Off-the-Shelf iPSC-derived CAR-T cells Containing Seven Functional Edits Overcome Antigen Heterogeneity, Improve Trafficking, and Withstand Immunosuppression Associated with Failed Tumor Treatment

Martin Hosking, Soheila Shirinbak, Bishwas Shrestha, Joy Grant, Kyla Omilusik, Hannah Keegan, Demetrio Cardenas, Angela Gentile, Shilpi Chandra, Loraine Loter, Lexe Linderhof, Nicholas Brookhouser, Stephanie Kennedy, Francisco Martinez, Loraine Campanati, Chris Ecker, Xu Yuan, Karina Palomares, Yi-Shin Lai, Lauren Fong, Yijia Pan, Shohreh Sikaroodi, Mark Jelcic, Philip Chu, Amit Mehta, Layton Smith, Eigen Peralta, Tom Lee, Ramzey Abujarour, Raedun Clarke, Bahram Valamehr

Fate Therapeutics, Inc., San Diego, CA, USA

Introduction

Although chimeric antigen receptor (CAR) T-cell therapy has shown remarkable efficacy in liquid tumors, its wider application to solid tumors has been hampered by tumor-associated antigen (TAA) heterogeneity, inefficient CAR-T cell trafficking to the tumor, and immunosuppression inherent to the tumor microenvironment. Moreover, the often-dysfunctional and heterogenous yield of highly-edited (e.g. >2 transgenes) primary, donor-derived CAR-T cells necessary to address these obstacles has limited their efficacy and wider clinical investigation.

T-cell derived induced pluripotent stem cells (TiPSCs) were engineered to express a CAR targeting a novel TAA domain and an interleukin-7 receptor fusion protein (IL7RF) under the regulation of the T-cell receptor alpha chain constant locus. Additionally, these TiPSCs were engineered to express TGFβ-signal redirection receptor (TGFβ-SRR), high-affinity non-cleavable CD16A (hnCD16), and CXCR2 within the CD38 locus enabling TGFβ resistance, secondary TAA targeting via antibody-dependent cell cytotoxicity (ADCC), and solid tumor specific trafficking, respectively. Engineered TiPSCs were differentiated into alpha-beta T (iT) cells, uniformly expressing all engineered transgenes and completely lacking both CD38 and T-cell receptor expression, avoiding the potential risk of graft-versus-host disease in an allogeneic setting (**Fig 1**).

Summary of results:

- ✓ Mitigation of tumor associated antigen heterogeneity and escape with potent and sustained CAR and CAR/hnCD16 mediated anti-tumor efficacy (**Fig 2 & 3**)
- ✓ Resistance to TGFβ-dependent suppression of effector function (**Fig 4**)
- ✓ Functional migration to CXCR2 ligands enriched within solid tumors (**Fig 5**)
- ✓ Potent *in vivo* efficacy of engineered CAR iT cells either as a monotherapy or in combination with therapeutic antibodies and *in vivo* hnCD16 activation (**Fig 6**)

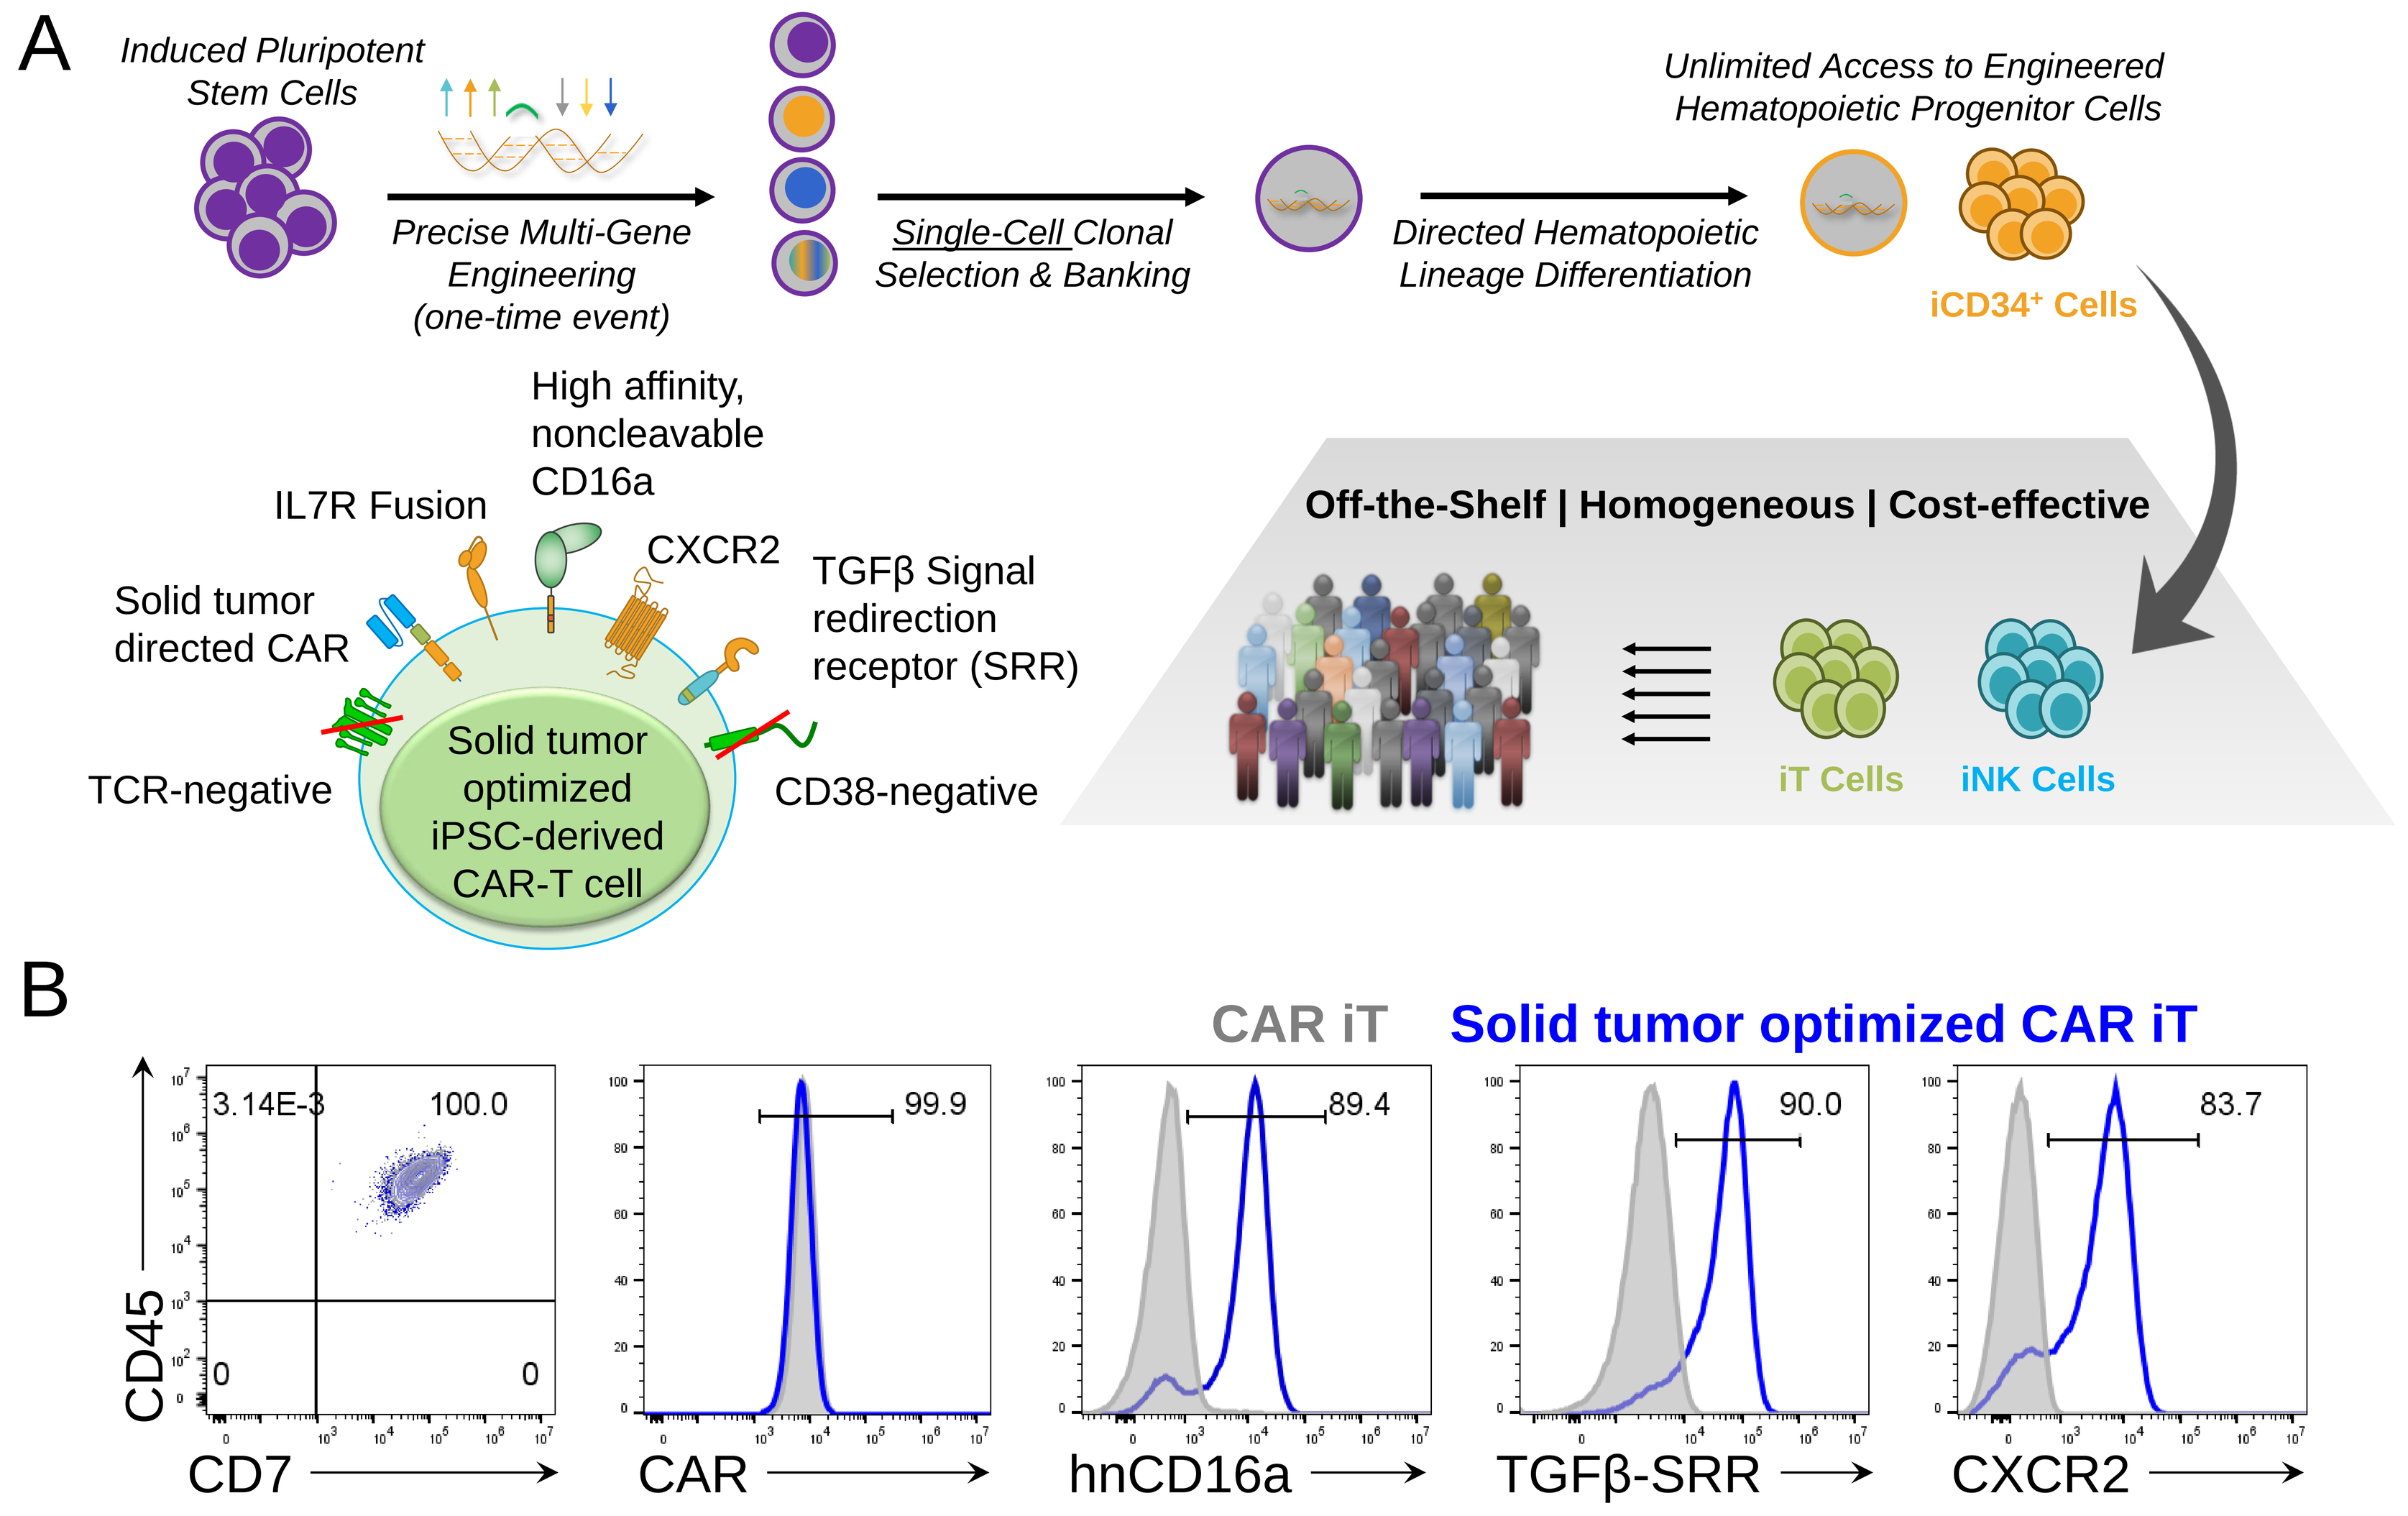
A
Induced Pluripotent Stem Cells
Precise Multi-Gene Engineering (one-time event)
Single-Cell Clonal Selection & Banking
Directed Hematopoietic Lineage Differentiation
Unlimited Access to Engineered Hematopoietic Progenitor Cells
iCD34+ Cells
High affinity, noncleavable CD16a
IL7R Fusion
CXCR2
Solid tumor directed CAR
TGFβ Signal redirection receptor (SRR)
TCR-negative
Solid tumor optimized iPSC-derived CAR-T cell
CD38-negative
Off-the-Shelf | Homogeneous | Cost-effective
iT Cells
iNK Cells
B
CAR iT
Solid tumor optimized CAR iT
3.14E-3
100.0
0
0
99.9
89.4
90.0
83.7
CD45
CD7
CAR
hnCD16a
TGFβ-SRR
CXCR2

Figure 1. Generation of off-the shelf iPSC derived CAR-T cells engineered for enhanced efficacy in solid tumors. (A) Overview of Fate's proprietary IPSC platform for the generation of precisely and uniformly engineered effector NK and/or T cells containing at total of seven functional edits tailored for solid tumor efficacy. (B) Representative flow cytometry demonstrating lymphoid commitment ($CD45^{High}CD7^{High}$) and high and homogenous CAR, hnCD16, TGFβ-SRR, and CXCR2 expression.

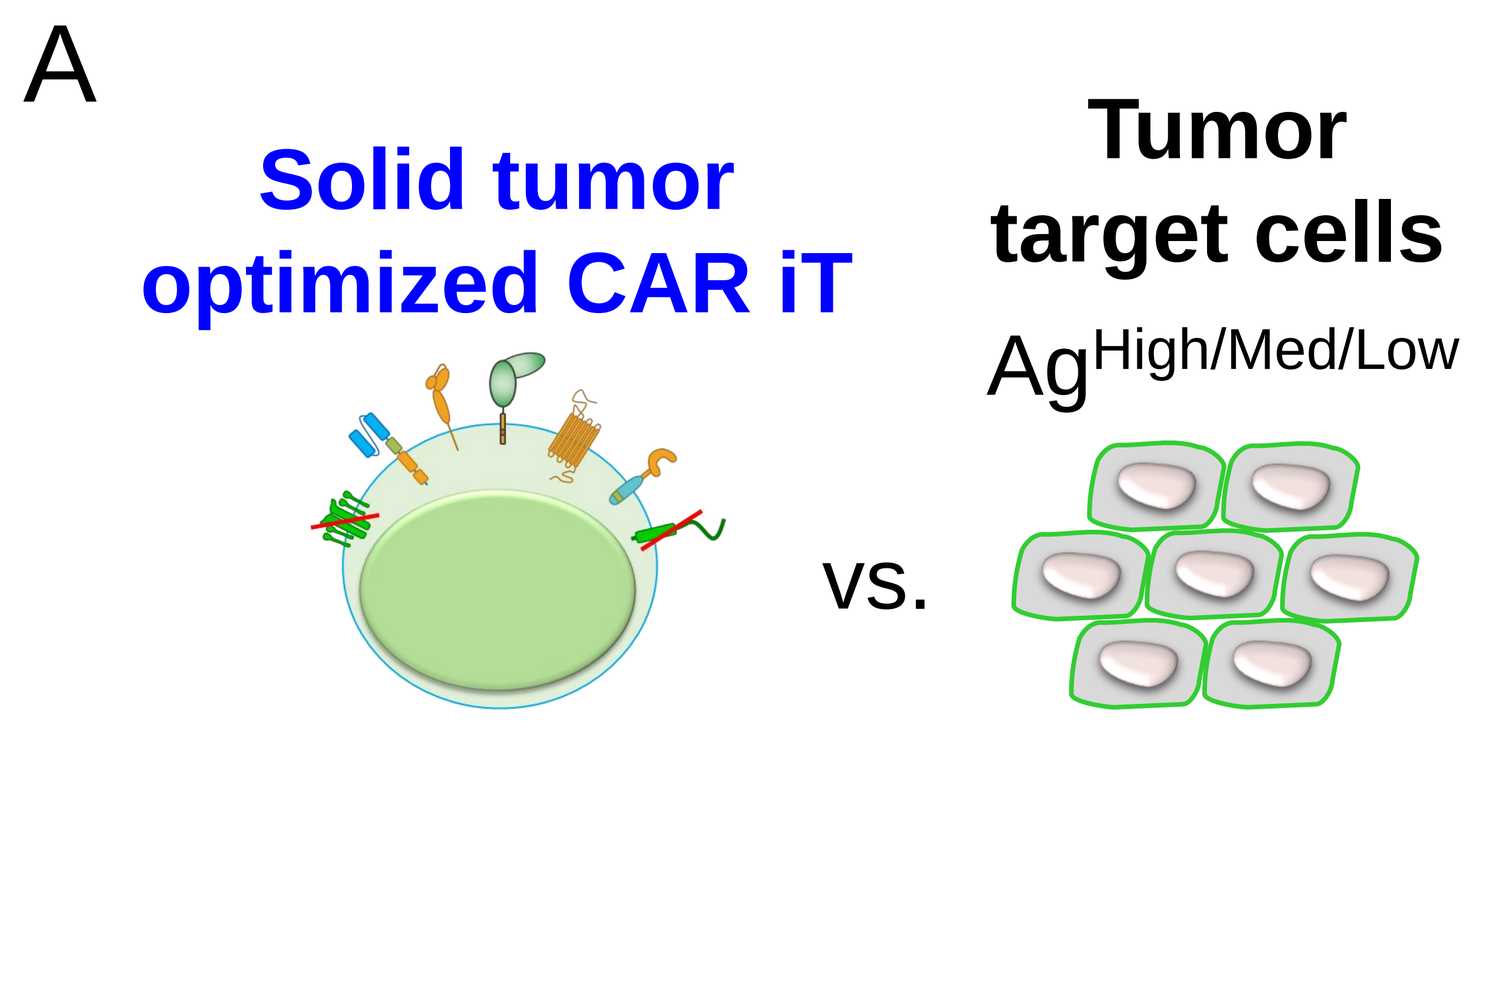
A
Solid tumor optimized CAR iT
vs.
Tumor target cells
Ag High/Med/Low

Figure 2. Robust CAR-dependent anti-tumor efficacy across multiple indications and antigen levels. (A) Experimental system to evaluate CAR-mediated efficacy of solid tumor optimized CAR iT cells against tumor targets from multiple solid tumor indications and with varying CAR antigen (1º TAA) levels. (B) The cytolytic efficacy of CAR iTs at a 2:1 Effector:Target in xCELLigence assay, demonstrating robust efficacy on antigen diverse targets from multiple tumor indications.

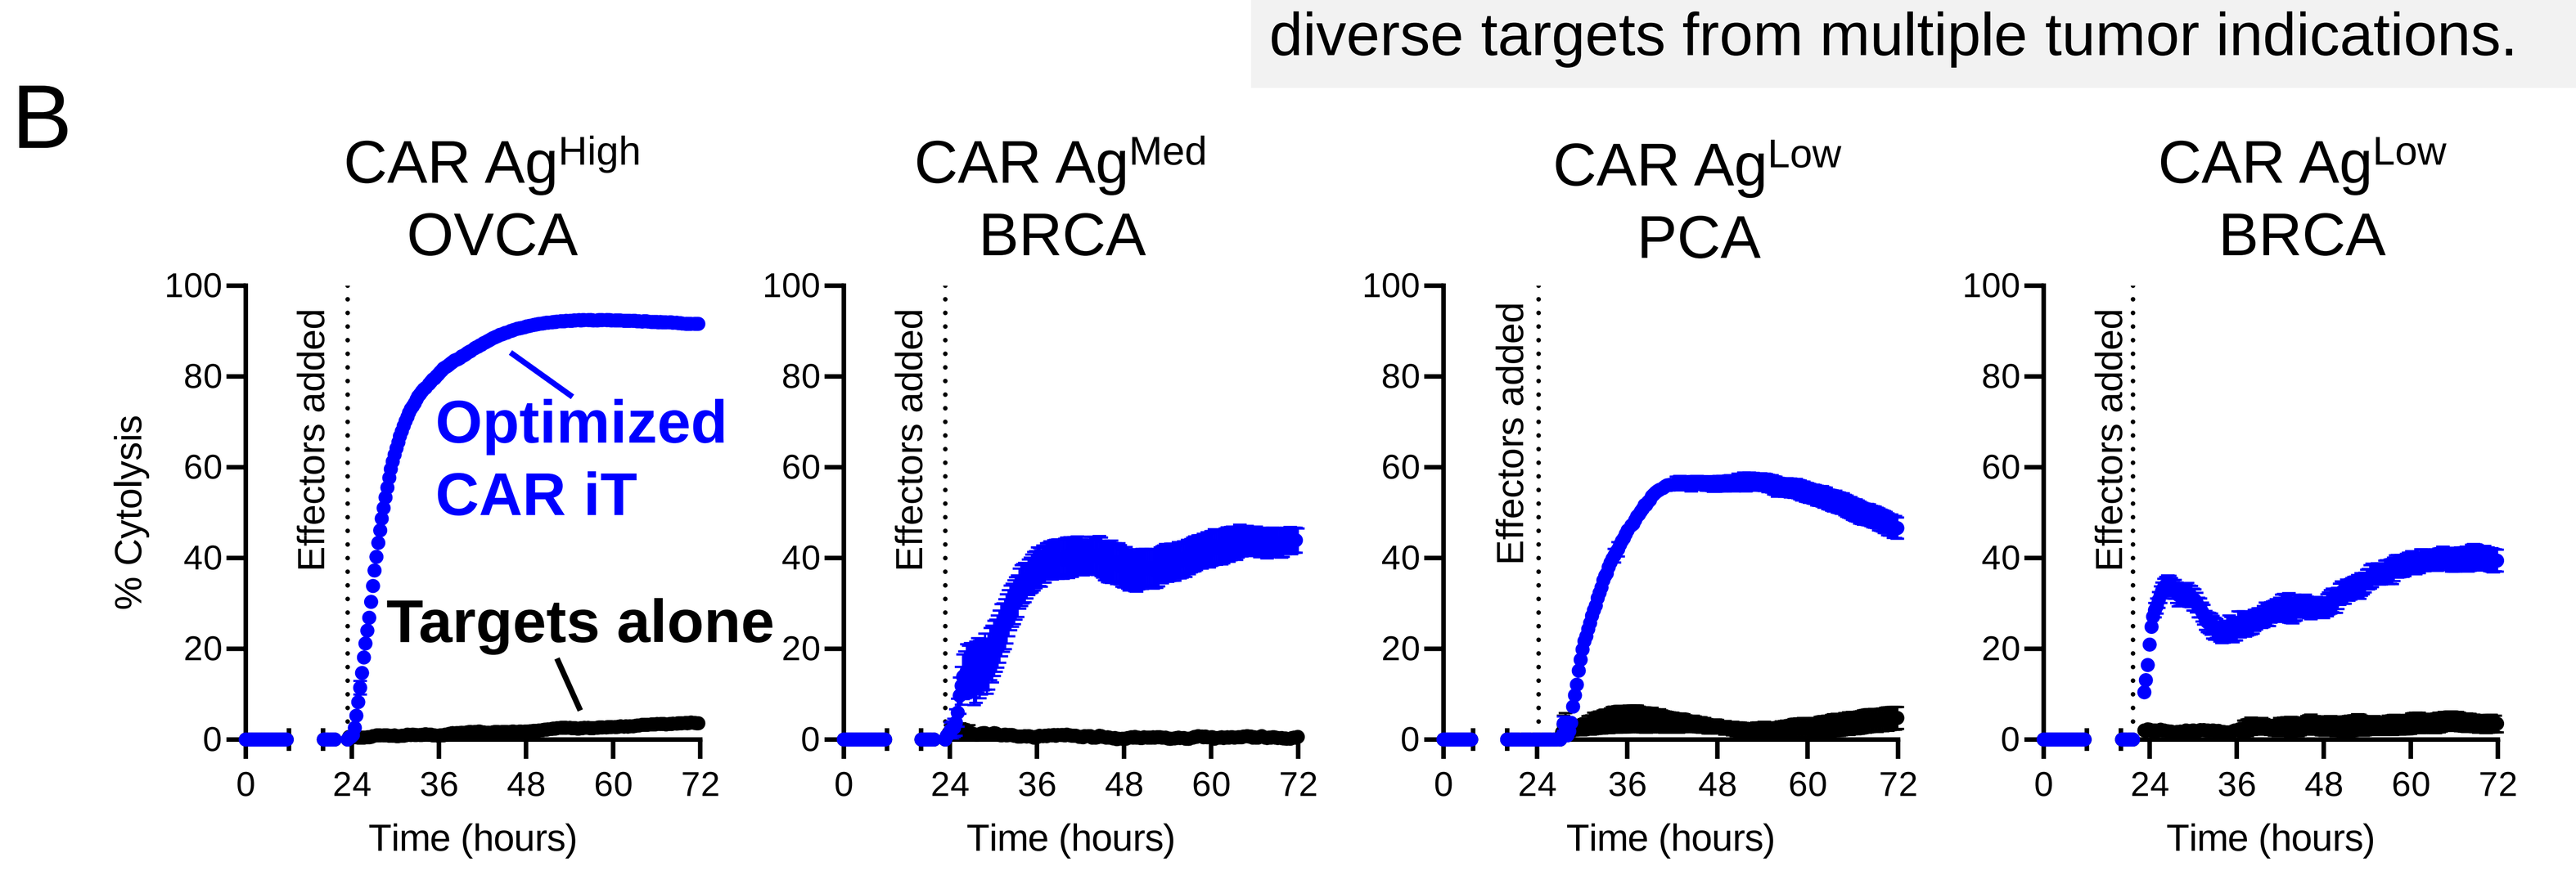
B
CAR Ag High
OVCA
CAR Ag Med
BRCA
CAR Ag Low
PCA
CAR Ag Low
BRCA
% Cytolysis
Effectors added
Optimized CAR iT
Targets alone
Time (hours)

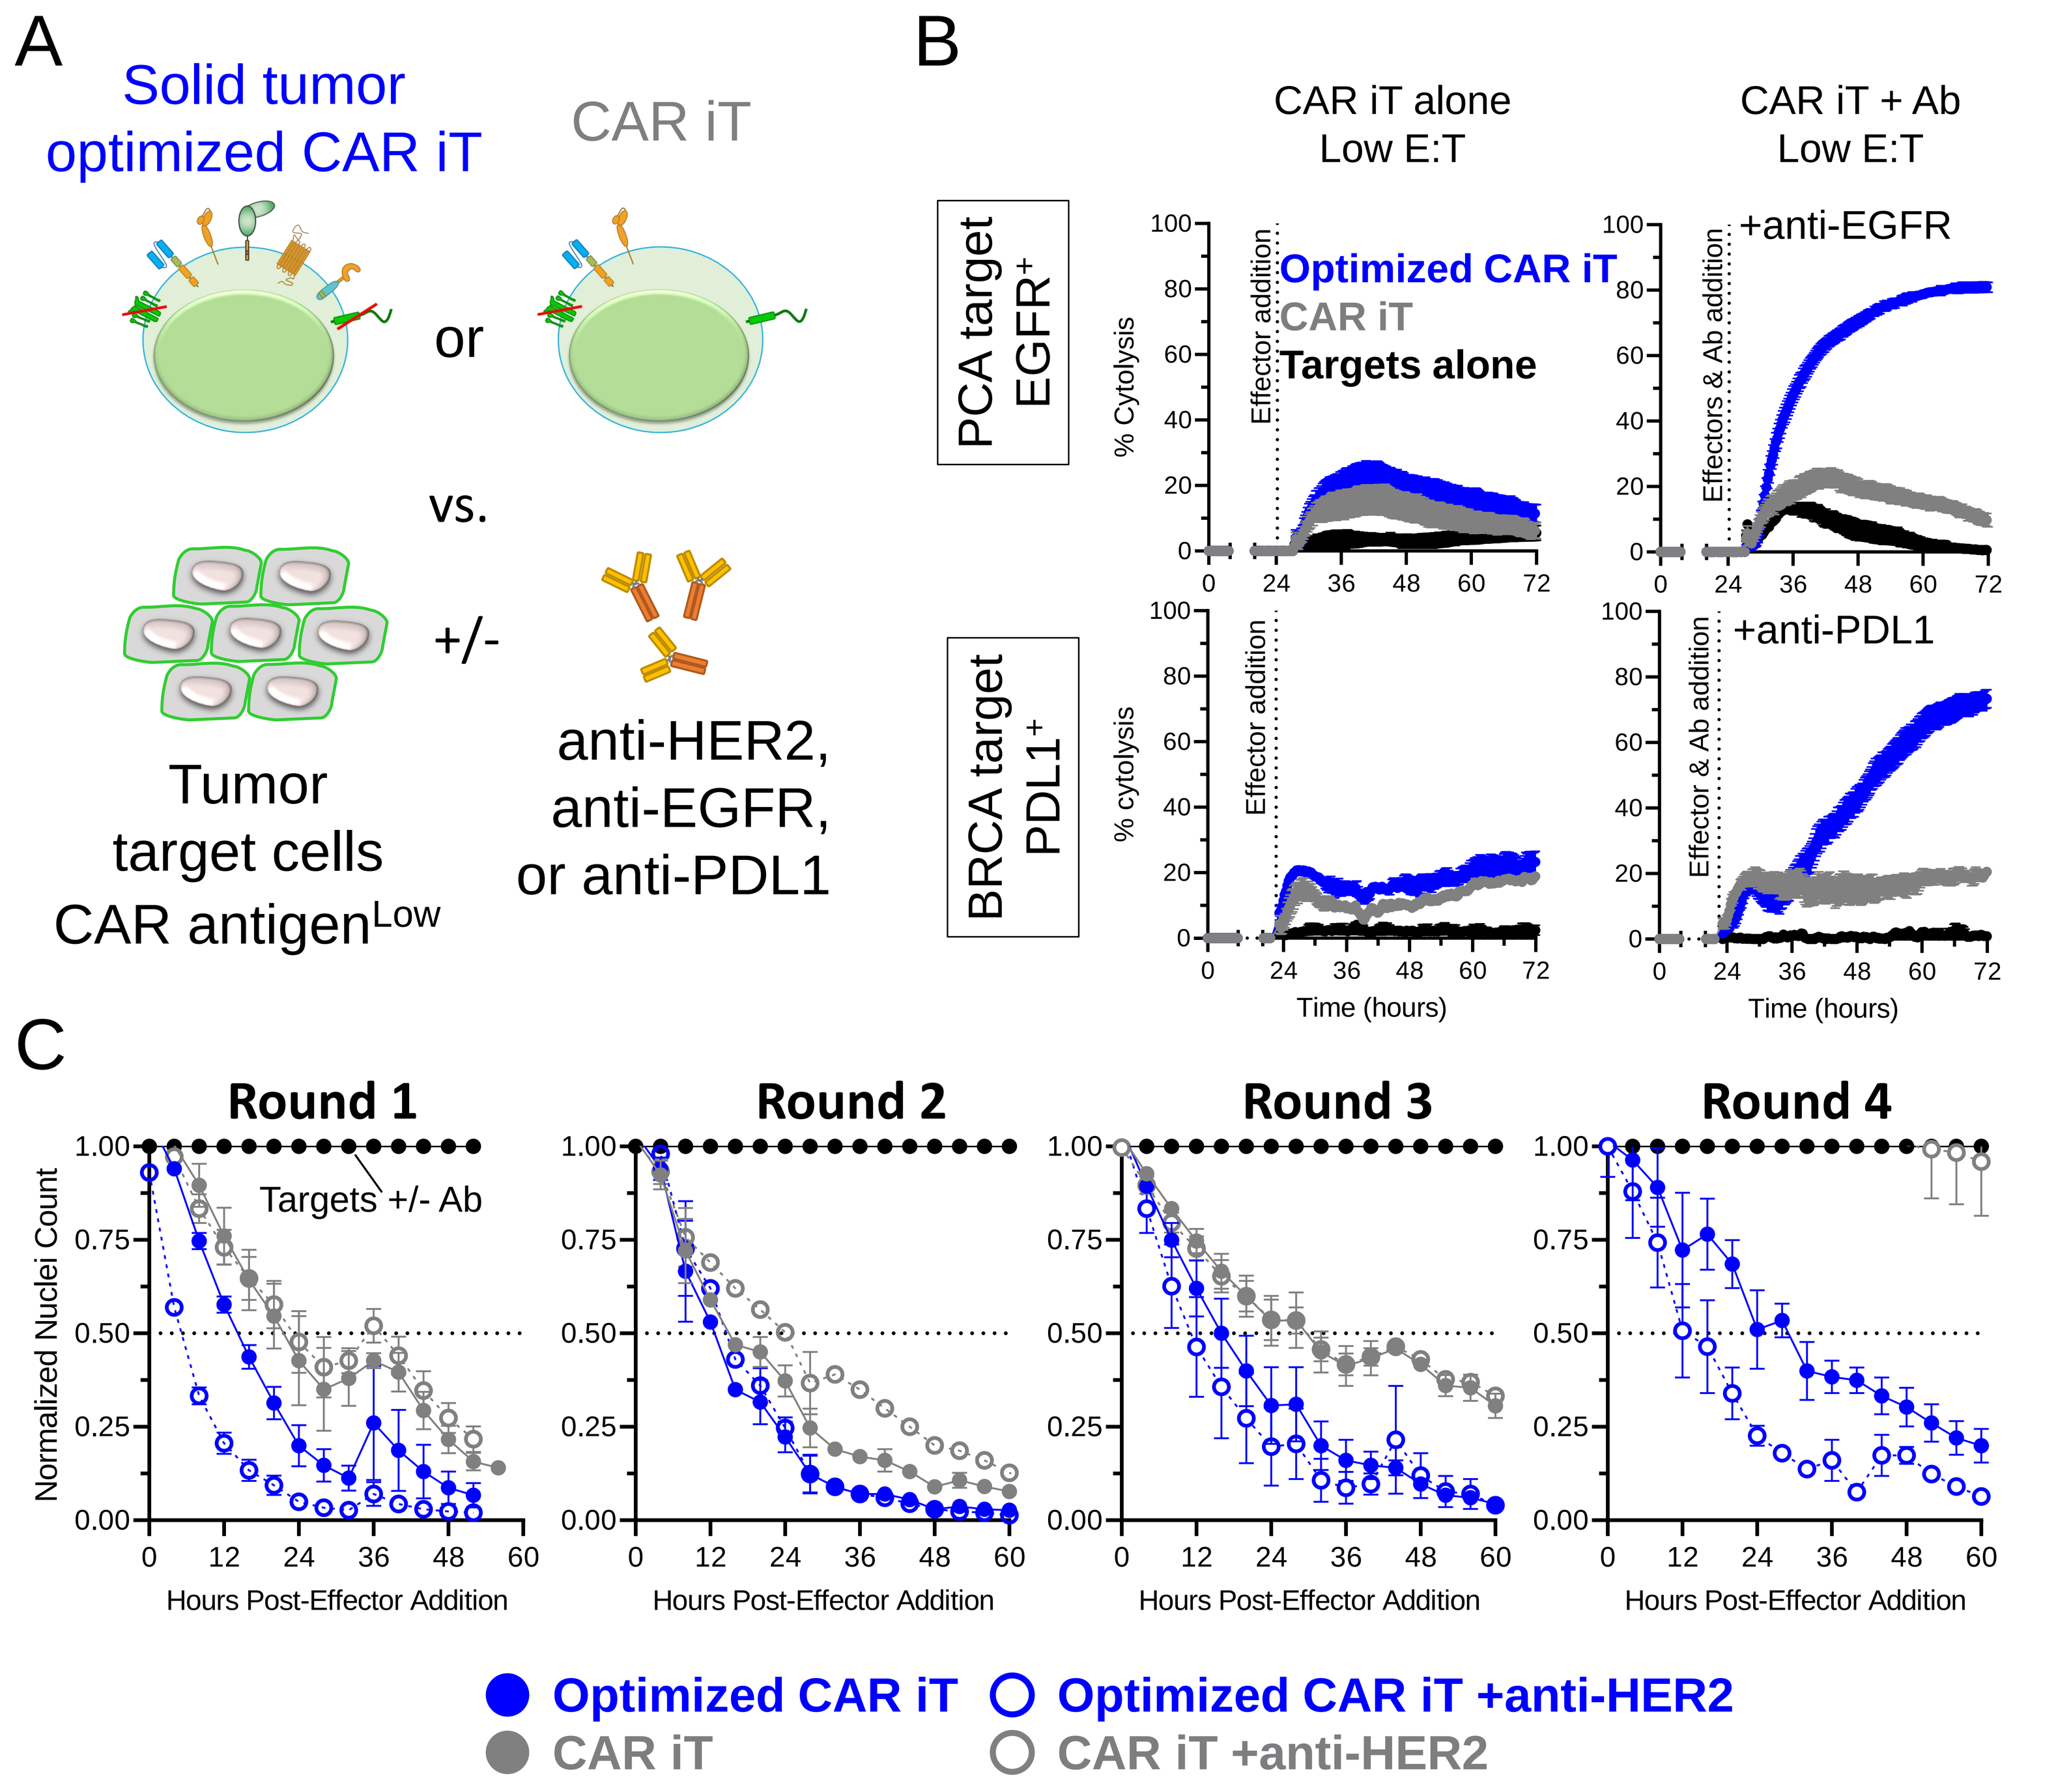
A
Solid tumor optimized CAR iT
CAR iT
or
vs.
+/-
Tumor target cells CAR antigen Low
anti-HER2, anti-EGFR, or anti-PDL1
B
CAR iT alone Low E:T
CAR iT + Ab Low E:T
PCA target EGFR+
BRCA target PDL1+
Optimized CAR iT
CAR iT
Targets alone
+anti-EGFR
+anti-PDL1
Effector addition
Effectors & Ab addition
Effector & Ab addition
% Cytolysis
Time (hours)
C
Round 1
Round 2
Round 3
Round 4
Targets +/- Ab
Normalized Nuclei Count
Hours Post-Effector Addition
Optimized CAR iT
Optimized CAR iT +anti-HER2
CAR iT
CAR iT +anti-HER2

Figure 3. CAR mediated anti-tumor efficacy is seen over multiple rounds of tumor challenge and is enhanced with hnCD16 activation and therapeutic antibodies. (A) Experimental overview to evaluate CAR +/- hnCD16 coactivation - mediated efficacy of solid tumor optimized CAR iT cells against tumor targets with varying CAR antigen and 2º TAA levels. (B) The cytolytic efficacy of CAR iTs and solid tumor optimized CAR iTs at [1:1] (top row) or [1:2] (bottom row) [Effector:Target] ratio +/- therapeutic antibody on CAR Ag^{Low} tumor targets was determined via xCELLigence assay. (C) The cytolytic efficacy of CAR iTs (Gray) and solid tumor optimized CAR iTs (Blue) [1:1] +/- therapeutic antibody was assessed via Incucyte assay with an effector:target reset of fresh tumor targets at every round.

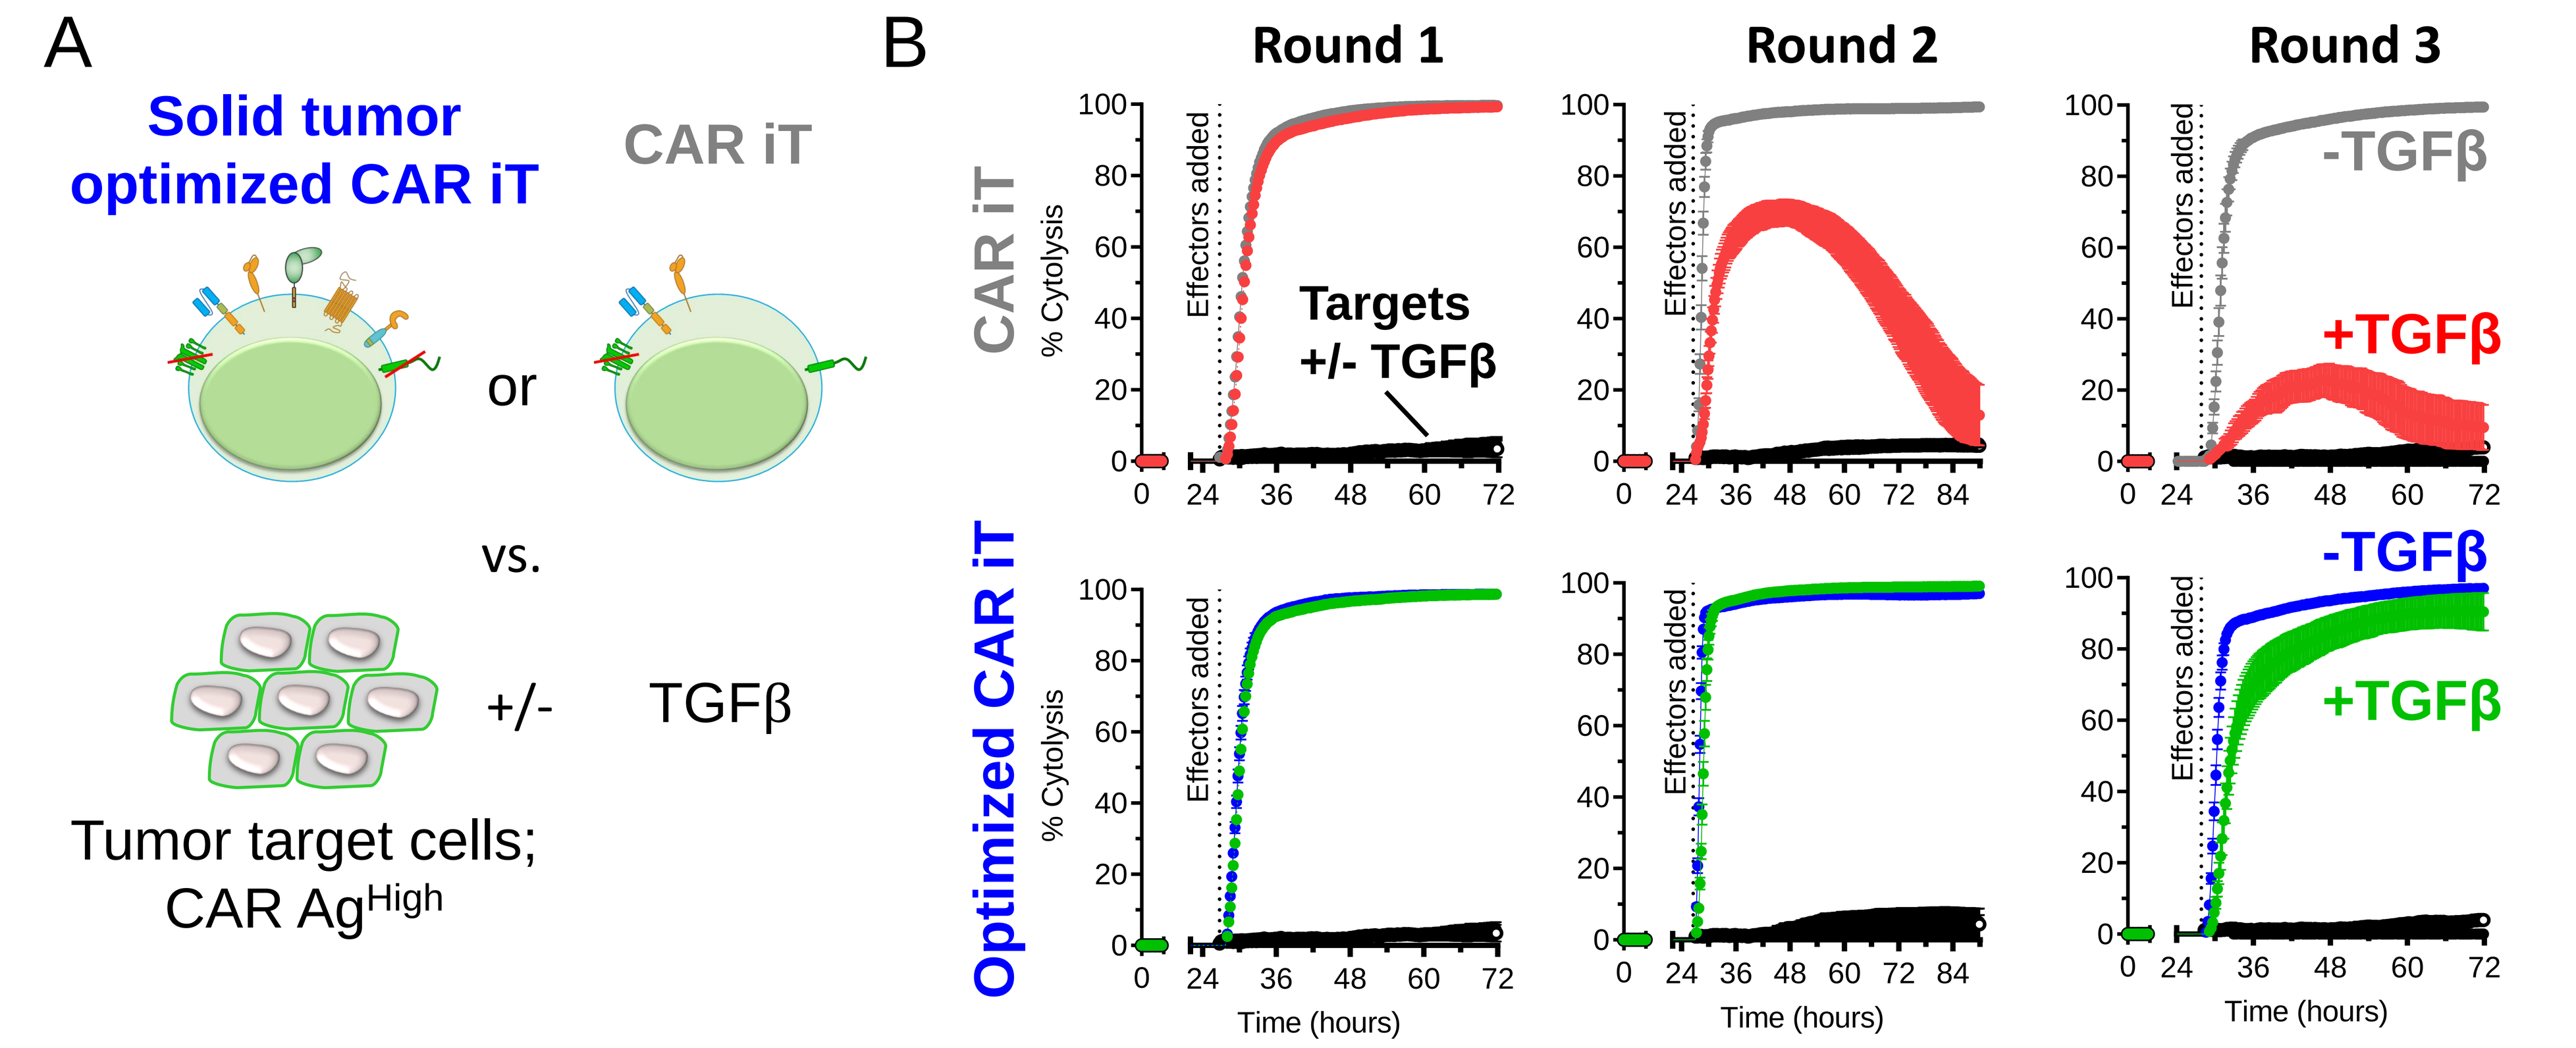
A
Solid tumor optimized CAR iT
CAR iT
or
vs.
+/-
TGFβ
Tumor target cells; CAR Ag High
B
Round 1
Round 2
Round 3
CAR iT
Optimized CAR iT
% Cytolysis
Effectors added
Targets +/- TGFβ
-TGFβ
+TGFβ
Time (hours)

Figure 4. Resistance to TGFβ – mediated effector suppression via TGFβ-SRR over multiple rounds of tumor challenge. (A) Experimental overview to demonstrate resistance to TGFβ – mediated effector suppression of solid tumor optimized CAR iT cells over multiple rounds of tumor challenge. (B) CAR iT (top row) and solid tumor optimized (bottom row) CAR iT cells were co-cultured with CAR Ag^{High} tumor target cells [2.5:1] +/- TGFβ for multiple rounds of tumor challenge. Cytolytic efficacy was evaluated via xCELLigence assay.

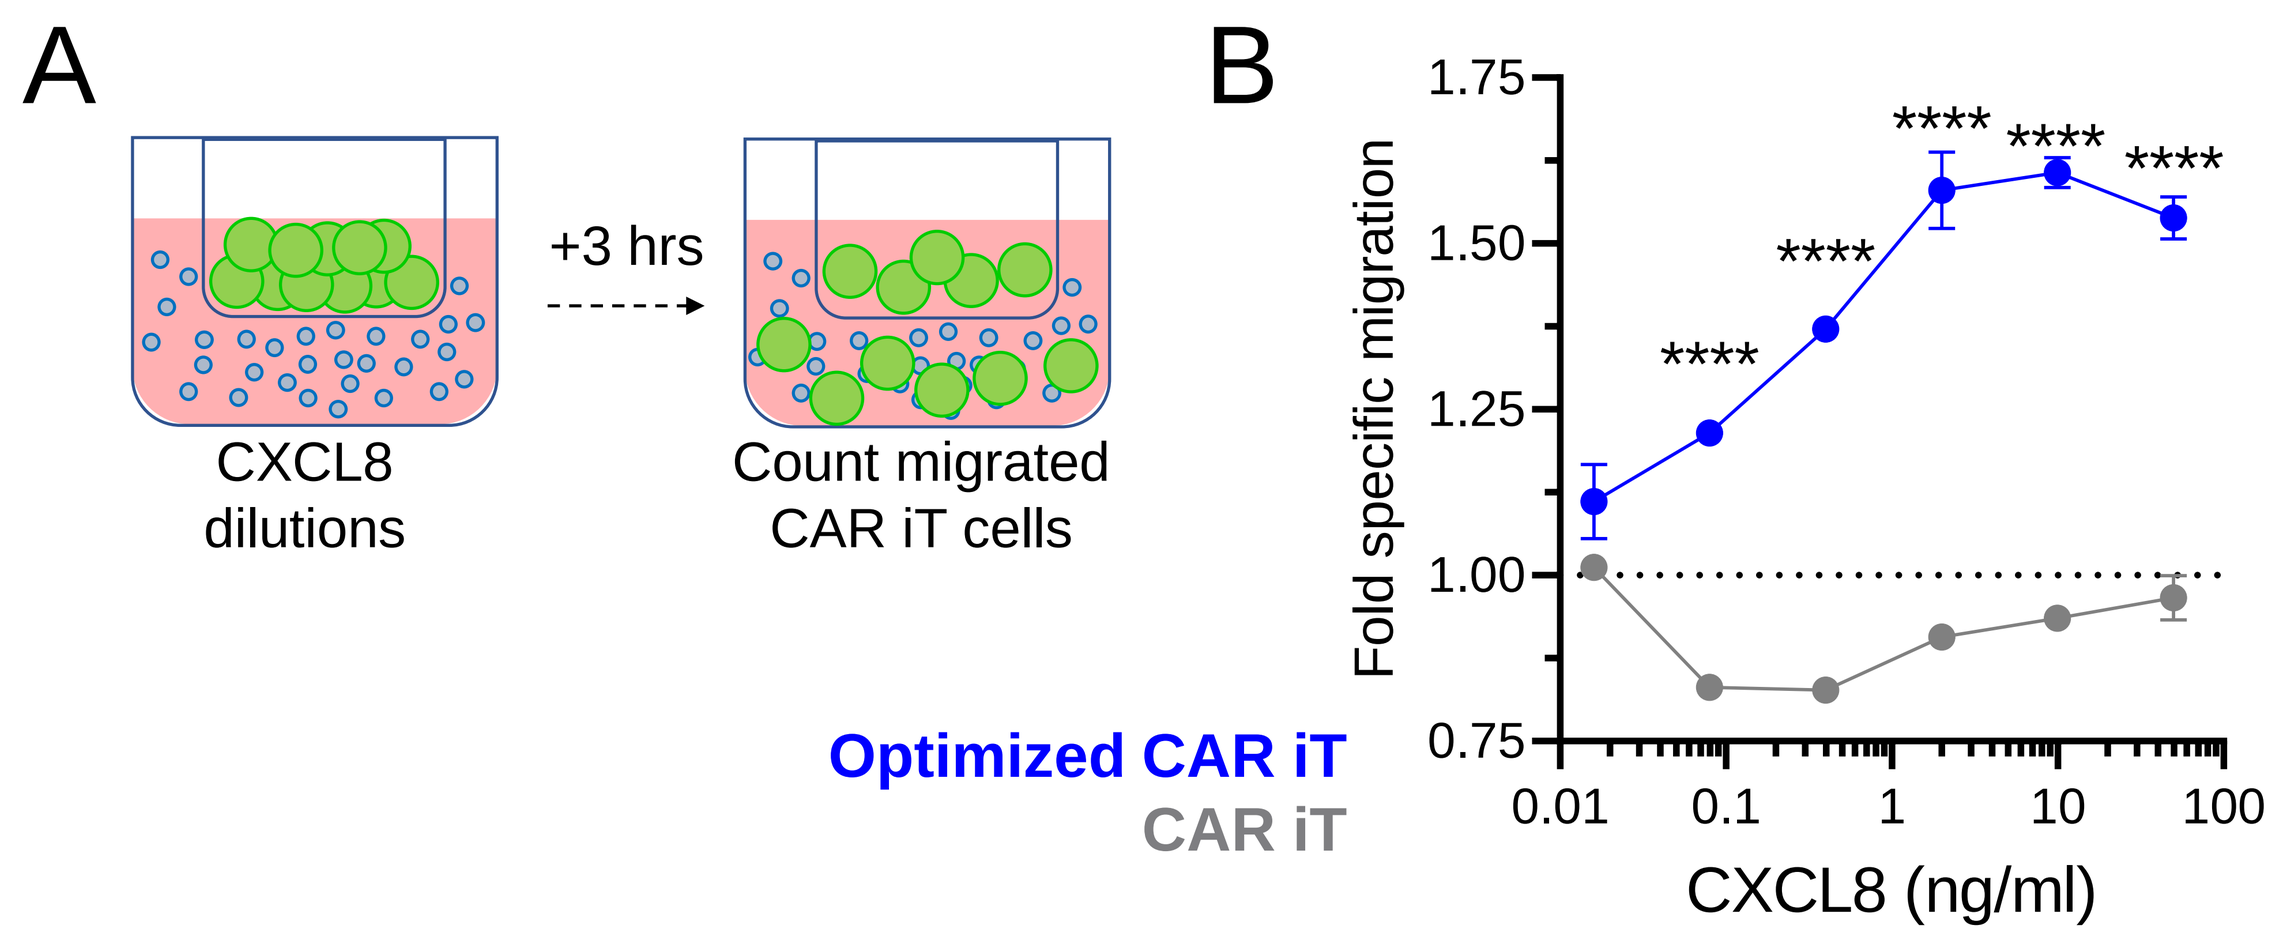
A
+3 hrs
CXCL8 dilutions
Count migrated CAR iT cells
B
Fold specific migration

CXCL8 (ng/ml)
Optimized CAR iT
CAR iT

Figure 5. CXCR2 expression enables CXCL8 – specific migration. (A) Transwell assay experimental overview to evaluate CXCR2 – dependent migration. (B) Transwell migration assays with CAR iT (gray) and solid tumor optimized CAR iT cells (blue) demonstrating specific and dose dependent migration by CXCR2+ optimized CAR iT cells to recombinant, human CXCL8.

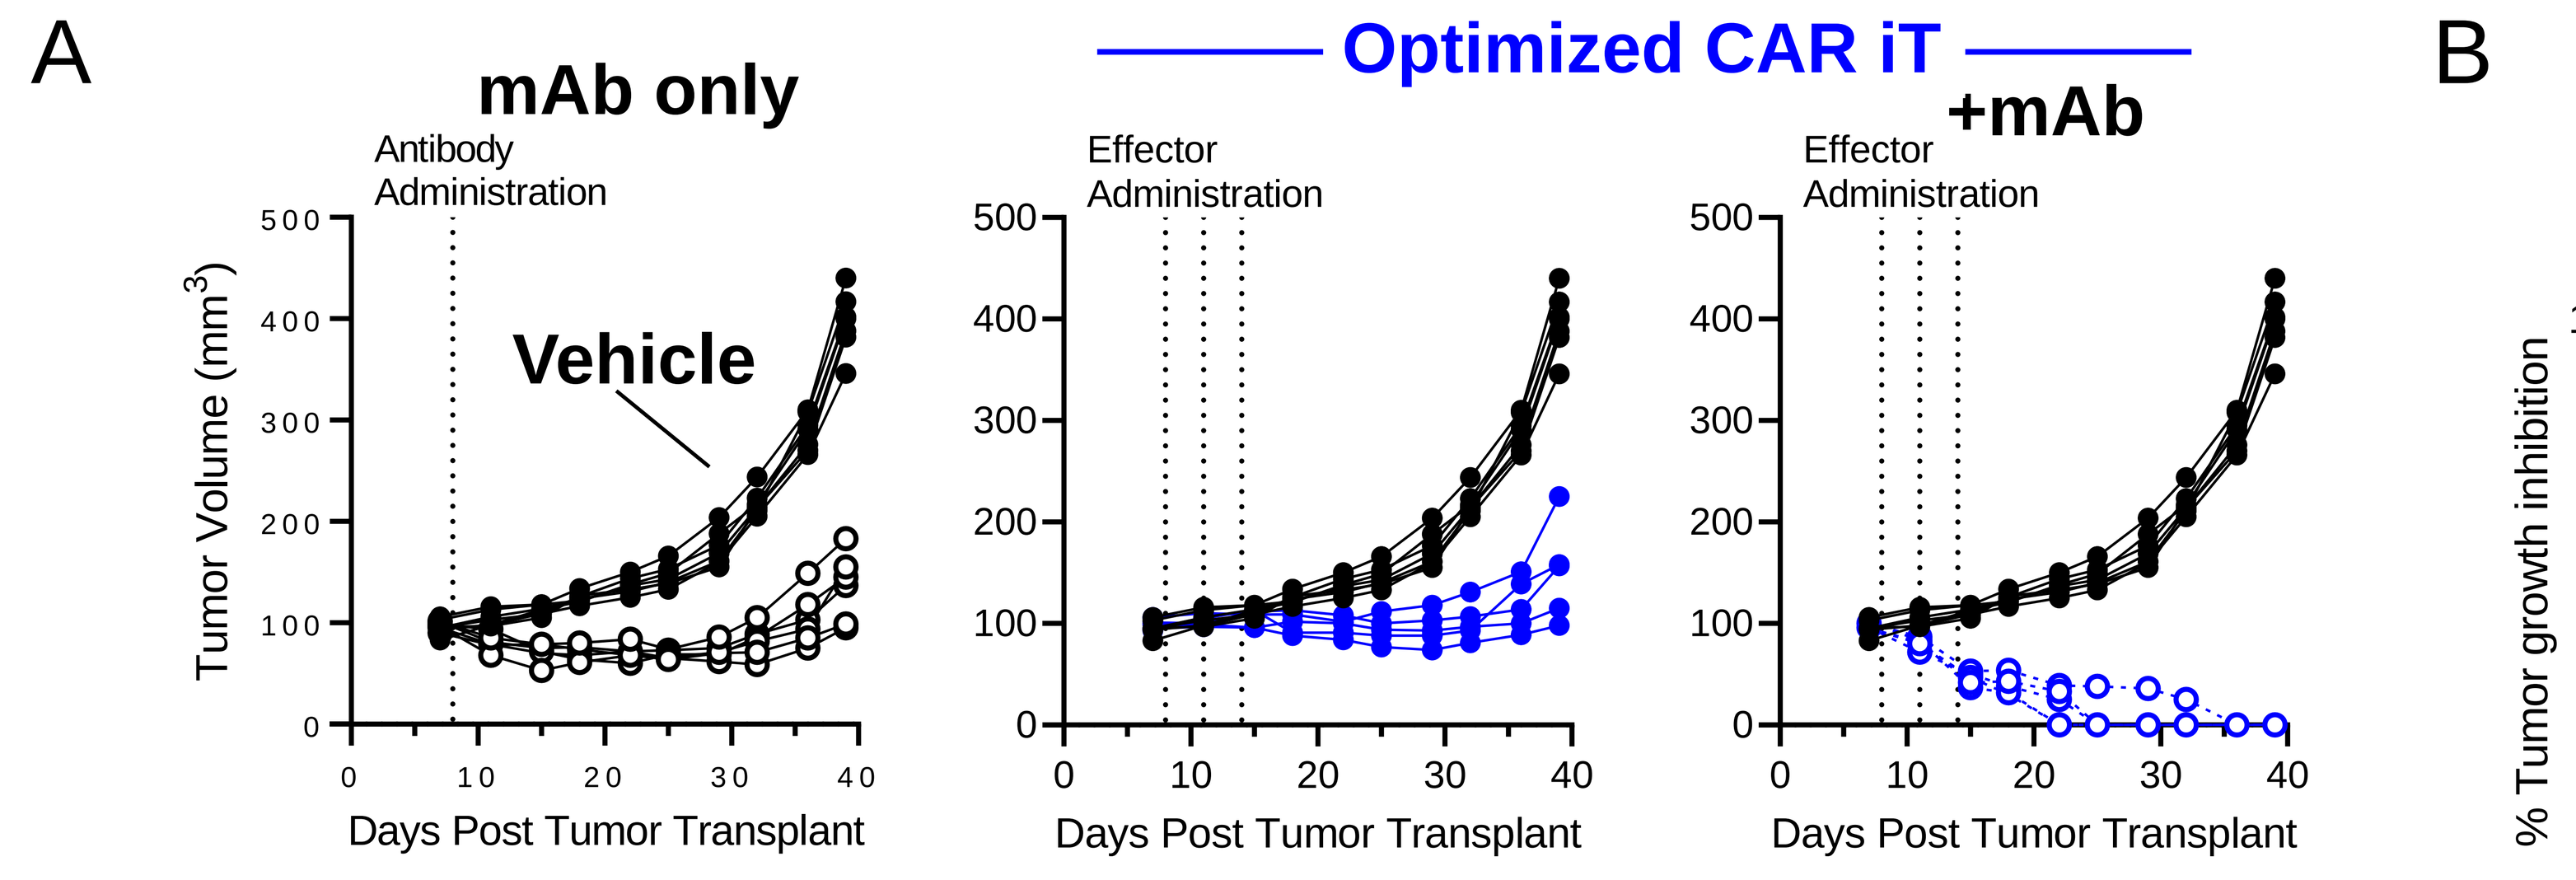
A
mAb only
Optimized CAR iT
+mAb
Antibody Administration
Effector Administration
Vehicle
Tumor Volume (mm3)
Days Post Tumor Transplant

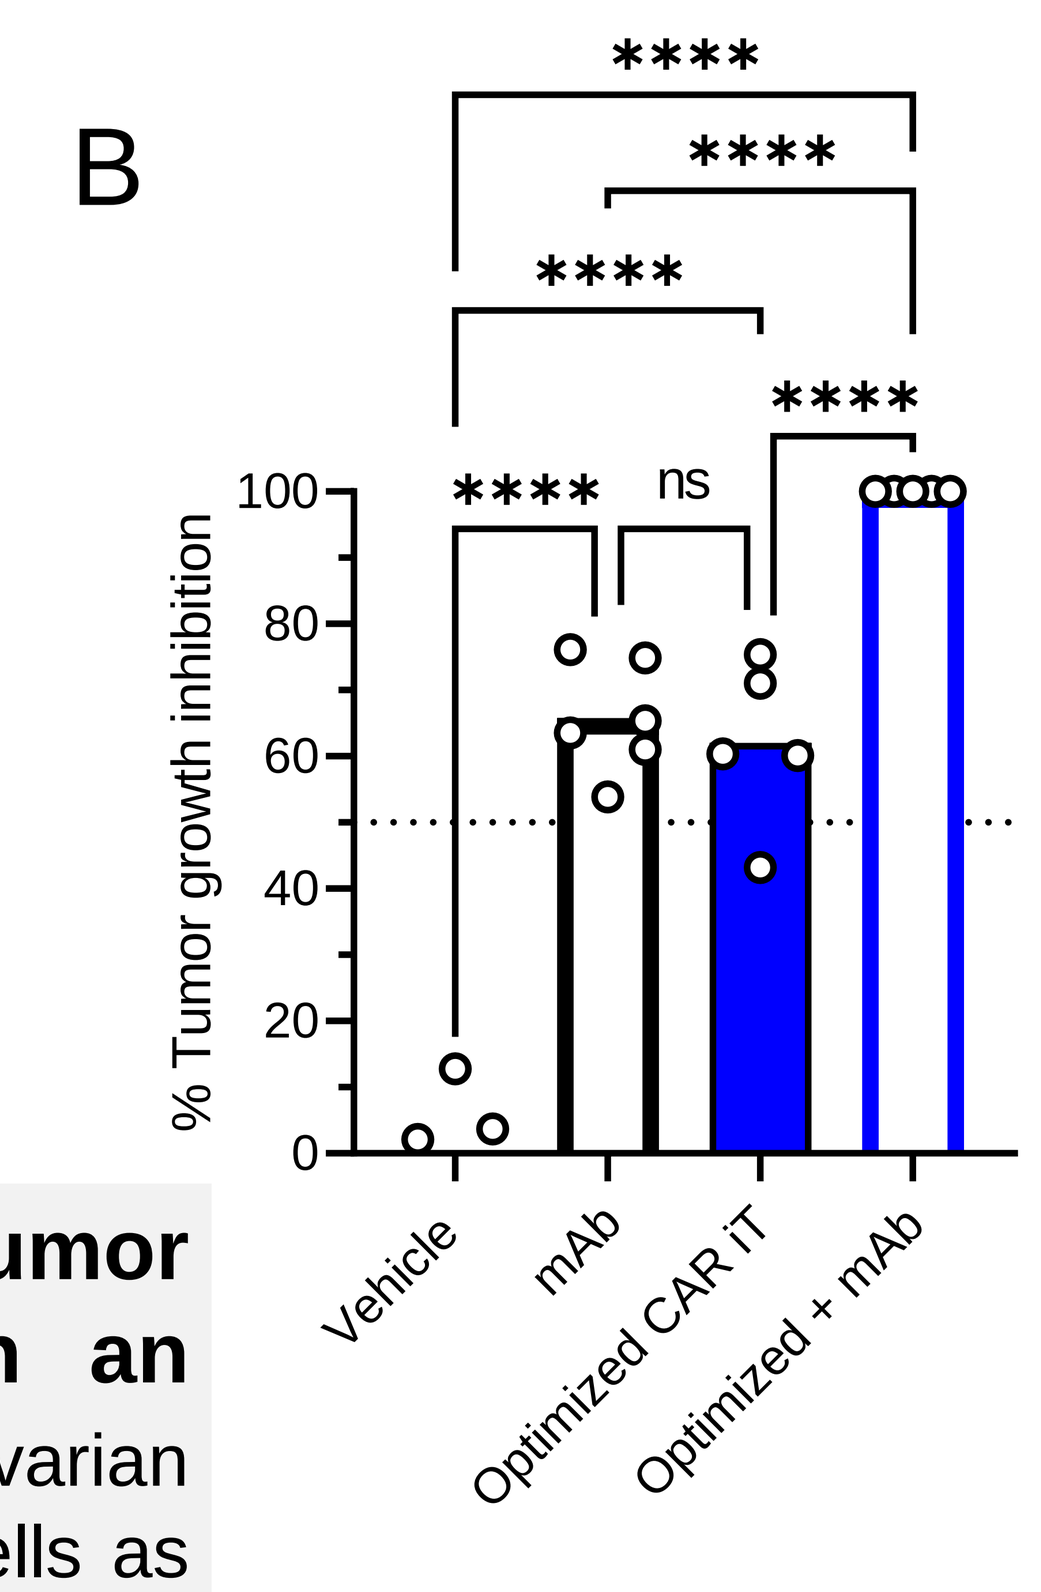
B

ns
% Tumor growth inhibition
Vehicle
mAb
Optimized CAR iT
Optimized + mAb

Figure 6. Solid tumor optimized CAR iT cells show enhanced anti-tumor efficacy in vivo by CAR and CAR/hnCD16 complementation in an aggressive ovarian xenograft model. (A) NSG mice bearing established ovarian xenografts were treated with antibody and/or solid tumor optimized (blue) CAR iT cells as indicated. Tumor volume was recorded and is shown for each individual recipient. (B) Tumor growth inhibition was calculated at 39 days post tumor inoculation.

Conclusion

These results demonstrate that a multiplex-engineered CAR-iT cell product, uniquely manufactured to be available off-the-shelf, can be tailored specifically to overcome common barriers observed in solid tumors, including the ability to preferentially traffic to the tumor, to promote tumor microenvironment resistance, and to elicit potent and enhanced anti-tumor activity in both *in vitro* and *in vivo* settings.