Characterization of Engineered Macrophages and other Myeloid Cells Differentiated from CD34+ Hematopoietic Progenitor Cells Derived from Pluripotent Stem Cells

S. LaBarge, A. Sims, J. Sandoval, B. Kandar, E. Sung, M. Denholtz, J. Martin, E. Diaz, A. Bagri, A. Witty, B. Valamehr

Fate Therapeutics, Inc., San Diego, CA

Executive Summary

Human induced pluripotent stem cells (hiPSCs) exhibit unlimited self-renewal potential and have the capacity to differentiate into somatic cell derivatives of all three germ layers, making them an ideal source for generation of cellular therapies. hiPSCs also address the inherent variability that arises from the use of patient- or donor-sourced cells which can lead to drug product inconsistencies that impact safety, efficacy, and therapeutic utility. To leverage these attributes, we have established a platform that enables multiplexed engineering of hiPSCs at the single-cell level and have developed a proprietary stage-specific differentiation protocol to support definitive hematopoiesis for the derivation of CD34+ hematopoietic progenitor (iCD34) cells. We have previously shown that these iCD34 cells exhibit efficient differentiation and expansion to lymphoid lineages, including Natural Killer (NK) and αβ T-cells

Additionally, iCD34 cells can efficiently give rise to myeloid populations including myeloid progenitor cells and macrophages. To this end, we have established a process for differentiating our engineered iCD34 cells into myeloid populations. Leveraging key aspects of definitive hematopoiesis, the iCD34 cells expand and differentiate into myeloid progenitor and uniformly express known macrophage markers including CD45, CD11b and CD14.

Graphical Representation

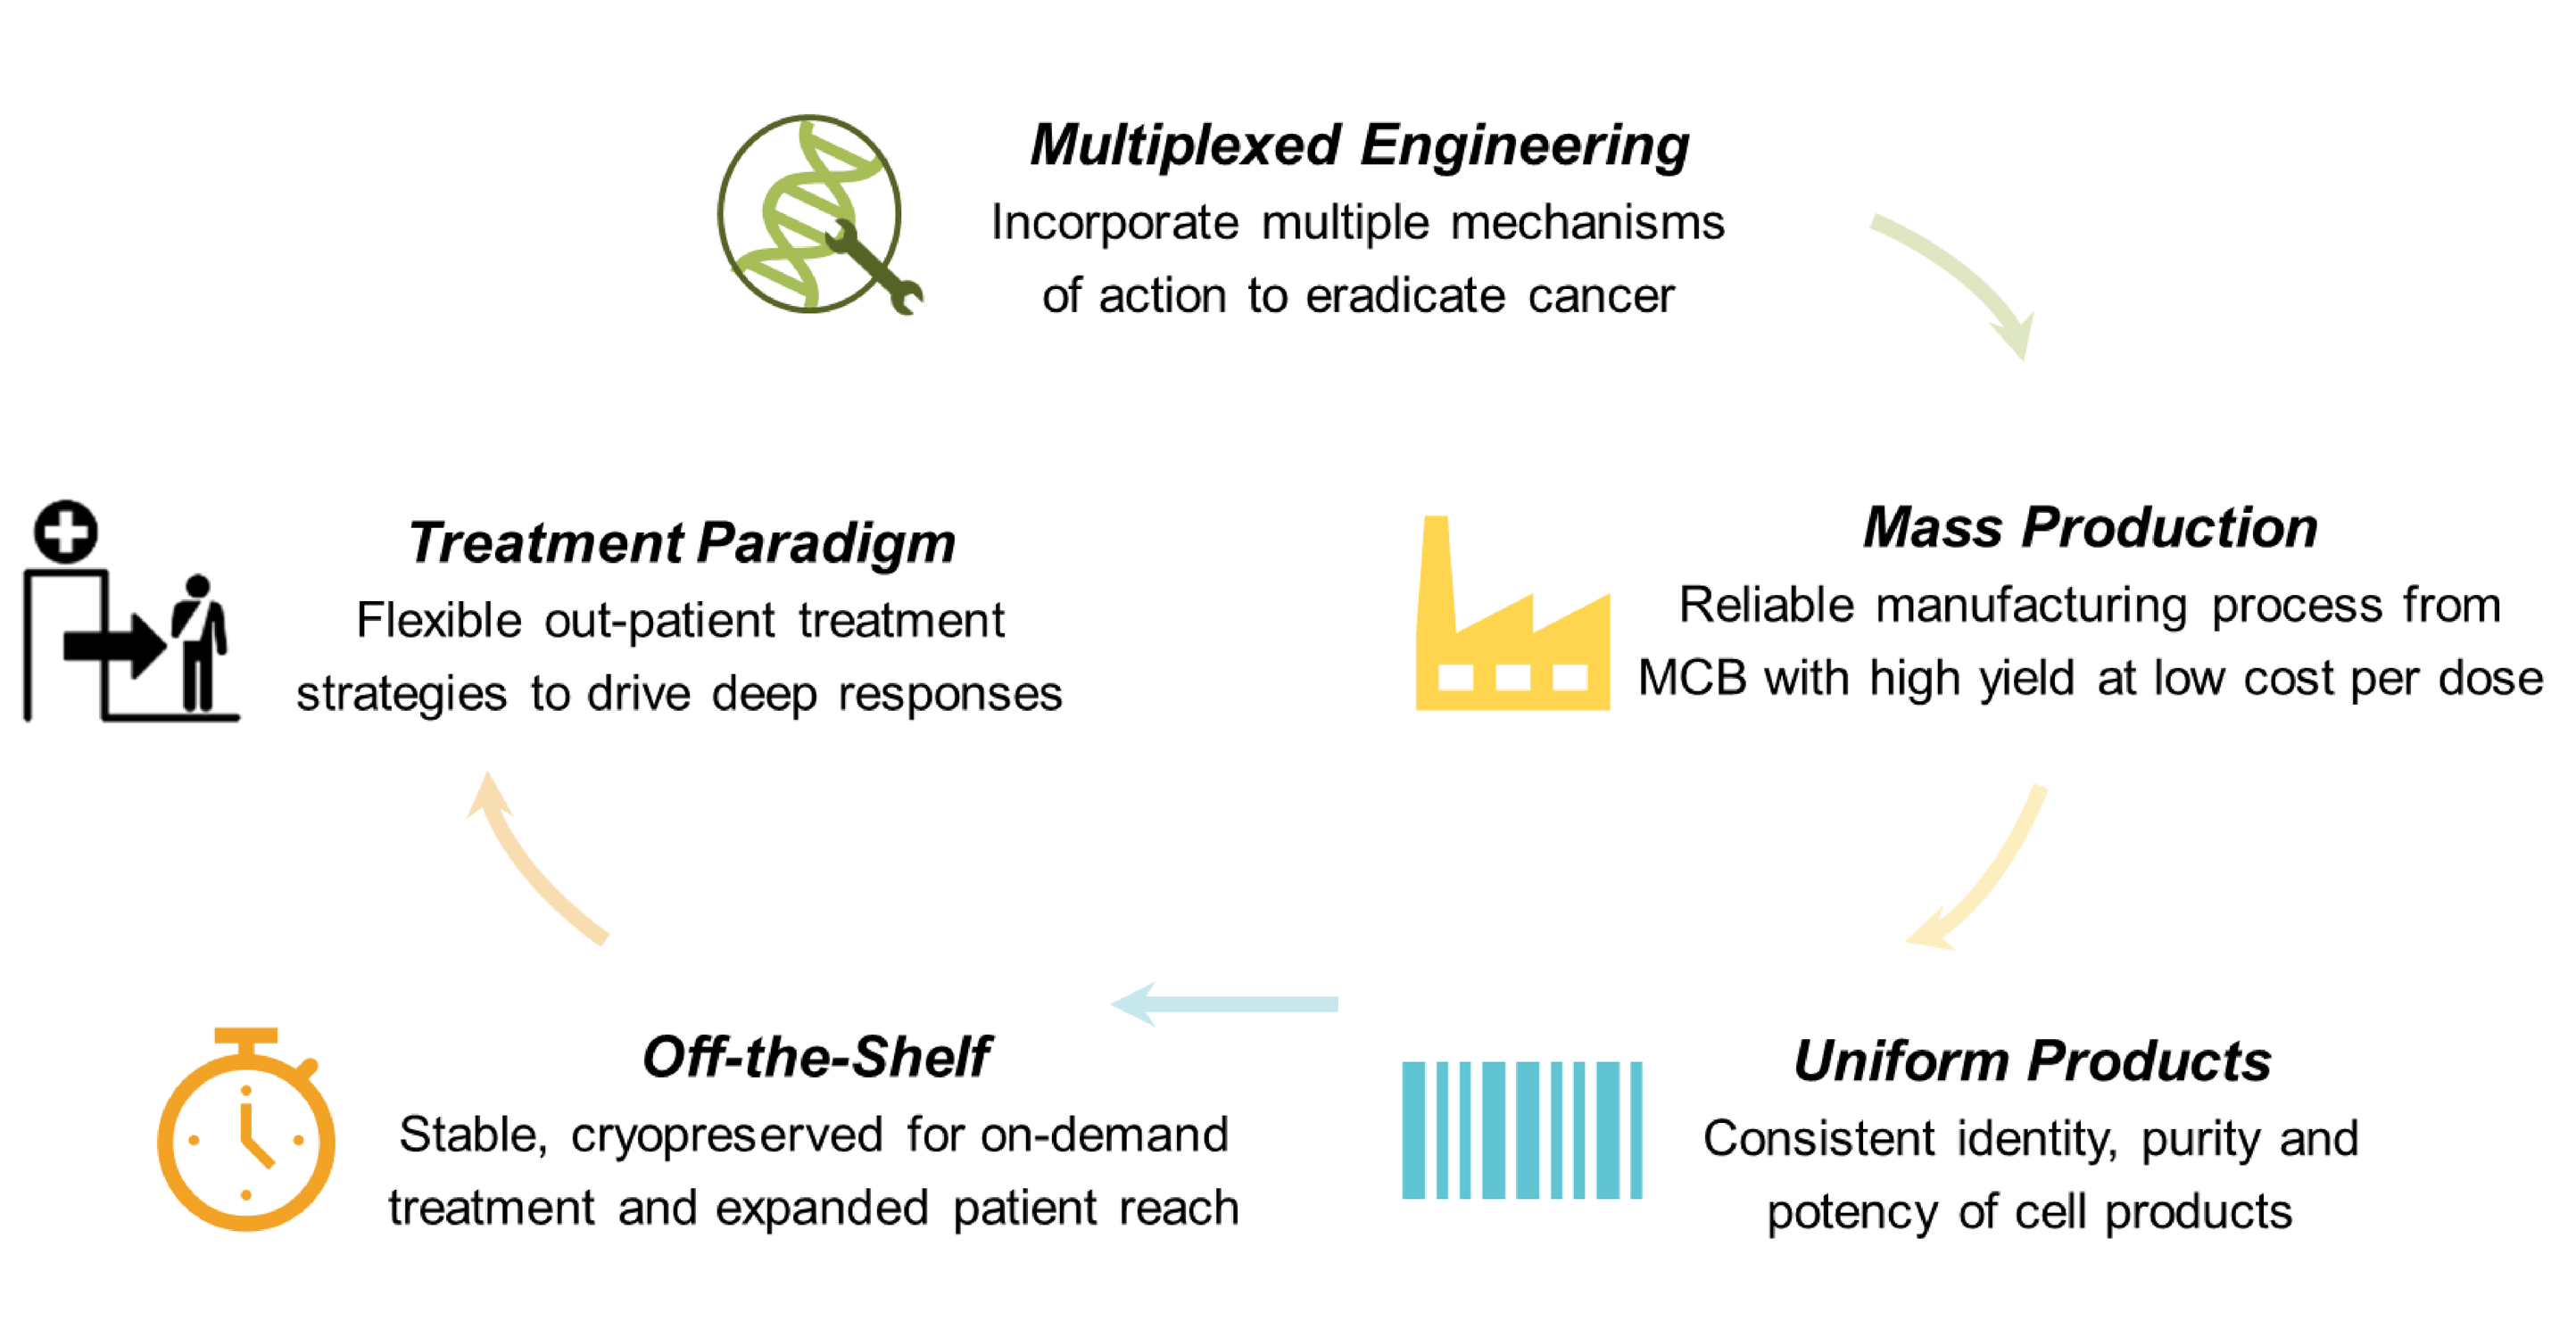
Multiplexed Engineering
Incorporate multiple mechanisms of action to eradicate cancer
Mass Production
Reliable manufacturing process from MCB with high yield at low cost per dose
Uniform Products
Consistent identity, purity and potency of cell products
Off-the-Shelf
Stable, cryopreserved for on-demand treatment and expanded patient reach
Treatment Paradigm
Flexible out-patient treatment strategies to drive deep responses

Conclusions

- iMacs can be made in uniform and consistent manner from hiPSCs, express canonical macrophage markers and display a gene expression profile like their peripheral blood derived counterparts.
- iMacs can acquire further specification based on environmental stimuli and can be broadly classified into M1-like/pro-inflammatory and M2-like/anti-inflammatory populations.
- Importantly, iMacs demonstrate macrophage function such as phagocytosis and the secretion of polarization specific cytokines.
- iMacs display biodistribution to various tissues, including the lung, following intravenous tail vein dosing.
- Collectively, the data confirm that the iCD34 cells generated from our hiPSC platform are multipotent hematopoietic progenitor cells and can efficiently expand and differentiate into lymphoid and myeloid lineages.

Results

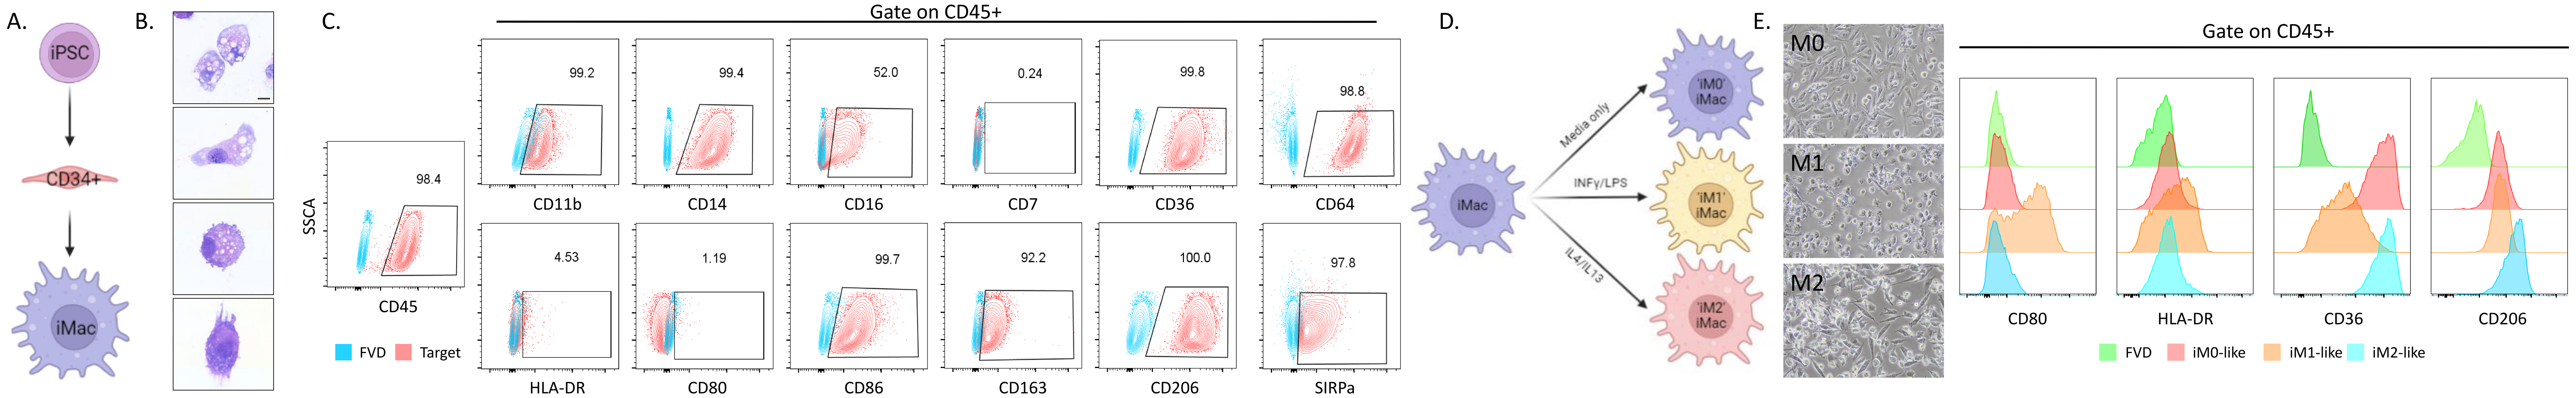
A.
iPSC
CD34+
iMac
B.
C.
Gate on CD45+
SSCA
CD45
98.4
FVD
Target
CD11b 99.2
CD14 99.4
CD16 52.0
CD7 0.24
CD36 99.8
CD64 98.8
HLA-DR 4.53
CD80 1.19
CD86 99.7
CD163 92.2
CD206 100.0
SIRPa 97.8
D.
iMac
Media only
INFγ/LPS
IL4/IL13
'iM0' iMac
'iM1' iMac
'iM2' iMac
E.
M0
M1
M2
Gate on CD45+
CD80
HLA-DR
CD36
CD206
FVD
iM0-like
iM1-like
iM2-like

Figure 1. Derivation, cell surface phenotype and polarization of Fate iPSC derived Macrophage. (A) iMacs are produced by directed differentiation of iPS cells to CD34+ hematopoietic progenitors and then to CD45+, CD11b+, CD14+ macrophages. (B) Giemsa stain of iPSC derived macrophages. (C) Flow cytometry of select macrophage markers in iMacs. (D) Schematic illustrating method to polarize iMacs with pro-inflammatory (M1) or anti-inflammatory (M2) stimuli. (E) Macrophages acquire further specification based on environmental stimuli and display M1-like/pro-inflammatory or M2-like/anti-inflammatory morphological changes and polarization specific cell surface marker expression. Cartoons created using BioRender.com.

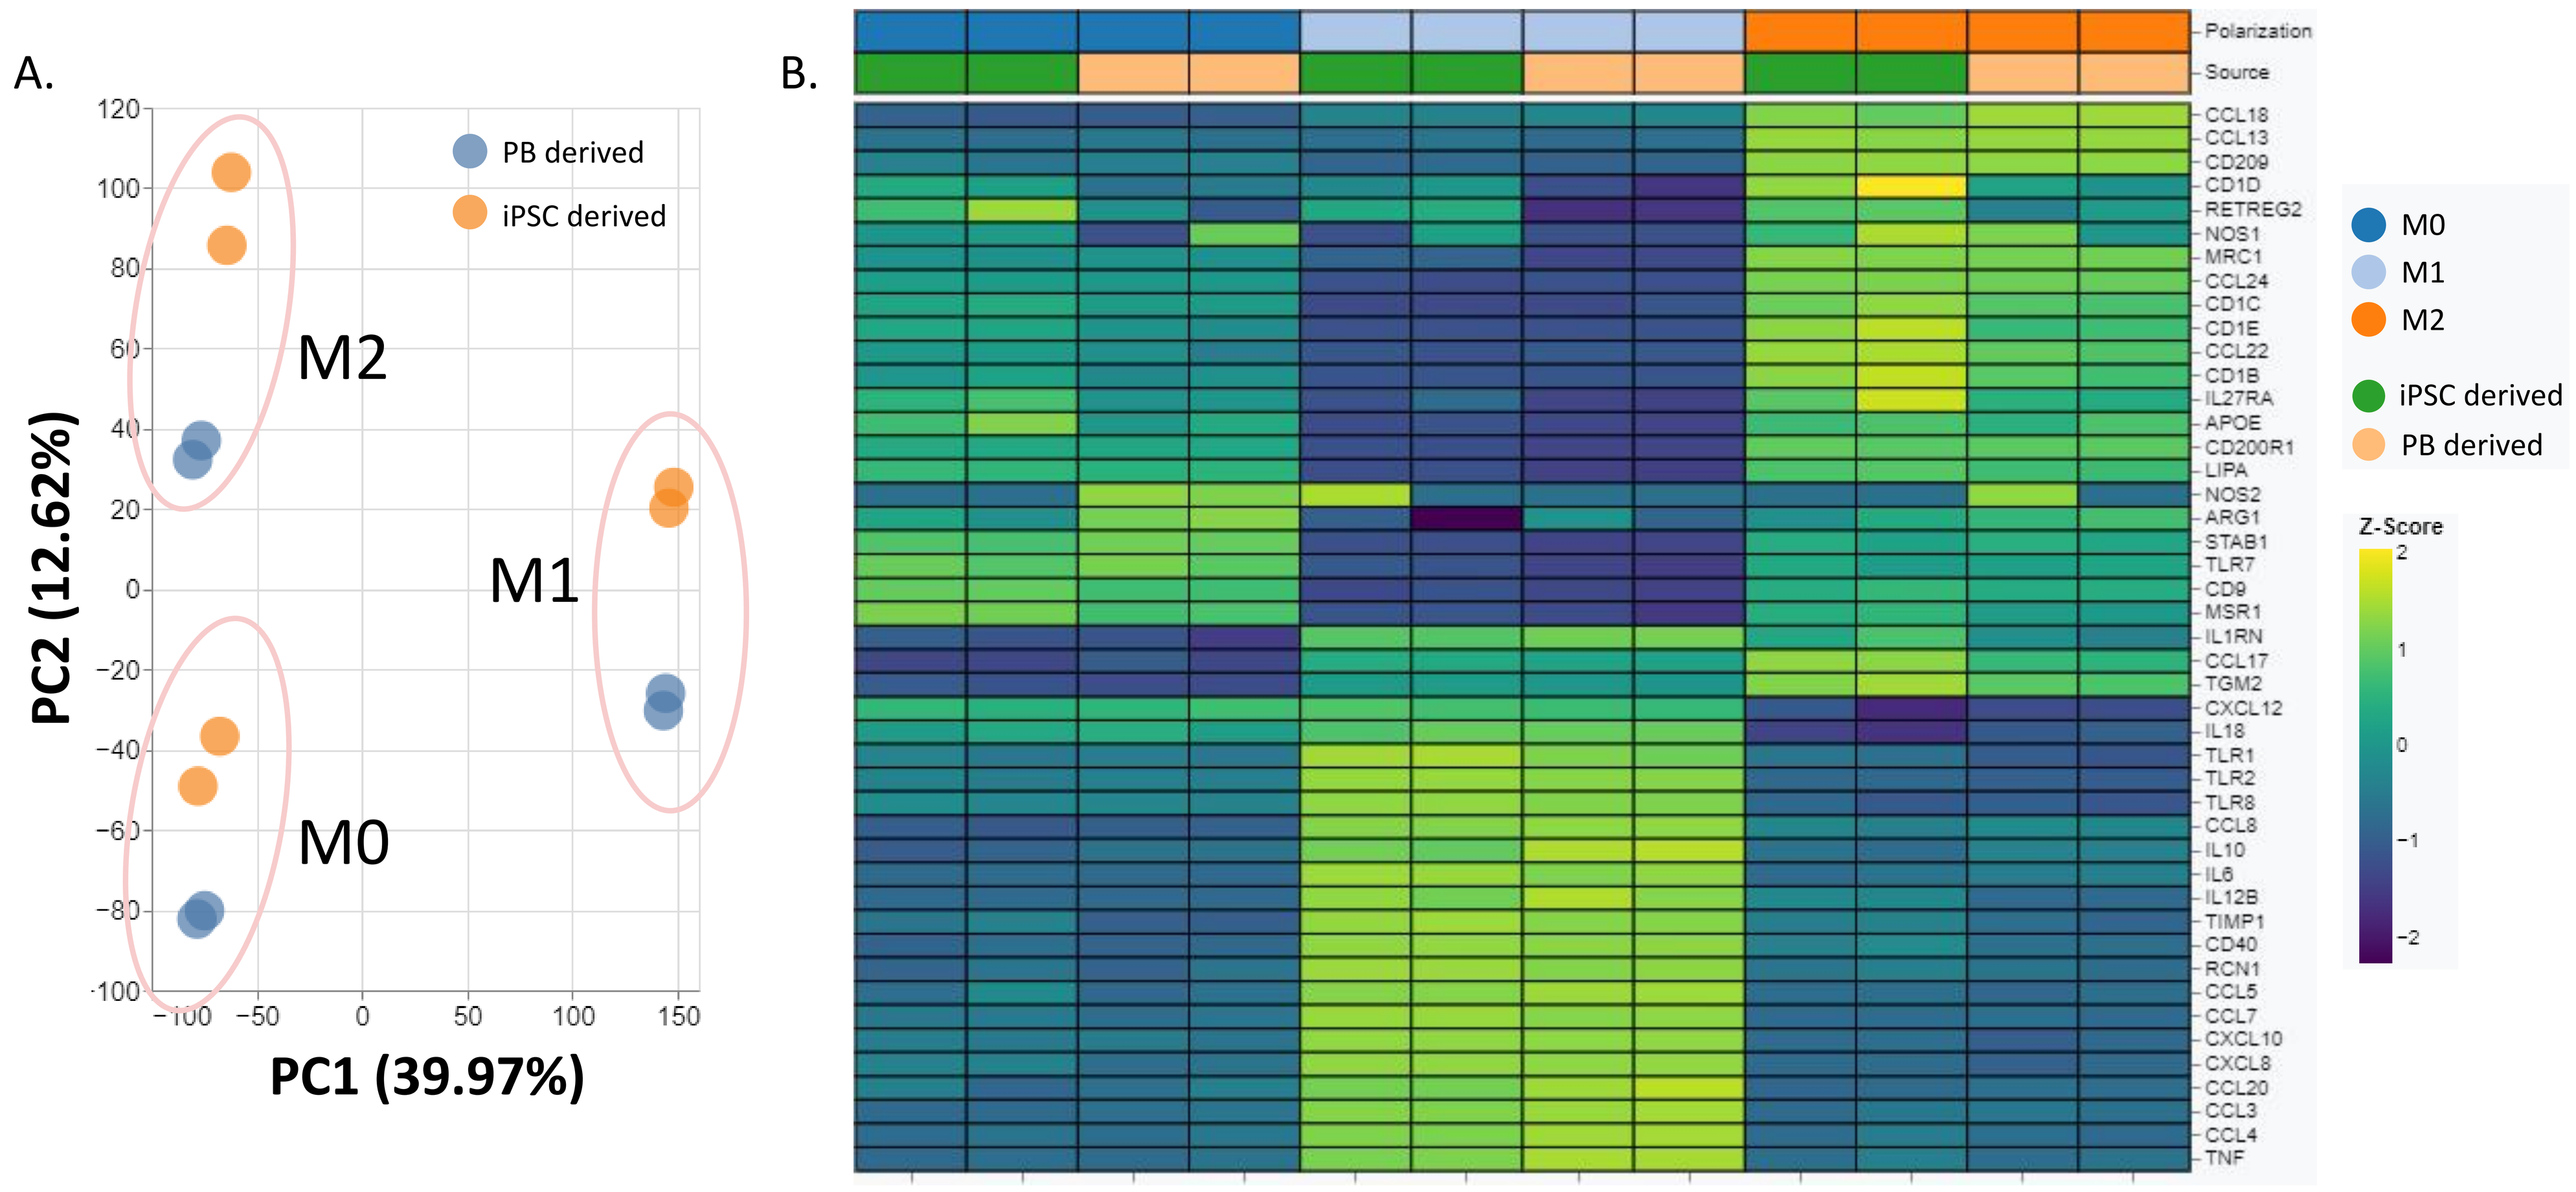
A.
PB derived
iPSC derived
M2
M1
M0
PC2 (12.62%)
PC1 (39.97%)
B.
Polarization
Source
CCL18
CCL13
CD209
CD1D
RETREG2
NOS1
MRC1
CCL24
CD1C
CD1E
CCL22
CD1B
IL27RA
APOE
CD200R1
LIPA
NOS2
ARG1
STAB1
TLR7
CD9
MSR1
IL1RN
CCL17
TGM2
CXCL12
IL18
TLR1
TLR2
TLR8
CCL8
IL10
IL6
IL12B
TIMP1
CD40
RCN1
CCL5
CCL7
CXCL10
CXCL8
CCL20
CCL3
CCL4
TNF
M0
M1
M2
iPSC derived
PB derived
Z-Score

Figure 2. Gene expression analysis of polarized iPSC derived and Peripheral Blood (PB) derived macrophages. iMacs and PB derived macrophages were polarized as in Figure 1D then analyzed through Bulk RNA sequencing. (A) Principal component analysis reveals primary clustering by polarization and not source. (B) Heatmap analysis of differentially expressed genes between macrophage source (iPSC or PB) and polarization state (M0, M1, M2).

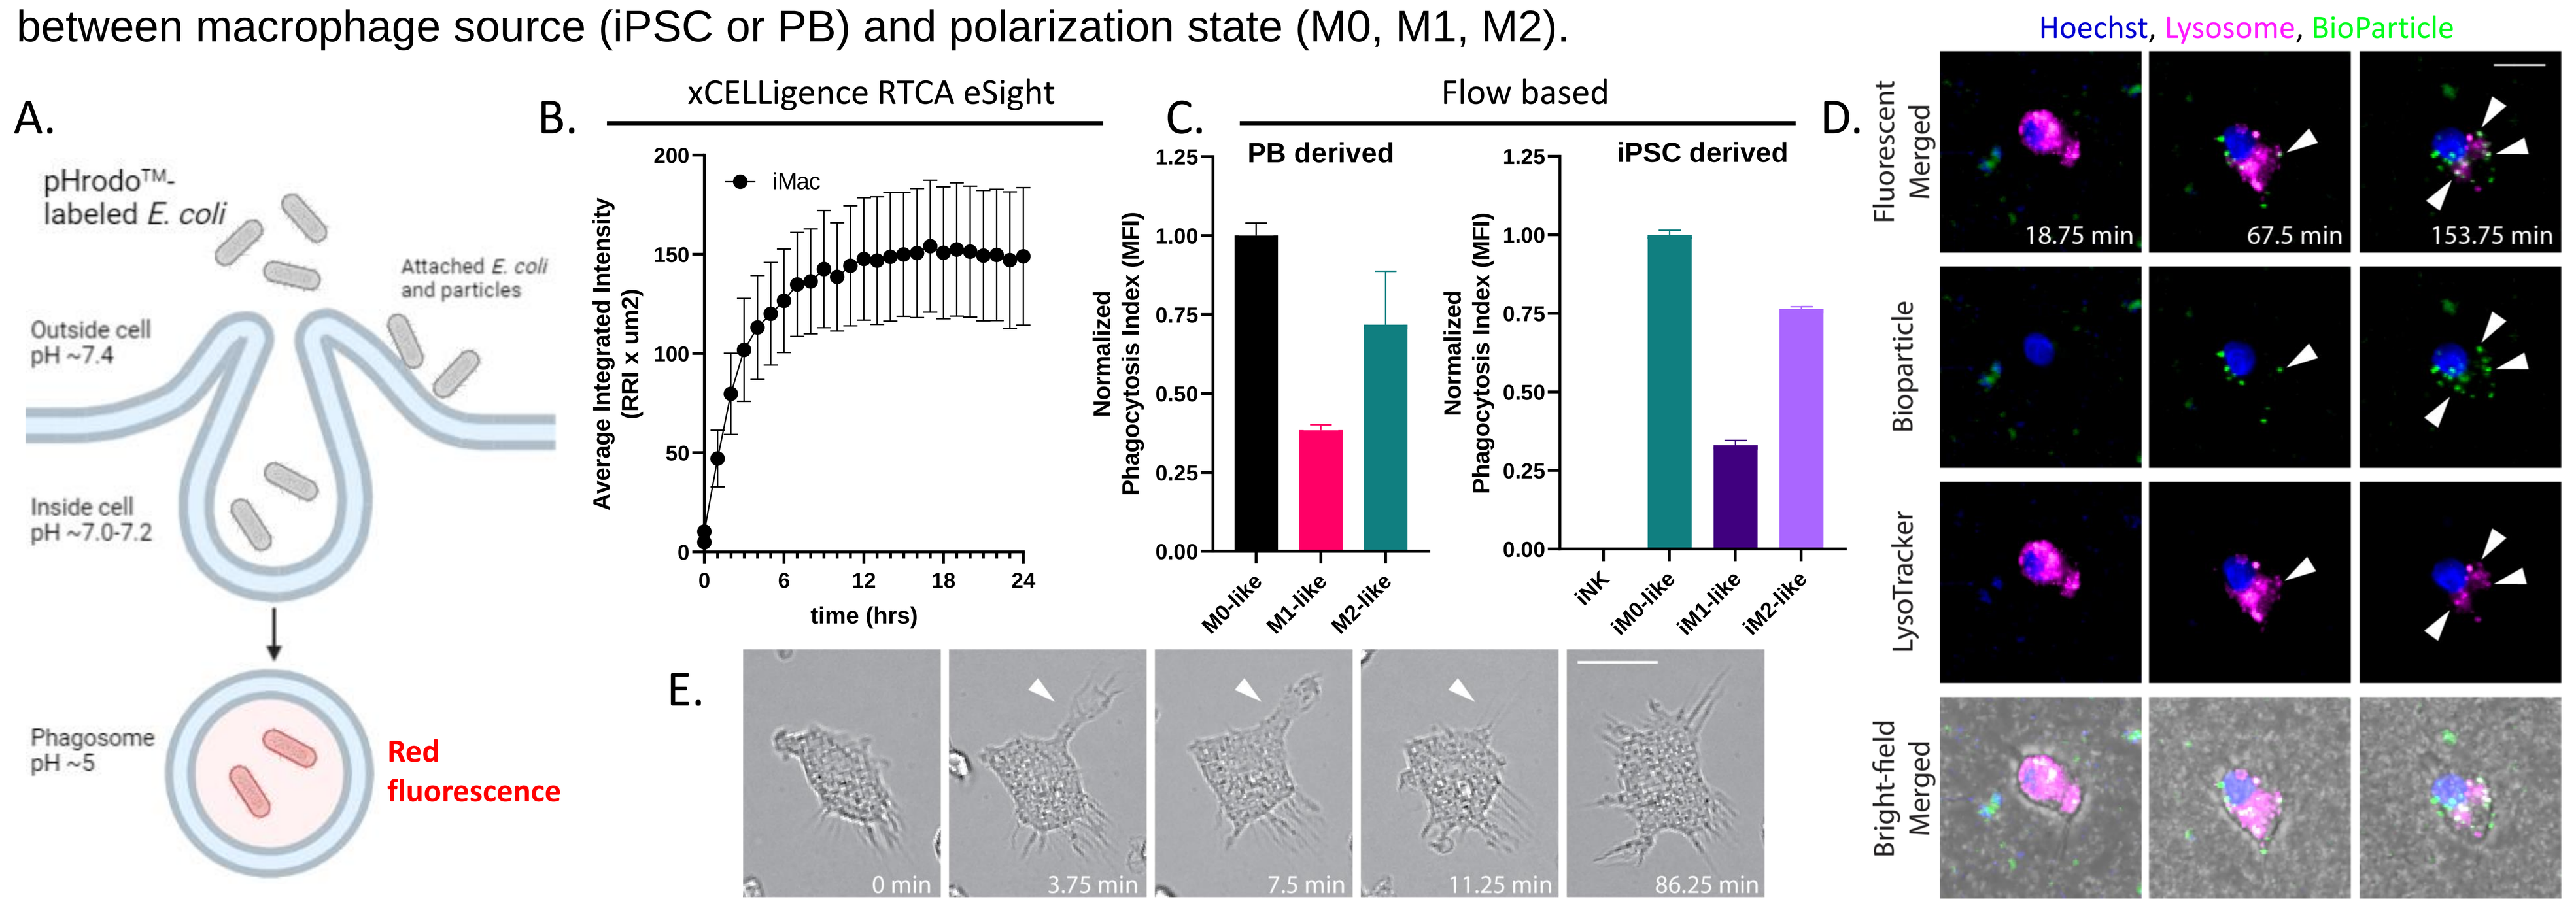
A.
pHrodo™-labeled E. coli
Attached E. coli and particles
Outside cell pH ~7.4
Inside cell pH ~7.0-7.2
Phagosome pH ~5
Red fluorescence
B.
xCELLigence RTCA eSight
iMac
Average Integrated Intensity (RRI x um2)
time (hrs)
C.
Flow based
PB derived
iPSC derived
Normalized Phagocytosis Index (MFI)
M0-like
M1-like
M2-like
iNK
iM0-like
iM1-like
iM2-like
D.
Hoechst, Lysosome, BioParticle
Fluorescent Merged
Bioparticle
LysoTracker
Bright-field Merged
18.75 min
67.5 min
153.75 min
E.
0 min
3.75 min
7.5 min
11.25 min
86.25 min

Figure 3. Fate iMacs display phagocytosis of pHrodo™ labelled *E. coli* bioparticles. (A) Schematic depicting mechanism of pHrodo-labelled E. coli phagocytosis. Briefly, Fate iMacs (M0, M1 or M2 generated via Figure 1D) or PB derived macrophages are incubated with pHrodo™ labelled E. coli bioparticles and analyzed on respective platforms. (B) Red Average Integrated Intensity (e.g., red fluorescence per um^2) measured by an xCELLigence RTCA eSight, over a 24 hr period (C) pHrodo fluorescence measured via flow cytometry after 1.5-hour incubation and (D) live cell imaging of iM2 phagocytosis using a Keyence Live Cell Imager depicting overlap of pHrodo labelled Bioparticle and lysosome. (E) Keyence live cell imaging of lamellipodia extension and retraction in iMacs incubated with pHrodo labelled E. coli bioparticle.

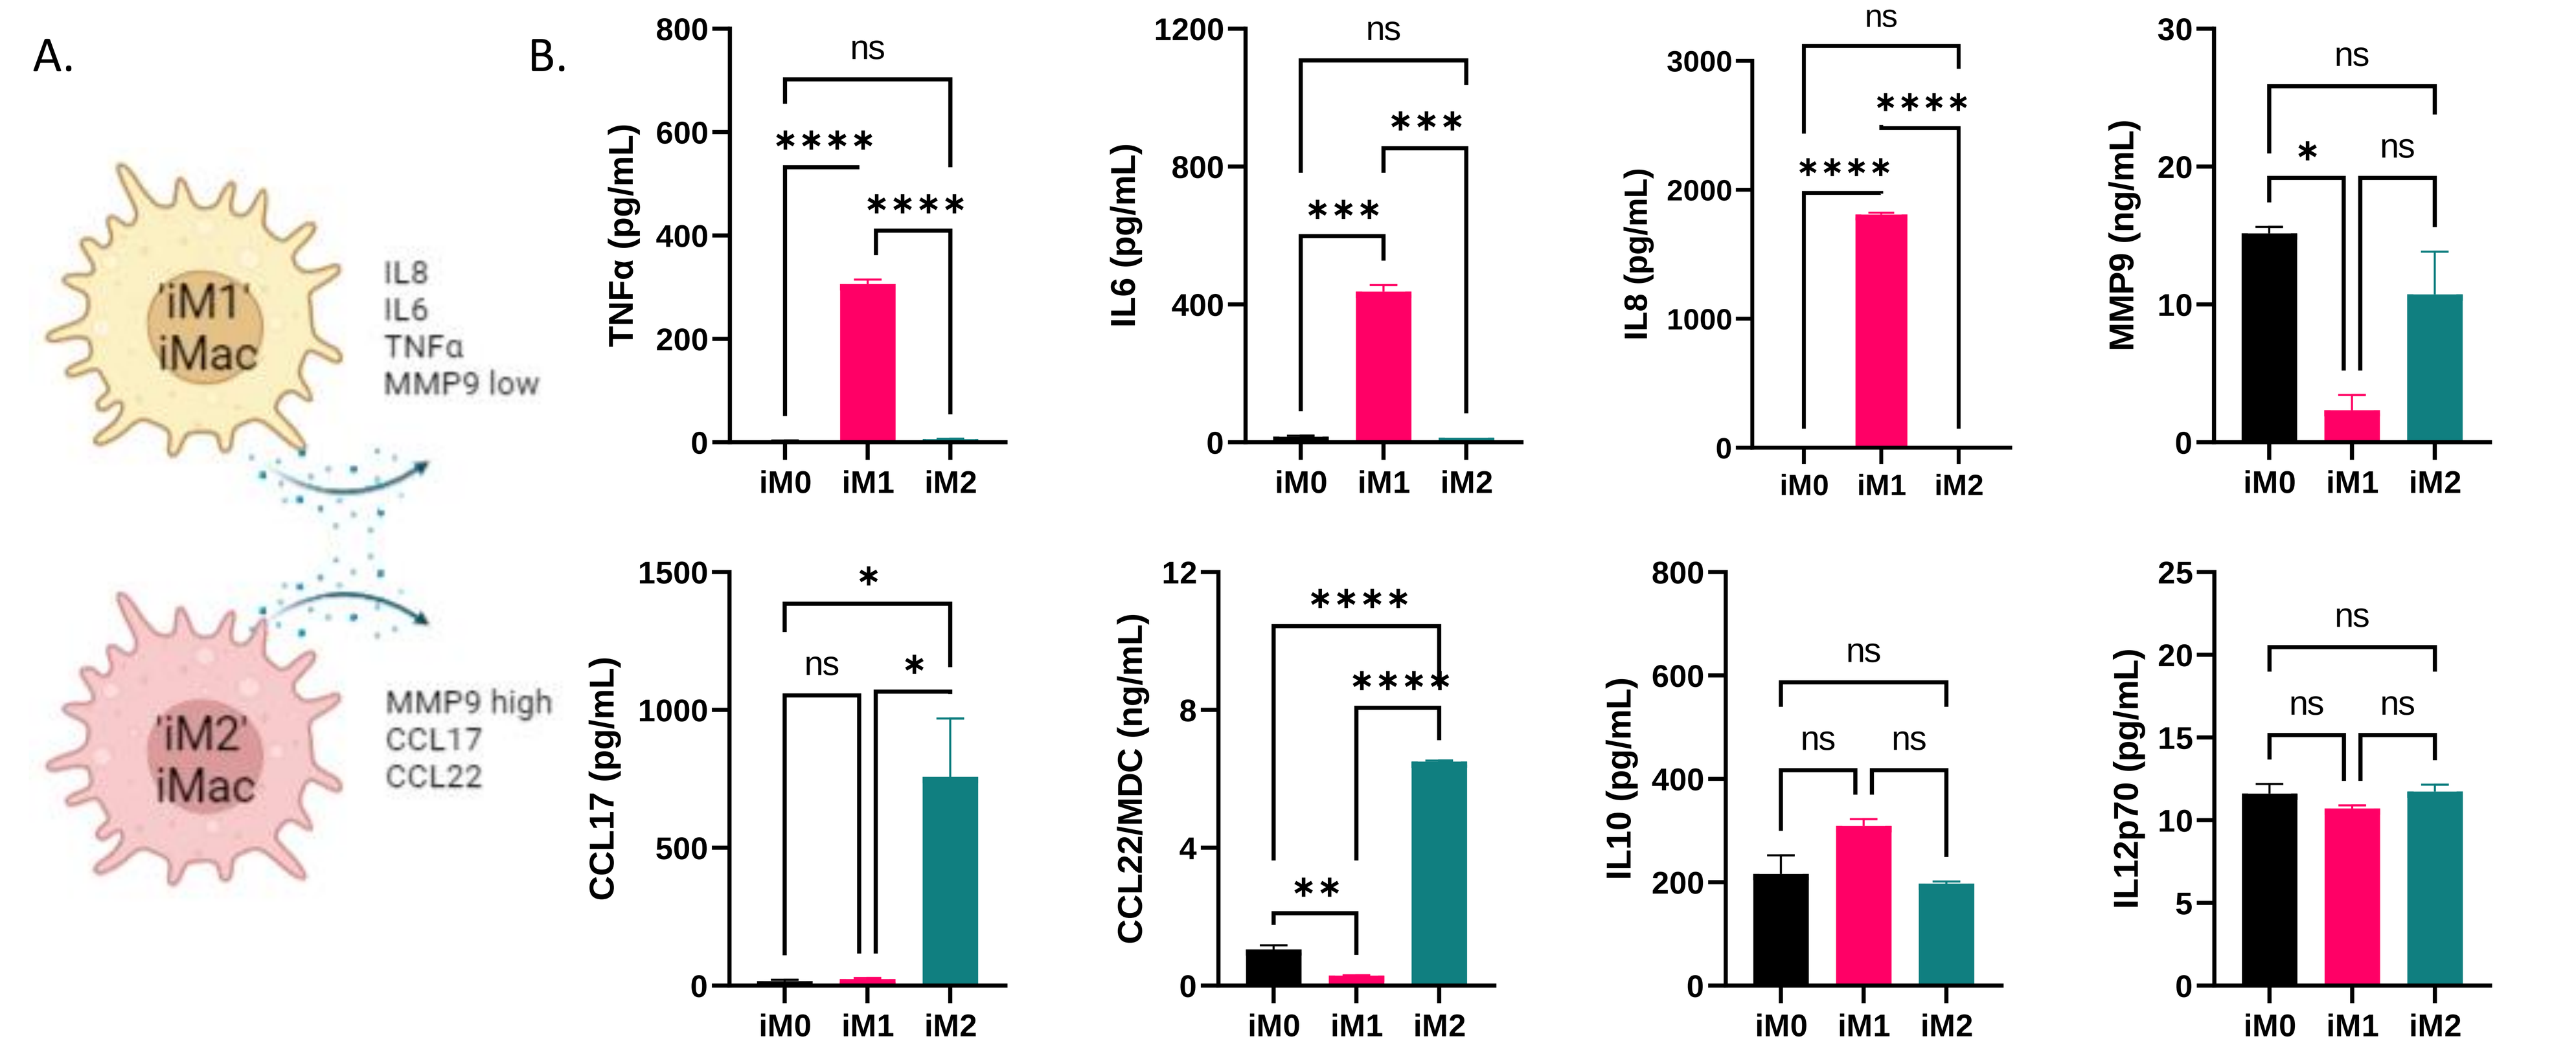
A.
'iM1' iMac
IL8
IL6
TNFα
MMP9 low
'iM2' iMac
MMP9 high
CCL17
CCL22
B.
TNFα (pg/mL)
IL6 (pg/mL)
IL8 (pg/mL)
MMP9 (ng/mL)
CCL17 (pg/mL)
CCL22/MDC (ng/mL)
IL10 (pg/mL)
IL12p70 (pg/mL)
iM0
iM1
iM2
ns

Figure 4. Fate iMacs display environmental specific cytokine release based on polarization state. Briefly, iMacs were polarized following Figure 1D. (A) Schematic depicting cytokine release profile of pro-inflammatory and anti-inflammatory polarized iMacs. Following polarization supernatant was collected and analyzed by R&D Quantikine ELISA kits for selected cytokines. (B) Quantified TNFα, IL6, IL8, MMP9, CCL17, CCL22, IL10, and IL12p70. Statistical analysis performed by One-way ANOVA with Tukey post-hoc test correction. *p<0.0332, **p<0.0021, ***p<0.0002, ****p<0.0001.

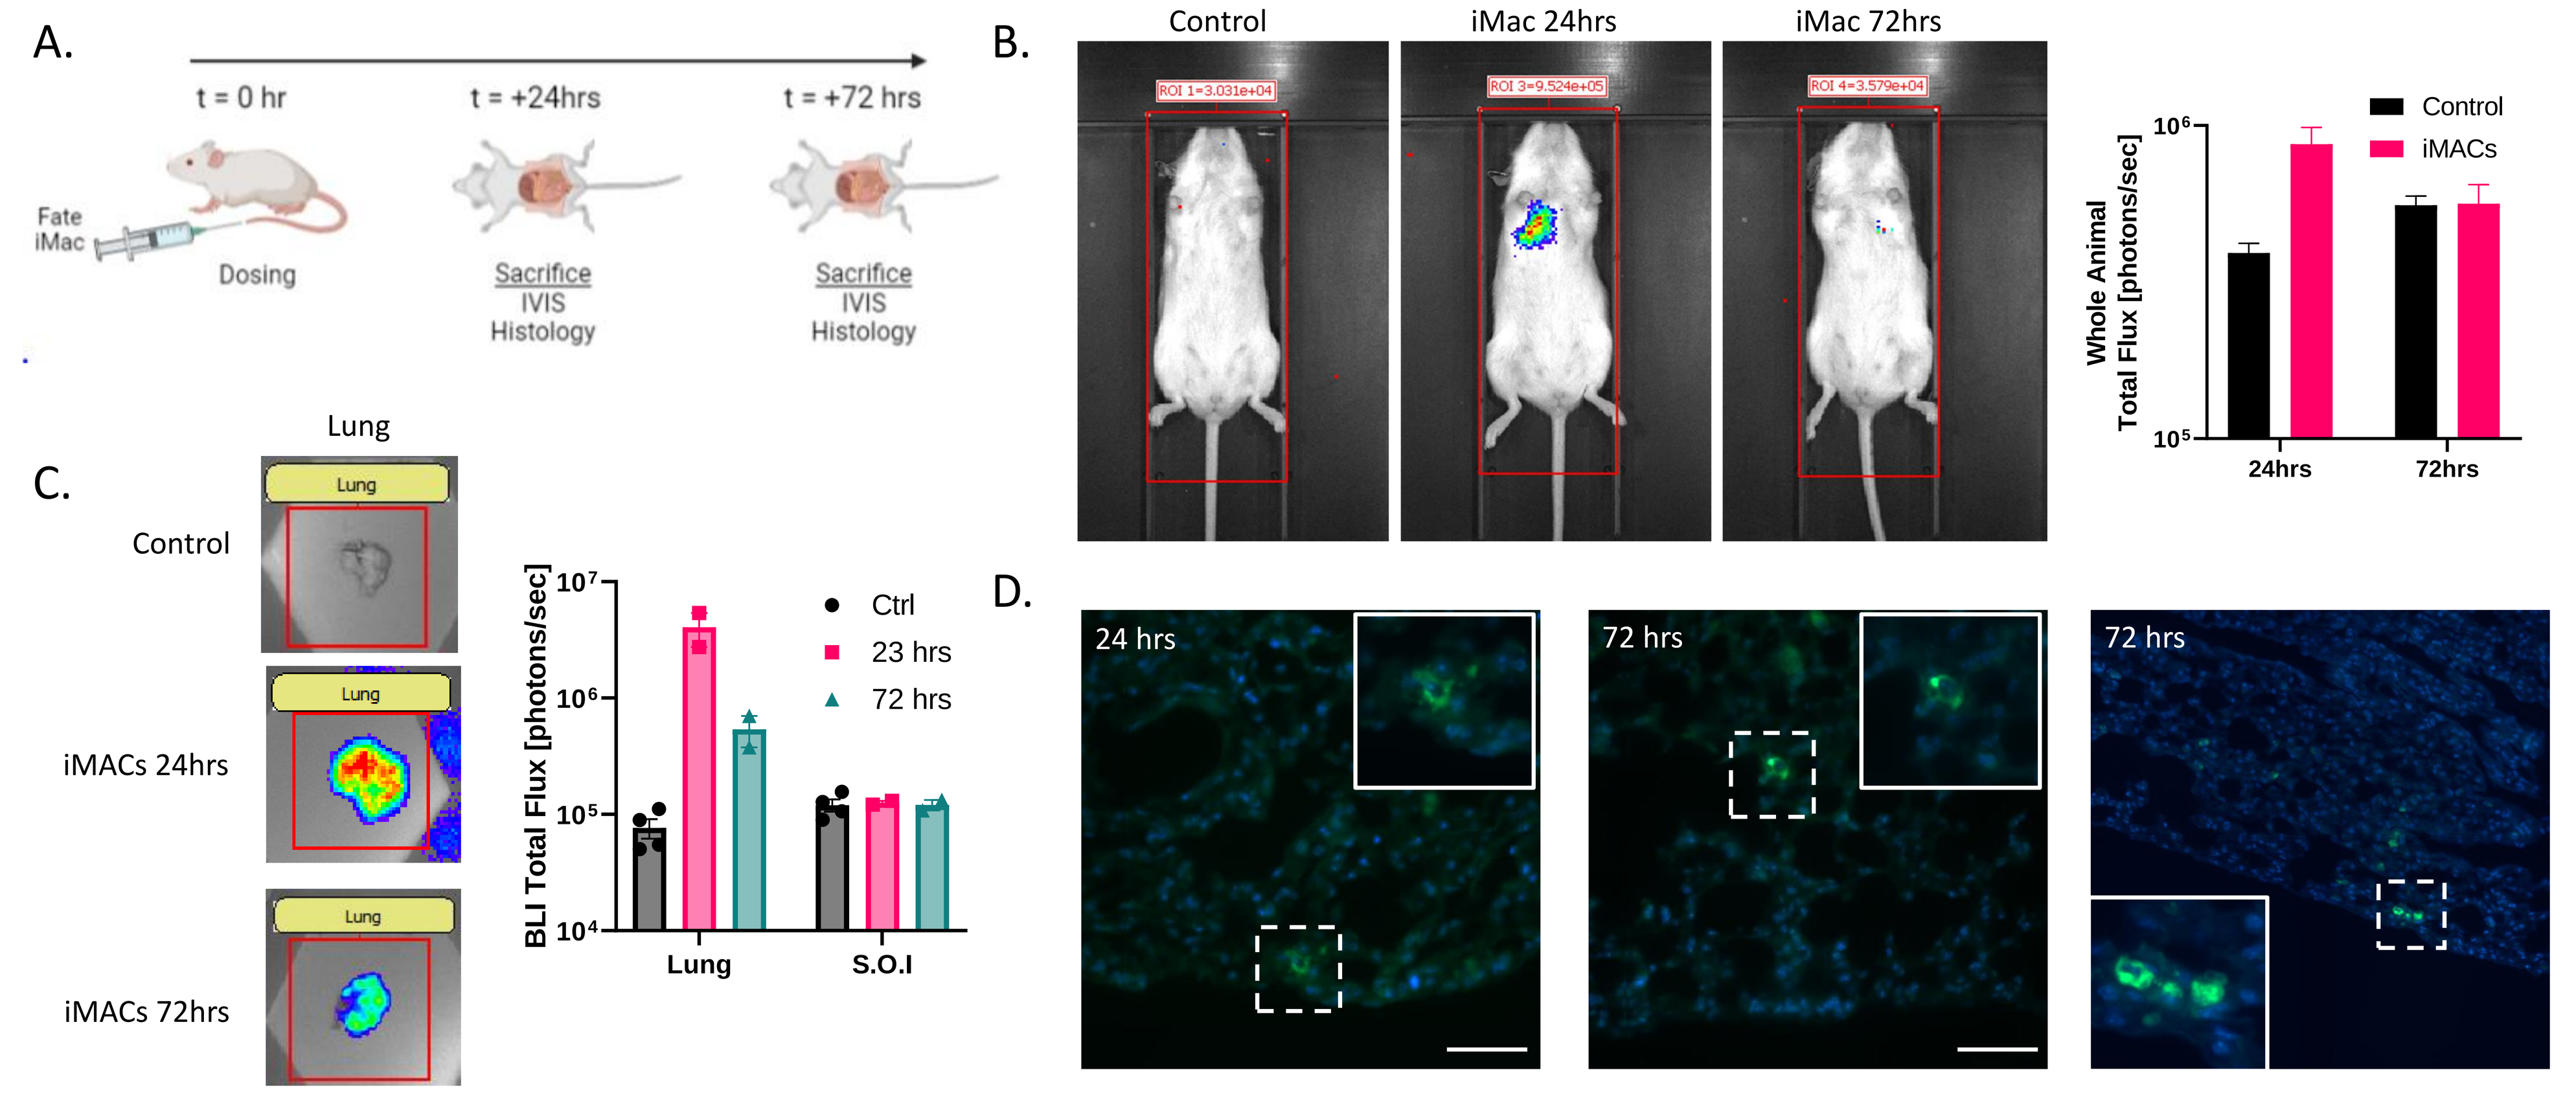
A.
t = 0 hr
t = +24hrs
t = +72 hrs
Fate iMac
Dosing
Sacrifice
IVIS
Histology
B.
Control
iMac 24hrs
iMac 72hrs
ROI 1=3.031e+04
ROI 3=9.524e+05
ROI 4=3.579e+04
Control
iMACs
Whole Animal Total Flux [photons/sec]
24hrs
72hrs
C.
Lung
Control
iMACs 24hrs
iMACs 72hrs
BLI Total Flux [photons/sec]
Ctrl
23 hrs
72 hrs
Lung
S.O.I
D.
24 hrs
72 hrs
72 hrs

Figure 5. Fate iMac Lung *in vivo* biodistribution. (A) Schematic depicting in vivo biodistribution study design. iMacs were I.V. dosed at time 0 and analyzed by in situ and ex vivo imaging and histology at 24 and 72 hours. (B) Whole body in situ IVIS imaging at 24 and 72 hours. (C) Ex vivo IVIS imaging of Lung at 24 and 72 hours. (D). Lung histology for GFP-iMacs at 24 and 72 hours.