Preclinical *in vivo* Model Development: Highlighting Success and Discussing Xenograft Advancements, a Step Closer in Predicting Patient Outcomes

Thomas Dailey, Jon Tuncel, Angela Gentile, Cara Bickers, Evelyn Diaz, Judy Martin, Steven Castro, Eric Sung, Alma Gutierrez, Jeffrey Chen, Melissandre Pache, Nancy Le, Brian Qua, Miguel Meza, Katie Restrepo, Soheila Shirinbak, Joy Grant, Martin Hosking, Peter Szabo, Layton H. Smith, Bahram Valamehr

Fate Therapeutics, Inc., San Diego, CA, USA

Introduction

Immunotherapy, in particular chimeric-antigen receptor (**CAR**) T cells, has been shown to be an effective strategy for the treatment of cancer. However, the full therapeutic potential of these innovative therapies has not yet been fulfilled. This is particularly true of complex solid tumors. In comparison with hematologic cancers, solid tumors are more complex with unique three-dimensional structures, an immunosuppressive microenvironment, and cellular heterogeneity that extends to the expression of tumor associated antigens. Therefore, robust preclinical models with a high degree of fidelity towards the clinical presentation of the tumor are required for the accurate evaluation and translation of next generation immunotherapies, including advancement of *in vivo* xenograft mouse models that more closely recapitulate disease as it is observed in the clinical setting.

Figure 1: Fate Therapeutics induced pluripotent stem cell (iPSC) product platform enables mass production of off-the-shelf, engineered, homogeneous cell products that can be administered with multiple doses to deliver more effective pharmacologic activity, including in combination with cycles of other cancer treatments.

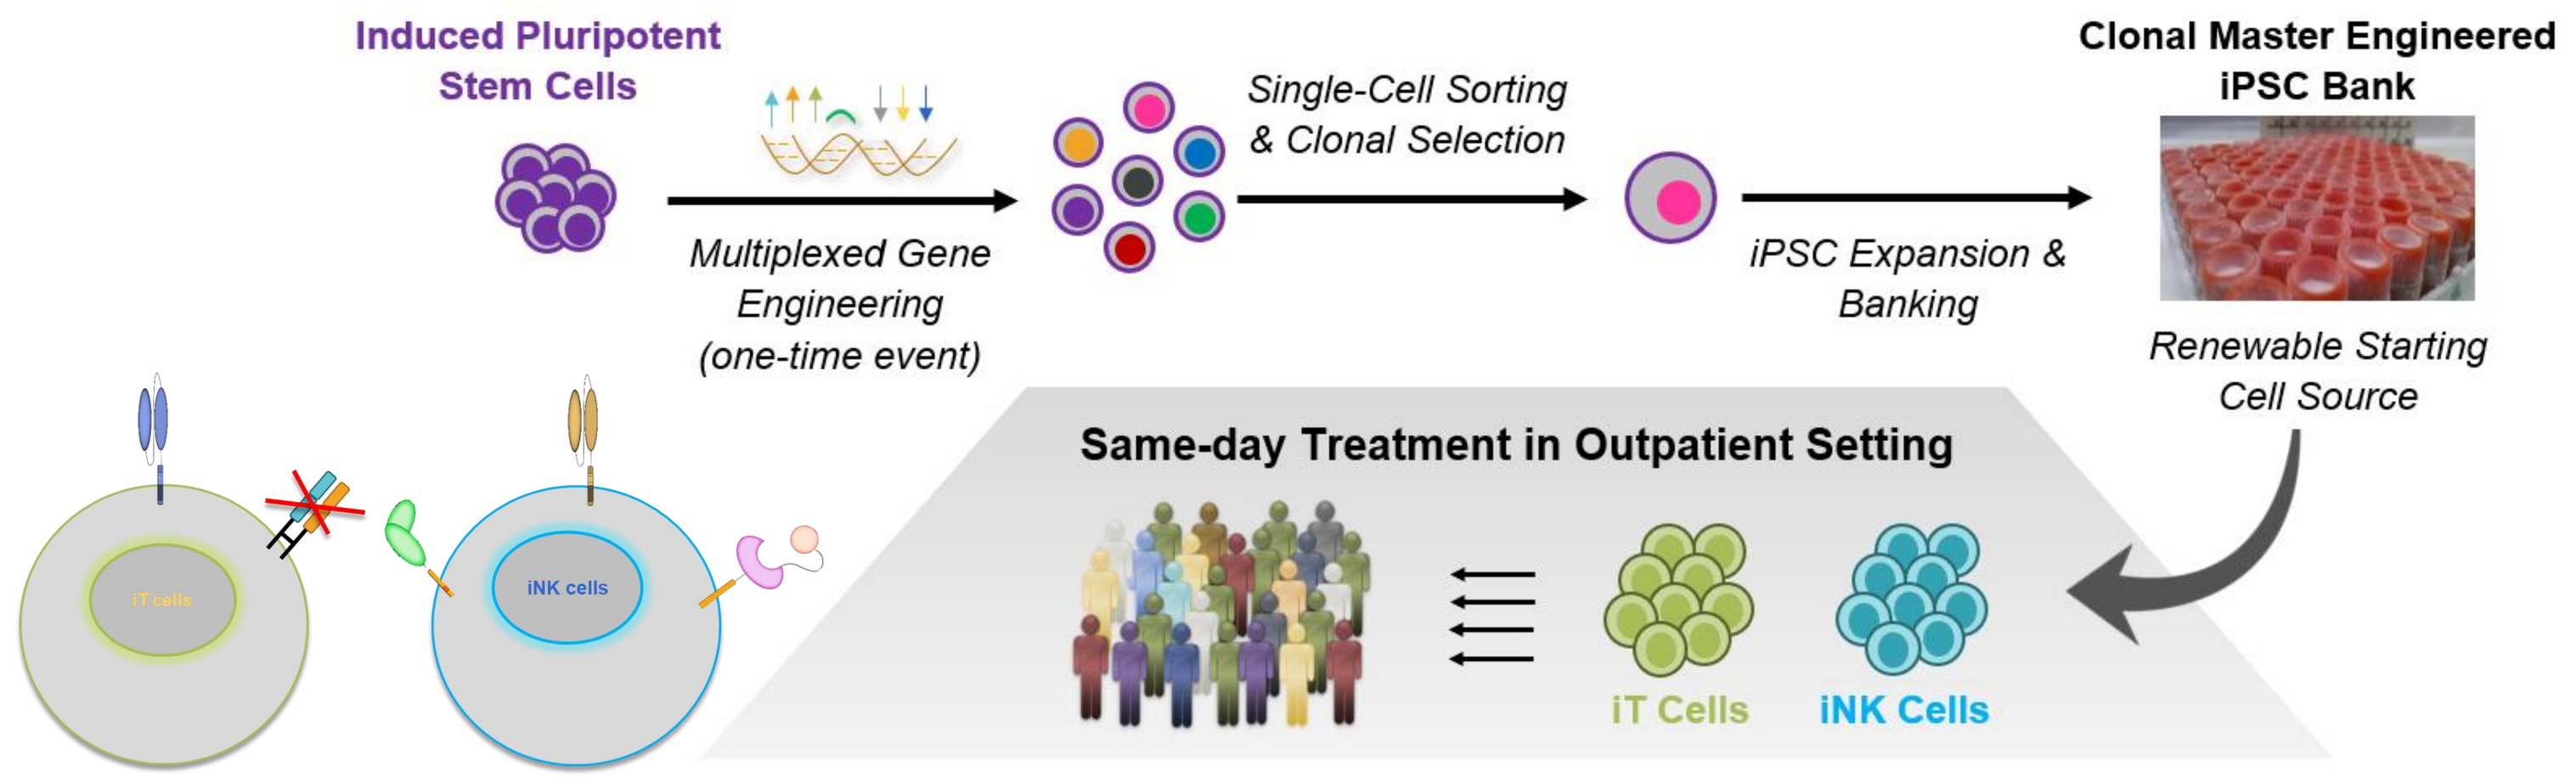
Induced Pluripotent Stem Cells
Multiplexed Gene Engineering (one-time event)
Single-Cell Sorting & Clonal Selection
iPSC Expansion & Banking
Clonal Master Engineered iPSC Bank
Renewable Starting Cell Source
Same-day Treatment in Outpatient Setting
iT Cells
iNK Cells

Summary

- Intraperitoneal xenograft tumor models facilitate investigation of tumor & effector cells interaction *in vivo,* providing insight into functional activity of the effector cells and synergistic interactions of co-administered drugs. iPSC-derived off-the-shelf CAR NK and T cells exhibit robust effector:target interaction and show effective anti-tumor activity.
- Intravenous xenograft tumor models are utilized for demonstrating the engraftment of tumor across the mouse and the ability of effector cells to traffic, seek out and eliminate tumor cells in a range of tissues. iPSC-derived off-the-shelf CAR NK and T cells exhibit the ability to traffic to various tissues and elicit robust tumor growth inhibition.
- Subcutaneous xenograft tumor models represent the challenges associated with bulky solid tumors. Effector cell penetration and residence within these tumors, and the effect of the tumor microenvironment on effector cell function, are best studied in these models. iPSC-derived off-the-shelf CAR NK and T cells exhibit efficient infiltration into the tumor and durable control of bulky tumor growth.
- To establish a new xenograft tumor model that mimics tumor escape and challenges seen in the clinic, we report the establishment of an orthotopic tumor model of triple negative breast cancer. The tumor growth kinetics and metastatic disease profile are consistent with various clinical observations. In addition, failure of primary CAR T cells to inhibit tumor growth, even though infiltration is seen, reflects the heterogeneity and aggressive nature of this tumor model and presents an opportunity to understand mechanisms of treatment resistance and ways to overcome tumor escape.

Figure 2: Intraperitoneal xenograft tumor models represent a unique opportunity to investigate, *in vivo*, the interaction between tumor and effector cells. Antitumor activity of both pCAR T cells and CAR iT cells is highlighted below.

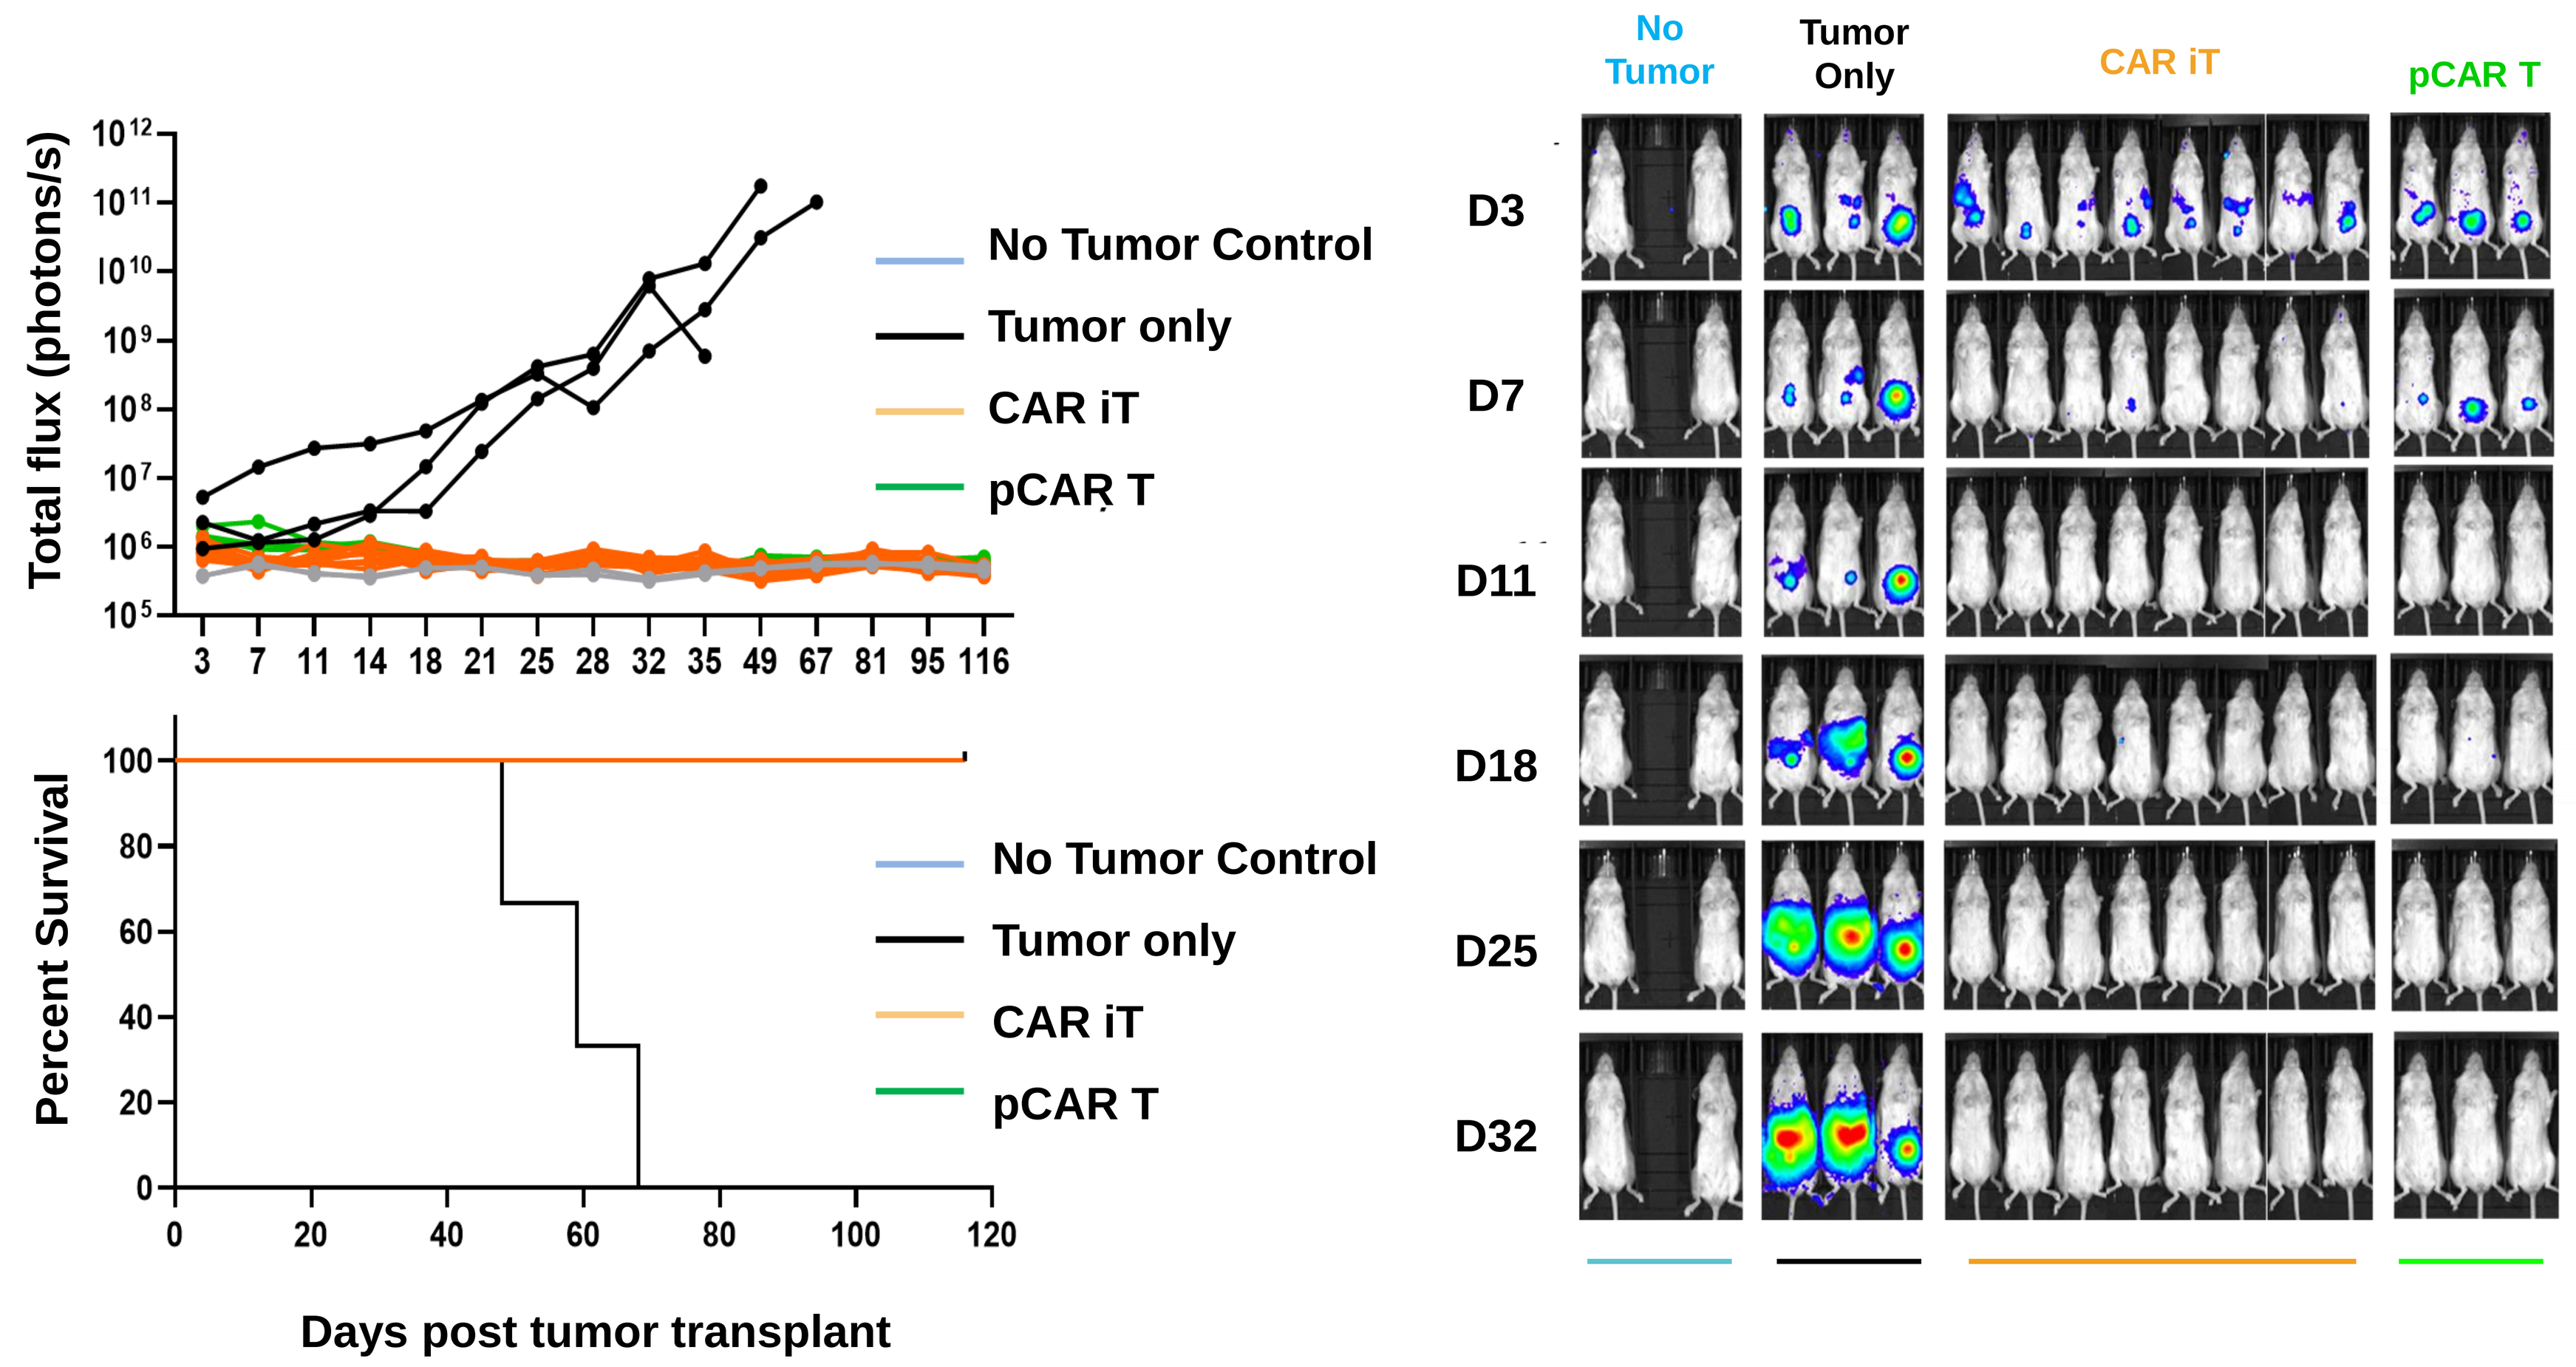
Total flux (photons/s)
No Tumor Control
Tumor only
CAR iT
pCAR T
Percent Survival
Days post tumor transplant
No Tumor
Tumor Only
CAR iT
pCAR T
D3
D7
D11
D18
D25
D32

Figure 3: Intravenous xenograft tumor models represent a stringent strategy to demonstrate the ability of effector cells to traffic throughout various tissues to find and kill disseminated tumor cells. Donor-to-donor variability of pCAR T cells and consistency of product performance from CAR iT cells derived from iPSCs is highlighted below.

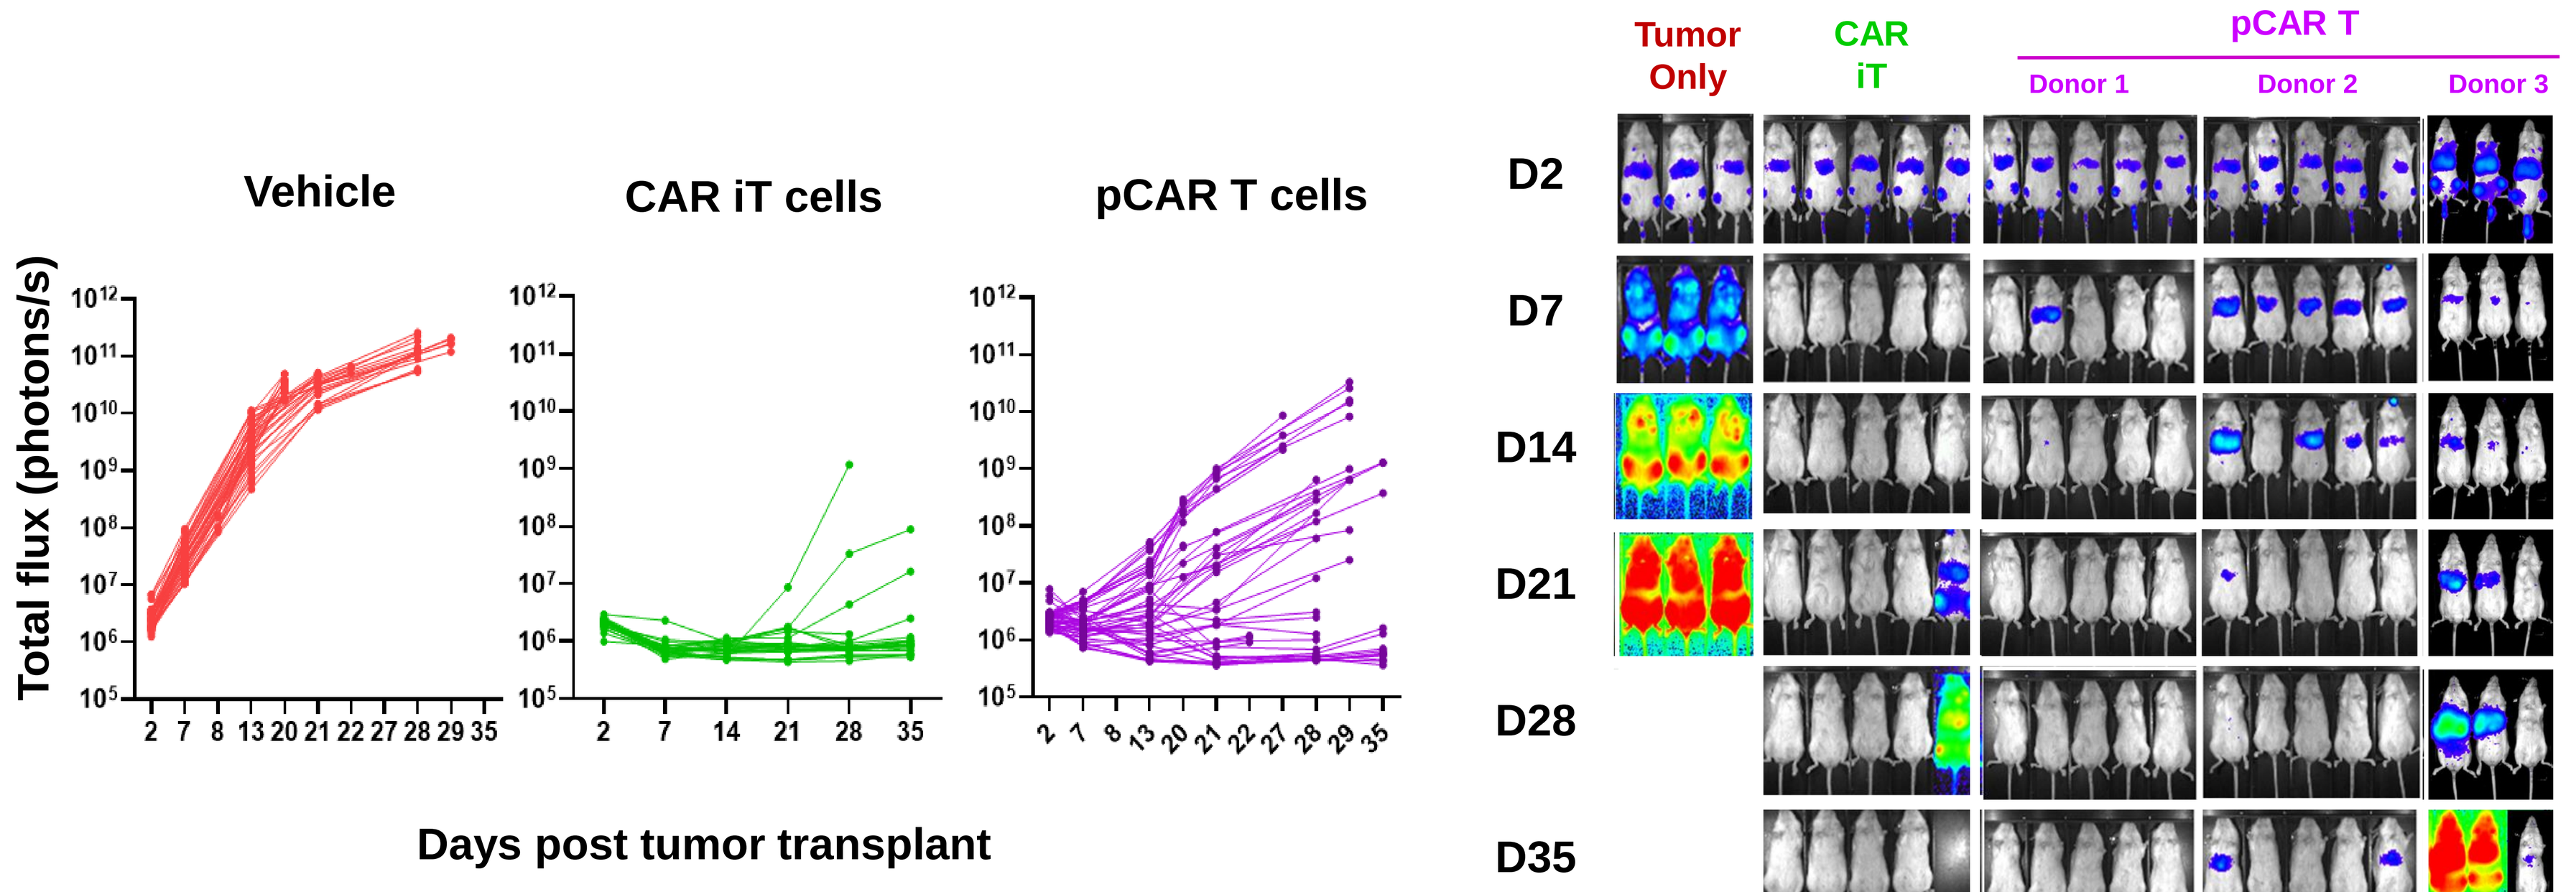
Vehicle
CAR iT cells
pCAR T cells
Total flux (photons/s)
Days post tumor transplant
Tumor Only
CAR iT
pCAR T
Donor 1
Donor 2
Donor 3
D2
D7
D14
D21
D28
D35

Figure 4: Subcutaneous xenograft tumor models represent a more physiological model of the tumor microenvironment facilitating investigation of effector cell homing and penetration of the tumor, as well as effects of tumor produced factors on effector cell function. Potent SKOV3 tumor control by CAR iT cells and their retention, accumulation and/or expansion in the tumor are presented below.

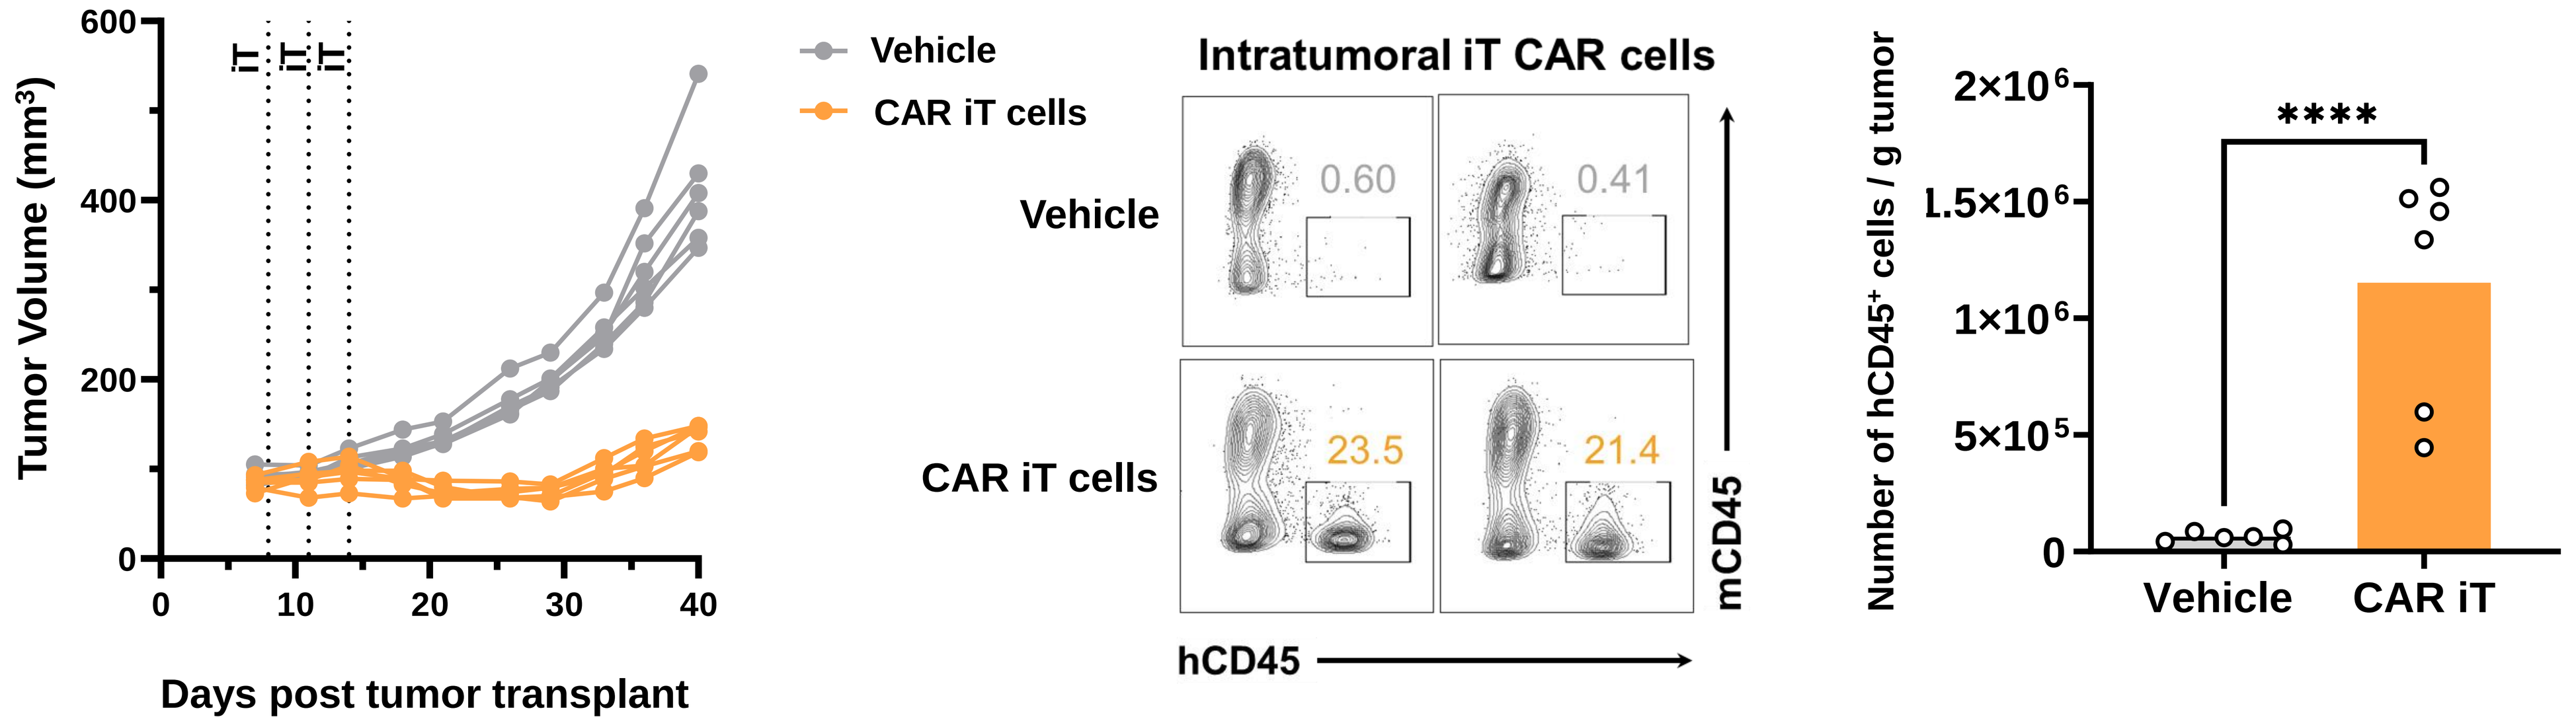
Tumor Volume (mm³)
Days post tumor transplant
Vehicle
CAR iT cells
Intratumoral iT CAR cells
0.60
0.41
23.5
21.4
hCD45
mCD45
Number of hCD45⁺ cells / g tumor

Vehicle
CAR iT

Figure 5: Establishment of metastatic breast cancer model as a proxy for patient disease progression. MDA-MB-231 over expressing CD19 produce three unique tumors: primary (⇧), secondary (⇧), and tertiary (⇧). Secondary and tertiary sites include lung and liver, recapitulating human disease where 31.4% and 26% of human patients show metastases. In addition, metastases were found in the right axillary lymph node, a hallmark sign for diagnosing and staging breast cancer in patients. The lack of tumor control by pCAR T cells suggests that this aggressive model may require multi-modal treatment regimens.

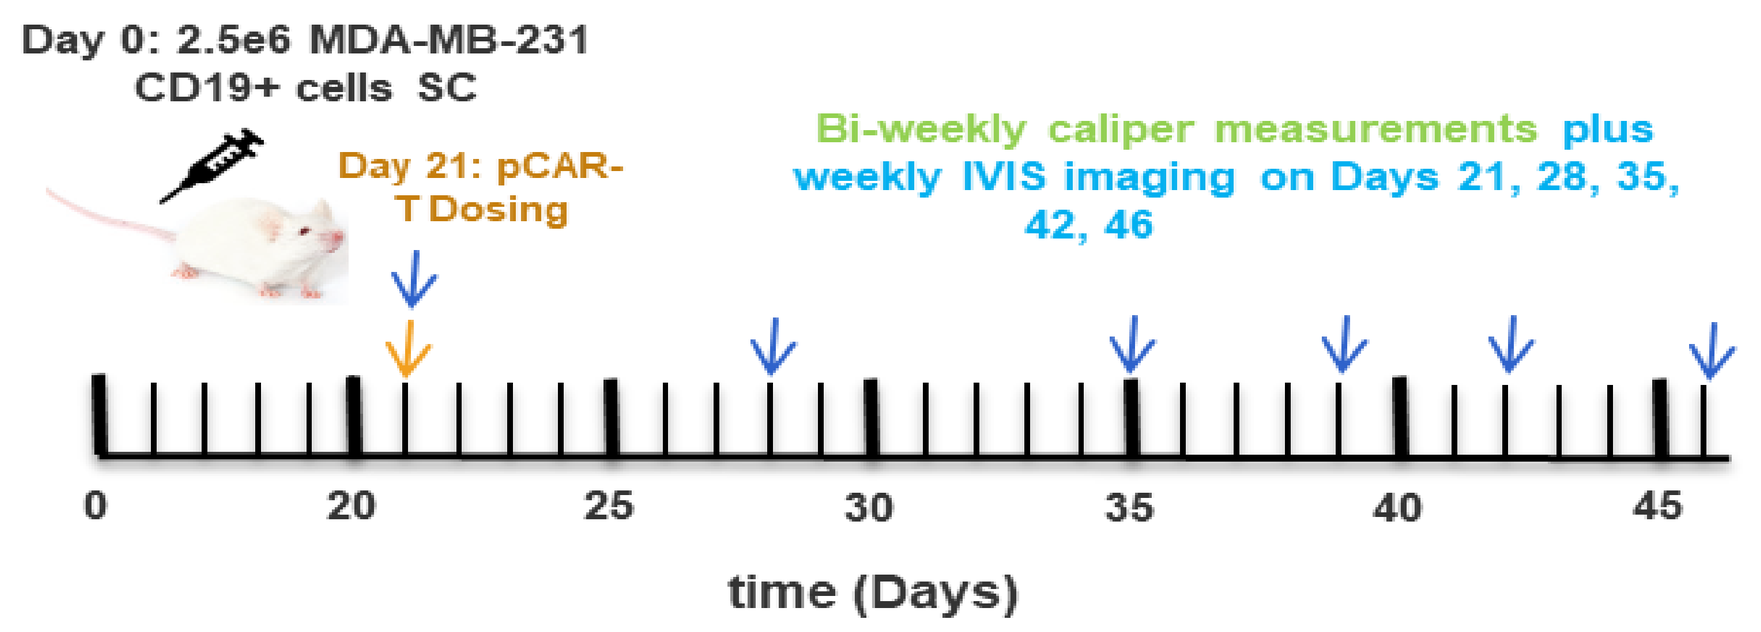
Day 0: 2.5e6 MDA-MB-231 CD19+ cells SC
Day 21: pCAR-T Dosing
Bi-weekly caliper measurements plus weekly IVIS imaging on Days 21, 28, 35, 42, 46
time (Days)

Group	n	Tumor	Tumor route	# Tumor cells	Implant volume	Effector	# Effector Cells
1	2	N/A	N/A	N/A	N/A	Vehicle	-
2	5	MDA-MB-231 CD19+	MFP #4	2.5e6	100 ul	Vehicle	-
3	5	MDA-MB-231 CD19+	MFP #4	2.5e6	100 ul	pCAR T	1 x 2e6

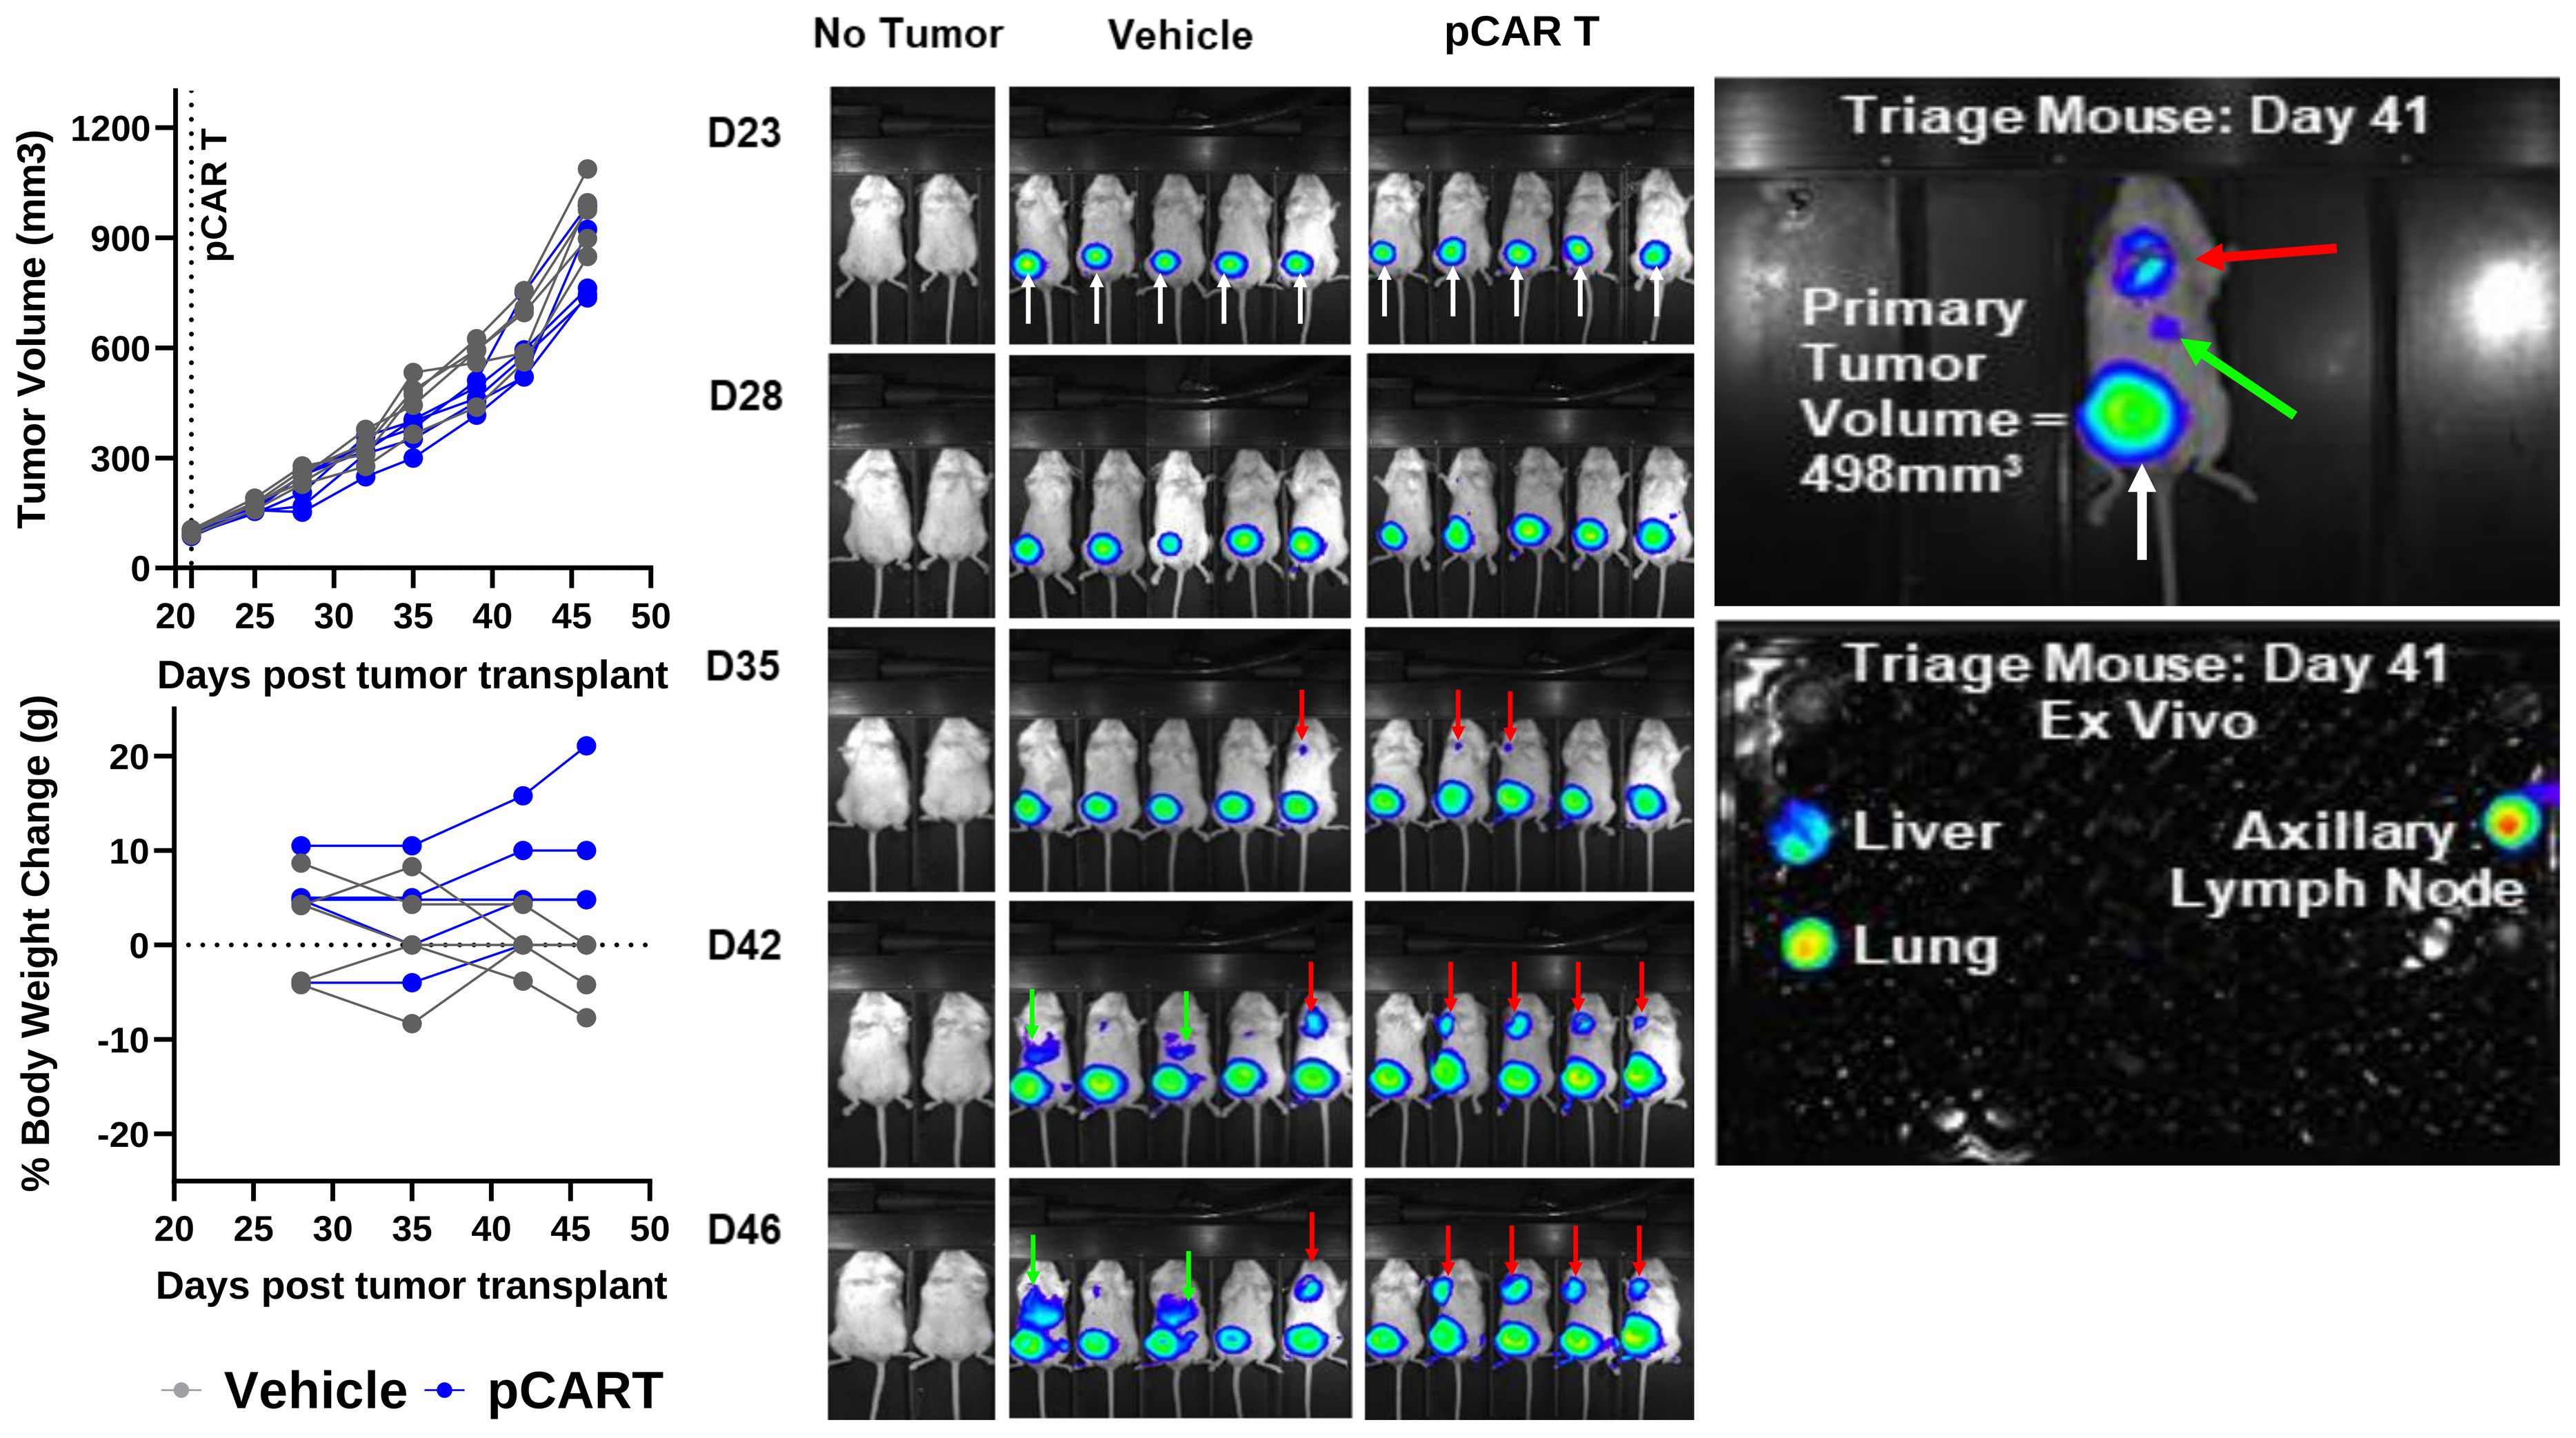
Tumor Volume (mm3)
pCAR T
Days post tumor transplant
% Body Weight Change (g)
Vehicle
pCART
No Tumor
Vehicle
pCAR T
D23
D28
D35
D42
D46
Triage Mouse: Day 41
Primary Tumor Volume = 498mm³
Triage Mouse: Day 41 Ex Vivo
Liver
Lung
Axillary Lymph Node

Figure 6: MDA-MB-231 overexpressing CD19 tumors show a highly heterogenous tumor with focal islands of inflammatory cells, areas of necrosis, and evidence of perivascular desmoplastic reaction. Immunohistochemical staining for human CD45⁺ cells shows that pCAR T cells penetrate the tumor widely, beyond the tumor surface, and into areas of necrotic border. This thorough distribution of pCAR T cells is inconsistent with the poor tumor control (below, left). Digital droplet (dd) PCR affords absolute quantification of human CD45⁺ DNA in tissues reflecting the total number of pCAR T cells in each tumor (below, right). ddPCR data shown below is consistent with the extent of pCAR T cell penetration of the tumor observed in the left-hand panel below.

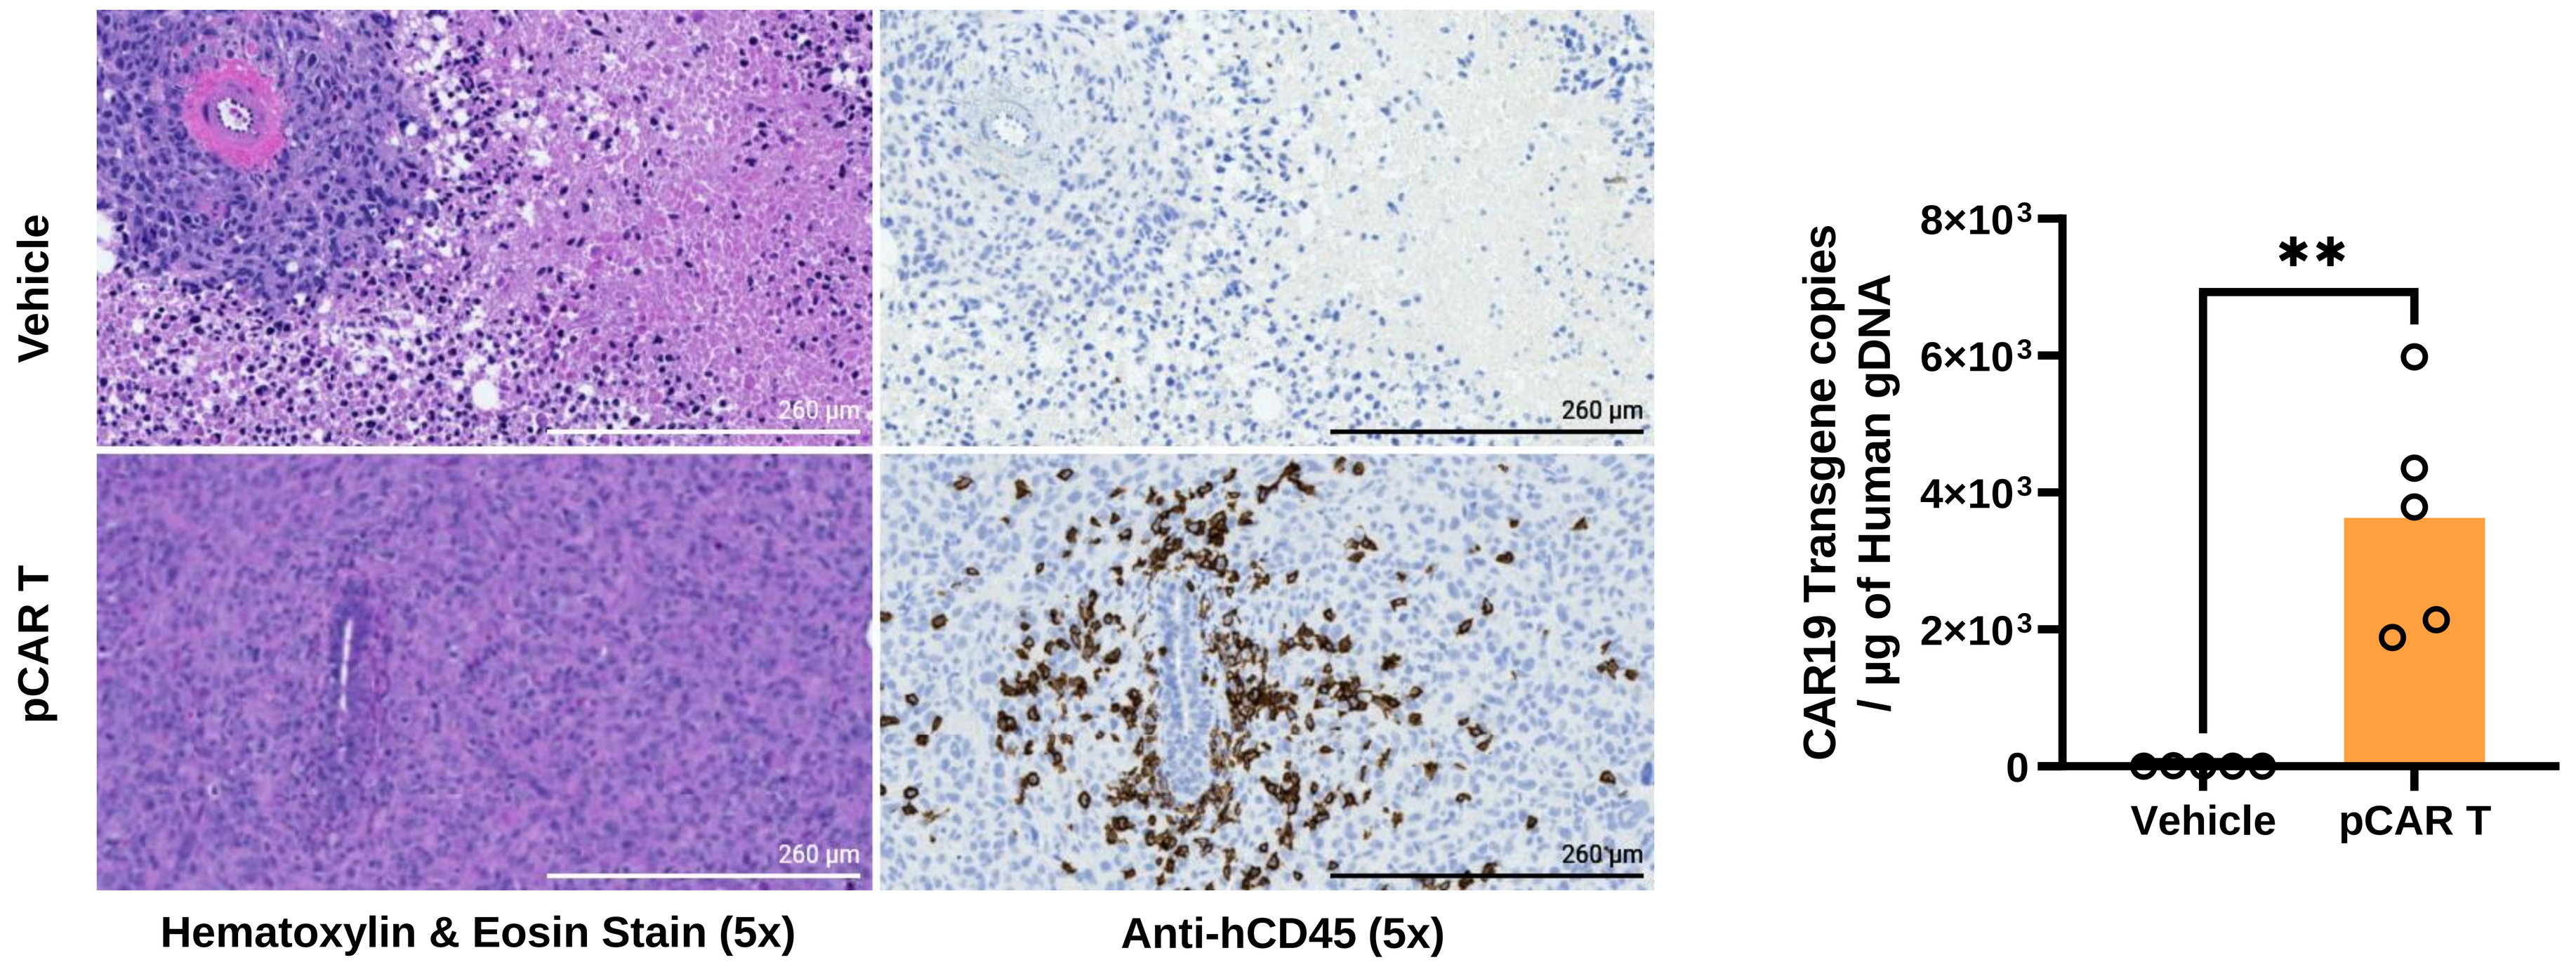
Vehicle
pCAR T
260 µm
Hematoxylin & Eosin Stain (5x)
Anti-hCD45 (5x)
CAR19 Transgene copies / µg of Human gDNA
**
Vehicle
pCAR T