Novel Immune Reconstitution Model Highlights the Importance of Stealth Strategies that Potentiate Effector Cell Function and Promote Functional Persistence of Next-Generation Adoptive Cell Therapies

K.L. Hayama[1], A.M. Williams[1], Y. Pan[1], B. Groff[1], R.M. Mbofung[1], E.S. Varady[1], X. Zong[1], L. Fong[1], N. Brookhouser[1], R. Abujarour[1], T. Lee[1], P.M. Szabo[1], L. Wong[1], S. Cooley[1], M. Mamonkin[2], KJ. Malmberg[3,4], R. Bjordahl[1], J.P. Goodridge[1], B. Valamehr[1]

1. Fate Therapeutics, Inc., San Diego, CA, USA 2. Baylor College of Medicine, Houston, TX, USA 3. Karolinska Institute, Stockholm, Sweden 4. University of Oslo, Norway

INTRODUCTION

Cyclophosphamide and Fludarabine (Cy/Flu)-based lymphodepletion conditioning (LDC) regimens are recognized as critical to inducing homeostatic cytokines and effecting immune system modulation to support adoptively transferred, cell-based cancer therapy. However, protracted LDC has been associated with poor immune reconstitution and increased susceptibility to opportunistic infections. Therefore, the next-generation of cancer cell therapies should prioritize reducing the requirement for LDC regimens.

Using a novel immune reconstitution in vitro assay, we provide promising evidence for the ability of our unique stealth strategies to support effector cell persistence and potentiation of function without the need for Cy/Flu-based LDC; including,

(1) engineering an alloimmune defense receptor (ADR) which targets 4-1BB expressed on alloreactive immune cells and provides a CD3ζ signaling boost to potentiate ADR-edited iPSC-derived NK (iNK) cells; (2) knockout of two undisclosed costimulatory ligands (L1/L2) to prevent immunological synapse formation; and (3) a combinatorial therapeutic strategy including CD38-null iNK cells with the addition of daratumumab, an anti-CD38 monoclonal antibody (mAb), to deplete alloreactive effector cells.

STRATEGIES TO REDUCE LYMPHO-CONDITIONING

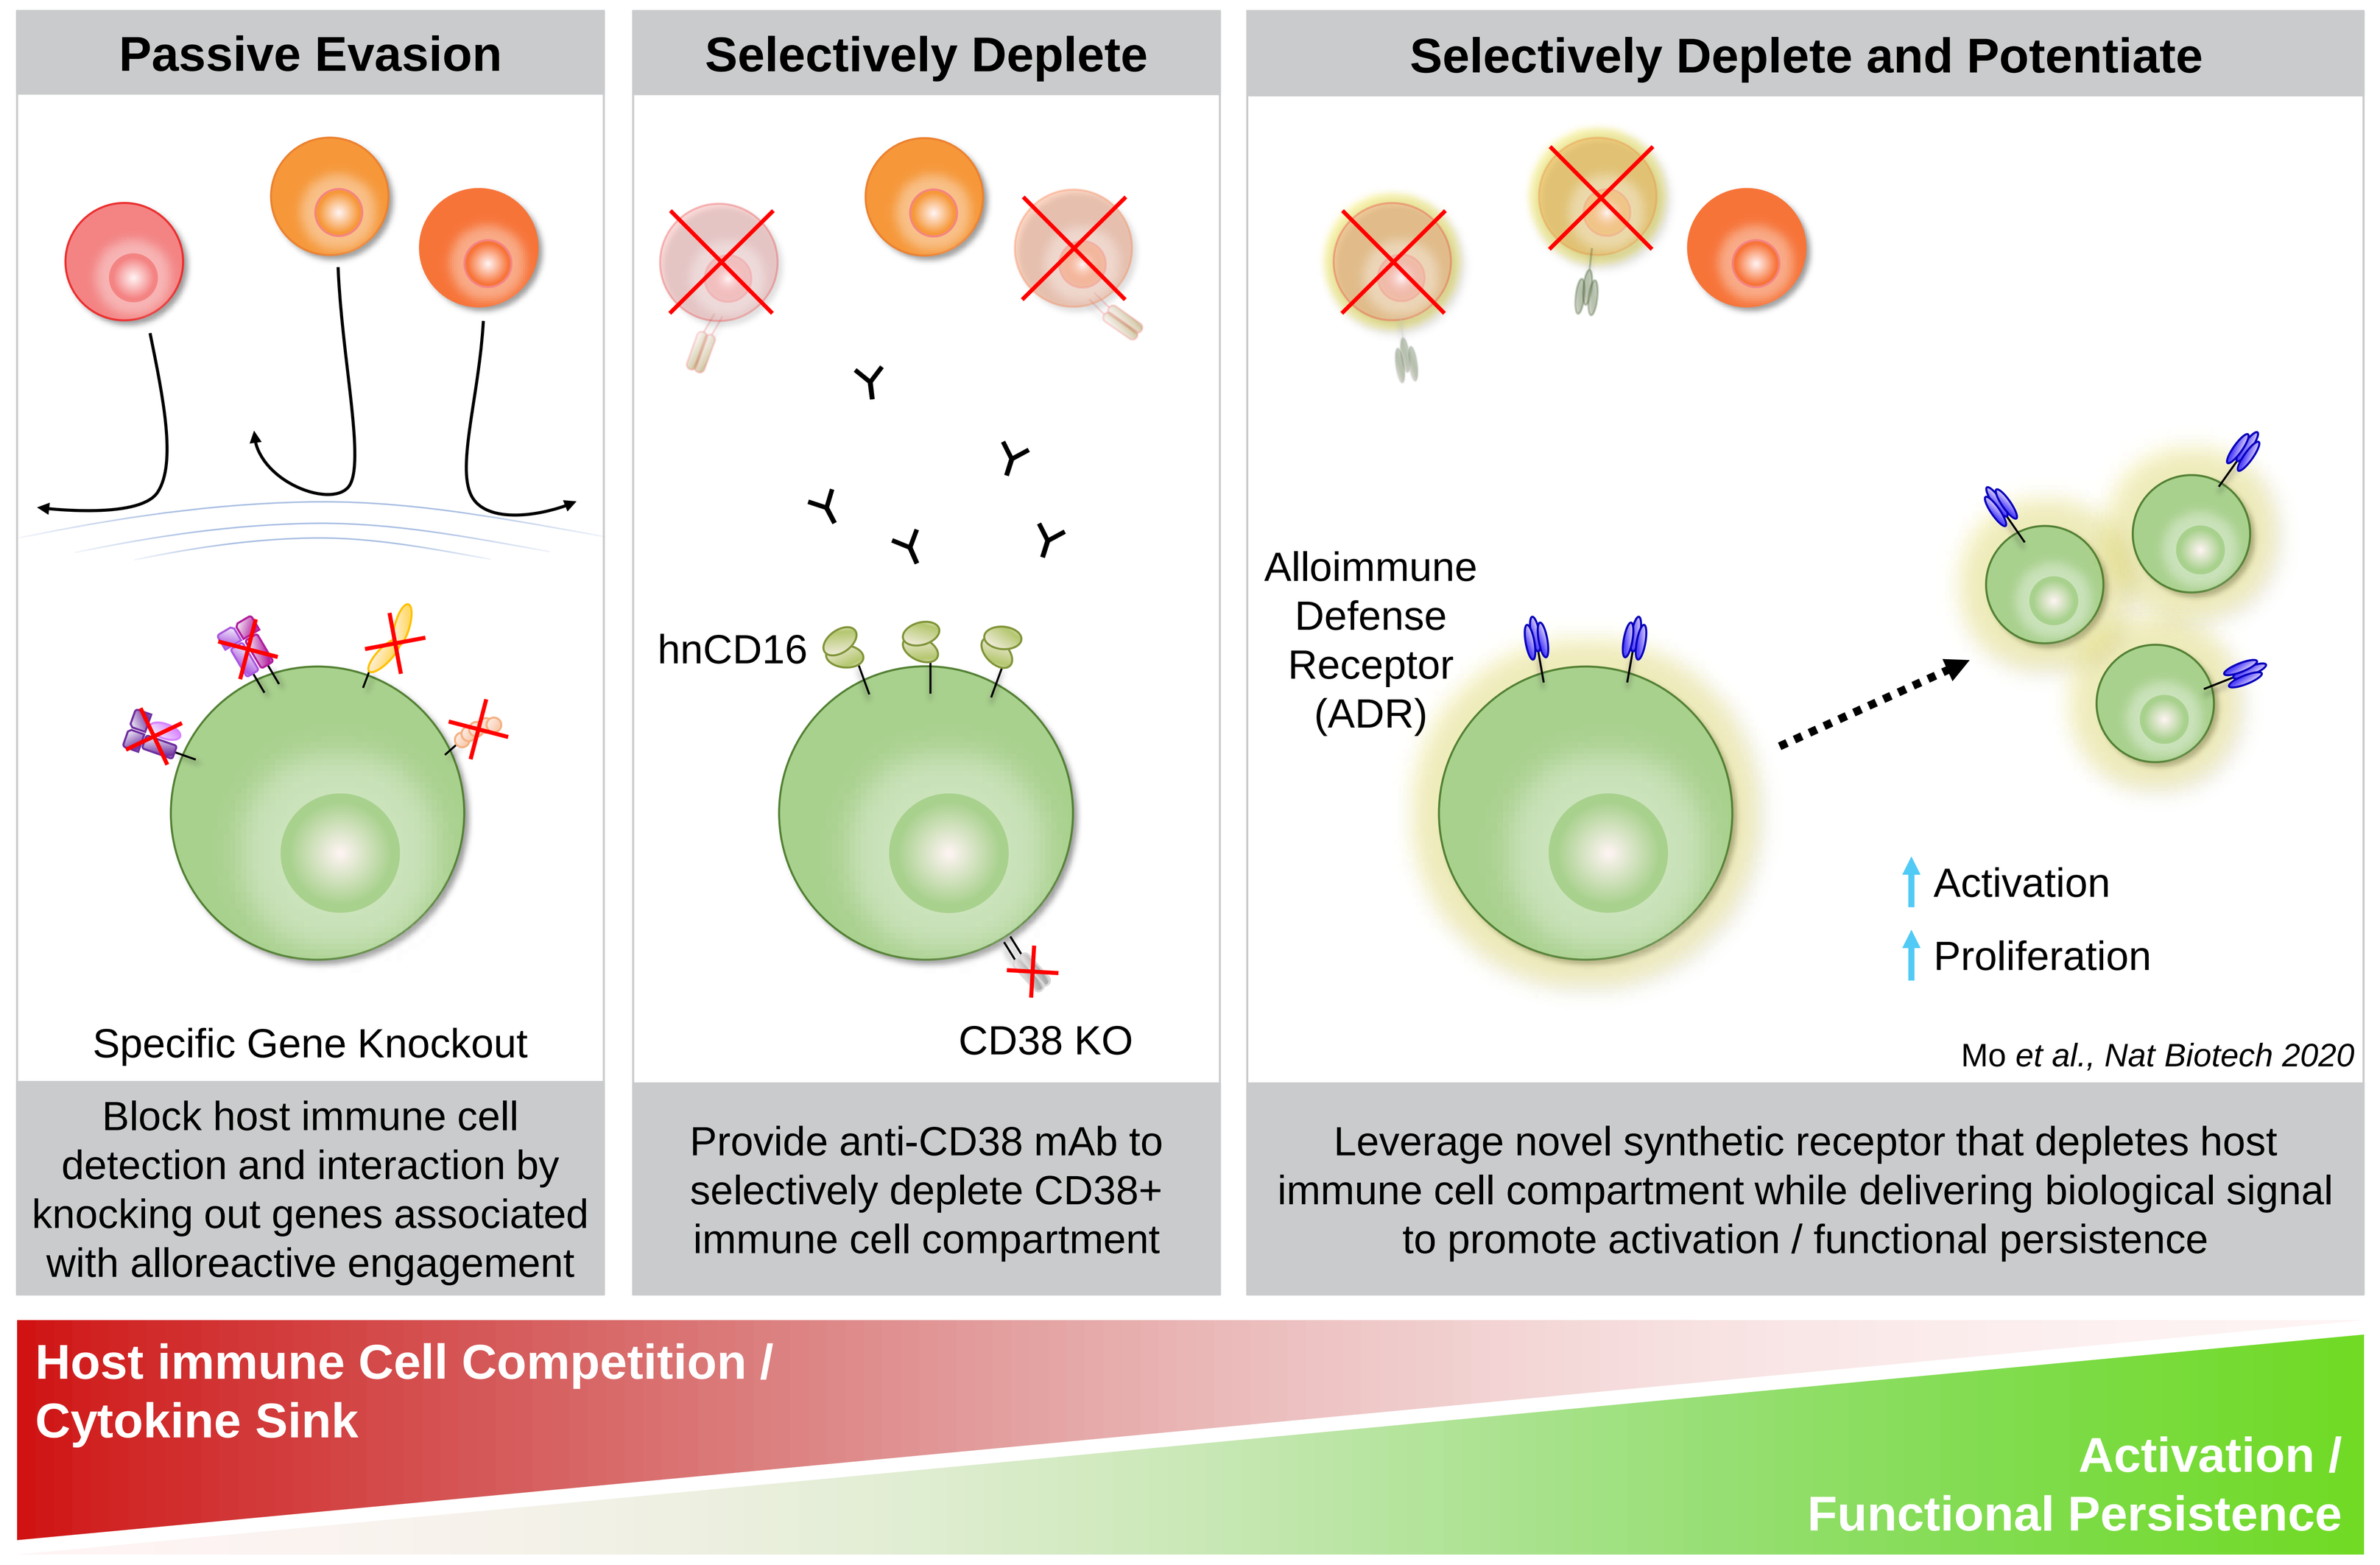
Passive Evasion
Selectively Deplete
Selectively Deplete and Potentiate
hnCD16
Alloimmune Defense Receptor (ADR)
Activation
Proliferation
Specific Gene Knockout
CD38 KO
Mo et al., Nat Biotech 2020
Block host immune cell detection and interaction by knocking out genes associated with alloreactive engagement
Provide anti-CD38 mAb to selectively deplete CD38+ immune cell compartment
Leverage novel synthetic receptor that depletes host immune cell compartment while delivering biological signal to promote activation / functional persistence
Host immune Cell Competition / Cytokine Sink
Activation / Functional Persistence

Mo *et al.*, Engineered off-the-shelf therapeutic T cells resist host immune rejection. Nature Biotechnology 2020
doi: 10.1038/s41587-020-0601-5

CONCLUSIONS

Our preclinical work has identified multiple approaches to eliminate the need for patient lympho-conditioning currently required to support adoptive cell therapy. Through the examination of novel stealth strategies in a unique allogeneic reconstitution model, we highlight:

- 4-1BB and CD38 are upregulated on activated effector cells, including alloreactive NK and T cells
- ADR uniquely potentiates iNK cells and extends functional persistence by selective targeting of 4-1BB-positive control or Cy/Flu-treated alloreactive immune cells
- MHC class I/II knockout (β2M/CIITA)-induced missing-self response by pbNKs is evaded by knocking out costimulatory ligand L1 and L2 or by selective depletion with daratumumab through ADCC
- Treating patients with daratumumab extends a window of opportunity for adoptive cell therapies after LDC by delaying immune reconstitution and preventing recovery of potentially alloreactive CD38$^+$ lymphocytes
- Combinatorial stealth strategies (e.g., ADR w/ daratumumab) can further enhance functional persistence

Collectively, the data demonstrate that unique stealth strategies that promote functional persistence and potentiate effector cell function have the potential to eliminate the need for Cy/Flu LDC. Notably, arming iNK cells with ADR extends functional persistence in the presence of an intact endogenous immune compartment.

RESULTS

Off-the-shelf, iPSC-derived NK cells armed with combinations of stealth edits to passively evade or selectively deplete alloreactive immune cells

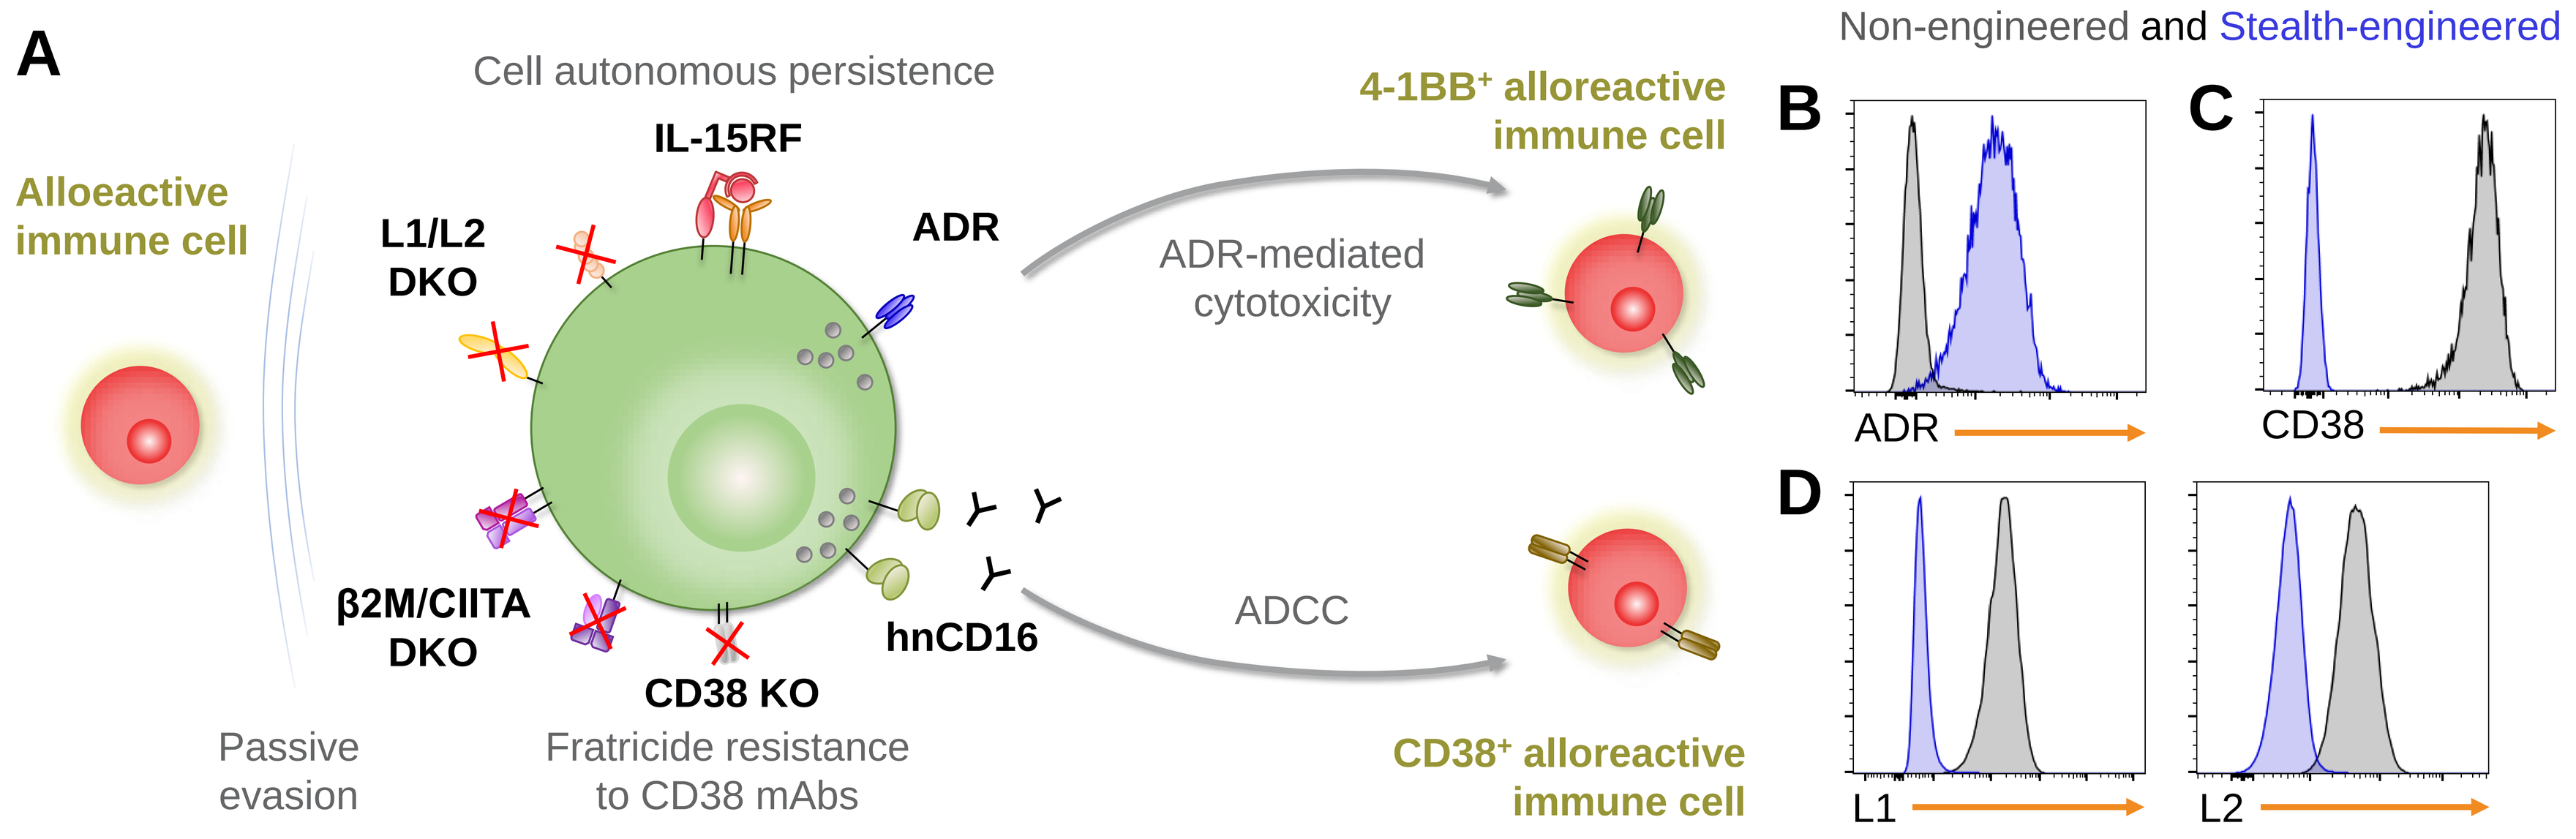
A
Cell autonomous persistence
IL-15RF
Alloeactive immune cell
L1/L2 DKO
ADR
β2M/CIITA DKO
CD38 KO
hnCD16
ADR-mediated cytotoxicity
ADCC
4-1BB+ alloreactive immune cell
CD38+ alloreactive immune cell
Passive evasion
Fratricide resistance to CD38 mAbs
Non-engineered and Stealth-engineered
B
C
D
ADR
CD38
L1
L2

Figure 1 ***Combinatorial edits for iPSC-derived NK cells.*** (A) iNK cells are engineered to express: (1) high-affinity, non-cleavable CD16 (hnCD16) to augment antibody dependent cellular cytotoxicity (ADCC); (2) an IL-15 receptor fusion (IL-15RF) to enhance NK functionality and persistence; (3) CD38 knockout to prevent anti-CD38 mAb-induced fratricide; (4) ADR to selectively target 4-1BB expressed on alloreactive cells; and/or (5) B2M/CIITA and L1/L2 double knockout (DKO) for passive evasion. (B-D) Three iNK stealth lines were evaluated for expression of engineered elements by flow cytometry as shown in blue for (B) ADR-armed, (C) CD38-null, and (D) costimulatory ligands L1/L2-null iNK cells.

ADR potentiates iPSC-derived NK cells by extending functional persistence and proliferation by selectively targeting 4-1BB-positive alloreactive immune cells

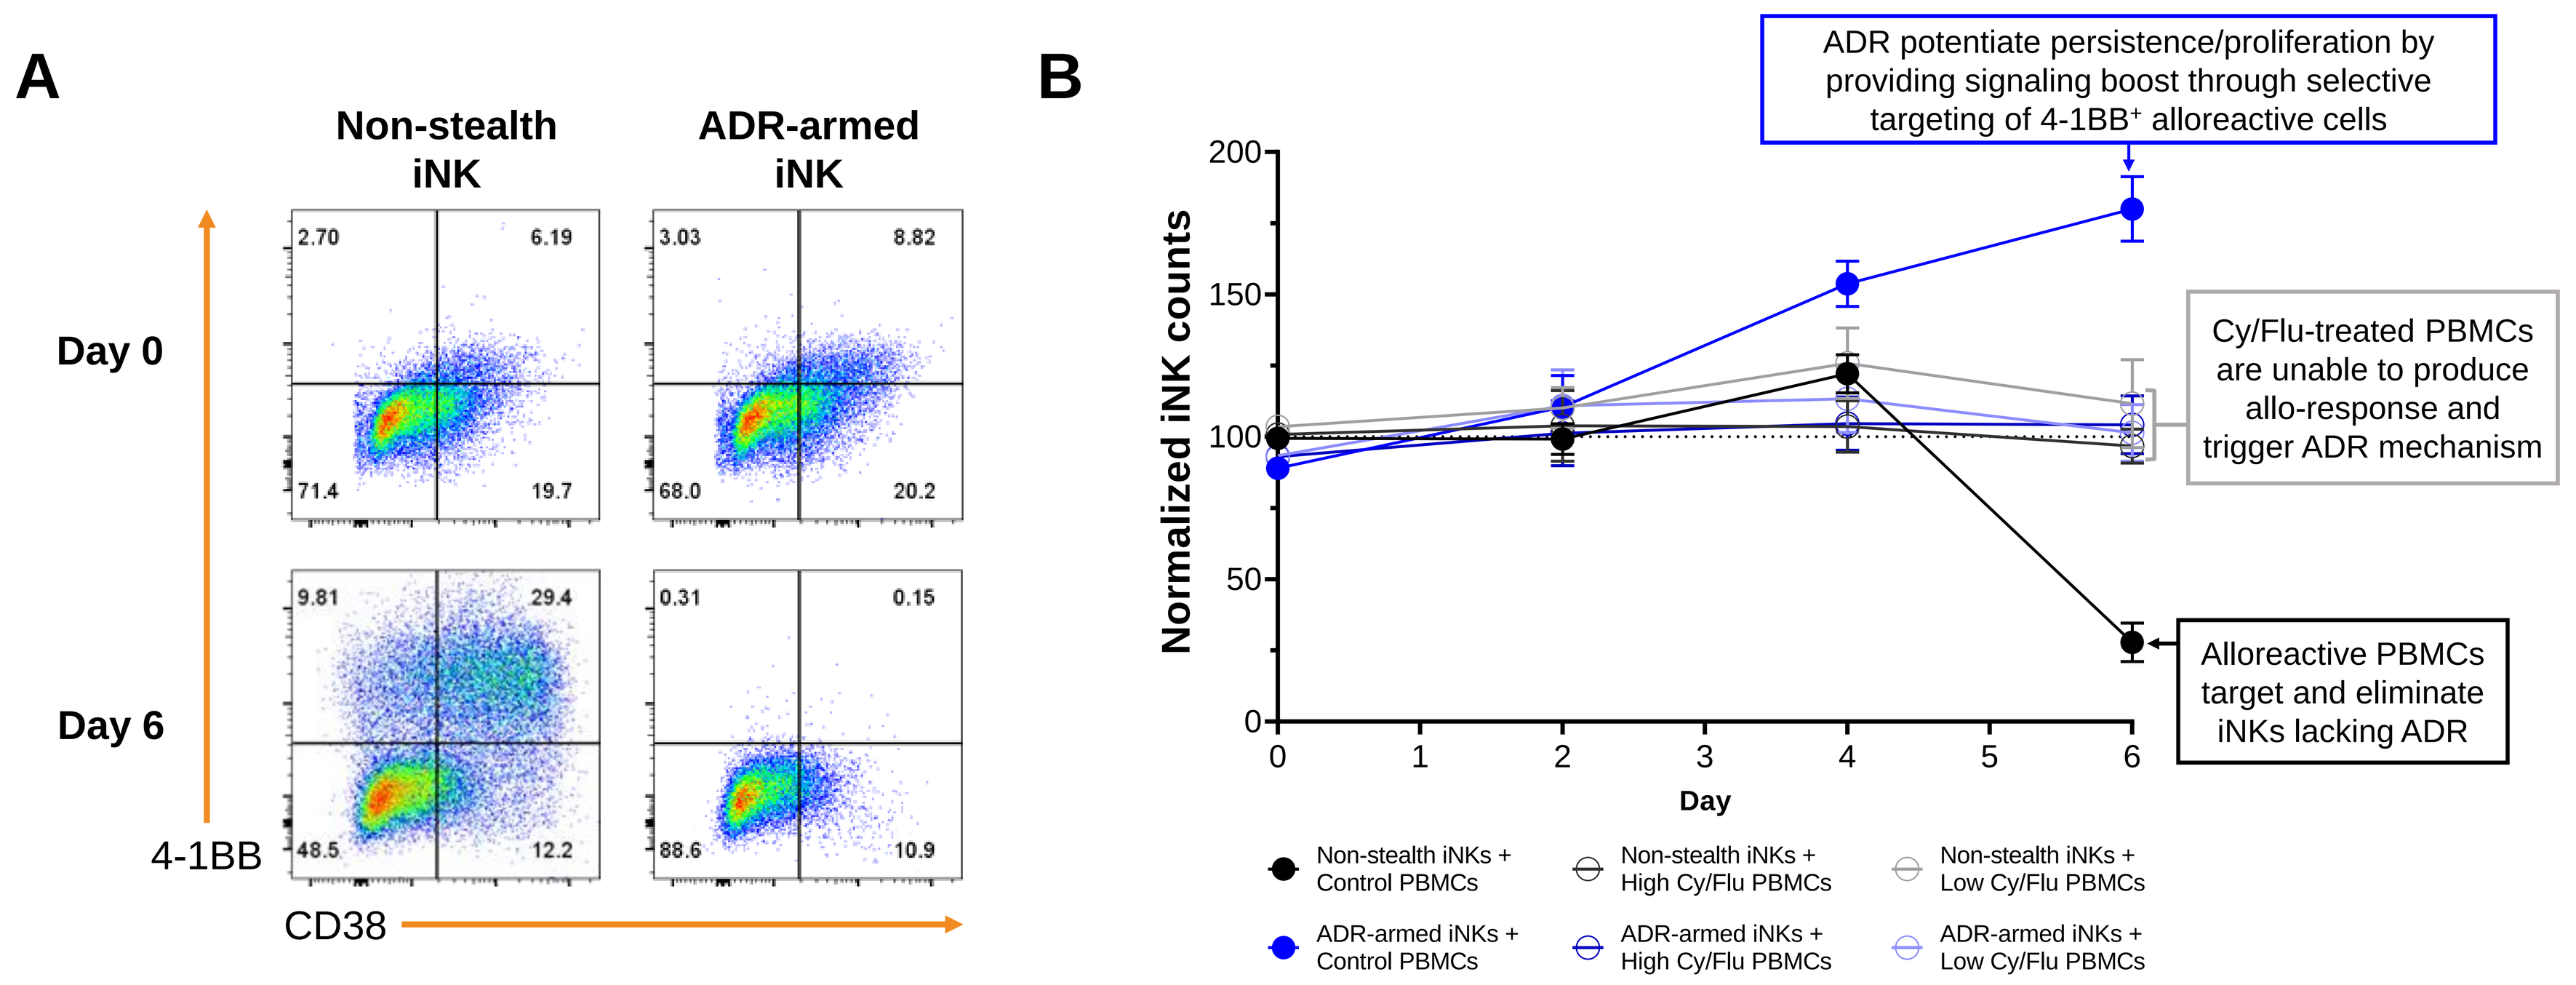
A
Non-stealth iNK
ADR-armed iNK
Day 0
Day 6
4-1BB
CD38
B
ADR potentiate persistence/proliferation by providing signaling boost through selective targeting of 4-1BB+ alloreactive cells
Cy/Flu-treated PBMCs are unable to produce allo-response and trigger ADR mechanism
Alloreactive PBMCs target and eliminate iNKs lacking ADR
Normalized iNK counts
Day
Non-stealth iNKs + Control PBMCs
Non-stealth iNKs + High Cy/Flu PBMCs
Non-stealth iNKs + Low Cy/Flu PBMCs
ADR-armed iNKs + Control PBMCs
ADR-armed iNKs + High Cy/Flu PBMCs
ADR-armed iNKs + Low Cy/Flu PBMCs

Figure 2 ***ADR potentiates and protects iNK cells from alloreactive immune cells.*** (A) Representative FACS plots of T cells after co-culture with iNK cells at 1:2 (E:T, iNK to PBMC) at day 6. (B) PBMCs were treated with phosphoramide mustard (cyclophosphamide) and fludarabine at two different dosages (1µg/ml and 0.125µg/ml respectively for low, 6µg/ml and 2µg/ml for high) for 24.hours, followed by 48 hours of rest in media, and then co-cultured with iNKs. iNK counts are normalized to alone wells.

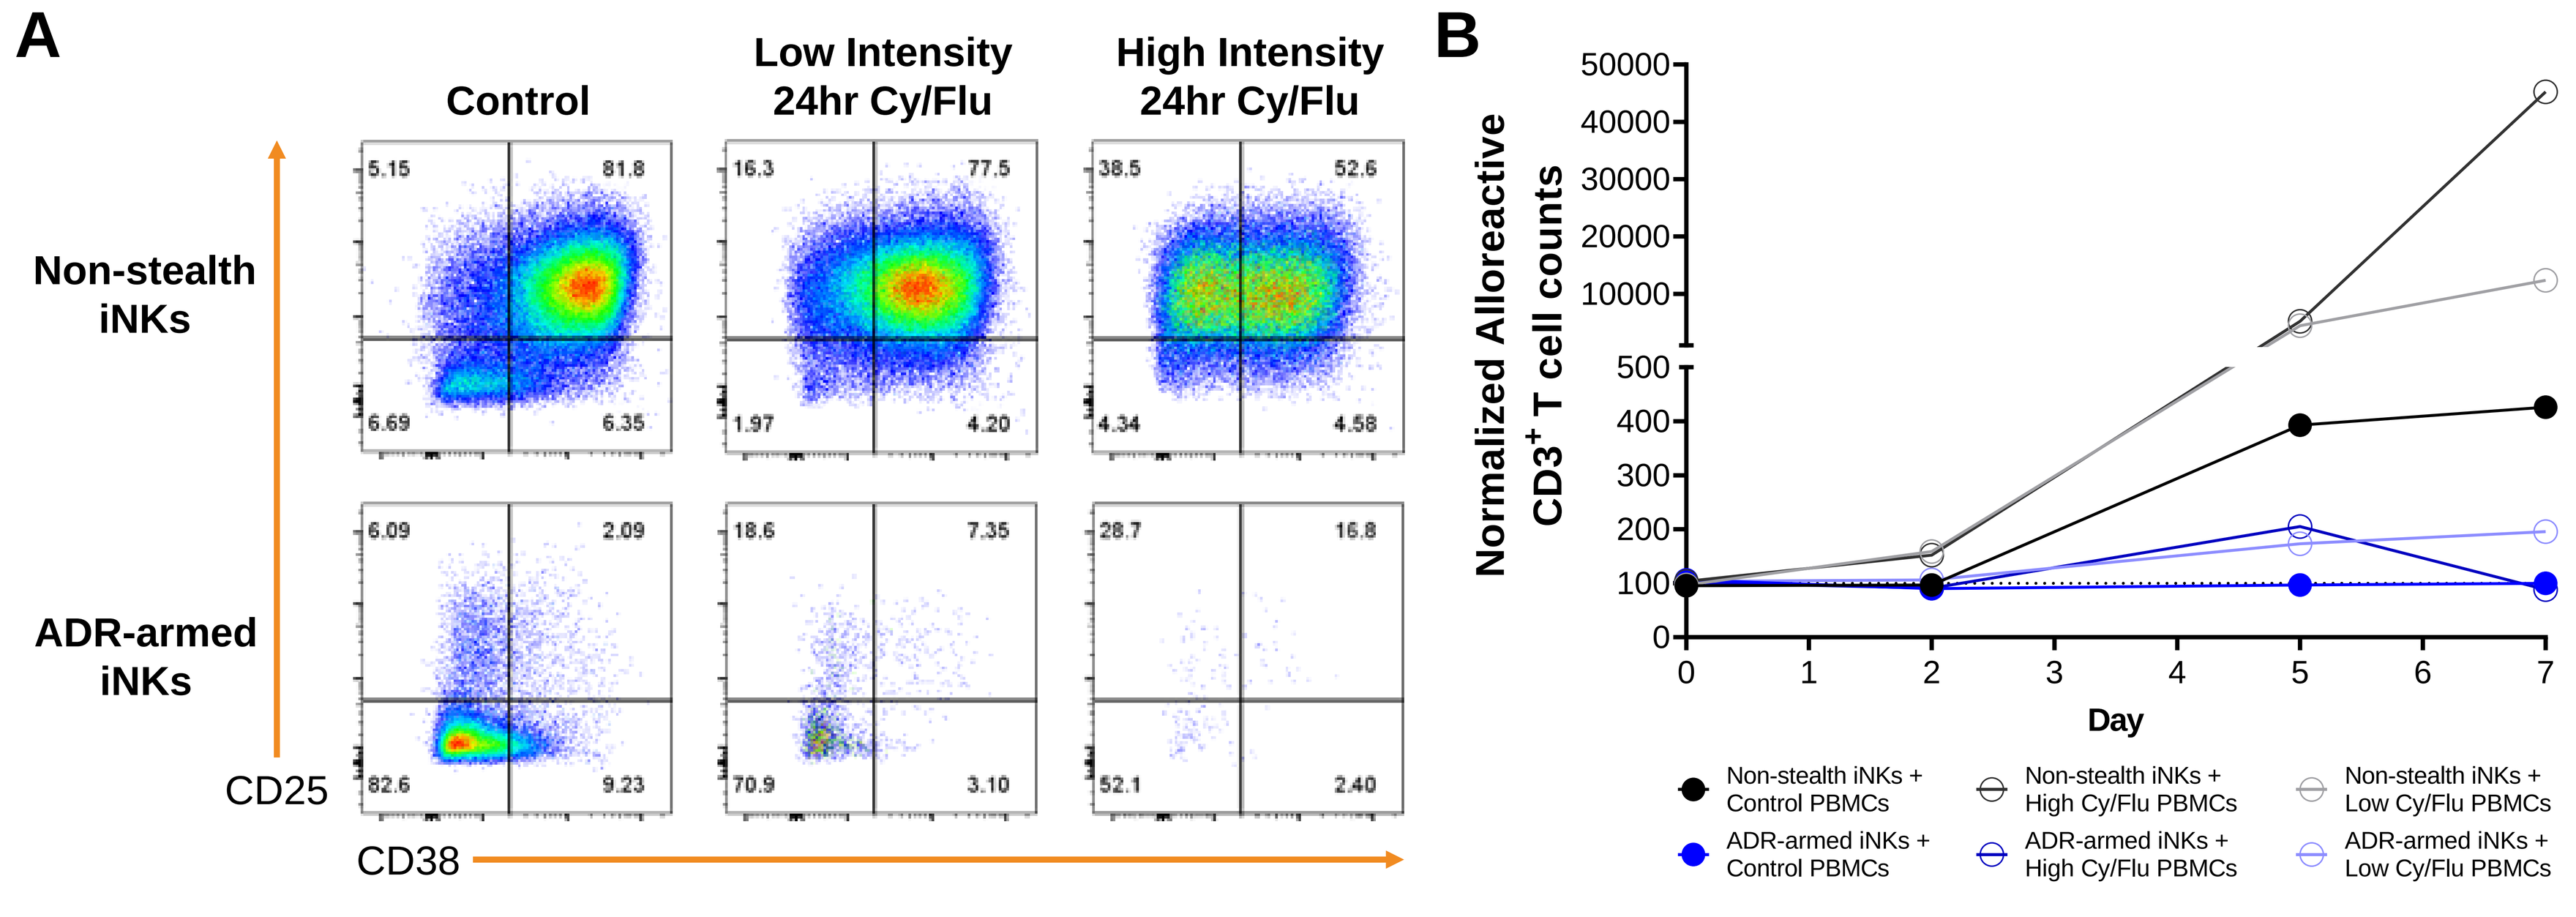
A
Control
Low Intensity 24hr Cy/Flu
High Intensity 24hr Cy/Flu
Non-stealth iNKs
ADR-armed iNKs
CD25
CD38
B
Normalized Alloreactive CD3+ T cell counts
Day
Non-stealth iNKs + Control PBMCs
Non-stealth iNKs + High Cy/Flu PBMCs
Non-stealth iNKs + Low Cy/Flu PBMCs
ADR-armed iNKs + Control PBMCs
ADR-armed iNKs + High Cy/Flu PBMCs
ADR-armed iNKs + Low Cy/Flu PBMCs

Figure 3 ***Distinct populations of alloreactive T cells present before and after Cy/Flu treatment are eliminated by ADR-armed iNK cells.*** PBMCs are treated with phosphoramide mustard (cyclophosphamide) and fludarabine treated at two different dosages (1µg/ml and 0.125µg/ml respectively for low, 3µg/ml and 1µg/ml for high intensity) for 24.hours and then directly co-cultured with iNKs at a 1:1 (E:T) ratio. (A) Representative FACS plots of T cells at day 7 examining CD38 and CD25 expression. (B) Alloreactive CD3$^+$ T cells normalized to alone wells are tracked across 7 days.

Costimulatory ligands L1/L2-null iNK cells evade MHC class I/II (β2M/CIITA) deficiency-induced missing self response from alloreactive peripheral blood NK (pbNK) cells

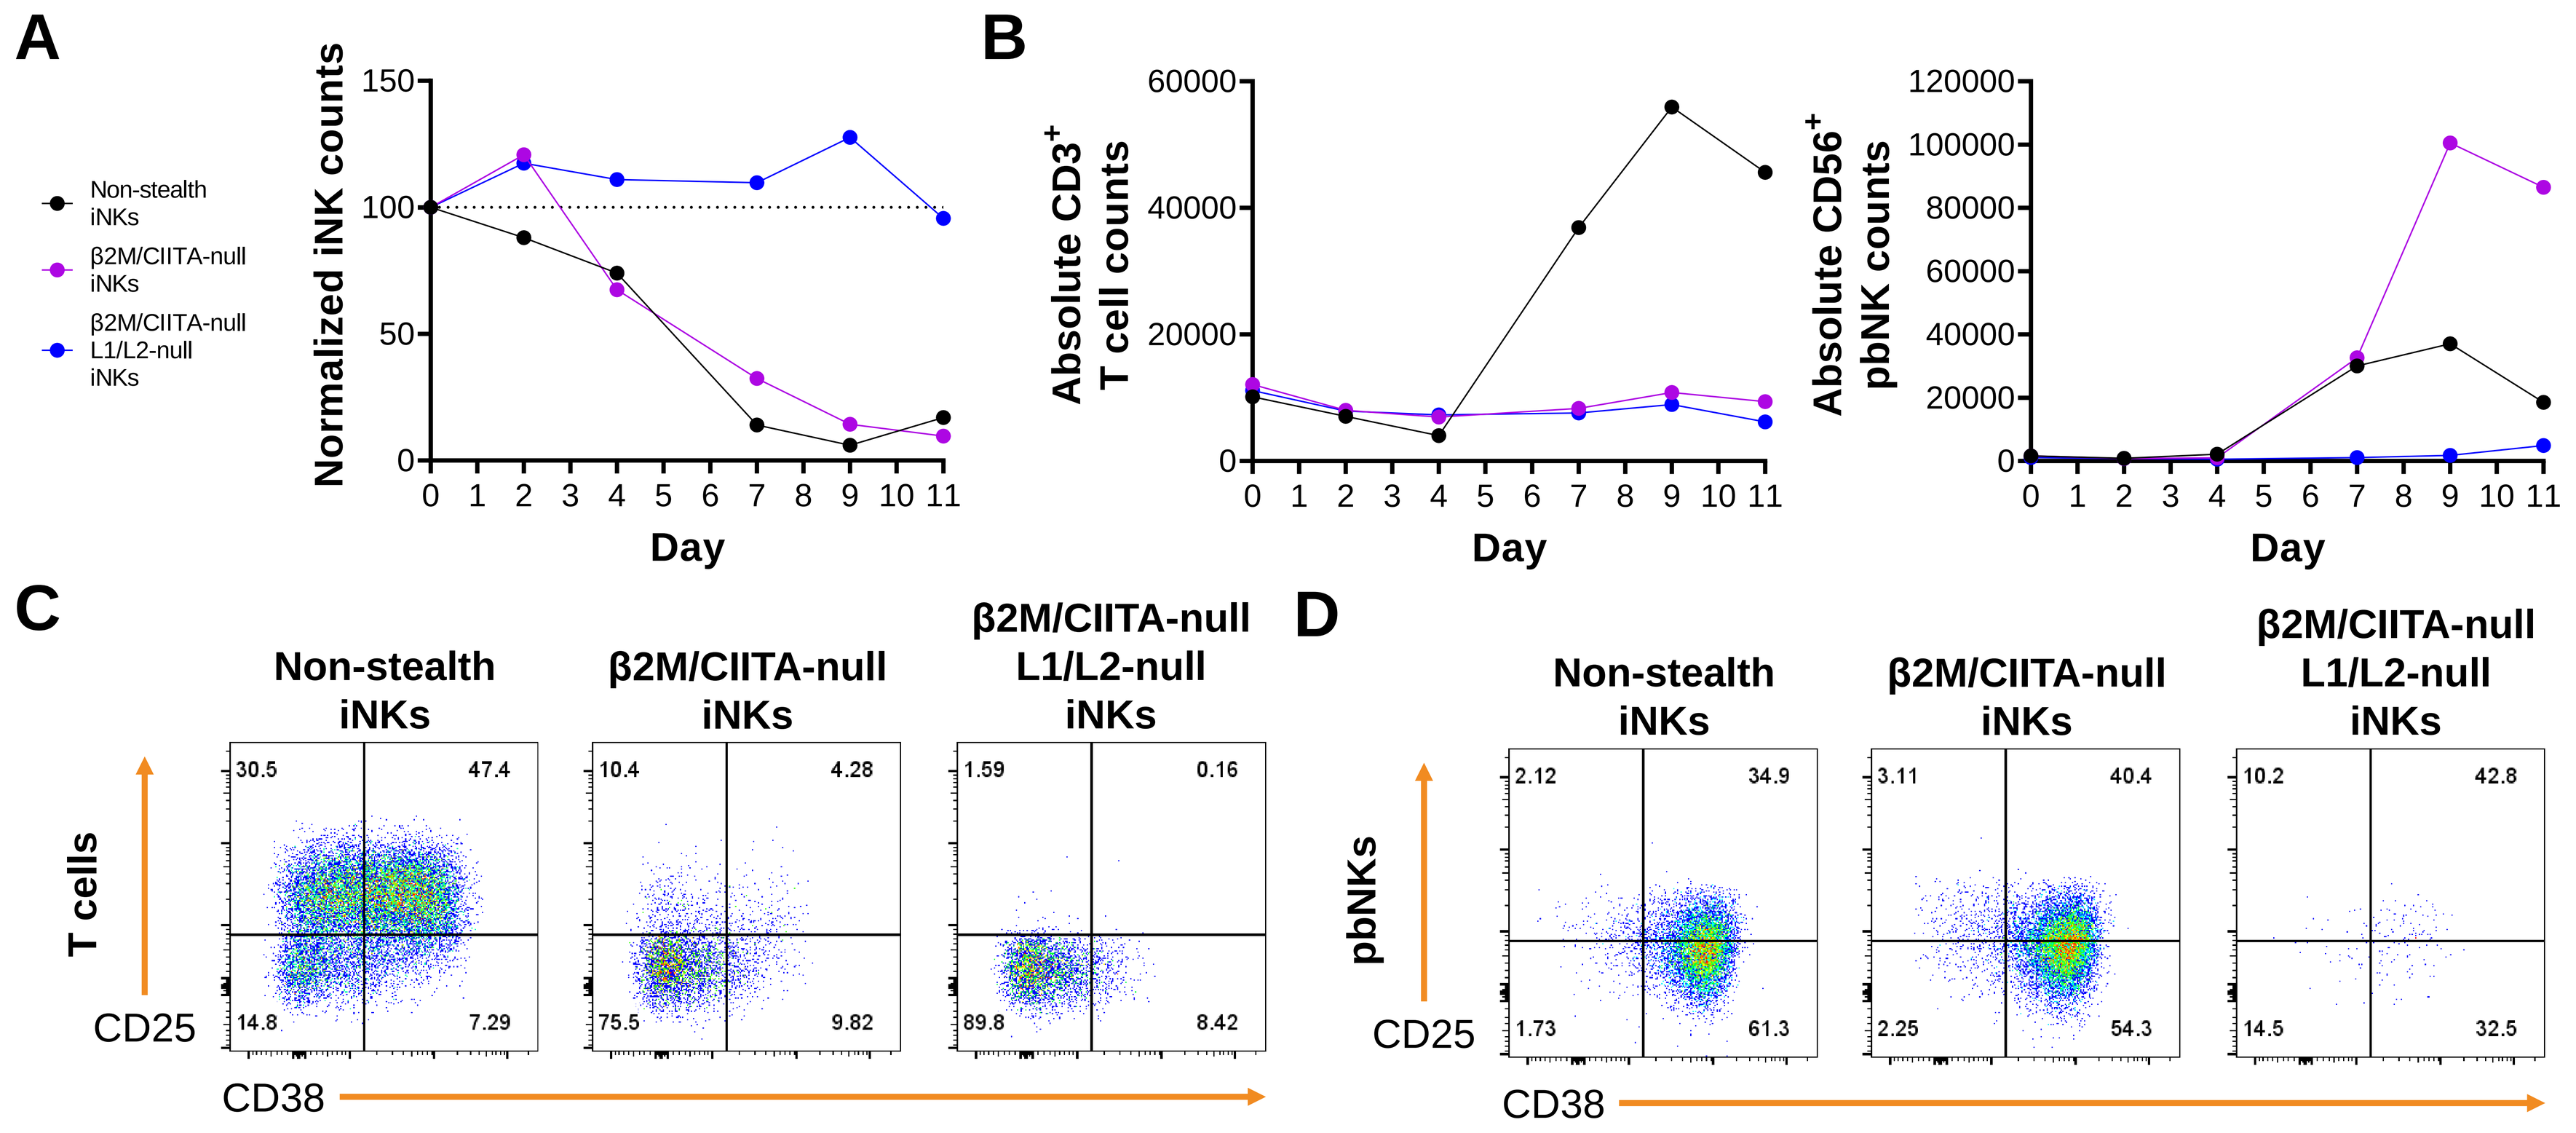
A
Non-stealth iNKs
β2M/CIITA-null iNKs
β2M/CIITA-null L1/L2-null iNKs
Normalized iNK counts
Day
B
Absolute CD3+ T cell counts
Absolute CD56+ pbNK counts
C
D
Non-stealth iNKs
β2M/CIITA-null iNKs
β2M/CIITA-null L1/L2-null iNKs
T cells
pbNKs
CD25
CD38

Figure 4 ***pbNK missing self response evaded in iNKs with double costimulatory ligand deficiency.*** iNK cells are engineered as β2M/CIITA-null or both β2M/CIITA and L1/L2-null. (A) iNKs are co-cultured with PBMCs at 1:1 and are normalized to their alone well temporal counterpart. (B) Absolute CD3$^+$ T cell and CD56$^+$ pbNK cell counts are quantified when co-cultured (1:1 for T cells, 2:1 for pbNKs) with iNKs. (C-D) Representative FACS plots examining CD25 and CD38 expression on (C) T cells and (D) pbNKs at 7 days after co-culture with iNKs.

Daratumumab provides an extended window of opportunity by delaying host immune reconstitution and eliminating potentially alloreactive CD38$^+$ T and NK cells

Figure 5 ***CD38-null iNK cells are resistant to alloreactive pbNK attack in the presence of daratumumab.*** Non-stealth and CD38/β2M/CIITA-null iNK cells are co-cultured with PBMCs with or without 10µg/ml of daratumumab at 1:1 and then tracked for iNK and CD38$^+$ pbNK counts across 9 days. (A) iNK cell counts normalized to alone wells. (B) Absolute CD38$^+$ pbNK cell counts.

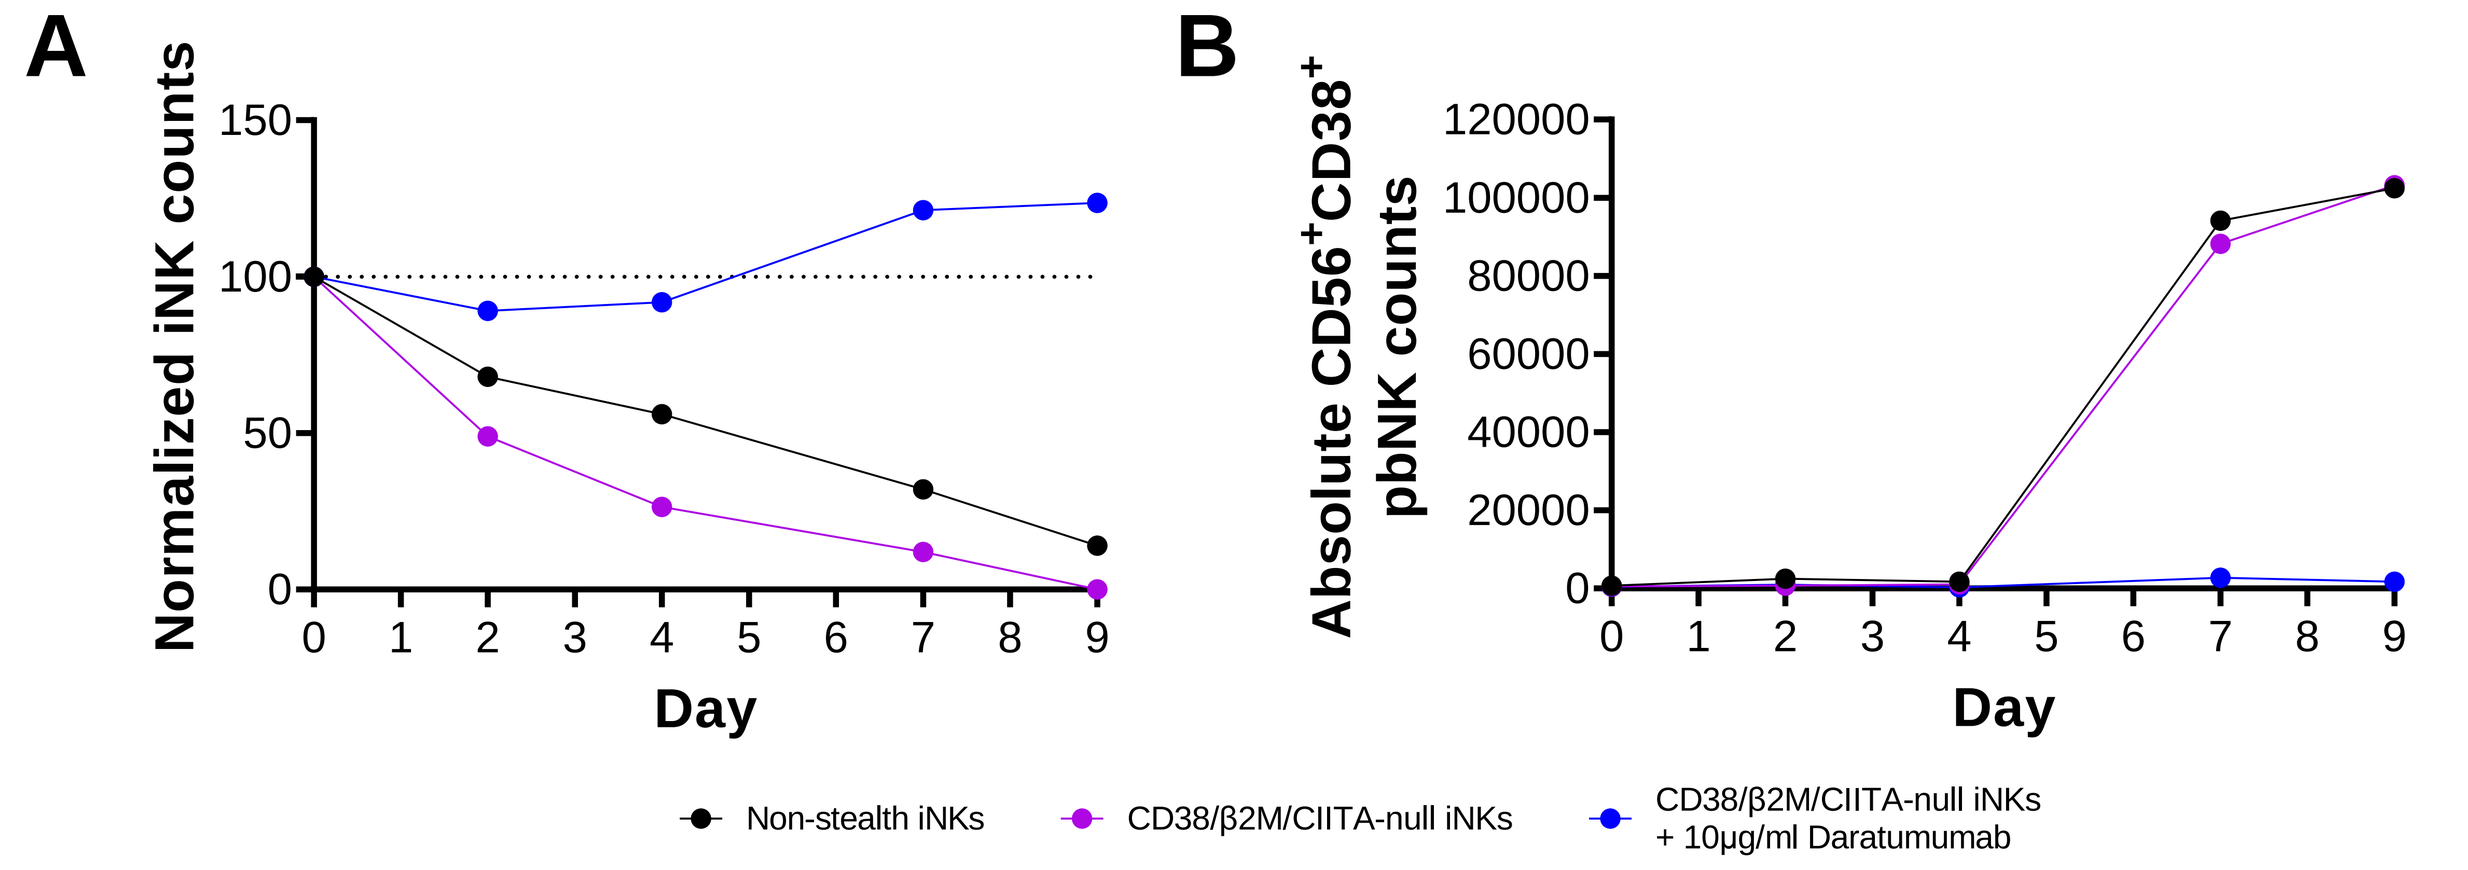
A
Normalized iNK counts
Day
B
Absolute CD56+CD38+ pbNK counts
Non-stealth iNKs
CD38/β2M/CIITA-null iNKs
CD38/β2M/CIITA-null iNKs + 10µg/ml Daratumumab

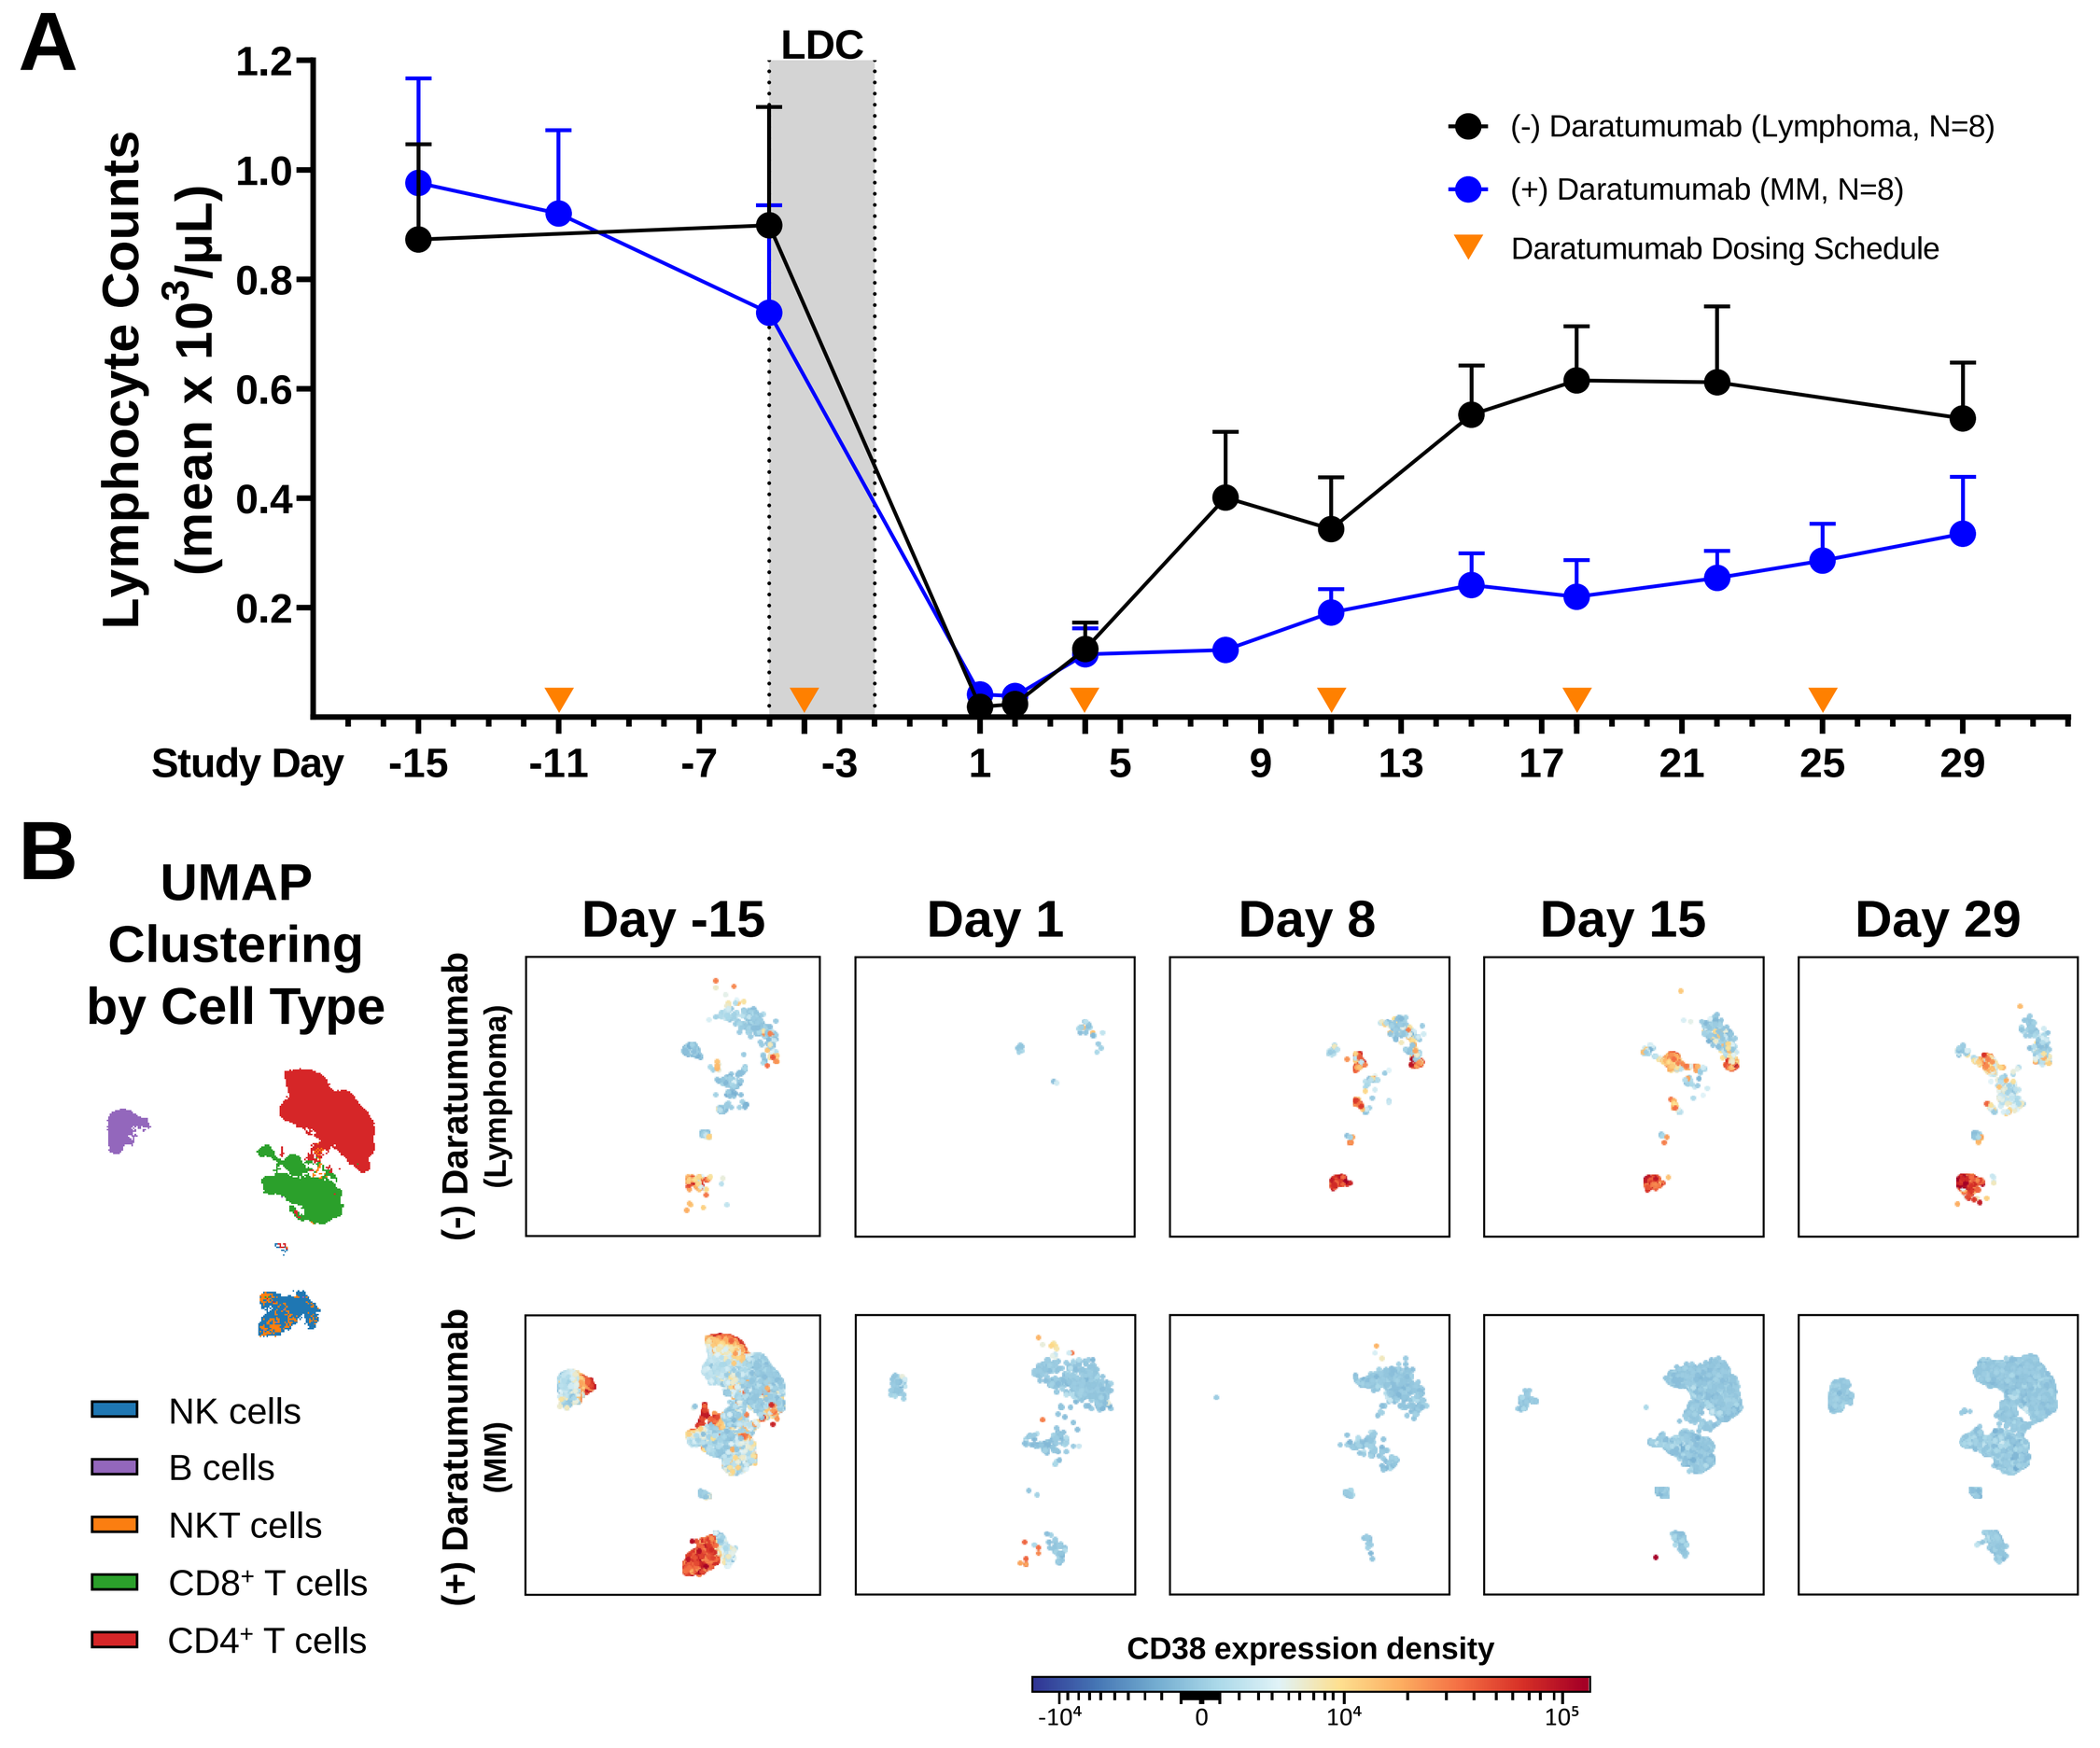
A
LDC
Lymphocyte Counts (mean x 10³/µL)
(-) Daratumumab (Lymphoma, N=8)
(+) Daratumumab (MM, N=8)
Daratumumab Dosing Schedule
Study Day
B
UMAP Clustering by Cell Type
Day -15
Day 1
Day 8
Day 15
Day 29
(-) Daratumumab (Lymphoma)
(+) Daratumumab (MM)
NK cells
B cells
NKT cells
CD8+ T cells
CD4+ T cells
CD38 expression density

Figure 6 ***Addition of daratumumab to lympho-depleting chemotherapy (LDC) delays host immune reconstitution.*** (A) Absolute lymphocyte profiles from lymphoma patients treated with FT516 in combination with rituximab (NCT04023071) and multiple myeloma (MM) patients treated with FT538 in combination with daratumumab (NCT04714372). The addition of daratumumab (▼) to FT538 reduces lymphocyte recovery post-LDC in MM patients. In contrast, lymphocyte recovery was faster and greater in the FT516 lymphoma patients who did not receive daratumumab. (B) Representative Uniform Manifold Approximation and Projection (UMAP) visualization of lymphocytes from a lymphoma patient with no daratumumab and a MM patient who received daratumumab prior to and weekly after LDC. The overlaid data files from each timepoint illustrate clustering of different cell types by color. Individual UMAP visualizations by timepoint demonstrate CD38 expression on endogenous CD4$^+$ T cells, CD8$^+$ T cells, B cells, and NK cells at screening. CD38 expressing T and NK cells recover fully in the lymphoma patient by Day 8, while majority of CD38$^+$ cells are eliminated in the MM patient and do not recover during the treatment.