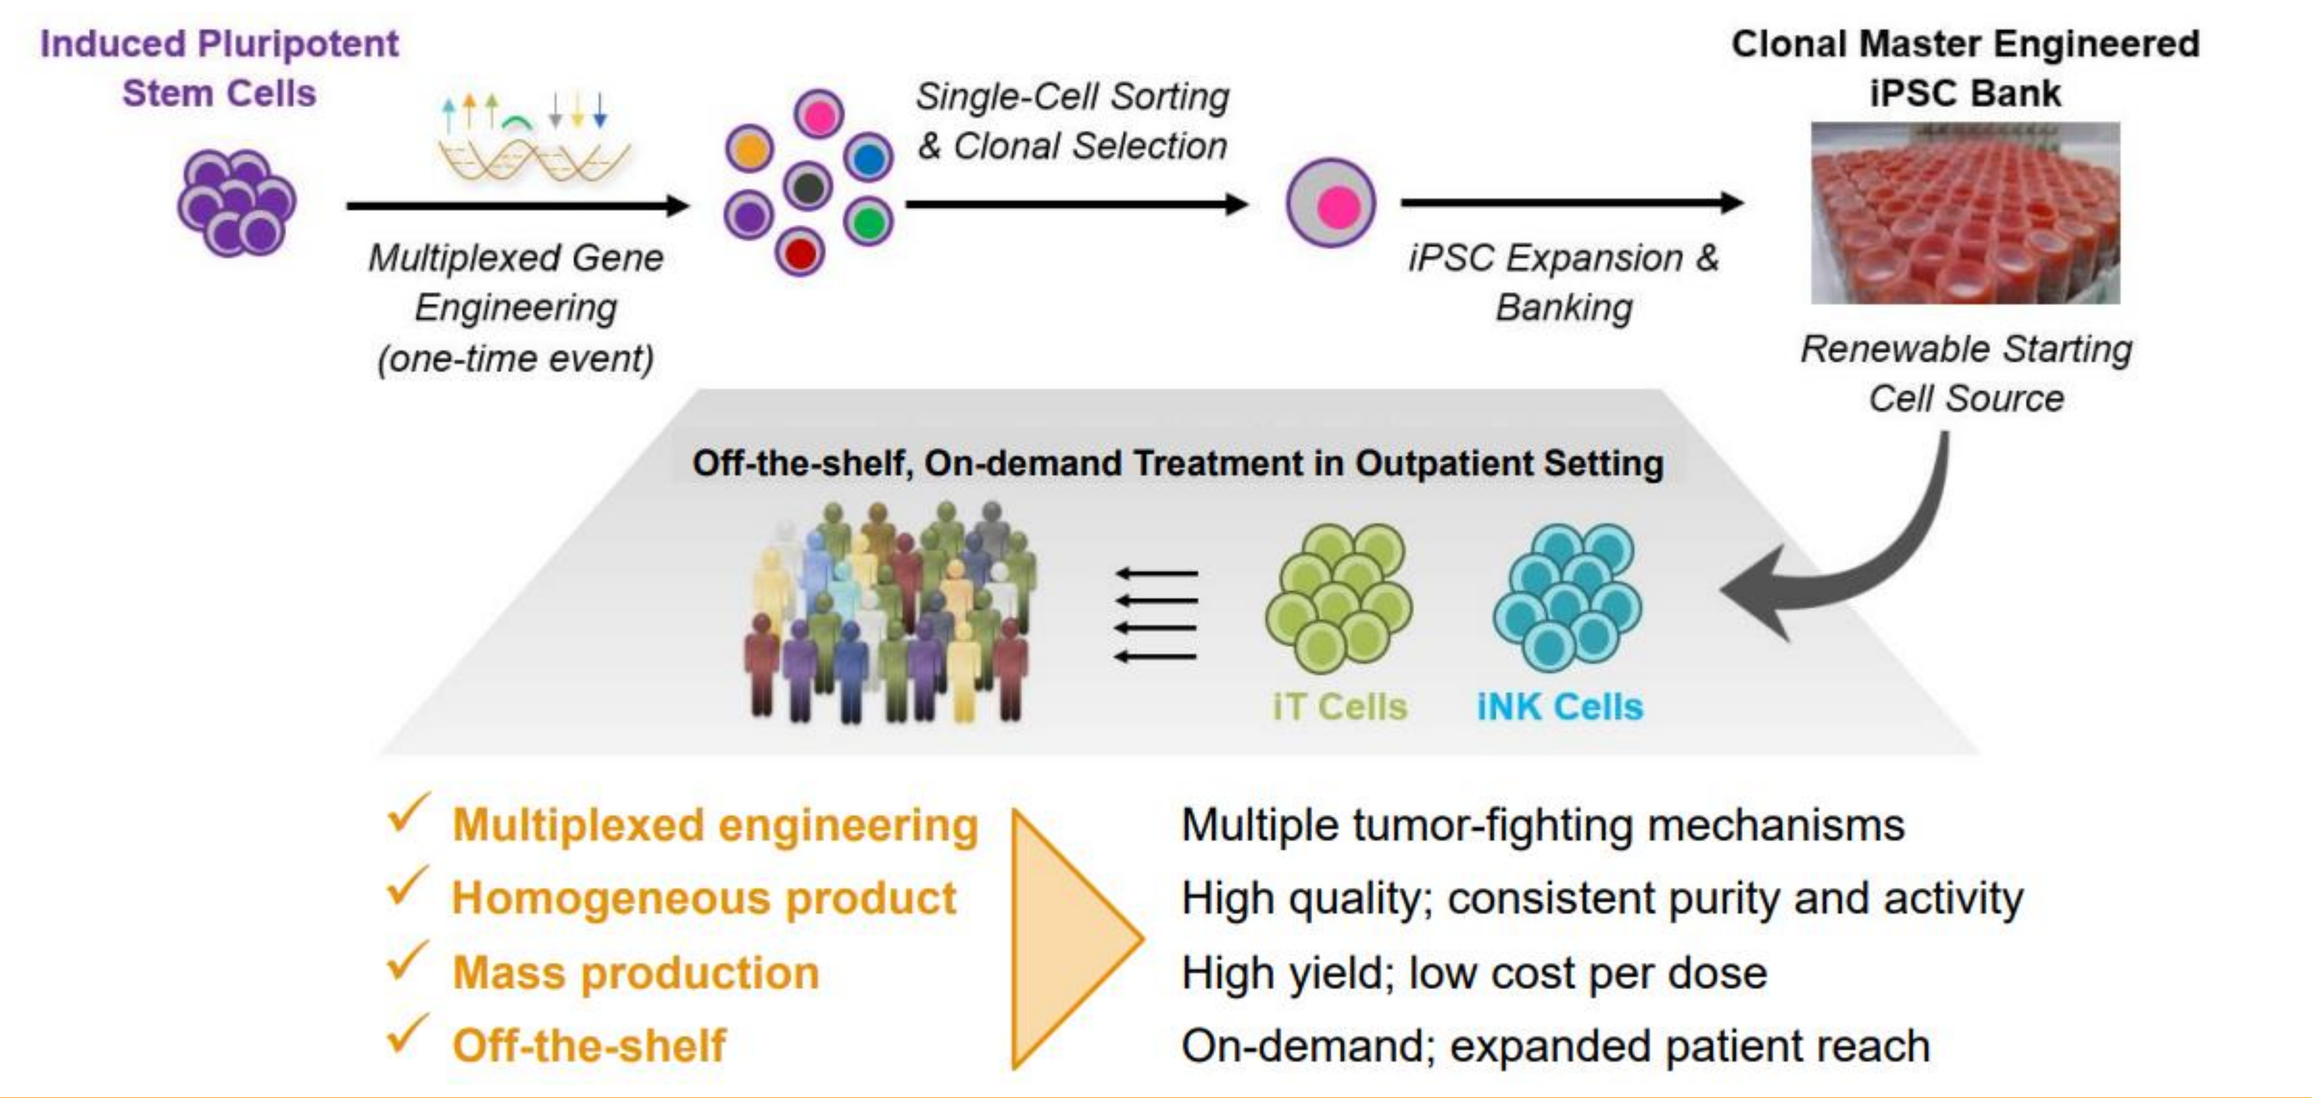
Induced Pluripotent Stem Cells
Multiplexed Gene Engineering (one-time event)
Single-Cell Sorting & Clonal Selection
iPSC Expansion & Banking
Clonal Master Engineered iPSC Bank
Renewable Starting Cell Source
Off-the-shelf, On-demand Treatment in Outpatient Setting
iT Cells
iNK Cells
✓ Multiplexed engineering
✓ Homogeneous product
✓ Mass production
✓ Off-the-shelf
Multiple tumor-fighting mechanisms
High quality; consistent purity and activity
High yield; low cost per dose
On-demand; expanded patient reach

Maintaining stability, quality, self-renewal and differentiation potential of engineered clonal iPSC master cell lines over-extended cryo-storage make them ideal and reliable starting material for mass manufacture of consistent and homogenous NK and T cell drug products

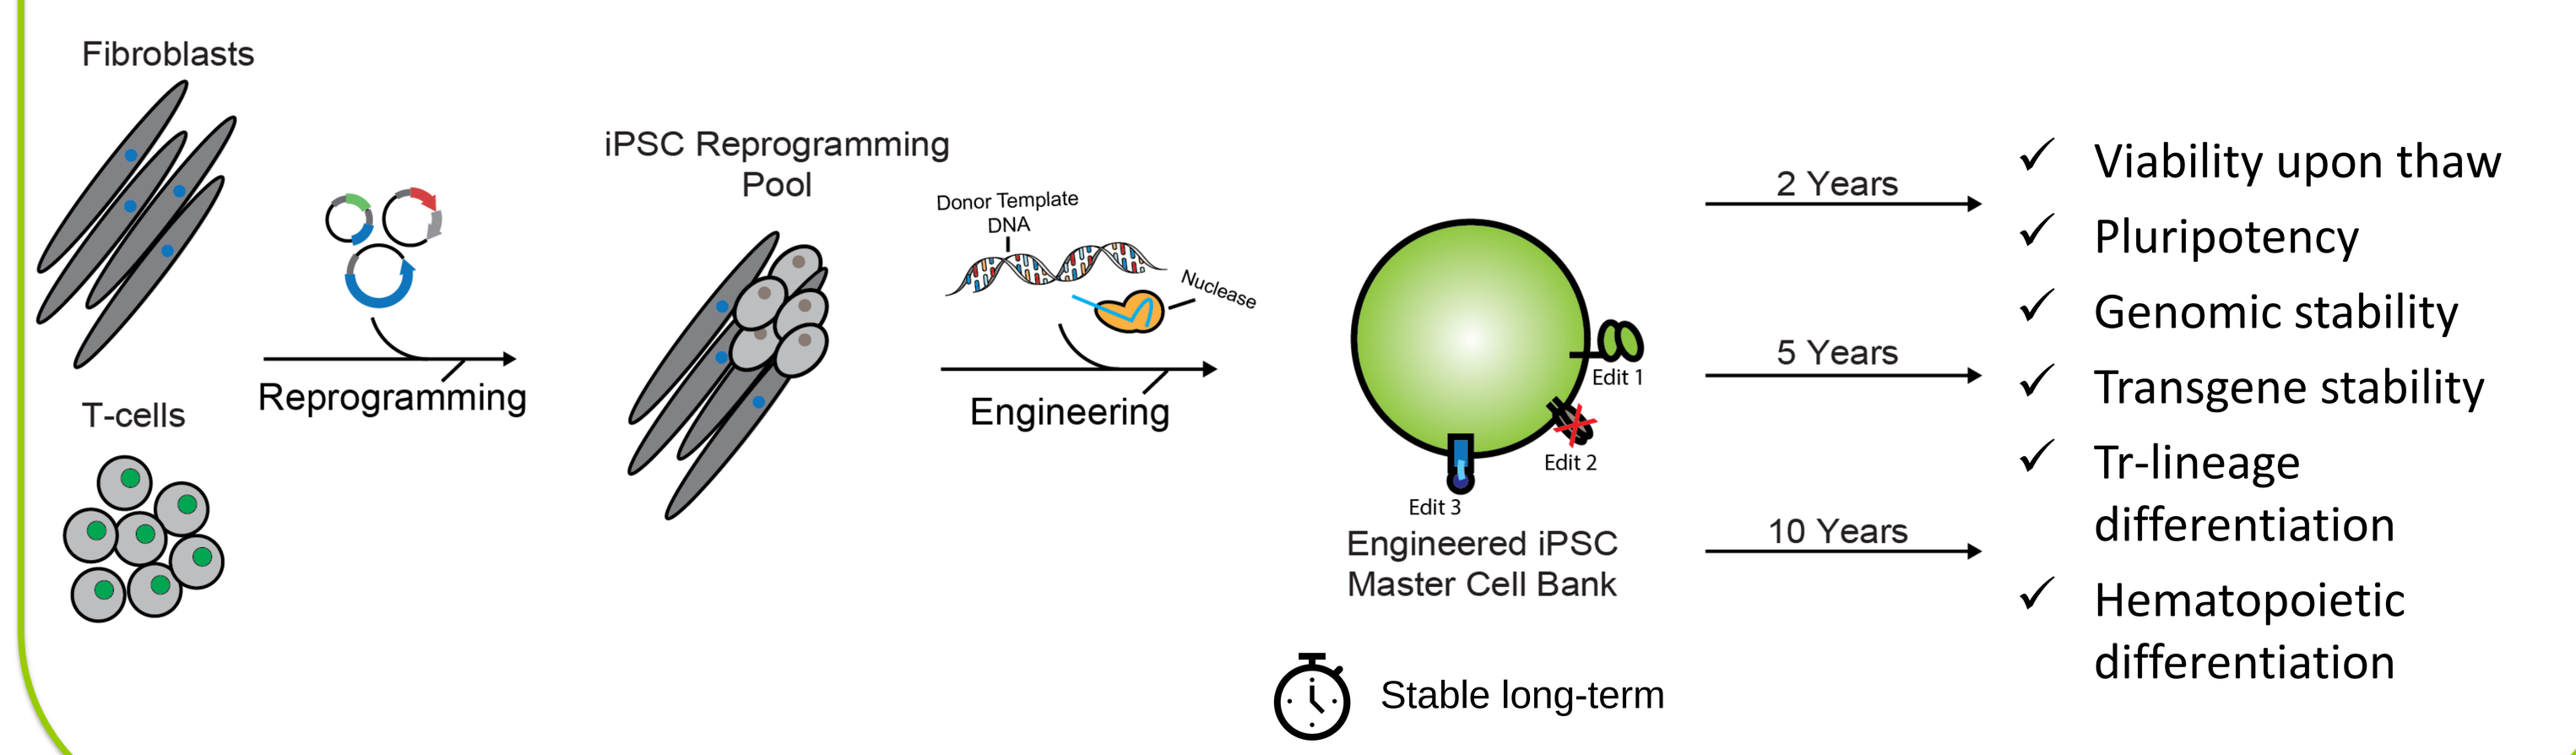
Fibroblasts
T-cells
Reprogramming
iPSC Reprogramming Pool
Donor Template DNA
Nuclease
Engineering
Edit 1
Edit 2
Edit 3
Engineered iPSC Master Cell Bank
Stable long-term
2 Years
5 Years
10 Years
✓ Viability upon thaw
✓ Pluripotency
✓ Genomic stability
✓ Transgene stability
✓ Tr-lineage differentiation
✓ Hematopoietic differentiation

Engineered iPSC master cell banks overcome issues inherent with engineering a bulk population including:

- ✓ Variable editing efficiencies
- ✓ Imprecise on-target edits
- ✓ Off-target mutations
- ✓ Copy number variability
- ✓ Random transgene insertions
- ✓ Heterogeneous edited population
- ✓ Genomic instability
- ✓ Need for repeat engineering

Objective

This study aims to evaluate long-term stability of iPSC banks following long-term cryo-storage (<-150˚C) over 10 years by assessing critical quality attributes including viability, recovery, purity, maintenance of pluripotency, genomic stability, and potency.

SUMMARY

- iPSCs derived using Fate's proprietary reprogramming and engineering methods are stable in cryo-storage over the period of the study (>10 years)
- Critical quality attributes such as viability, recovery, purity, genomic stability and potency are maintained over long-term cryo-storage
- Long-term stability of clonal iPSC banks is critical to maintain high quality, reliable and renewable starting material for the manufacture of highly consistent and homogenous off-the-shelf cellular drug products

RESULTS

Cryopreserved iPSC Banks Maintain Viability and Stable Pluripotent Phenotype Over 10 Years

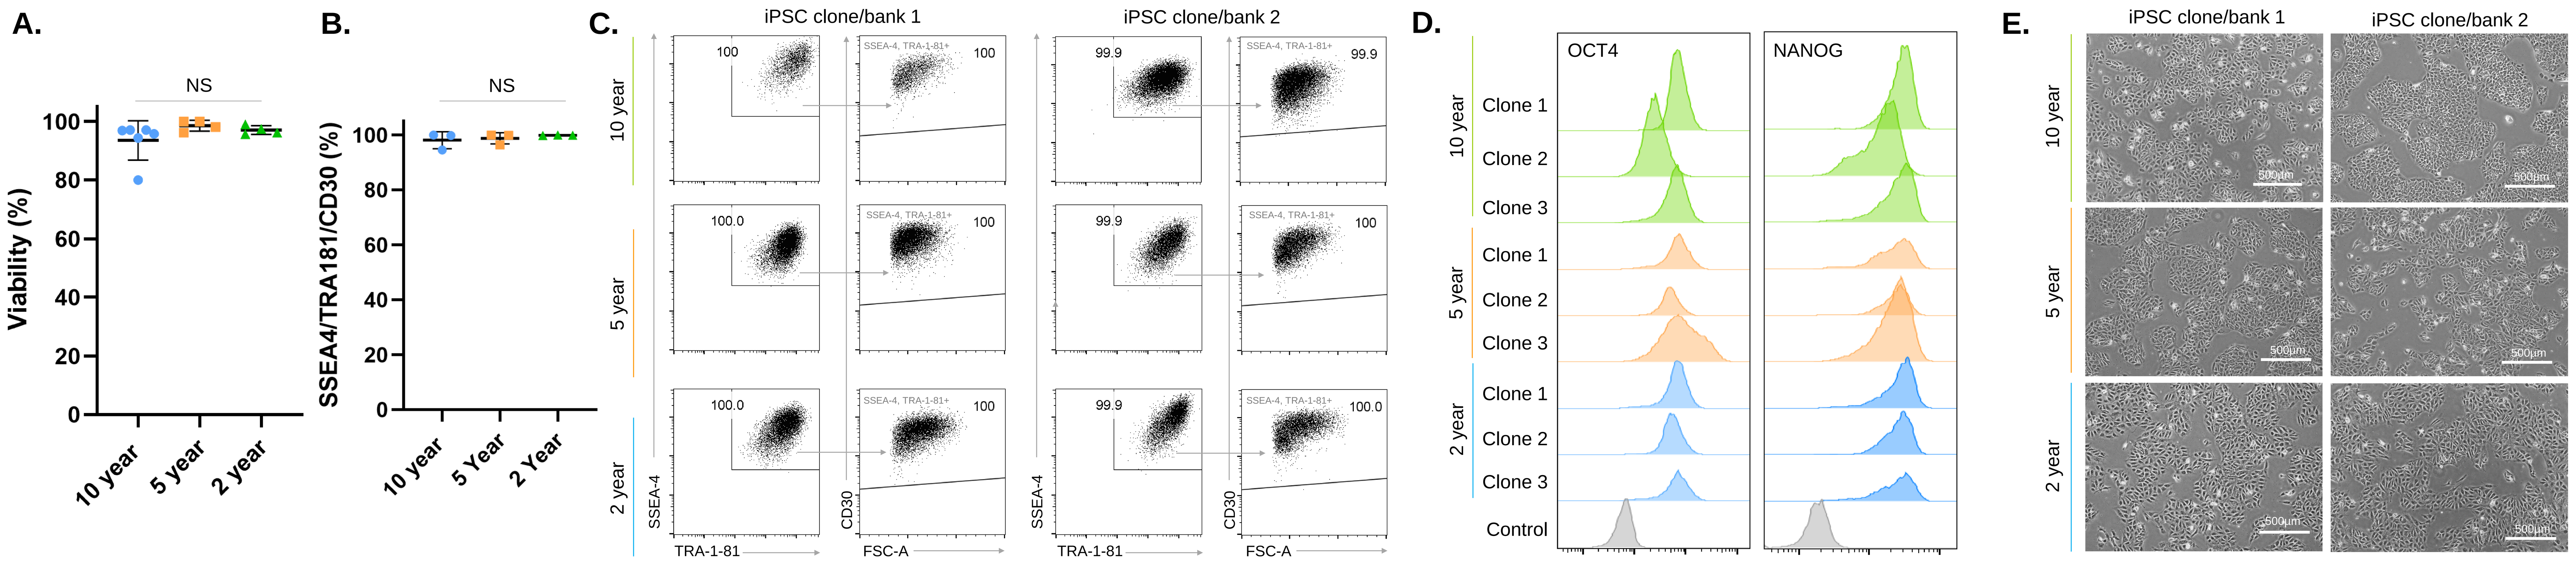
A.
B.
C.
D.
E.
NS
Viability (%)
SSEA4/TRA181/CD30 (%)
10 year
5 year
2 year
iPSC clone/bank 1
iPSC clone/bank 2
SSEA-4
TRA-1-81
CD30
FSC-A
OCT4
NANOG
Clone 1
Clone 2
Clone 3
Control

Figure 1. Analysis of iPSC viability and pluripotency expression after cryopreservation. (A) Cell viability in iPSC banks cryopreserved for 10, 5, or 2 years assessed immediately post-thaw. No significant difference in viability was seen between groups determined by one-way ANOVA analysis and Tukey multiple comparisons test. **(B)** Flow cytometry analysis of iPSC cultures revealed robust expression of pluripotency markers SSEA-4, TRA-1-81, and CD30. No significant differences in expression of pluripotency markers was seen between groups determined by one-way ANOVA and Tukey multiple comparisons test. **(C)** Representative post-thaw flow cytometry for iPSC banks cryopreserved for 10, 5, or 2 years. **(D)** All iPSC banks showed robust staining for the intracellular pluripotency regulators, OCT4 and NANOG, indicative of high-quality iPSCs. **(E)** Brightfield images of long-term cryopreserved iPSC post-thaw. iPSCs maintain pluripotent morphology after extended cryo-storage.

Pluripotency Gene Expression and Genomic Stability Maintained Over Long-term Cryo-storage

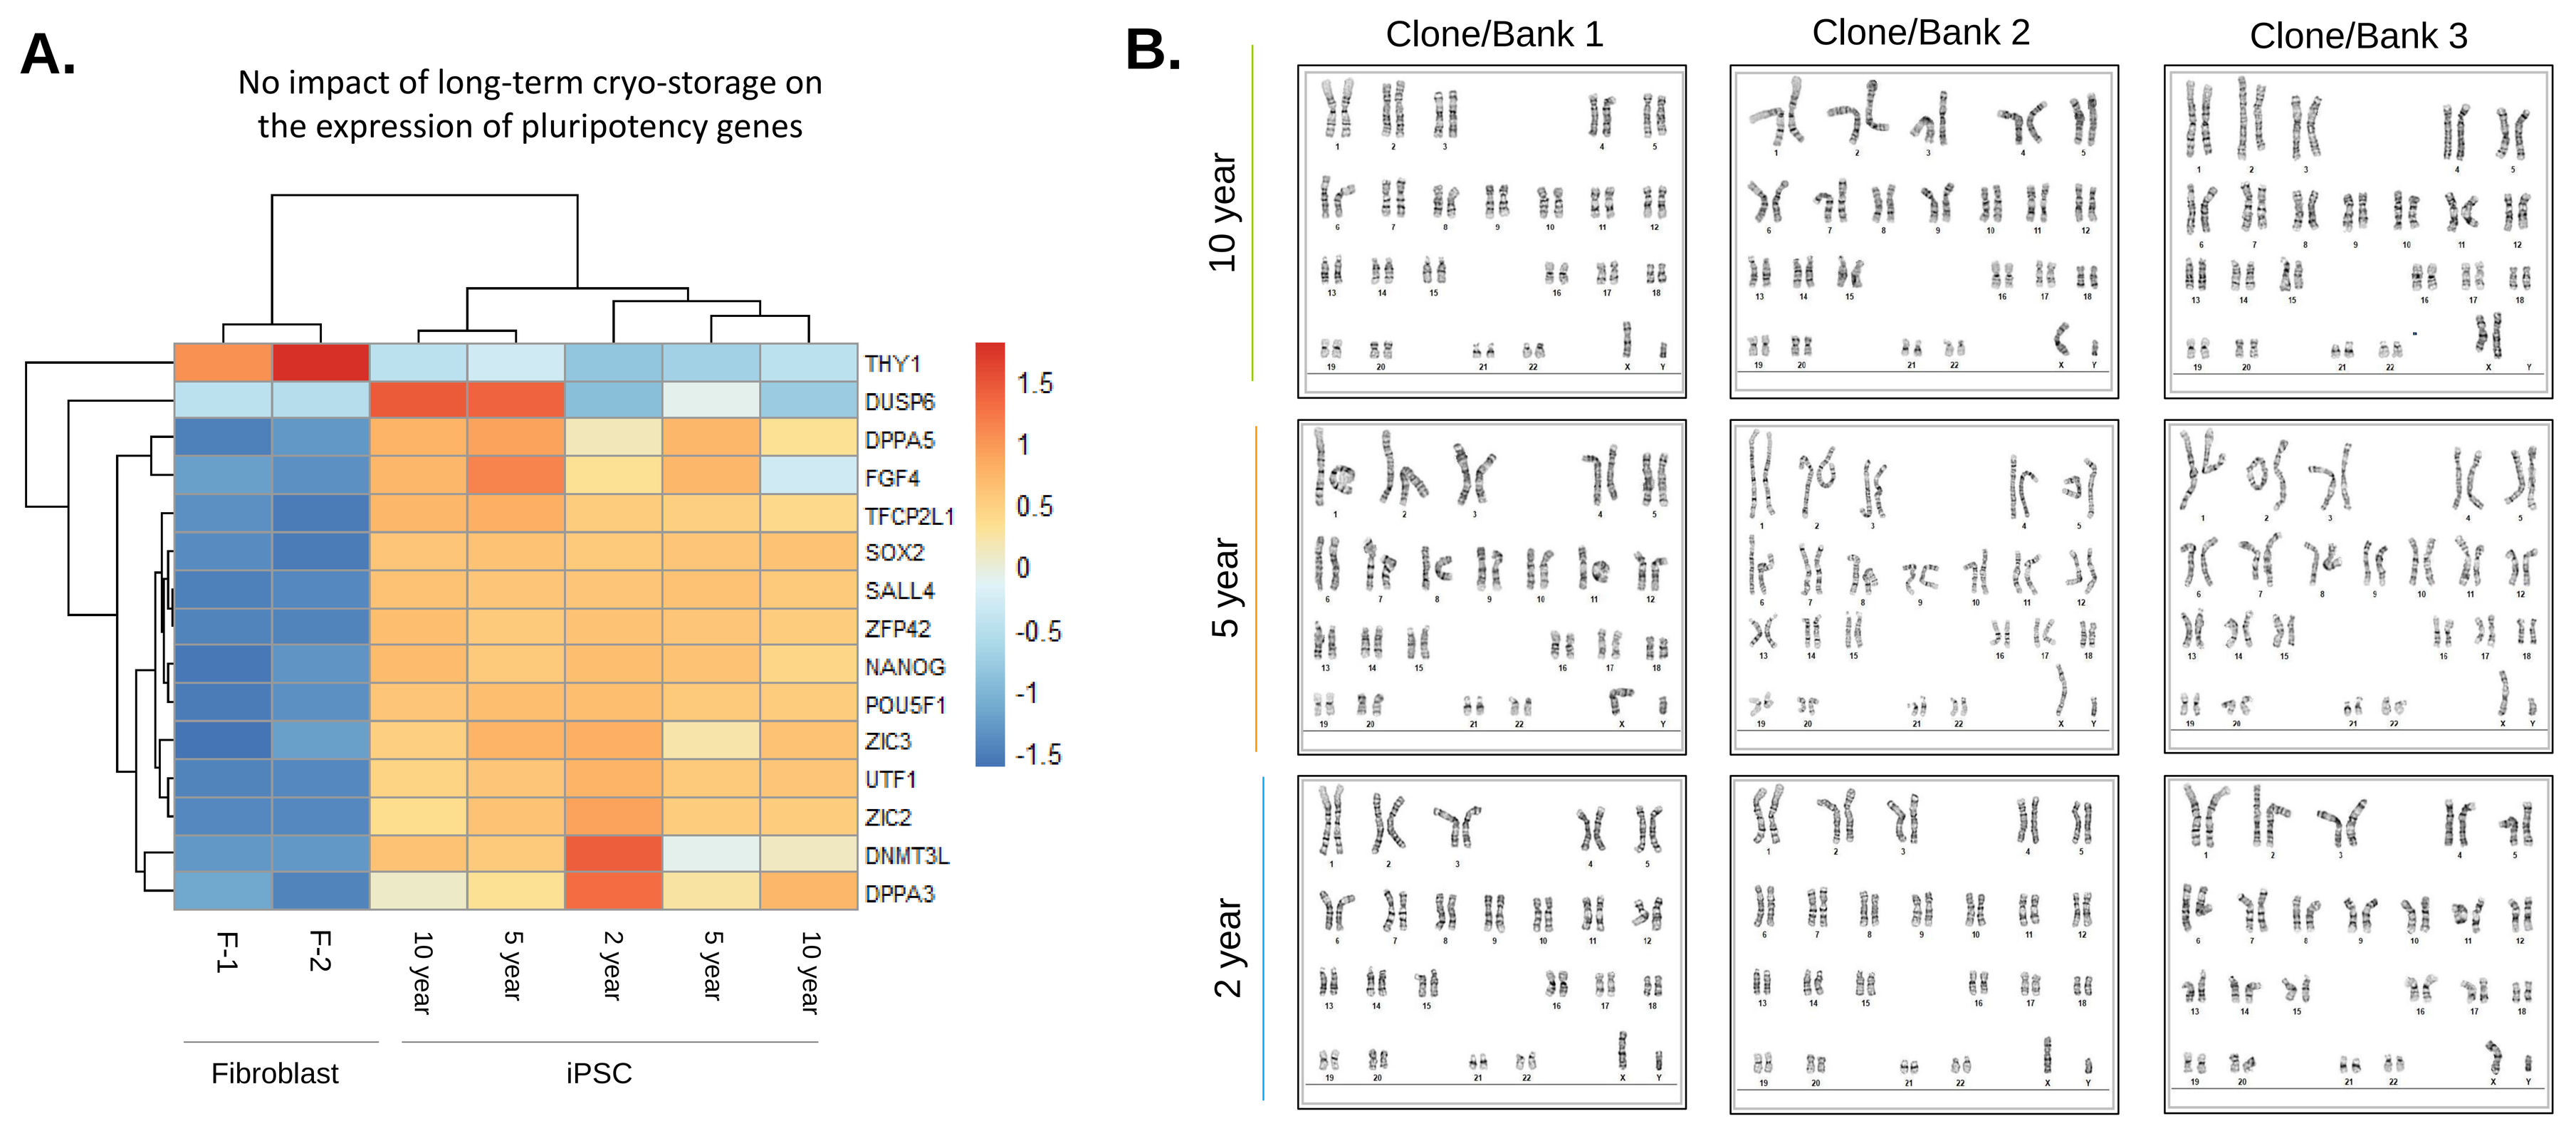
A.
No impact of long-term cryo-storage on the expression of pluripotency genes
THY1
DUSP6
DPPA5
FGF4
TFCP2L1
SOX2
SALL4
ZFP42
NANOG
POU5F1
ZIC3
UTF1
ZIC2
DNMT3L
DPPA3
F-1
F-2
Fibroblast
iPSC
B.
Clone/Bank 1
Clone/Bank 2
Clone/Bank 3
10 year
5 year
2 year

Figure 2. Maintenance of pluripotency gene expression and genomic stability of cryopreserved iPSCs. (A) Gene expression analysis by NanoString shows no significant impact on pluripotency gene expression in iPSC banks following long-term cryo-storage (>10 years vs 2 years). **(B)** Genomic stability of iPSC lines frozen for indicated years was assessed by G-banding after thaw and passage. All lines examined were shown to maintain a normal, euploid karyotype.

Transgene Expression is Stable Over Long-term Cryo-storage

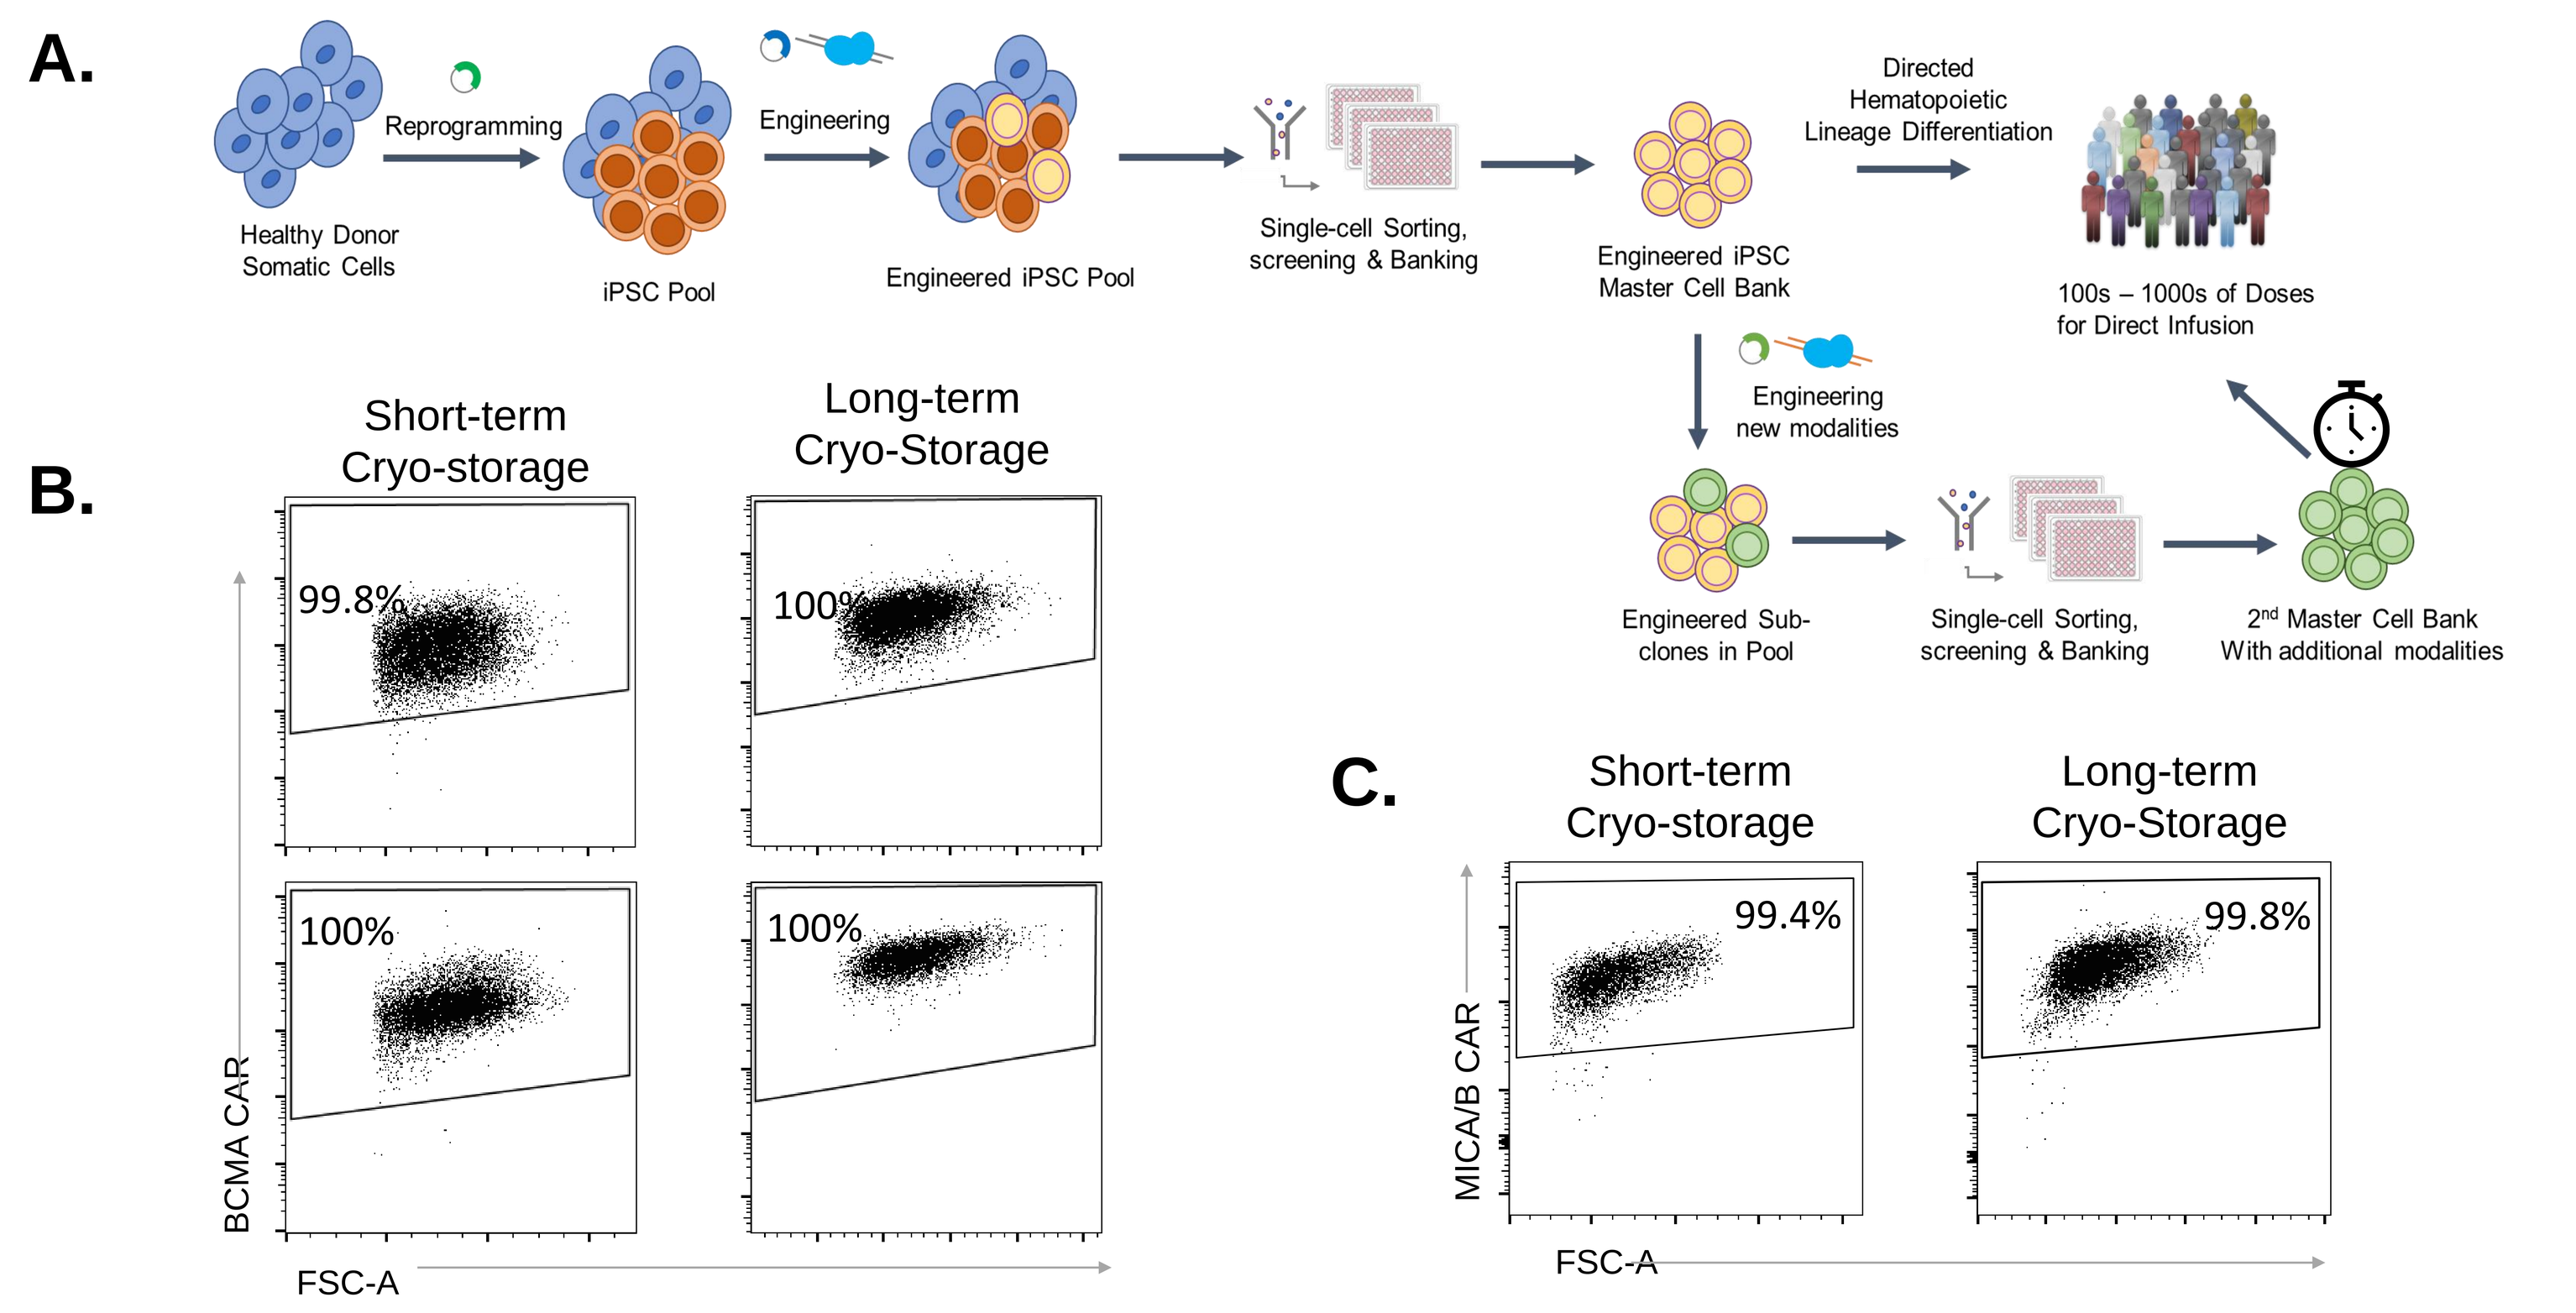
A.
Healthy Donor Somatic Cells
Reprogramming
iPSC Pool
Engineering
Engineered iPSC Pool
Single-cell Sorting, screening & Banking
Engineered iPSC Master Cell Bank
Directed Hematopoietic Lineage Differentiation
100s – 1000s of Doses for Direct Infusion
Engineering new modalities
Engineered Sub-clones in Pool
2nd Master Cell Bank With additional modalities
B.
Short-term Cryo-storage
Long-term Cryo-Storage
99.8%
100%
BCMA CAR
FSC-A
C.
99.4%
99.8%
MICA/B CAR

Figure 3. Phenotypic characterization of engineered iPSC lines after short- and long-term cryo-storage. (A) Engineered iPSC lines expressing diverse transgenes were cryopreserved for either 2 **(C)** or 3 **(B)** years and assessed for stability of transgene expression by flow cytometry. All engineered lines showed stable expression of the transgenes.

iPSC Banks Maintain Robust Trilineage Differentiation Potential After Long-Term Cryo-storage

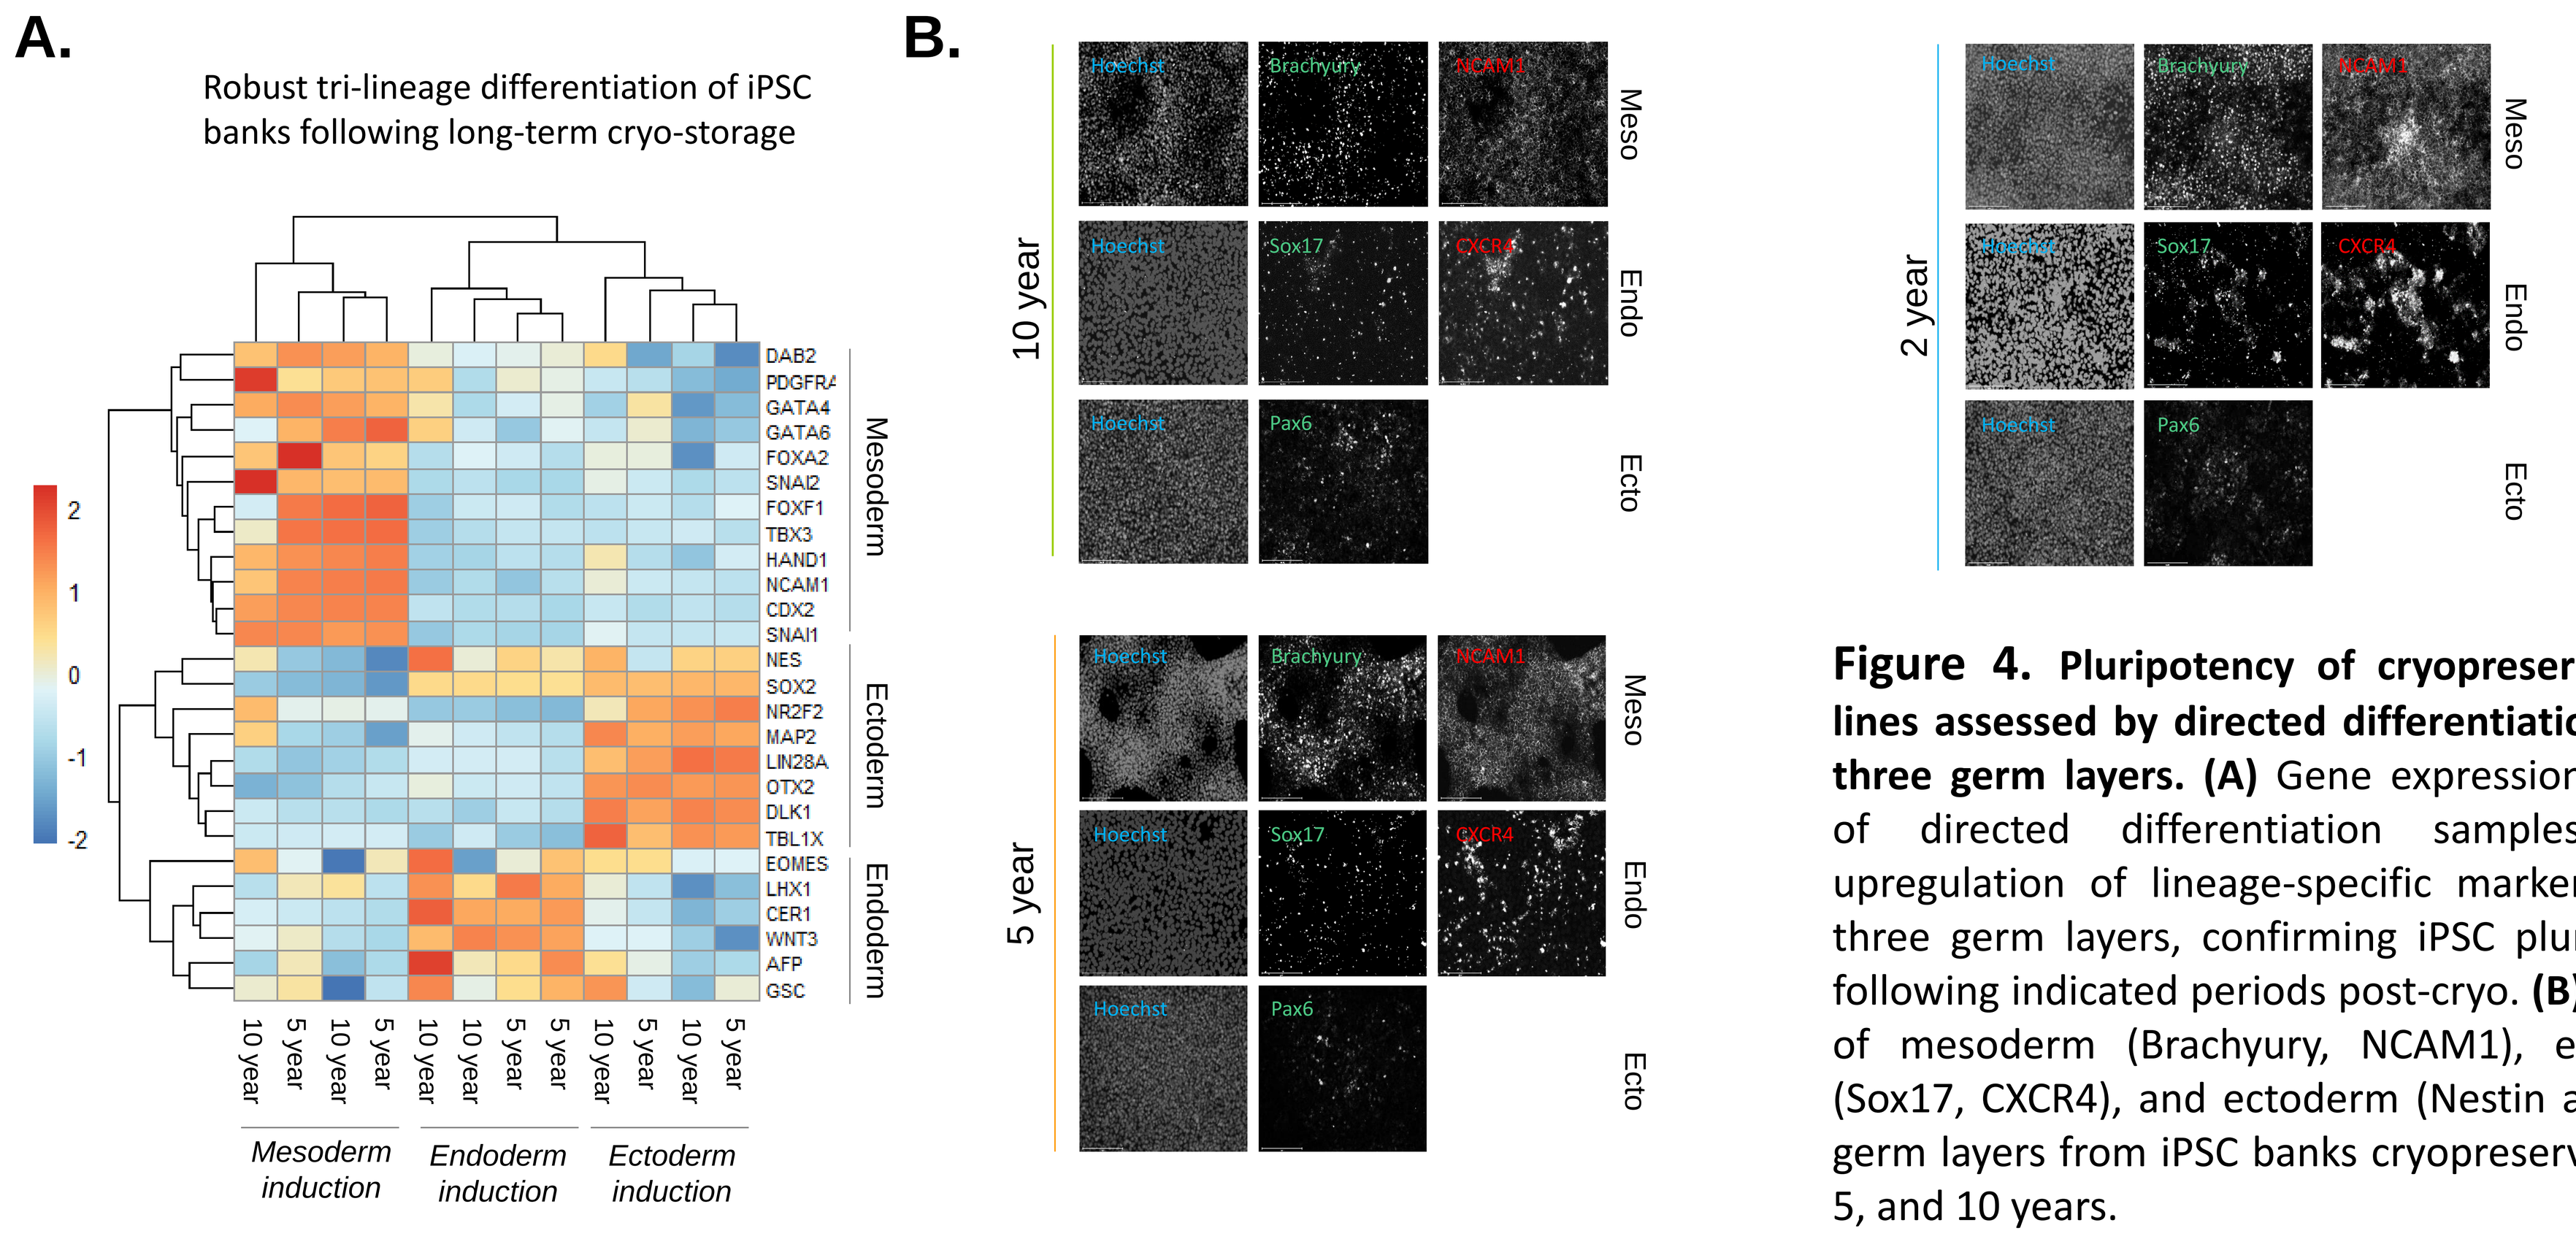
A.
Robust tri-lineage differentiation of iPSC banks following long-term cryo-storage
Mesoderm
Ectoderm
Endoderm
Mesoderm induction
Endoderm induction
Ectoderm induction
B.
10 year
5 year
2 year
Hoechst
Brachyury
NCAM1
Sox17
CXCR4
Pax6
Meso
Endo
Ecto

Figure 4. Pluripotency of cryopreserved iPSC lines assessed by directed differentiation to the three germ layers. (A) Gene expression analysis of directed differentiation samples shows upregulation of lineage-specific markers for all three germ layers, confirming iPSC pluripotency following indicated periods post-cryo. **(B)** Markers of mesoderm (Brachyury, NCAM1), endoderm (Sox17, CXCR4), and ectoderm (Nestin and Pax6) germ layers from iPSC banks cryopreserved for 2, 5, and 10 years.

Robust Hematopoietic Differentiation Potential After Long-Term Cryo-storage

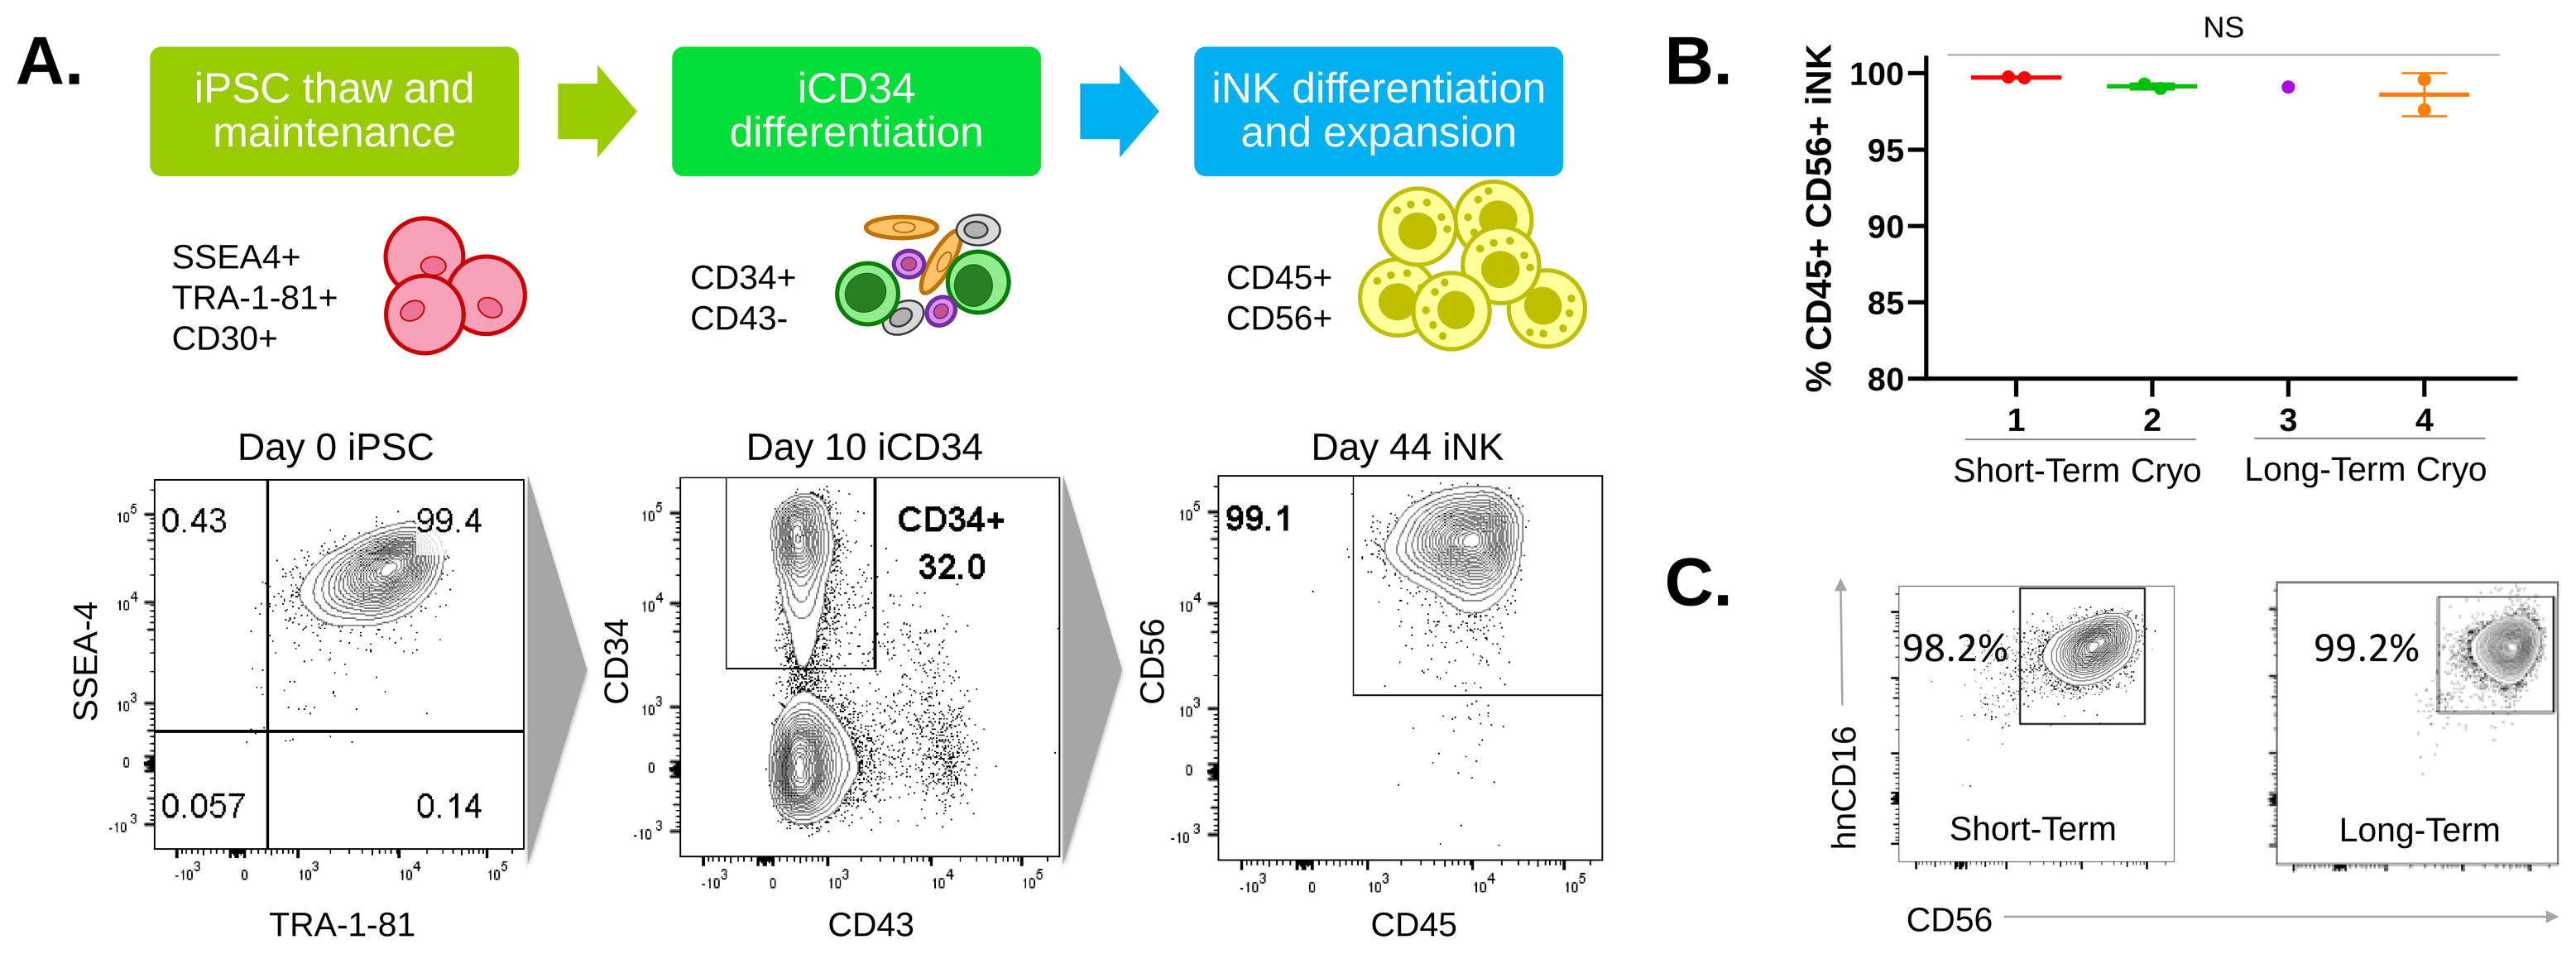
A.
iPSC thaw and maintenance
iCD34 differentiation
iNK differentiation and expansion
SSEA4+ TRA-1-81+ CD30+
CD34+ CD43-
CD45+ CD56+
Day 0 iPSC
Day 10 iCD34
Day 44 iNK
SSEA-4
TRA-1-81
CD34
CD43
CD56
CD45
B.
NS
% CD45+ CD56+ iNK
Short-Term Cryo
Long-Term Cryo
C.
98.2%
99.2%
hnCD16
Short-Term
Long-Term

Figure 5. Long-term cryopreserved iPSCs maintain robust hematopoietic differentiation potential after long-term storage. (A) NK cells are produced by directed differentiation of SSEA4+TRA-1-81+ iPSC cells to CD34+ hematopoietic progenitors and then to CD45+CD56+ NK cells. **(B)** Long-term cryopreserved iPSCs maintain the capacity to differentiate to a homogenous population of CD45+CD56+ iNK cells in multiple independent rounds of iNK differentiation and expansion. **(C)** iNKs derived from long- term cryopreserved iPSCs (right panel) maintain stable expression of the engineered transgene, high-affinity, non-cleavable CD16 Fc receptor (hnCD16) compared to iNKs derived from short-term cryopreserved iPSCs (left panel).